AB928, a Novel Dual Adenosine Receptor Antagonist, Combined With Chemotherapy or AB122 (anti-PD-1) in Patients With Advanced Tumors: Preliminary Results From Ongoing Phase I Studies

Powderly J[1], de Souza P[2], Gutierrez R[3], Horvath L[4], Seitz L[5], Ashok D[5], Park A[5], Walters M[5], Karakunnel JJ[5], Berry W[5], Rieger A[5], Garofalo A[5], Lai DW[5], Chaudhry A[6]

[1]Carolina BioOncology Institute, Huntersville, NC, USA; [2]Western Sydney University, Campbelltown, NSW Australia; [3]The Angeles Clinic and Research Institute, Los Angeles, CA, USA; [4]Chris O'Brien Lifehouse, Camperdown, NSW Australia; [5]Arcus Biosciences, Inc., Hayward, CA, USA; [6]Summit Cancer Center, Spokane, WA, USA

Contact: Melissa Paoloni, Email: mpaoloni@arcusbio.com

ASCO 2019
Abstract # 2604

Introduction

The tumor microenvironment (TME) contains high levels of immunosuppressive adenosine (ADO), which binds to and activates the A_{2a} receptor ($A_{2a}R$) and A_{2b} receptors ($A_{2b}R$) on immune cells, leading to an ineffective anti-tumor response (**Figure 1**). CD73 enzyme and tissue non-specific alkaline phosphatase (TNAP) are primarily responsible for the conversion of extracellular adenosine mono-phosphate (AMP) to ADO.

Figure 1. Adenosine Pathway

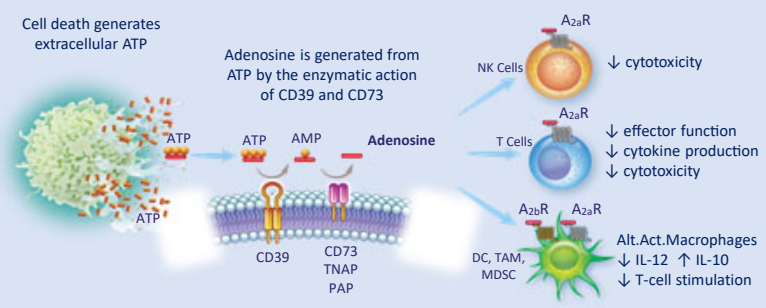

Cell death generates extracellular ATP
Adenosine is generated from ATP by the enzymatic action of CD39 and CD73
ATP
ATP
AMP
Adenosine
ATP
CD39
CD73
TNAP
PAP
NK Cells
A2aR
↓ cytotoxicity
T Cells
A2aR
↓ effector function
↓ cytokine production
↓ cytotoxicity
A2bR
A2aR
DC, TAM, MDSC
Alt.Act.Macrophages
↓ IL-12 ↑ IL-10
↓ T-cell stimulation

AB928 is a selective, small-molecule, dual $A_{2a}R/A_{2b}R$ antagonist with minimal penetration across the blood brain barrier. It was specifically designed to potently block the immunosuppressive effects of ADO in the TME. Preclinically, combining AB928 with either chemotherapy or anti-PD-1 resulted in greater tumor control, suggesting AB928 may have additive activity when paired with either of these agents in cancer patients.

Previously, we completed a dose-escalation study of AB928 in healthy volunteers. AB928 was shown to be well tolerated at all dose levels tested from 10 mg to 200 mg, once-daily dosing. We determined that a daily dose of 150 mg provides ≥ 90% receptor coverage at all time points assessed (Seitz et al., Investigational New Drugs (2018); *https://doi.org/10.1007/s10637-018-0706-6*). Pharmacokinetic (PK) and pharmacodynamic (PD) evaluations supported a starting dose of AB928 at 75 mg once daily in the ongoing combination trials in oncology patients (this poster).

Methods

- Three global phase 1/1b disease-specific platform studies are assessing the safety, tolerability, PK, PD, and clinical activity of AB928 in combination with chemotherapy (chemo) or AB122 (α-PD-1).
- Eligible participants have advanced solid tumors, an ECOG performance status of 0 or 1, and could be treatment naïve or have received up to 5 lines of prior therapy.
- AB928 was administered orally once daily in increasing doses starting at 75 mg with the intent to define the RP2D for each combination of chemotherapy (pegylated liposomal doxorubicin (PLD) or mFOLFOX), or AB122.
- Safety assessments include identification of adverse events (AEs) and dose limiting toxicities (DLTs), as well as changes from baseline in vital signs, clinical laboratory parameters, physical examination findings, ECOG performance, and ECG results. Safety analyses are based upon all participants who receive at least 1 dose of AB928.
- Tumor and peripheral blood samples were collected to describe the PK profile and assess the PD effects of AB928 in combination.
- Biomarker evaluations include gene sequence and expression, protein quantification, and T cell profiling before and after treatment with AB928 combinations.
- Clinical activity will be determined per RECIST v1.1 or appropriate response criteria in tumor-specific dose-expansion cohorts.

Trial Design

Table 1. List of AB928 Combination Studies in Oncology Subjects

Study Number	NCT Number	Combination	Tumor Type In Escalation*
AB928-002	NCT03719326	AB928 + PLD	TNBC or OC
AB928-003	NCT03720678	AB928 + mFOLFOX	GEC or CRC
AB928-005	NCT03629756	AB928 + AB122 (240 mg Q2W)	Solid tumors

*TNBC=Triple Negative Breast Cancer, OC=Ovarian Cancer, GEC=Gastro-Esophageal Cancer, CRC=Colorectal Cancer

Each study is made up of a dose-escalation and dose-expansion phase.

- **Dose Escalation (n=12-18)**: Escalating doses of AB928 (**Figure 2**) administered orally once daily (QD) in combination with chemotherapy backbone or AB122 (**Table 1**) will be assessed based on a 3+3 design.
- **Dose Expansion (n=15-40)**: RP2D of AB928 in combination will be used in tumor-specific dose-expansion cohorts.

Figure 2. Study Scheme

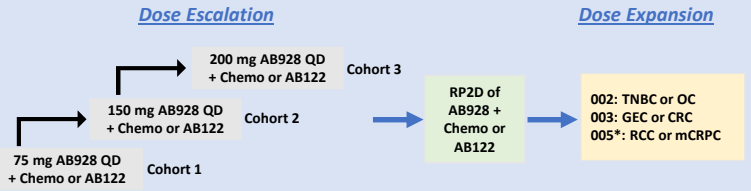

Dose Escalation
Dose Expansion
75 mg AB928 QD + Chemo or AB122
Cohort 1
150 mg AB928 QD + Chemo or AB122
Cohort 2
200 mg AB928 QD + Chemo or AB122
Cohort 3
RP2D of AB928 + Chemo or AB122
002: TNBC or OC
003: GEC or CRC
005*: RCC or mCRPC

*RCC=Renal Cell Carcinoma, mCRPC=metastatic Castration-Resistant Prostate Adenocarcinoma

Results

Demographics and Patient Characterization

As of 17 May 2019 (data cut-off), a total of 26 participants have been treated with AB928 combination therapy in studies: AB928-002 (AB928+PLD), n=7; AB928-003 (AB928+mFOLFOX), n=7; and AB928-005 (AB928+AB122), n=12.

Most participants were white and non-Hispanic, median age varied, and 77% were female (**Table 2**); 23% had received prior immunotherapy (α-PD(L)-1 or α-CTLA4).

In dose escalation, a total of 5/26 (19%) participants have discontinued AB928 treatment, most due to disease progression. As of the data cut-off, 21/26 (81%) participants were still on study, the majority in early evaluation.

Table 2. Dose Escalation Patient Demographics and Characterization

Characteristics		AB928-002		AB928-003		AB928-005		
		Cohort 1 (n=3)	Cohort 2 (n=4)	Cohort 1 (n=4)	Cohort 2 (n=3)	Cohort 1 (n=3)	Cohort 2 (n=6)	Cohort 3 (n=3)
Median (range) age, years		66 (40-68)	67 (57-80)	52 (37-59)	48 (39-62)	73 (56-79)	64 (50-66)	71 (56-77)
Biological sex, n (%)	Female	3 (100%)	4 (100%)	2 (50%)	1 (33%)	2 (67%)	5 (83%)	3 (100%)
	Male	0 (0%)	0 (0%)	2 (50%)	2 (67%)	1 (33%)	1 (17%)	0 (0%)
Tumor types*		OC n=3	OC n=4	CRC n=4	CRC n=3	OC, EC, Bladder	CRC n=3, EC n=2, NET	Unk, PDAC, OC
Prior LOT (median)		3		3		2		
Time on Treatment for mITT** (range, weeks)		7.1-12.3	TE***	2.4-14.3	4.1-6.3	5.3-42	6.3-21	1.1-8

*OC=Ovarian Cancer, CRC=Colorectal Cancer, EC=Endometrial Cancer, NET=Neuroendocrine Tumor, Unk=not yet defined in eDC, PDAC=Pancreatic Ductal Adenocarcinoma; **mITT=modified intent-to-treat population; ***TE=too early to assess

Safety and Tolerability

AEs are collected from the start of therapy until up to 90 days after the last dose of AB928 in combination or until initiation of a new systemic anticancer therapy. All AEs are graded according to NCI CTCAE v5.0.

Table 3. Summary of Treatment-Emergent Adverse Events (TEAEs) in AB928 Dose Escalation

Patients with	AB928-002		AB928-003		AB928-005		
	Cohort 1 (n=3)	Cohort 2 (n=4)	Cohort 1 (n=4)	Cohort 2 (n=3)	Cohort 1 (n=3)	Cohort 2 (n=6)	Cohort 3 (n=3)
TEAEs, n (%)	3 (100%)	2 (50%)	4 (100%)	3 (100%)	3 (100%)	3 (50%)	0
AB928-related TEAEs, n (%)	2 (66.7%)	1 (25%)	3 (75%)	1 (33.3%)	3 (100%)	2 (33%)	0
Grade 1-2, n (%)	3 (100%)	2 (50%)	4 (100%)	3 (100%)	3 (100%)	3 (50%)	0
AB928-related Grade 1-2, n (%)	2 (66.7%)	1 (25%)	3 (75%)	1 (33.3%)	3 (100%)	2 (33%)	0
Grade 3-4, n (%)	2 (66.7%)	0	4 (100%)	0	0	0	0
AB928-related Grade 3-4, n (%)	0	0	1 (25%)	0	0	0	0
SAE, n (%)	1 (33.3%)	0	2 (50%)	0	2 (67%)	0	0
AB928-related SAE, n (%)	0	0	0	0	0	0	0
DLT*, n	0	0	0	0	0	1	0

* The DLT period across all AB928 combination studies is 28 days.

The only DLT observed was due to Gr 2 rash in cohort 2 (AB928 + AB122) resulting in < 20% of drug administered.

Thus far, a maximum tolerated dose of AB928 in combination has not been reached.

The overall AB928-related AE profile is shown in **Table 4**. No Grade 4 or 5 AB928-related AEs were reported across the studies.

Table 4. AB928-related ≥ Grade 3 Adverse Event Profile by Treatment Group

AE Preferred Term (Grade 3 or above)	AB928-002		AB928-003		AB928-005		
	Cohort 1 (n=3)	Cohort 2 (n=4)	Cohort 1 (n=4)	Cohort 2 (n=3)	Cohort 1 (n=3)	Cohort 2 (n=6)	Cohort 3 (n=3)
Anemia	0	1	1	0	0	0	0
Fatigue	0	0	1	0	0	0	0
Leukopenia	0	1	0	0	0	0	0
Nausea	0	0	1	0	0	0	0
Neutropenia	0	1	0	0	0	0	0

AB928 PK/PD in Oncology Subjects is Comparable to That Seen in Healthy Volunteers

The inhibition of $A_{2a}R$-mediated effects by AB928 was determined in blood samples from all AB928 studies by the decreased phosphorylation of CREB (pCREB) following stimulation *ex vivo* with 5 µM of the adenosine receptor agonist NECA (5'-N-ethylcarboxamidoadenosine). At each PD time point, levels of pCREB in CD8⁺ T cells were assessed by phosphoflow cytometry. pCREB inhibition following AB928 dosing in each subject was calculated by normalizing against the NECA response observed at the pre-dose time point.

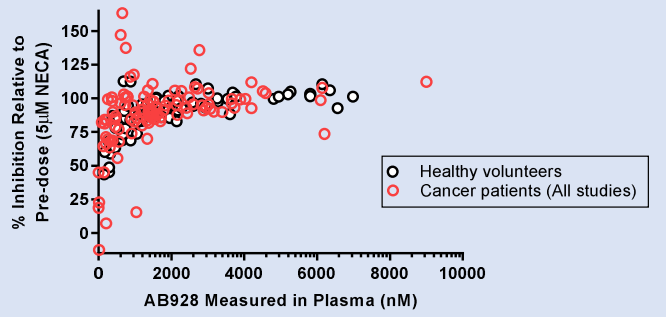

% Inhibition Relative to Pre-dose (5µM NECA)
AB928 Measured in Plasma (nM)
Healthy volunteers
Cancer patients (All studies)

Figure 3: PK/PD correlations are shown from oncology subjects enrolled in AB928 combination trials with chemotherapy or AB122 (red circles) and from healthy volunteers (black circles). Data from oncology subjects and healthy volunteers are not statistically different (p= 0.91, Kolmogorov-Smirnov model).

Evidence of Immune Engagement in a Subject With a CD73 High, PD-L1 Low, TMB Low, Ovarian Carcinoma

Across the AB928 combination studies, tumor biopsies were obtained at screening or as archival (w/n 6 months) tissue. On-treatment biopsies were collected around Day 43, if medically feasible. The example below illustrates the planned biomarker characterization of all participants.

The following information is from a subject with ovarian carcinoma in study 005 (AB928+AB122) (**Figures 4-6**):

- Diagnosed May 2016; received 3 prior lines of therapy (Carbo/PLD, PARPi, Carbo/Gemzar) in the recurrent/metastatic setting, naïve to α-PD(L)-1 treatment
- Baseline tumor characteristics: low TMB, PD-L1 TPS 1% and CPS 4% (by 22C3), MSS

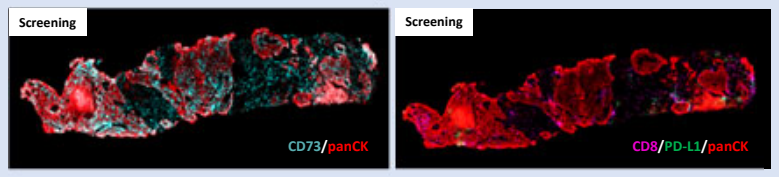

Screening
CD73/panCK
Screening
CD8/PD-L1/panCK

Figure 4: Histologic images of tumor biopsies at screening (above). Immunostaining identifies biopsy cores with cancerous cells (red) which have high levels of CD73 (>50% positive cells, top left), low PD-L1 expression, and minimal CD8⁺ T cell infiltration (top right).

AB928 + AB122 Induced Tumoral CD8⁺ T Cell Infiltration and Effector T Cell Gene Signature

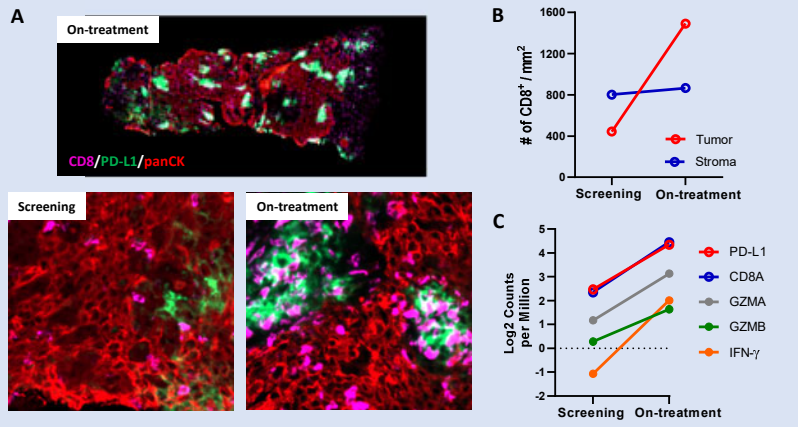

A
On-treatment
CD8/PD-L1/panCK
Screening
On-treatment
B
of CD8⁺ / mm²
Tumor
Stroma
Screening
On-treatment
C
Log2 Counts per Million
PD-L1
CD8A
GZMA
GZMB
IFN-γ
Screening
On-treatment

Figure 5: A, CD8 density in screening and on-treatment biopsies (tumor=panCK positive; stroma=panCK negative). B, A large increase is seen in intra-tumoral CD8⁺ T cells with no additional accumulation in the stroma. C, Expression (mRNA) of effector T cell genes, defined by CD8A, Granzyme A/B, IFN-γ and PD-L1, increase in the on-treatment biopsy.

AB928 + AB122 Induced Expansion of Intra-tumoral T Cell Clones That Were Not Detectable at Screening

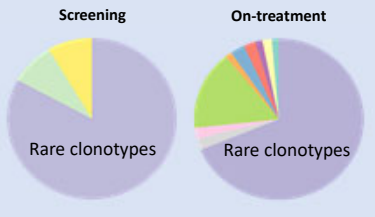

Screening
On-treatment
Rare clonotypes
Rare clonotypes

Figure 6: TCR clonality in screening and on-treatment biopsies by RNA-Seq. Chart segments represent unique T cell clonotypes identified within the bulk population. Clonotypes representing less than 2% of the population are pooled in the largest segment. Analysis of the on-treatment biopsy indicates clonal expansion of specific T cell clonotypes as well as enhanced diversity.

This subject has remained on treatment for 42 weeks with stable disease. Biomarker characterization suggests enhanced intra-tumoral immune infiltration, effector T cell gene signature and TCR clonality post-treatment with AB928 and AB122.

Tumor Response and Disease Stabilization After Treatment With AB928 + AB122

Early clinical data from the dose-escalation portion of the AB928 + AB122 trial. Study-eligible participants have advanced tumor types with traditionally low levels of response to α-PD(L)-1 therapies.

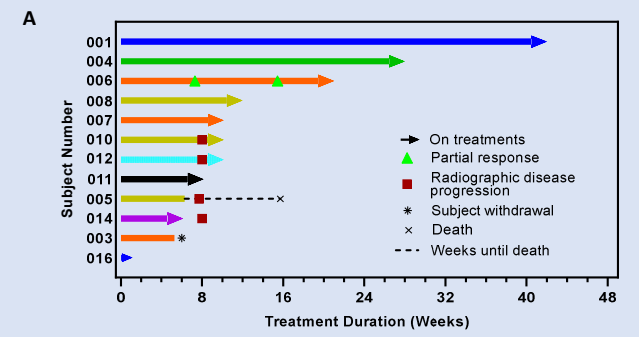

A
Subject Number
001
004
006
008
007
010
012
011
005
014
003
016
On treatments
Partial response
Radiographic disease progression
Subject withdrawal
Death
Weeks until death
Treatment Duration (Weeks)

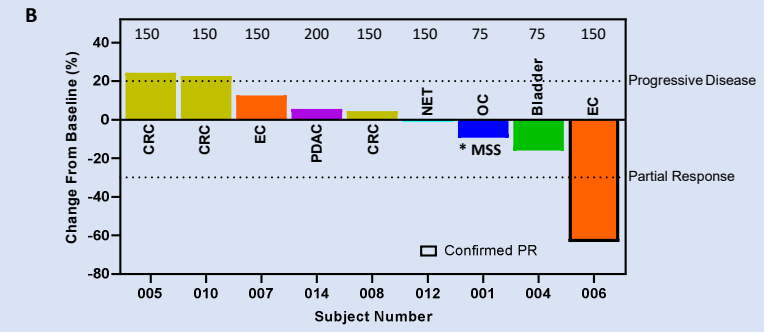

B
Change From Baseline (%)
150
150
150
200
150
150
75
75
150
CRC
CRC
EC
PDAC
CRC
NET
OC
Bladder
EC
* MSS
Progressive Disease
Partial Response
Confirmed PR
005
010
007
014
008
012
001
004
006
Subject Number

Figure 7: A, Tumor response for individual subjects by weeks. Each bar represents one subject in the study. Colors in the swimlane plot match the tumor types defined in the waterfall plot, except for one undefined histology (subject 011). B, Best percentage change from baseline. A total number of 9 subjects (RECIST evaluable) had 1 or more disease assessments. There is one confirmed partial response in a mixed Mullerian endometrial cancer (EC) participant (subject 006). The *OC participant (MSS=Microsatellite Stable) is the same subject depicted in **Figures 4-6**.

Conclusions

- Early escalation of AB928 in combination with chemotherapy or AB122 demonstrates a favorable safety profile.
- AB928 PK/PD in oncology subjects is comparable to that of healthy volunteers.
- Preliminary biomarker characterization illustrates increased CD8⁺ T cell infiltrate into the tumor, associated with an elevated effector T cell signature and T cell clonal expansion in a CD73-high tumor microenvironment.
- Initial clinical evaluation of AB928 in combination with AB122 demonstrates tumor responses and disease stabilization in advanced tumor types with traditionally low levels of response to α-PD(L)-1 therapies.

Acknowledgements

We thank the Principal Investigators, site staff, and study participants for their efforts on behalf of these studies. We acknowledge Dan DiRenzo, Akshata Udyavar, Mike Farias and Steve Young for their contributions to the biomarker analyses. Lastly we thank Fangfang Yin and Melissa Paoloni for their assistance with the poster's development.