Efficacy and Safety of AB928 Plus Modified FOLFOX-6 in Participants With Metastatic Colorectal Cancer: Initial Results at the Recommended Dose for Expansion (ARC-3)

M Cecchini,[1] M Modiano,[2] F Braiteh,[3] OS Gardner,[4] HN Gilbert,[4] D DiRenzo,[4] L Seitz,[4] MJ Walters,[4] F Yin,[4] R Woloski,[4] MC Paoloni,[4] and KY Chung[5]

[1]Yale University School of Medicine, New Haven, CT; [2]Arizona Blood and Cancer Specialists, Tucson, AZ; [3]Comprehensive Cancer Centers of Nevada, Las Vegas, NV; [4]Arcus Biosciences, Inc., Hayward, CA; [5]Prisma Health Cancer Institute, ITOR, Greenville, SC

Poster # LB-387
Abstract # 9953

INTRODUCTION

The Adenosine Axis in Cancer

In response to platinum chemotherapy, dying tumor cells release high levels of ATP into the tumor microenvironment (TME) where CD39 and CD73 convert ATP to adenosine (**Figure 1**).[1,2] By binding adenosine receptors 2a and 2b ($A_{2a}R$ and $A_{2b}R$) expressed on immune cells, adenosine promotes immunosuppression by inhibiting critical components of the antitumor immune response and ultimately enables tumor cells to evade destruction.[2] Initial research focused on $A_{2a}R$ as the most relevant adenosine receptor in cancer physiology; however, $A_{2b}R$ signaling mediates unique functions, such as activation of MAPK signaling.[3] Thus, adenosine receptor blockade may have the potential to overcome adenosine-dependent immunosuppression and lead to enhanced therapeutic efficacy of some chemotherapeutic agents.[2]

Figure 1. Critical Role of Adenosine Pathway in Immunosuppressive Tumor Microenvironment

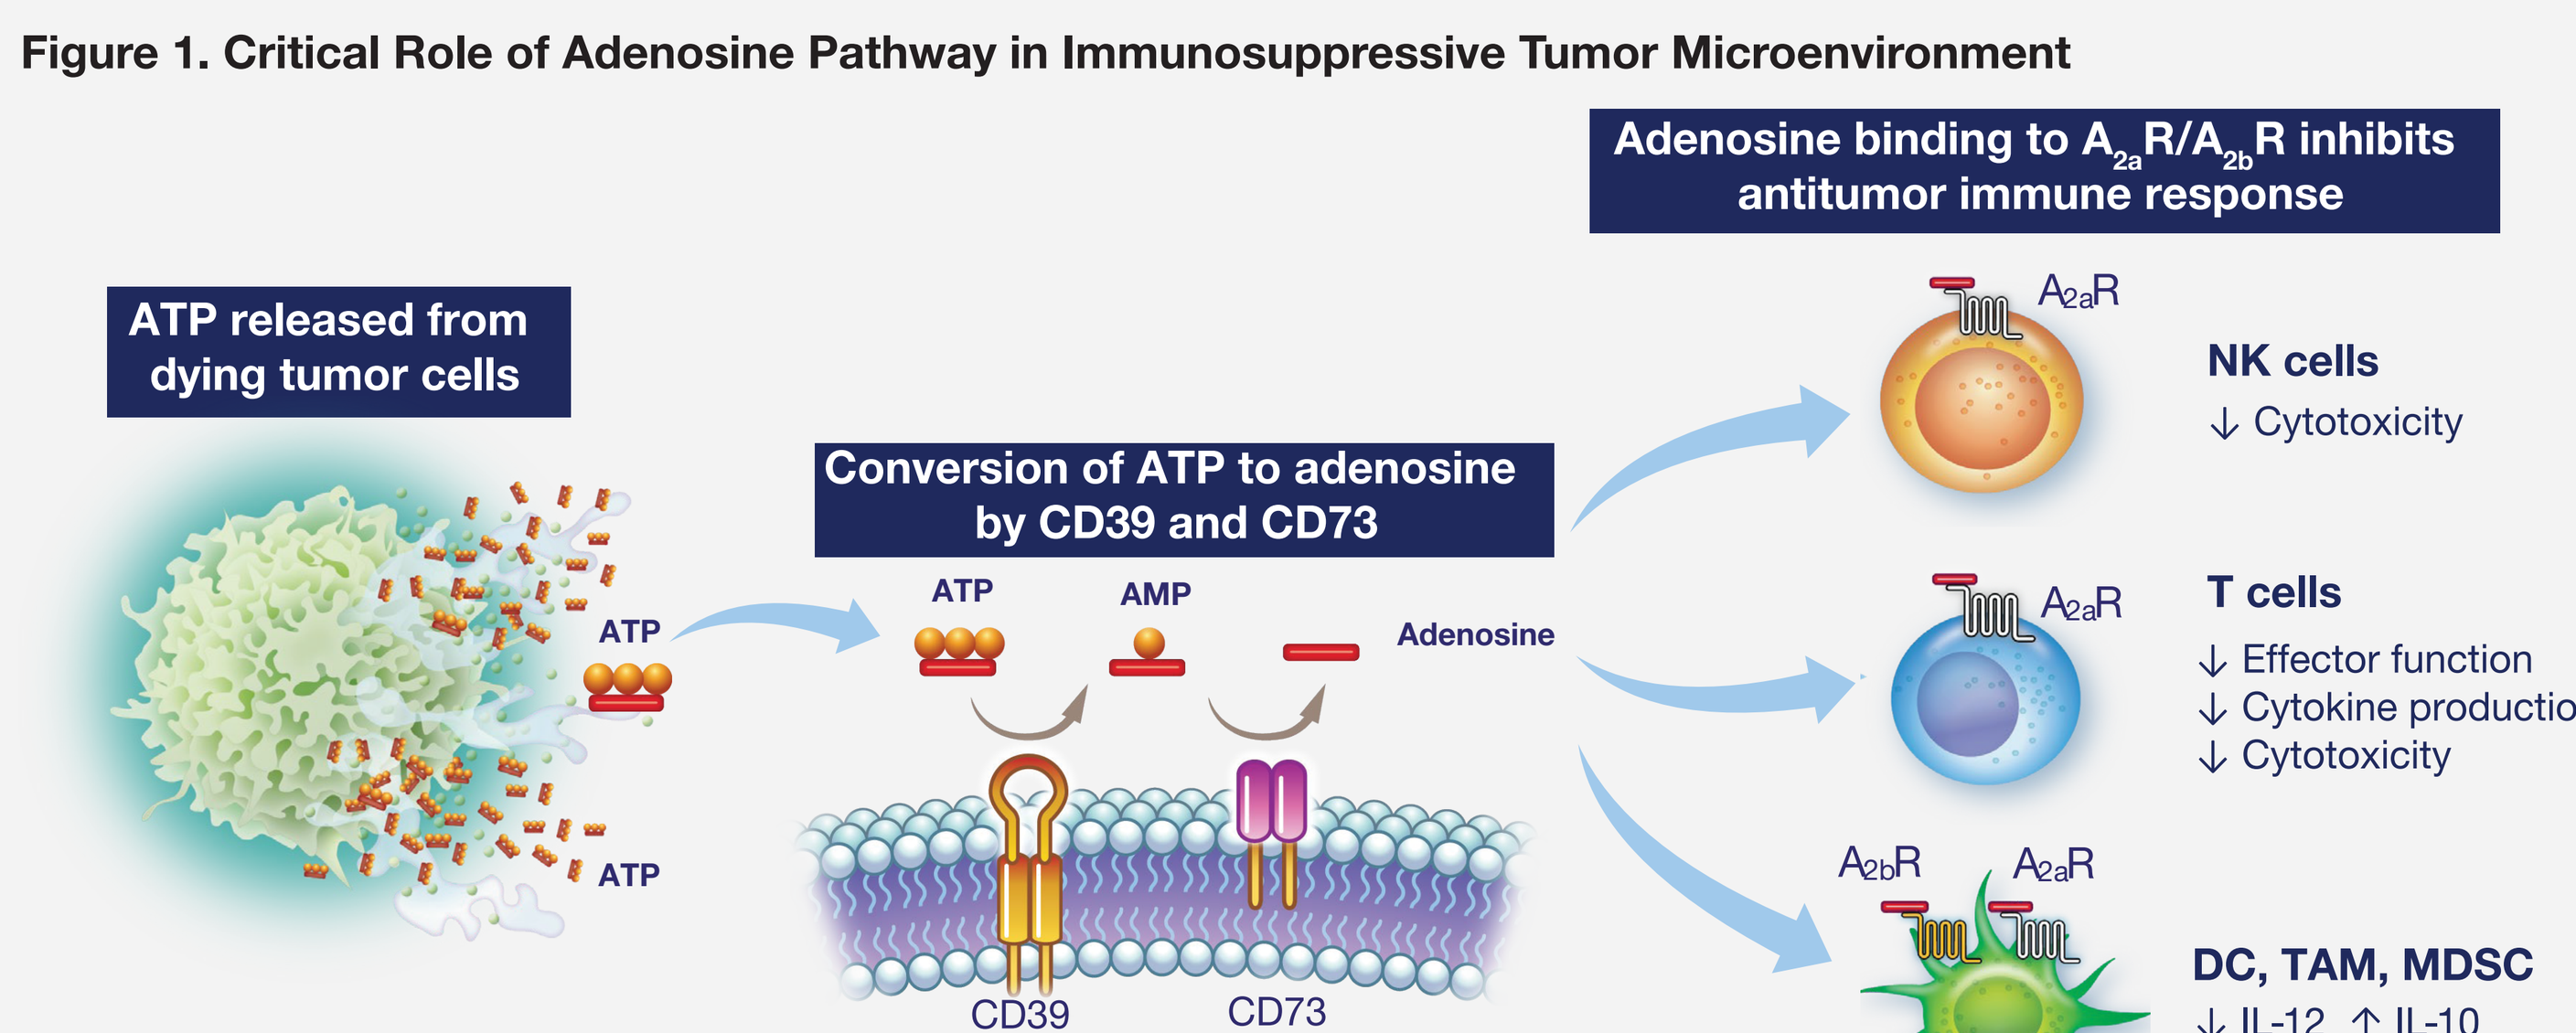

Adenosine binding to A2aR/A2bR inhibits antitumor immune response
ATP released from dying tumor cells
Conversion of ATP to adenosine by CD39 and CD73
ATP
AMP
Adenosine
CD39
CD73
TNAP
A2aR
A2bR
NK cells
↓ Cytotoxicity
T cells
↓ Effector function
↓ Cytokine production
↓ Cytotoxicity
DC, TAM, MDSC
↓ IL-12 ↑ IL-10
↓ T-cell stimulation

ATP, adenosine triphosphate; $A_{2a}R/A_{2b}R$, adenosine receptors 2a/2b; DC, dendritic cell; MDSC, myeloid-derived suppressor cells; NK, natural killer; TAM, tumor-associated macrophage.

AB928 is an orally bioavailable, small-molecule, selective dual antagonist of $A_{2a}R$ and $A_{2b}R$ that was specifically designed to block the immunosuppressive effects associated with high adenosine concentration within the TME; it is the only adenosine receptor antagonist in active clinical trials that potently blocks $A_{2b}R$. Currently, there are 4 ongoing global phase 1/1b disease-specific platform studies to assess safety, tolerability, pharmacokinetics (PK), pharmacodynamics (PD), and preliminary clinical activity of AB928 in combination with chemotherapy and/or anti-PD-1 antibody[4]

- Based on dose escalation data from these studies, AB928 150 mg once daily (QD) was selected as the recommended dose for expansion (RDE) based on PK, PK/PD correlation, and a well-tolerated safety profile of AB928 + chemo/immunotherapy

ARC-3 Study Rationale

Platinum-based chemotherapy, specifically 5-fluorouracil plus oxaliplatin (FOLFOX), is a standard of care (SOC) treatment for patients with metastatic colorectal cancer (mCRC).[5] Despite recent therapeutic advances, patients with mCRC have a 5-year survival rate of 15%, which leaves a great unmet need for novel mCRC treatments with improved safety, enhanced efficacy, and that can induce durable clinical benefits.[6]

- In multiple analyses of human tumors, CRC has been shown to have some of the highest expression levels of CD73 and $A_{2b}R$ compared with other tumor types[7,8]
- Additionally, *KRAS*, *BRAF*, and *EGFR* mutations found in mCRC are associated with CD73 overexpression[9,10] (**Figure 2**)
- *KRAS* and *BRAF* mutant tumors not only produce higher levels of adenosine but may also respond, in an autocrine $A_{2b}R$-mediated fashion, to those increased adenosine levels by activating growth pathways synergistic with the oncogenic mutation
- In preclinical studies, AB928 + oxaliplatin synergistically inhibited murine tumor growth and increased the number of intratumoral CD8+ T cells[11]
- For these reasons, patients with mCRC may be particularly sensitive to AB928 in combination with SOC chemotherapy

Figure 2. Correlation of EGFR, KRAS, and BRAF Mutations with Elevated CD73 Expression

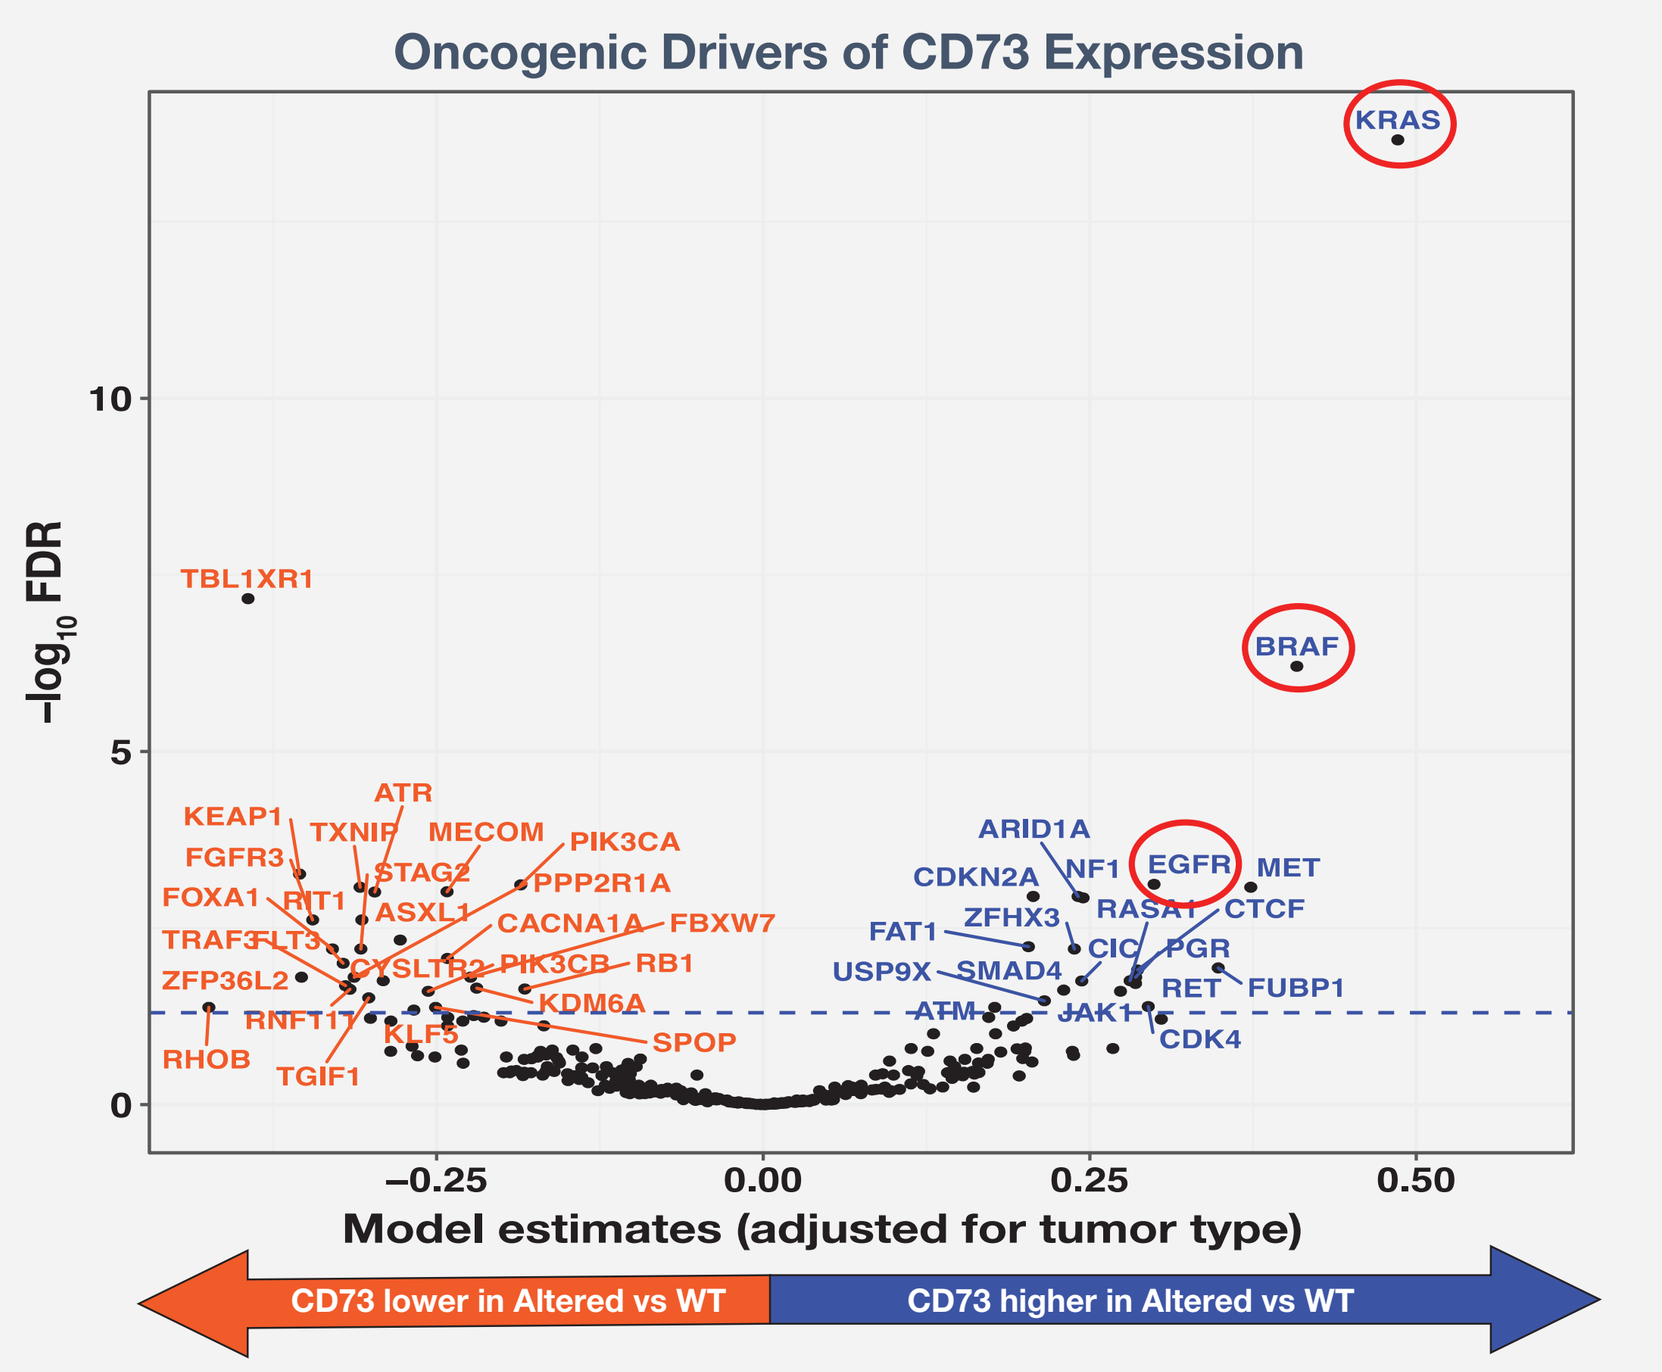

Oncogenic Drivers of CD73 Expression
KRAS
BRAF
EGFR
TBL1XR1
MET
CTCF
ARID1A
CDKN2A
NF1
RASA1
ZFHX3
FAT1
USP9X
SMAD4
CIC
PGR
RET
FUBP1
ATM
JAK1
CDK4
ATR
KEAP1
TXNIP
MECOM
PIK3CA
FGFR3
STAG2
PPP2R1A
FOXA1
RIT1
ASXL1
CACNA1A
FBXW7
TRAF3
FLT3
PIK3CB
RB1
ZFP36L2
CYSLTR2
KDM6A
RNF111
KLF5
SPOP
RHOB
TGIF1
10
5
0
−0.25
0.00
0.25
0.50
$-\log_{10}$ FDR
Model estimates (adjusted for tumor type)
CD73 lower in Altered vs WT
CD73 higher in Altered vs WT

FDR, false discovery rate; WT, wild type.

METHODS

Study Design

ARC-3 (NCT03720678) is an ongoing, Phase 1/1b, open-label, dose-escalation and dose-expansion study to evaluate safety, tolerability, PK, and clinical activity of AB928 + mFOLFOX-6 in patients with advanced mCRC (**Figure 3**).

- **Dose escalation (gastroesophageal cancer [GEC] or CRC):** AB928 (75 or 150 mg QD) + standard mFOLFOX-6 was evaluated in a 3+3 design; patients were monitored for dose-limiting toxicities (DLTs) for 28 days
- **Dose expansion (CRC only):** Enrolled patients will receive AB928 at the RDE in combination with standard mFOLFOX-6

Figure 3. ARC-3 Study Design

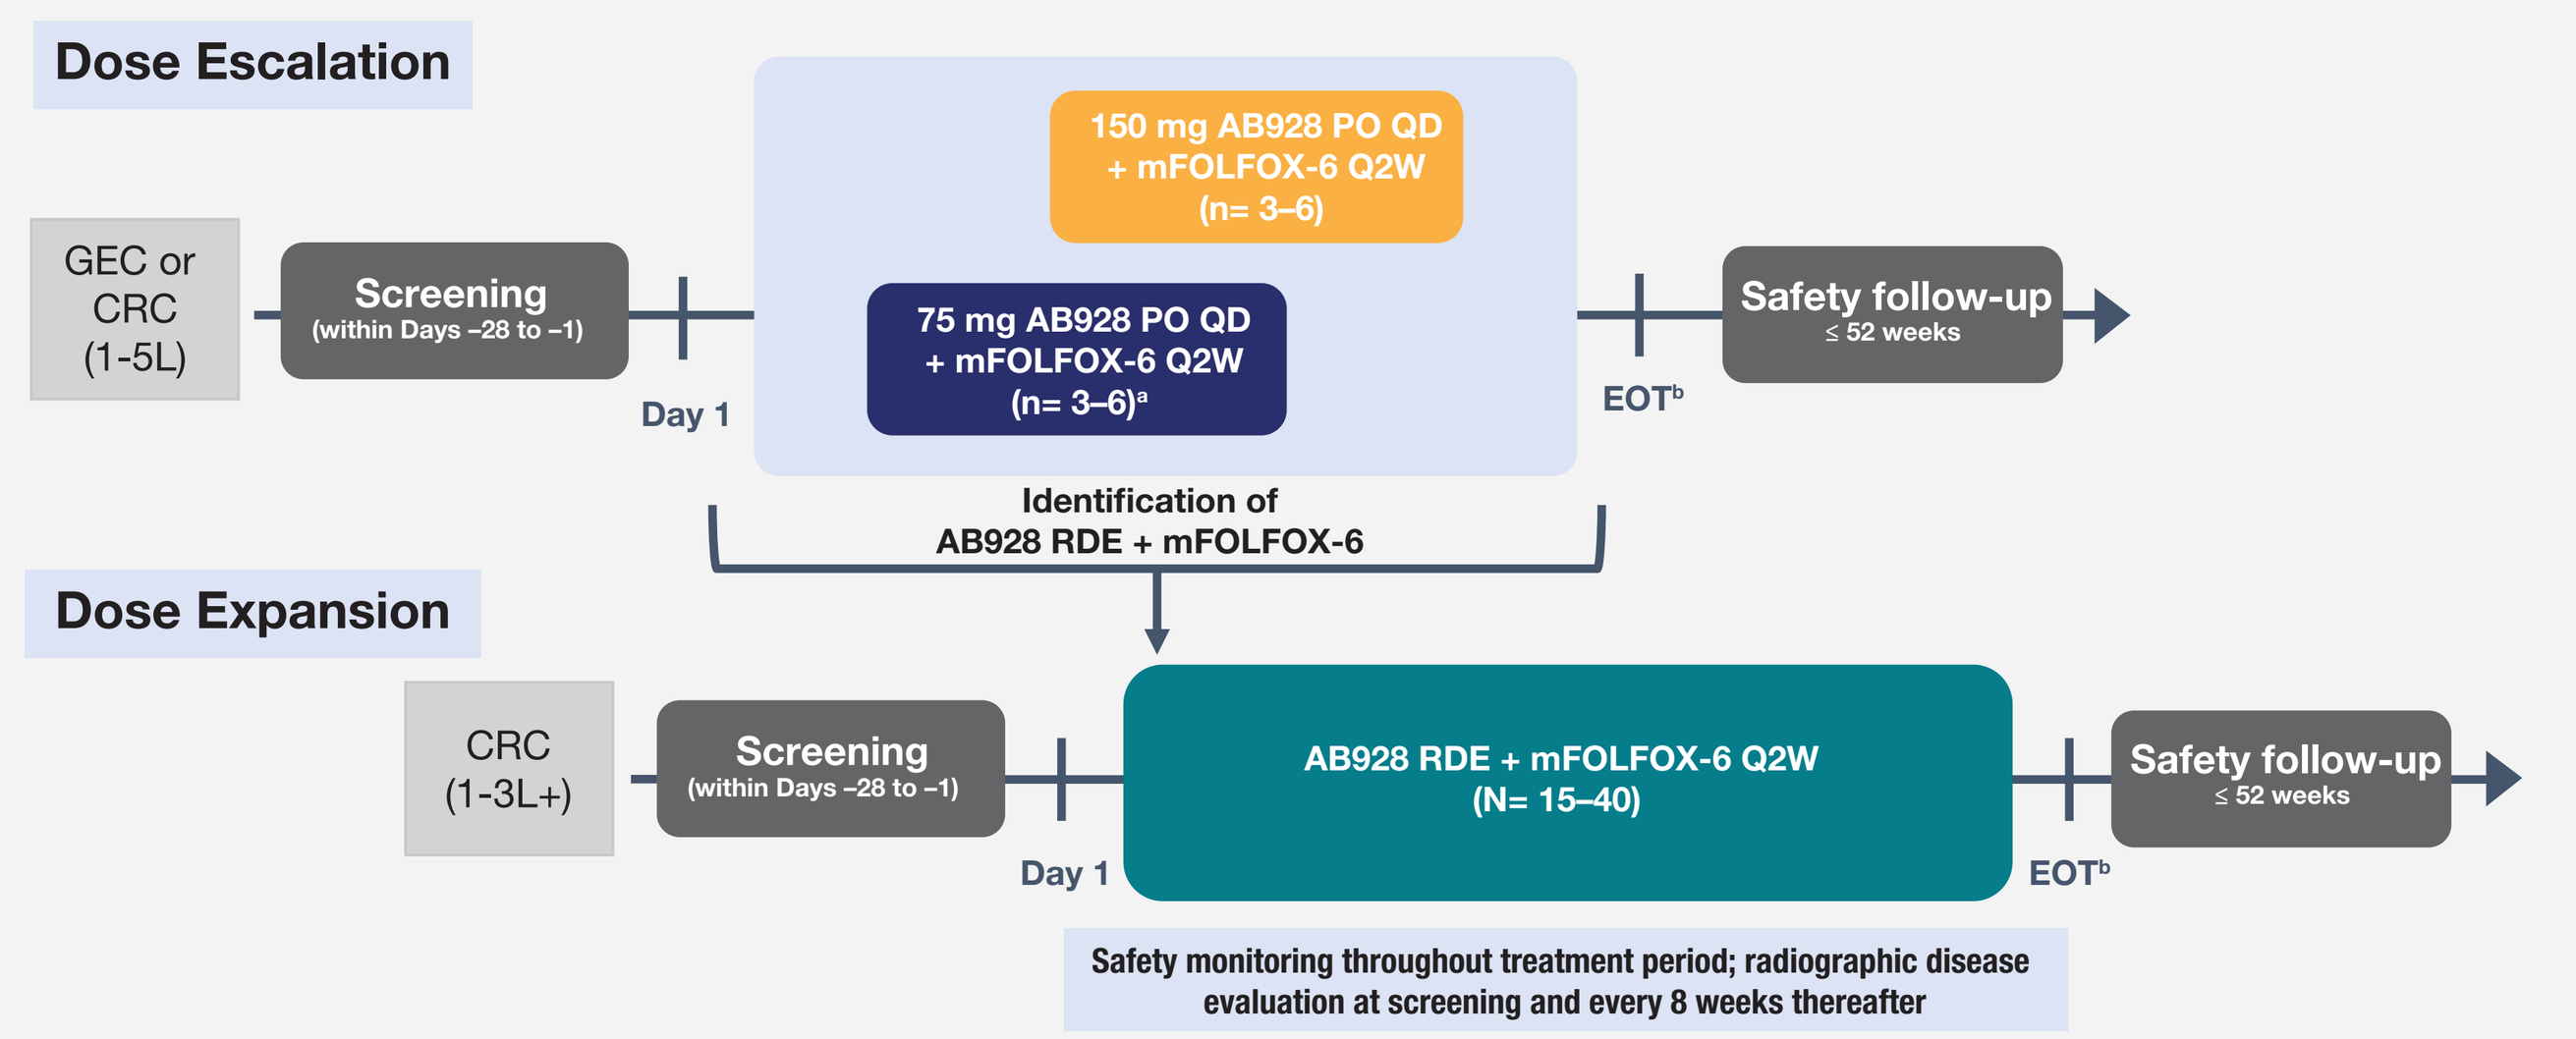

Dose Escalation
GEC or CRC (1-5L)
Screening (within Days −28 to −1)
Day 1
150 mg AB928 PO QD + mFOLFOX-6 Q2W (n= 3–6)
75 mg AB928 PO QD + mFOLFOX-6 Q2W (n= 3–6)a
EOTb
Safety follow-up ≤ 52 weeks
Identification of AB928 RDE + mFOLFOX-6
Dose Expansion
CRC (1-3L+)
Screening (within Days −28 to −1)
Day 1
AB928 RDE + mFOLFOX-6 Q2W (N= 15–40)
EOTb
Safety follow-up ≤ 52 weeks
Safety monitoring throughout treatment period; radiographic disease evaluation at screening and every 8 weeks thereafter

[a] When a minimum of 3 patients evaluable for toxicity completed the DLT evaluation period for a given AB928 dose, subsequent patients may be enrolled at the same, lower, or higher AB928 dose, or a dose may be chosen as the RDE that has not exceeded the MTD; [b] Patients will continue to receive study drug until disease progression or toxicity as assessed by the investigator; mFOLFOX-6 regimen: oxaliplatin: 85 mg/m^2 IV Q2W; leucovorin 400 mg/m^2 IV Q2W; and 5-FU 400 mg/m^2 IV bolus + 2,400 mg/m^2 (continuous 46-hour infusions on Days 1-2)[12,13] 5-FU, 5-fluorouracil; CRC, colorectal cancer; DLT, dose-limiting toxicity; EOT, end of treatment; GEC, gastroesophageal cancer; IV, intravenously; MTD, maximum tolerated dose; Q2W, every 2 weeks; PO, orally; QD, once daily; RDE, recommended dose for expansion.

ARC-3 Design Features

- Primary objective is safety and tolerability of AB928 + mFOLFOX-6 with secondary objectives that include clinical activity
- Eligible patients have histologically confirmed GEC (dose escalation only) or CRC that is metastatic or locally advanced and unresectable; ≥ 1 measurable lesion per RECIST v1.1; ECOG performance status 0-1
- Baseline archival tumor specimens or biopsies and on-treatment biopsies (if medically feasible) are collected from all patients to evaluate their immune composition and disease characteristics before and after treatment
- Patients receive AB928 + mFOLFOX-6 in combination until progression, unacceptable toxicity, or investigator decision
 - 5-fluorouracil and oxaliplatin may be discontinued due to SOC guidelines; AB928 and/or other study treatments may be continued until the aforementioned criteria are met

Statistical Analysis

- Safety analyses included all patients who received at least 1 dose of AB928
 - Summary statistics were provided for treatment-emergent AEs (TEAEs) and serious AEs (TESAEs), TEAE severity, and AE relationship to study drugs
- Efficacy analyses included all patients who were enrolled and assigned to receive AB928
 - Clinical activity was assessed according to RECIST v1.1 criteria
 - Disease control rate (DCR) was defined as the percentage of patients with a best overall response of complete response (CR), partial response (PR), or stable disease (SD) for ≥ 2 disease assessments

RESULTS

Patient Baseline Characteristics

As of May 8, 2020, 35 patients have received AB928 + mFOLFOX-6: 75 mg AB928 (n = 4) or 150 mg AB928 (n = 31)

- 150 mg AB928 QD was selected as the RDE based on PK, PK/PD correlation, and a well tolerated safety profile in the dose escalation portion of the study

For all patients, the mean age was 53 years; most patients were white (83%) and non-Hispanic (94%; **Table 1**). Most patients were late line; 22/23 patients who received prior treatment for metastatic disease had prior FOLFOX and/or FOLFIRI.

Table 1. Patient Demographics and Characteristics

Characteristics	Dose Escalation: 75 mg AB928 QD + mFOLFOX-6 Q2W (n = 4)	Dose Escalation: 150 mg AB928 QD + mFOLFOX-6 Q2W (n = 7)	Dose Expansion: 150 mg AB928 QD + mFOLFOX-6 Q2W (n = 24)	Total (N = 35)
Mean age (SD), years	50.0 (9.4)	52.9 (9.7)	53.4 (10.7)	52.9 (10.1)
Sex, male, n (%)	2 (50)	5 (71)	11 (46)	18 (51)
Race, n (%)				
White	3 (75)	5 (71)	21 (88)	29 (83)
Black	1 (25)	2 (29)	1 (4)	4 (11)
Asian	0	0	2 (8)	2 (6)
Ethnicity, Hispanic, n (%)	0	1 (14)	1 (4)	2 (6)
Prior therapies for metastatic disease, n (%)				
0	0	0	12 (50)	12 (34)
1	2 (50)	0	2 (8)	4 (11)
2	0	1 (14)	6 (25)	7 (20)
3+	2 (50)	6 (86)	4 (17)	12 (34)
Median prior treatments for metastatic disease, (range)	2 (1-3)	4 (2-7)	0.5 (0-4)	2 (0-7)

Q2W, once every 2 weeks; QD, once daily; SD, standard deviation.

Available baseline tumor samples (n = 17) analyzed internally by whole exome sequencing were all microsatellite stable with low or intermediate tumor mutational burden.

RESULTS

Safety Analyses

As shown in **Table 2**, all patients reported ≥ 1 TEAE and 11 TEAEs were reported by > 30% of patients; the most common TEAEs were fatigue (66%), nausea (60%), and diarrhea (49%)

- AB928-related TEAEs occurred in 27/35 (77%) of patients
 - The majority of events were Grade 1 or 2
- Ten patients reported ≥ Grade 3 AB928-related TEAEs that were also possibly related to mFOLFOX-6: neutropenia (n = 5), diarrhea (n = 2), AST increased (n = 2), fatigue (n = 1), nausea (n = 1), hyperglycemia (n = 1), anemia (n = 1), acute kidney injury (n = 1)

Eight patients (23%) reported ≥ 1 TESAE. One patient experienced a Grade 3 TESAE of acute kidney injury that was deemed related to AB928 and oxaliplatin; as a result, AB928 dosing was interrupted and oxaliplatin was withdrawn and the event resolved in 14 days. Three patients (9%) had TEAEs resulting in AB928 discontinuation; none were deemed related to AB928.

Table 2: Treatment Emergent Adverse Events

Patients, n (%)	Dose Escalation: 75 mg AB928 QD + mFOLFOX-6 Q2W (n = 4)	Dose Escalation: 150 mg AB928 QD + mFOLFOX-6 Q2W (n = 7)	Dose Expansion: 150 mg AB928 QD + mFOLFOX-6 Q2W (n = 24)	Total (N = 35)
Any TEAE	4 (100)	7 (100)	24 (100)	35 (100)
Grade ≥ 3	4 (100)	7 (100)	15 (63)	26 (74)
Any TESAE	2 (50)	3 (43)	3 (13)	8 (23)
Grade ≥ 3	2 (50)	3 (43)	3 (13)	8 (23)
AB928-related TEAEs[a]	3 (75)	6 (86)	18 (75)	27 (77)
Grade ≥ 3	2 (50)	1 (14)	7 (29)	10 (29)
AB928-related TESAEs[a]	0	0	1 (4)	1 (3)
Grade ≥ 3	0	0	1 (4)	1 (3)
Any study treatment d/c due to TEAEs	1 (25)	2 (29)	6 (25)	9 (26)
AB928 discontinuation due to TEAEs	0	1 (14)	2 (8)	3 (9)
Deaths due to TEAEs	0	0	1 (4)[b]	1 (3)
TEAEs in > 30% of all patients				
Fatigue	2 (50)	5 (71)	16 (67)	23 (66)
Nausea	3 (75)	5 (71)	13 (54)	21 (60)
Diarrhea	1 (25)	6 (86)	10 (42)	17 (49)
Neutropenia	1 (25)	3 (43)	10 (42)	14 (40)
Anemia	1 (25)	4 (57)	9 (38)	14 (40)
Thrombocytopenia	1 (25)	4 (57)	8 (33)	13 (37)
Neuropathy peripheral	2 (50)	1 (14)	10 (42)	13 (37)
AST increased	1 (25)	4 (57)	8 (33)	13 (37)
ALT increased	1 (25)	3 (43)	9 (38)	13 (37)
Decreased appetite	1 (25)	4 (57)	7 (29)	12 (34)
Constipation	1 (25)	2 (29)	8 (33)	11 (31)

[a] Events may also be considered related to some components of the mFOLFOX-6 treatment regimen; [b] Unexplained death on Day 2 of first treatment cycle was considered a suspected unexpected serious adverse reaction by the sponsor. ALT, alanine aminotransferase; AST, aspartate aminotransferase; QD, once daily; Q2W, once every 2 weeks; TEAE, treatment emergent adverse event; TESAE, treatment emergent serious adverse event.

Clinical Activity

As of May 8, 2020, 16/35 (46%) of patients remained on active treatment (**Figure 4**): 8 are 1L or 2L patients, 8 are 3L+ patients

- Median time on treatment was 16.1 weeks (range: 2–46 weeks) for patients in the dose escalation and 18.2 weeks (range: 0-32 weeks) for those in the dose expansion
- Median time on treatment for patients by treatment line was as follows:
 - 1L patients (n = 12): 20.1 weeks (range: 0.29–32 weeks)
 - 2L patients (n = 4): 9.2 weeks (range: 4.29–16.1 weeks)
 - 3L+ patients (n = 19): 16.9 weeks (range: 1.71–46.0 weeks)
- *BRAF* and *KRAS* mutation/wild type status is shown for those patients with available data (**Figure 4**, left panels); additional biomarker characterization is ongoing

Figure 4. Time on AB928 + mFOLFOX-6 Treatment

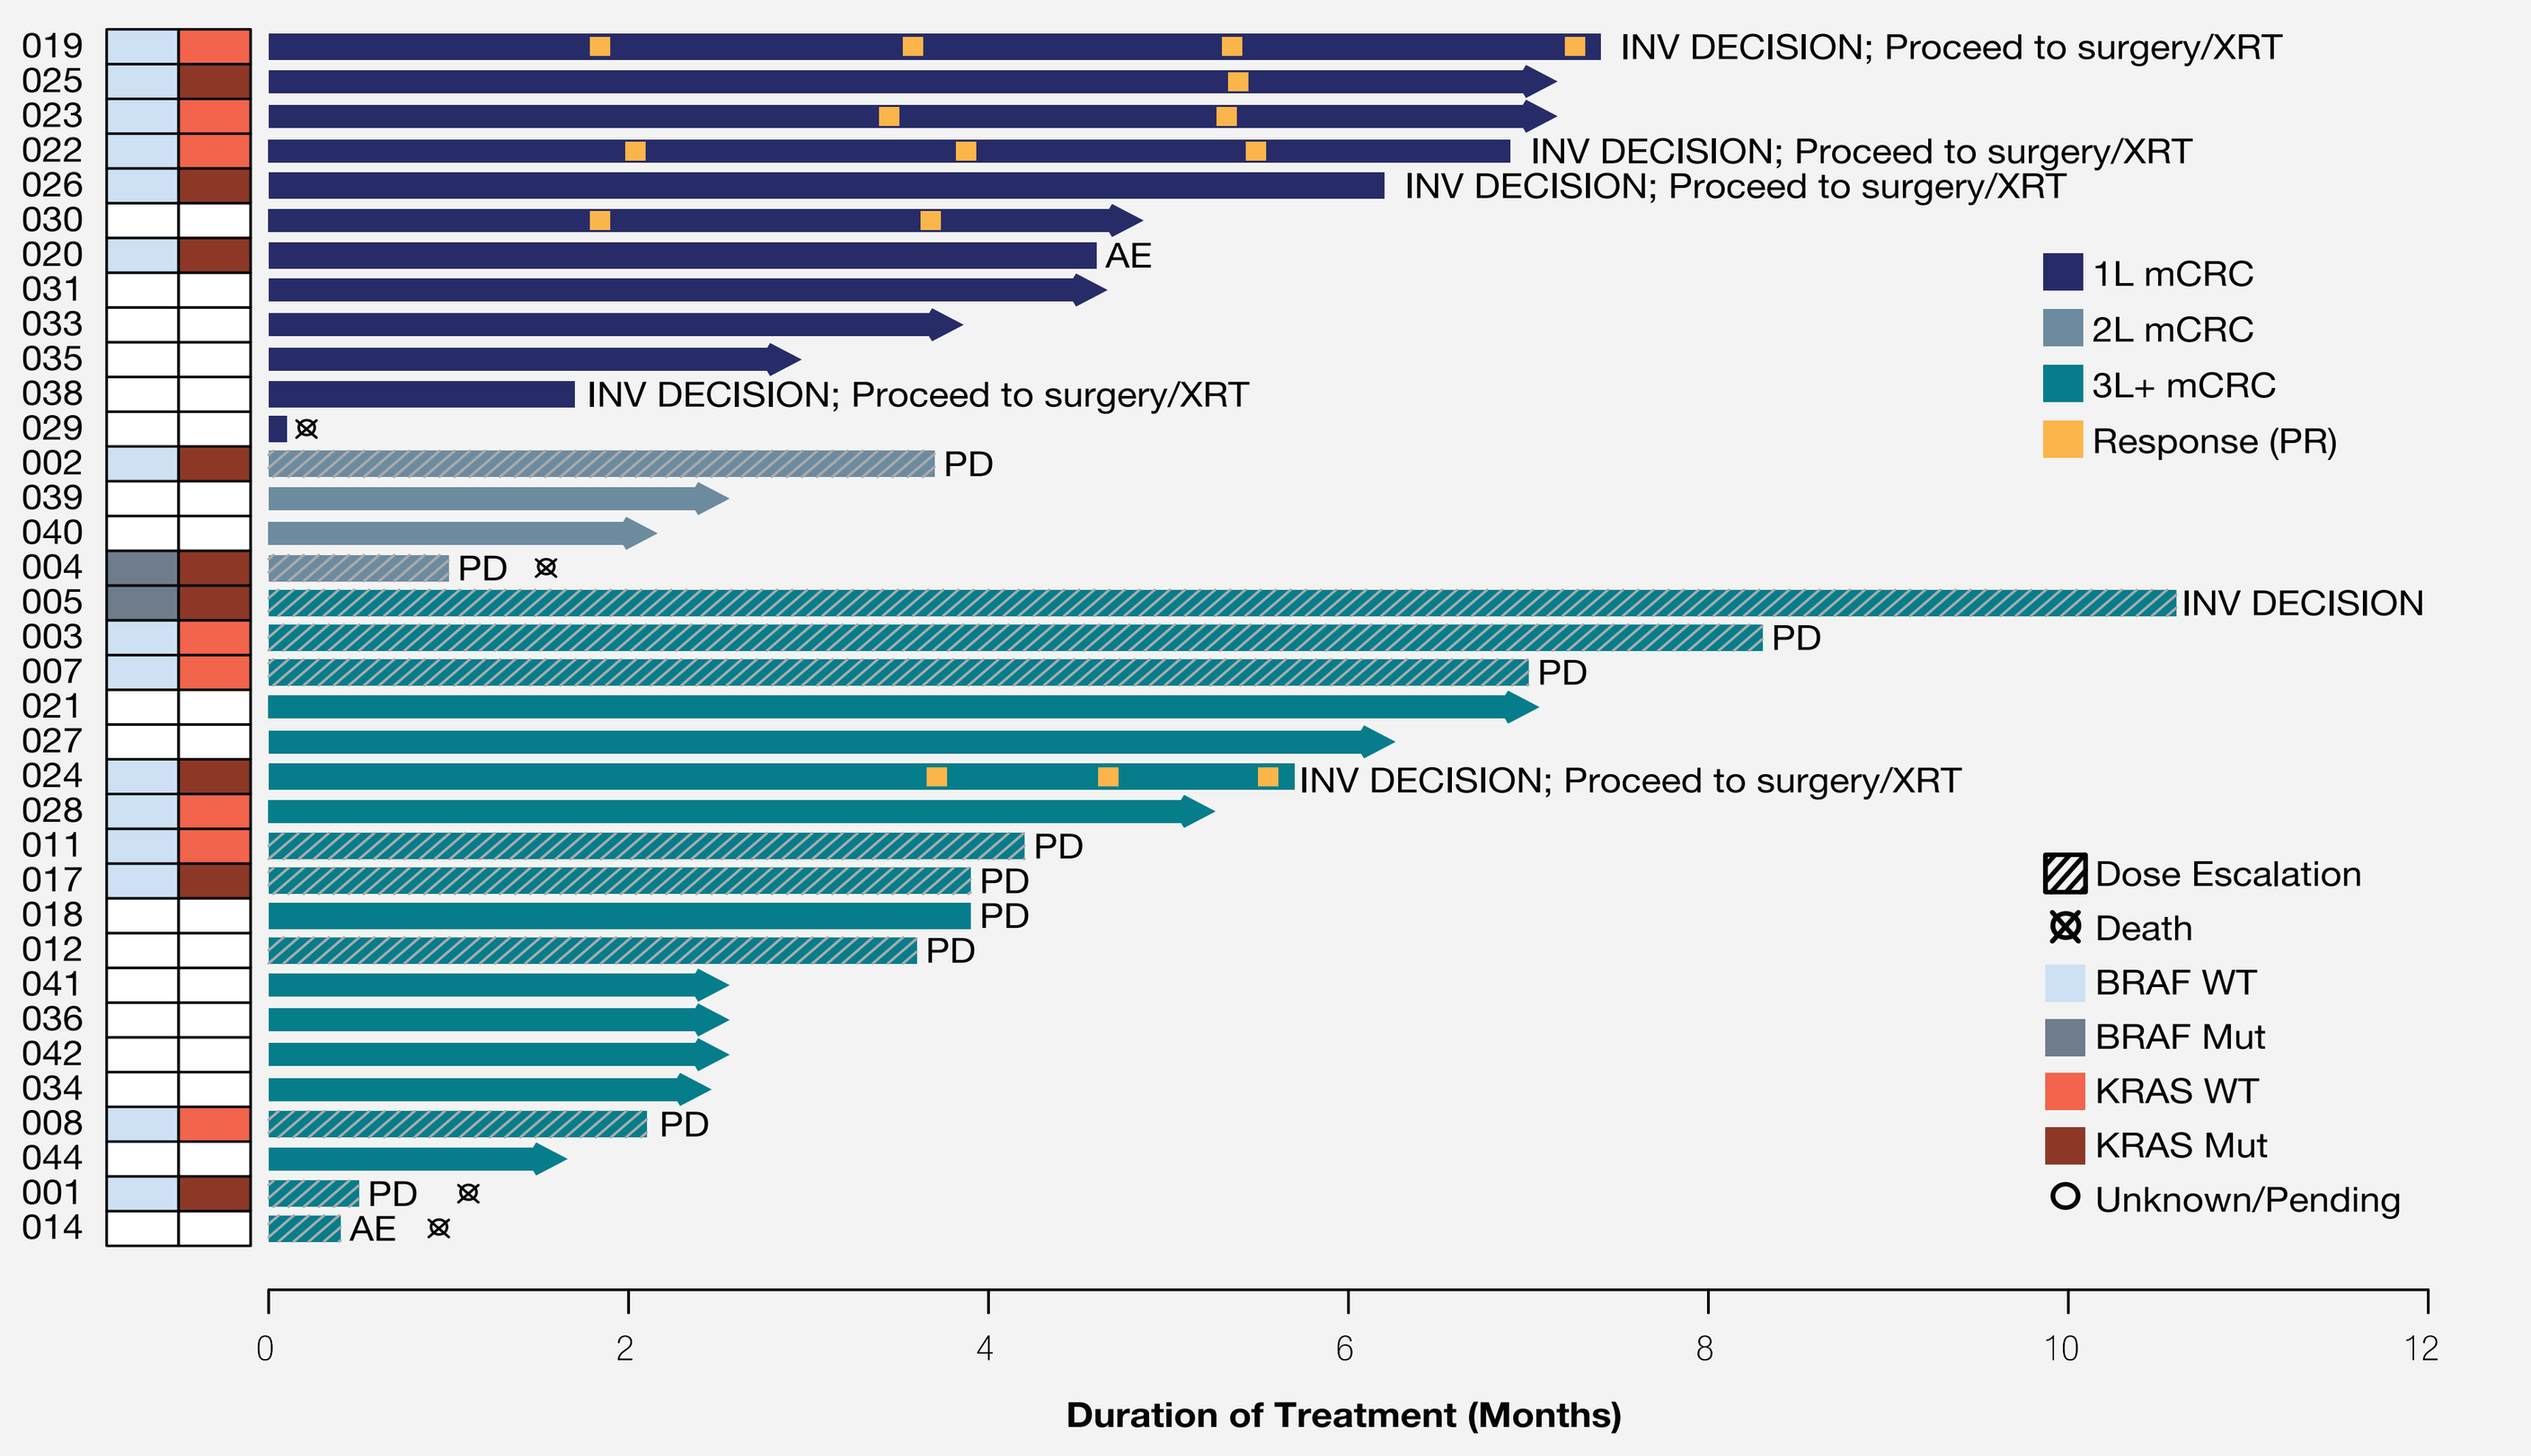

019
025
023
022
026
030
020
031
033
035
038
029
002
039
040
004
005
003
007
021
027
024
028
011
017
018
012
041
036
042
034
008
044
001
014
INV DECISION; Proceed to surgery/XRT
INV DECISION; Proceed to surgery/XRT
INV DECISION; Proceed to surgery/XRT
AE
INV DECISION; Proceed to surgery/XRT
PD
PD
INV DECISION
PD
PD
INV DECISION; Proceed to surgery/XRT
PD
PD
PD
PD
PD
PD
AE
1L mCRC
2L mCRC
3L+ mCRC
Response (PR)
Dose Escalation
Death
BRAF WT
BRAF Mut
KRAS WT
KRAS Mut
Unknown/Pending
0
2
4
6
8
10
12
Duration of Treatment (Months)

AE, adverse event; INV DECISION, investigator decision for discontinuation; mCRC, metastatic colorectal cancer; Mut, mutation; PD, progressive disease; XRT, radiotherapy; WT, wild type.

RESULTS

Best Overall Response

Best percentage change from baseline in the sum of target lesions is shown in **Figure 5.** Of patients with SD, 9/24 (38%) had tumor shrinkage of ≥ 15%.

- Investigator-assessed DCR was 76% with 6 PRs and 10 SDs for ≥ 2 disease assessments
- For 11 patients treated with ≥ 2 prior lines of therapy for metastatic disease (3L+), DCR was 64% with 1 PR and 6 SDs

Two patients had CRs of target lesions; both received 150 mg QD AB928 + mFOLFOX-6 as 1L mCRC treatment, then discontinued the study at the investigator's discretion to undergo surgical resection of remaining non-target lesions.

- Patient 019: 2 target lesions (60 mm) at baseline; PR at Days 56, 109, and 163; CR on Day 221
- Patient 022: 4 target lesions (64 mm) at baseline; PR at Day 62; CR on Days 118 and 167

Given deep responses and the potential for definitive cure, 3 additional patients (including a 3L+ patient) proceeded to surgical resection with or without chemo/radiotherapy at the investigator's recommendation.

Figure 5. Best Percentage Change from Baseline in Sum of Target Lesions

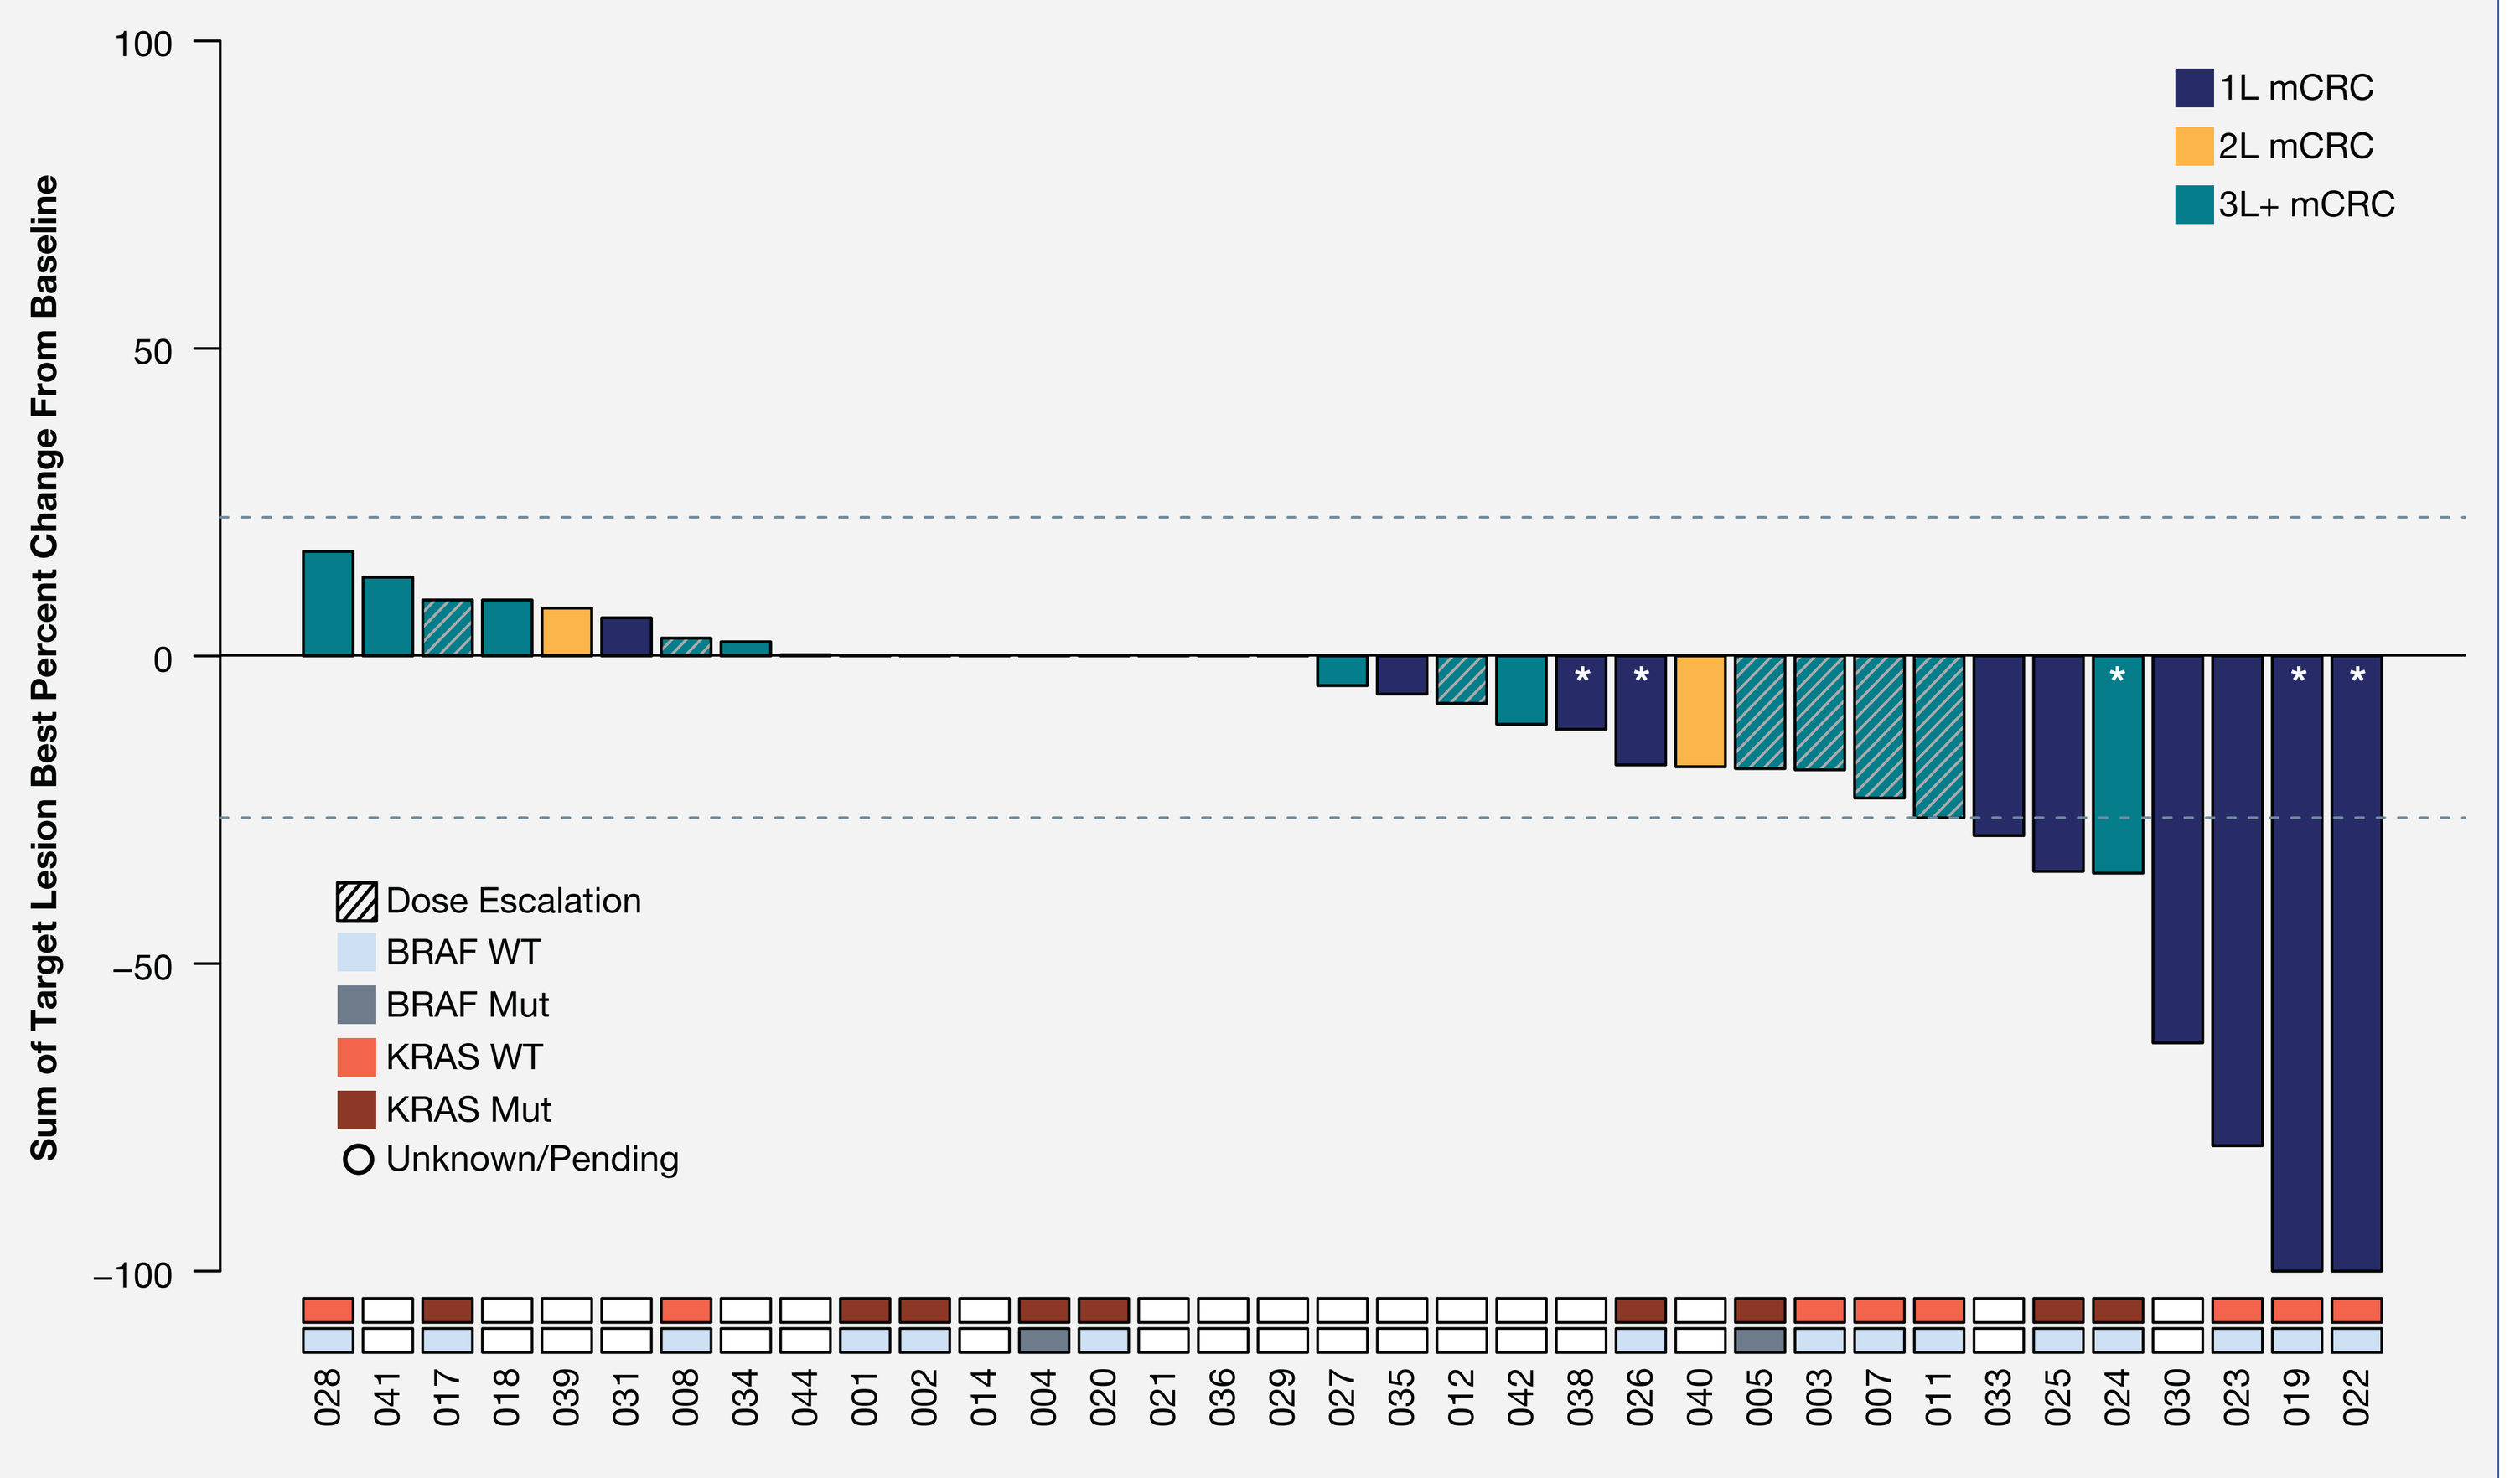

100
50
0
−50
−100
Sum of Target Lesion Best Percent Change From Baseline
1L mCRC
2L mCRC
3L+ mCRC
Dose Escalation
BRAF WT
BRAF Mut
KRAS WT
KRAS Mut
Unknown/Pending
028
041
017
018
039
031
008
034
044
001
002
014
004
020
021
036
029
027
035
012
042
038
026
040
005
003
007
011
033
025
024
030
023
019
022

* Investigator decision to proceed with definitive surgery and/or chemo/radiotherapy. mCRC, metastatic colorectal carcinoma; Mut, mutation; WT, wild type.

CONCLUSIONS

- AB928 with mFOLFOX-6 has been well tolerated without significant evidence of additive toxicity in patients with mCRC
- Combination treatment was associated with disease control in all patients with 1 baseline disease assessment (n = 30), including those with microsatellite stable disease and *RAS/BRAF* mutated mCRC
- Encouraging deep responses were observed across 1L to 3L+ patients; five patients discontinued the study at the investigator's discretion to undergo alternate therapy with curative intent
- Patients with late line disease previously treated with FOLFOX or FOLFIRI have shown clinical benefit (PR and/or SD > 4 months) with AB928 combination therapy; these encouraging results warrant further exploration
- Planned enrollment of up to 40 patients is proceeding based on early efficacy gates; as of May 8, 2020, 16 patients are continuing to receive study treatment
- Across the AB928 program, extensive tissue/blood biomarker characterization is ongoing

FUTURE DIRECTIONS

- Randomized studies of AB928 + standard care regimens are ongoing in late line mCRC
 - MORPHEUS-CRC (NCT03555149): Phase 1b/2; AB928 + atezolizumab ± regorafenib vs regorafenib monotherapy
- Future studies evaluating AB928 combination therapy in *RAS/BRAF* mutated mCRC and other advanced solid tumor types are in development

ACKNOWLEDGMENTS AND DISCLOSURES

The authors gratefully acknowledge the patients, their families, and their caregivers for their participation in this clinical trial. Additionally, they would like to thank the Principal Investigators and study staff for their efforts in conducting the study and Akshata Udyavar for her assistance with bioinformatics analyses. M Cecchini and M Modiano have no conflicts of interest to disclose; F Braiteh has received honoraria from Amgen, AstraZeneca, Bristol Myers Squibb, Genentech, Incyte, Merck, and Pfizer; has had a consulting/advisory role for Amgen, AstraZeneca, Eisai, Exelixis, Genentech, and Pfizer; participated in speakers' bureaus for Amgen, AstraZeneca, Bayer, Bristol Myers Squibb, Celgene, Genentech, Incyte, Lilly, Pfizer, and Puma Biotechnology; and has received funding for travel, accommodations, or expenses from Amgen, AstraZeneca, Bayer, Bristol Myers Squibb, Celgene, Eisai, Exelixis, Genentech, Incyte, Lilly, Pfizer, and Puma Biotechnology; OS Gardner, HN Gilbert, D DiRenzo, L Seitz, MJ Walters, F Yin, R Woloski, and MC Paoloni are/were employees of Arcus Biosciences, Inc. and may own company stock. KY Chung is an employee of Prisma Health Cancer Institute - ITOR. Medical writing assistance was provided by Amanda Martin, PhD, of Medical Expressions (Chicago, IL) and was funded by Arcus Biosciences.

REFERENCES

1. Martins I, et al. *Cell Cycle*. 2009;8(22):3723-3728. 2. Vijayan D, et al. *Nat Rev Cancer*. 2017;17(12):765. 3. Gao ZG, et al. *Int J Mol Sci*. 2019;20(20):5139. 4. Powderly J, et al. *ESMO 2019*. Abstract 4854. 5. Martini G, et al. *World J Gastroenterol*. 2017;23(26):4675-4688. 6. Siegel RL, et al. *CA Cancer J Clin*. 2020;70(1):7-30. 7. DiRenzo D, et al. *AACR 2019*. Abstract 3168. 8. DiRenzo D, et al. *SITC 2019*. Poster P557. 9. Udyavar A, et al. *AACR 2019*. Abstract 2526. 10. Ashok D, et al. *SITC 2019*. Poster P379. 11. Jaen J, et al. *SITC 2018*. Abstract 10724. 12. Cheeseman SL, et al. *Br J Cancer*. 2002;87(4):393-399. 13. Hochster HS, et al. *J Clin Oncol*. 2008;26(21):3523-3529.

Corresponding Author: Melissa Paoloni (mpaoloni@arcusbio.com)

June 22-24, 2020. AACR Virtual Annual Meeting.