Poster #5039

ARCUS BIOSCIENCES

ARC-6: A Phase 1b/2, Open-Label, Randomized Platform Study to Evaluate Efficacy and Safety of Etrumadenant (AB928)-Based Treatment Combinations in Patients with Metastatic Castrate-Resistant Prostate Cancer

Sumit K. Subudhi,[1] Johanna C. Bendell,[2] Michael Carducci,[3] Lisa M. Kopp,[4] Jennifer R. Scott,[4] Michele Grady,[4] Olivia S. Gardner,[4] David R. Wise[5]

[1]University of Texas MD Anderson Cancer Center, Houston, TX; [2]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [3]Johns Hopkins Sidney Kimmel Comprehensive Cancer Center, Baltimore, MD; [4]Arcus Biosciences, Inc., Hayward, CA; [5]New York University Perlmutter Cancer Center, New York, NY

INTRODUCTION

The Adenosine Axis in Cancer

- Standard-of-care (SOC) chemotherapy regimens may contribute to immunosuppression by elevating intratumoral levels of adenosine triphosphate (ATP) in the tumor microenvironment (TME) where the enzymes CD39 and CD73 successively convert ATP to adenosine; in prostate cancer, the highly expressed protein, prostatic acid phosphatase (PAP), produces additional adenosine[1,2] (**Figure 1**)
- By binding adenosine receptors 2a and 2b ($A_{2a}R$ and $A_{2b}R$) expressed on immune cells, adenosine promotes immunosuppression by inhibiting critical components of the antitumor immune response, ultimately enabling tumors to evade destruction[2]
- Additionally, $A_{2a}R$ signaling impairs the activation, proliferation, and cytotoxic activity of effector T cells[3]
- Initial research focused on $A_{2a}R$ as the most relevant adenosine receptor in cancer physiology; however, $A_{2b}R$ signaling through MAP kinase pathway activation mediates unique functions, such as cancer cell intrinsic survival and dendritic cell activation and function[4]
- Thus, targeting the adenosine axis in combination with standard chemotherapy regimens or immunotherapy may have a more profound effect on activating and inducing sustained antitumor immunity in prostate cancer[2]

Figure 1. Critical Role of Adenosine Pathway in the Immunosuppressive Tumor Microenvironment

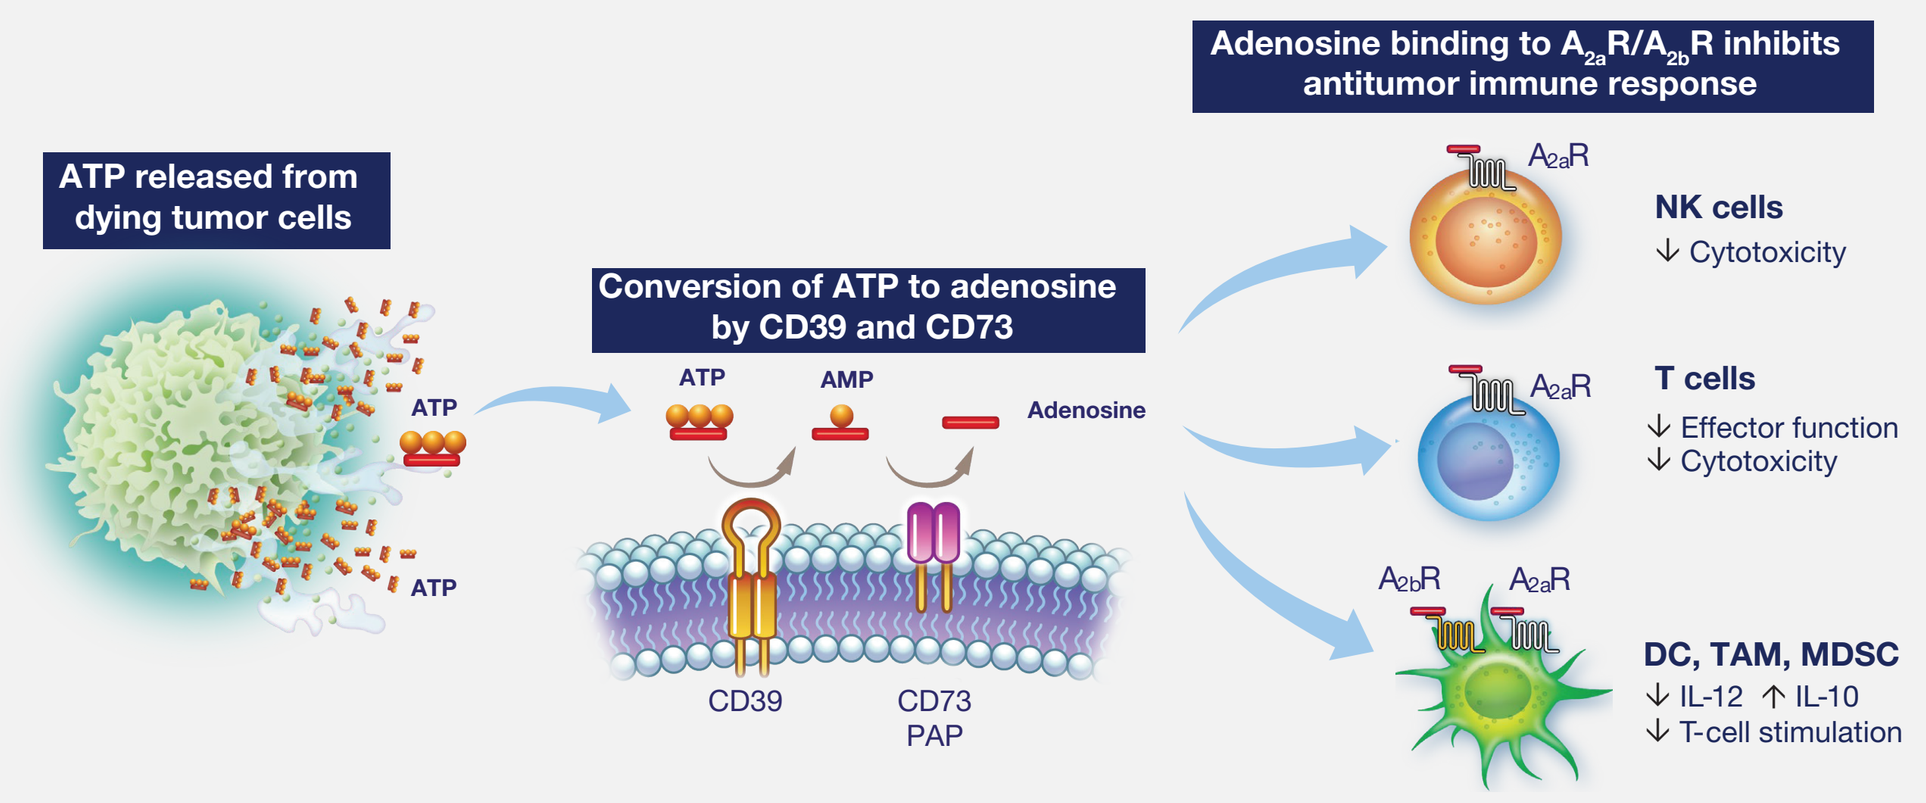
Adenosine binding to $A_{2a}R/A_{2b}R$ inhibits antitumor immune response
ATP released from dying tumor cells
Conversion of ATP to adenosine by CD39 and CD73
ATP
AMP
Adenosine
CD39
CD73
PAP
$A_{2a}R$
NK cells
↓ Cytotoxicity
T cells
↓ Effector function
↓ Cytotoxicity
$A_{2b}R$
DC, TAM, MDSC
↓ IL-12 ↑ IL-10
↓ T-cell stimulation

AMP, adenosine monophosphate; ATP, adenosine triphosphate; $A_{2a}R/A_{2b}R$, adenosine receptors 2a/2b; DC, dendritic cell; IL, interleukin; MDSC, myeloid-derived suppressor cells; NK, natural killer; PAP, prostatic acid phosphatase; TAM, tumor-associated macrophage.

- Etrumadenant is an orally bioavailable, small-molecule, selective dual antagonist of $A_{2a}R$ and $A_{2b}R$ that was specifically designed to block the immunosuppressive effects associated with high adenosine concentration within the TME; it is the most advanced adenosine receptor antagonist in active clinical trials that potently and selectively blocks $A_{2a}R$ and $A_{2b}R$
- There are several ongoing randomized Phase 1b/2 studies to assess safety, tolerability, pharmacokinetics (PK), pharmacodynamics (PD), and preliminary clinical activity of etrumadenant in combination with chemotherapy and/or anti-programmed cell death protein 1 (PD-1) antibody[5-8]
 - Based on dose-escalation data, etrumadenant 150 mg once daily (QD) was selected as the recommended dose for expansion (RDE) based on PK, PK/PD correlation, and a well-tolerated safety profile of etrumadenant + chemo/immunotherapy
- Recently, in patients with late-line metastatic colorectal cancer, etrumadenant in combination with chemotherapy demonstrated an objective response rate (ORR) as well as median progression-free survival and overall survival (OS) that compare favorably with current SOC treatments[7]

ARC-6 Study Rationale

- In prostate tumors, PAP plays a similar role to CD73 in converting adenosine monophosphate (AMP) to adenosine[2]
 - Serum PAP levels are increased in patients with prostate cancer, particularly in those with metastatic disease, compared with healthy adults[9]
- In metastatic castrate-resistant prostate cancer (mCRPC), adenosine axis expression and myeloid signatures were negatively correlated with OS, with $A_{2b}R$ being one of the most prognostic genes evaluated in a gene expression dataset[10]
- Combination therapy that includes antagonism of the adenosine and PD-1/programmed death-ligand 1 (PD-L1) pathways with chemotherapy may hold promise for enhancing treatment efficacy without additional toxicity
 - In early clinical trials, the combination of $A_{2a}R$ and PD-L1 inhibition has antitumor activity in some patients with late-line mCRPC[11]
 - Early data suggest that etrumadenant in combination with zimberelimab (AB122), a human monoclonal antibody (mAb) targeting PD-1, is well tolerated and has clinical benefit in heavily pretreated patients across tumor types (NCT03846310)[5]
 - In patients with mCRPC, docetaxel plus anti–PD-1 therapy has clinical activity in some patients[12], but there remains an unmet need for efficacy in a greater proportion of patients, especially in those that are late-line
- Efficacy of docetaxel plus immunotherapy may be limited by the immunosuppression caused by high levels of adenosine in the prostate TME; thus, adding an adenosine receptor antagonist such as etrumadenant to chemo/immunotherapy may enhance antitumor activity in mCRPC

METHODS

ARC-6 Study Design

- ARC-6 (NCT04381832) is an ongoing, Phase 1b/2, randomized, open-label, multi-cohort study to evaluate the efficacy and safety of etrumadenant combination therapy in patients with mCRPC (**Figure 2**)
 - The data presented herein represent initial efficacy and safety results for etrumadenant + zimberelimab in combination with the SOC backbone, docetaxel (EZD); enrollment for the Phase 1b EZD arm is complete
 - Randomized Phase 2 enrollment, EZD vs docetaxel, was initiated in January 2021 and is ongoing

Figure 2. ARC-6 Study Design

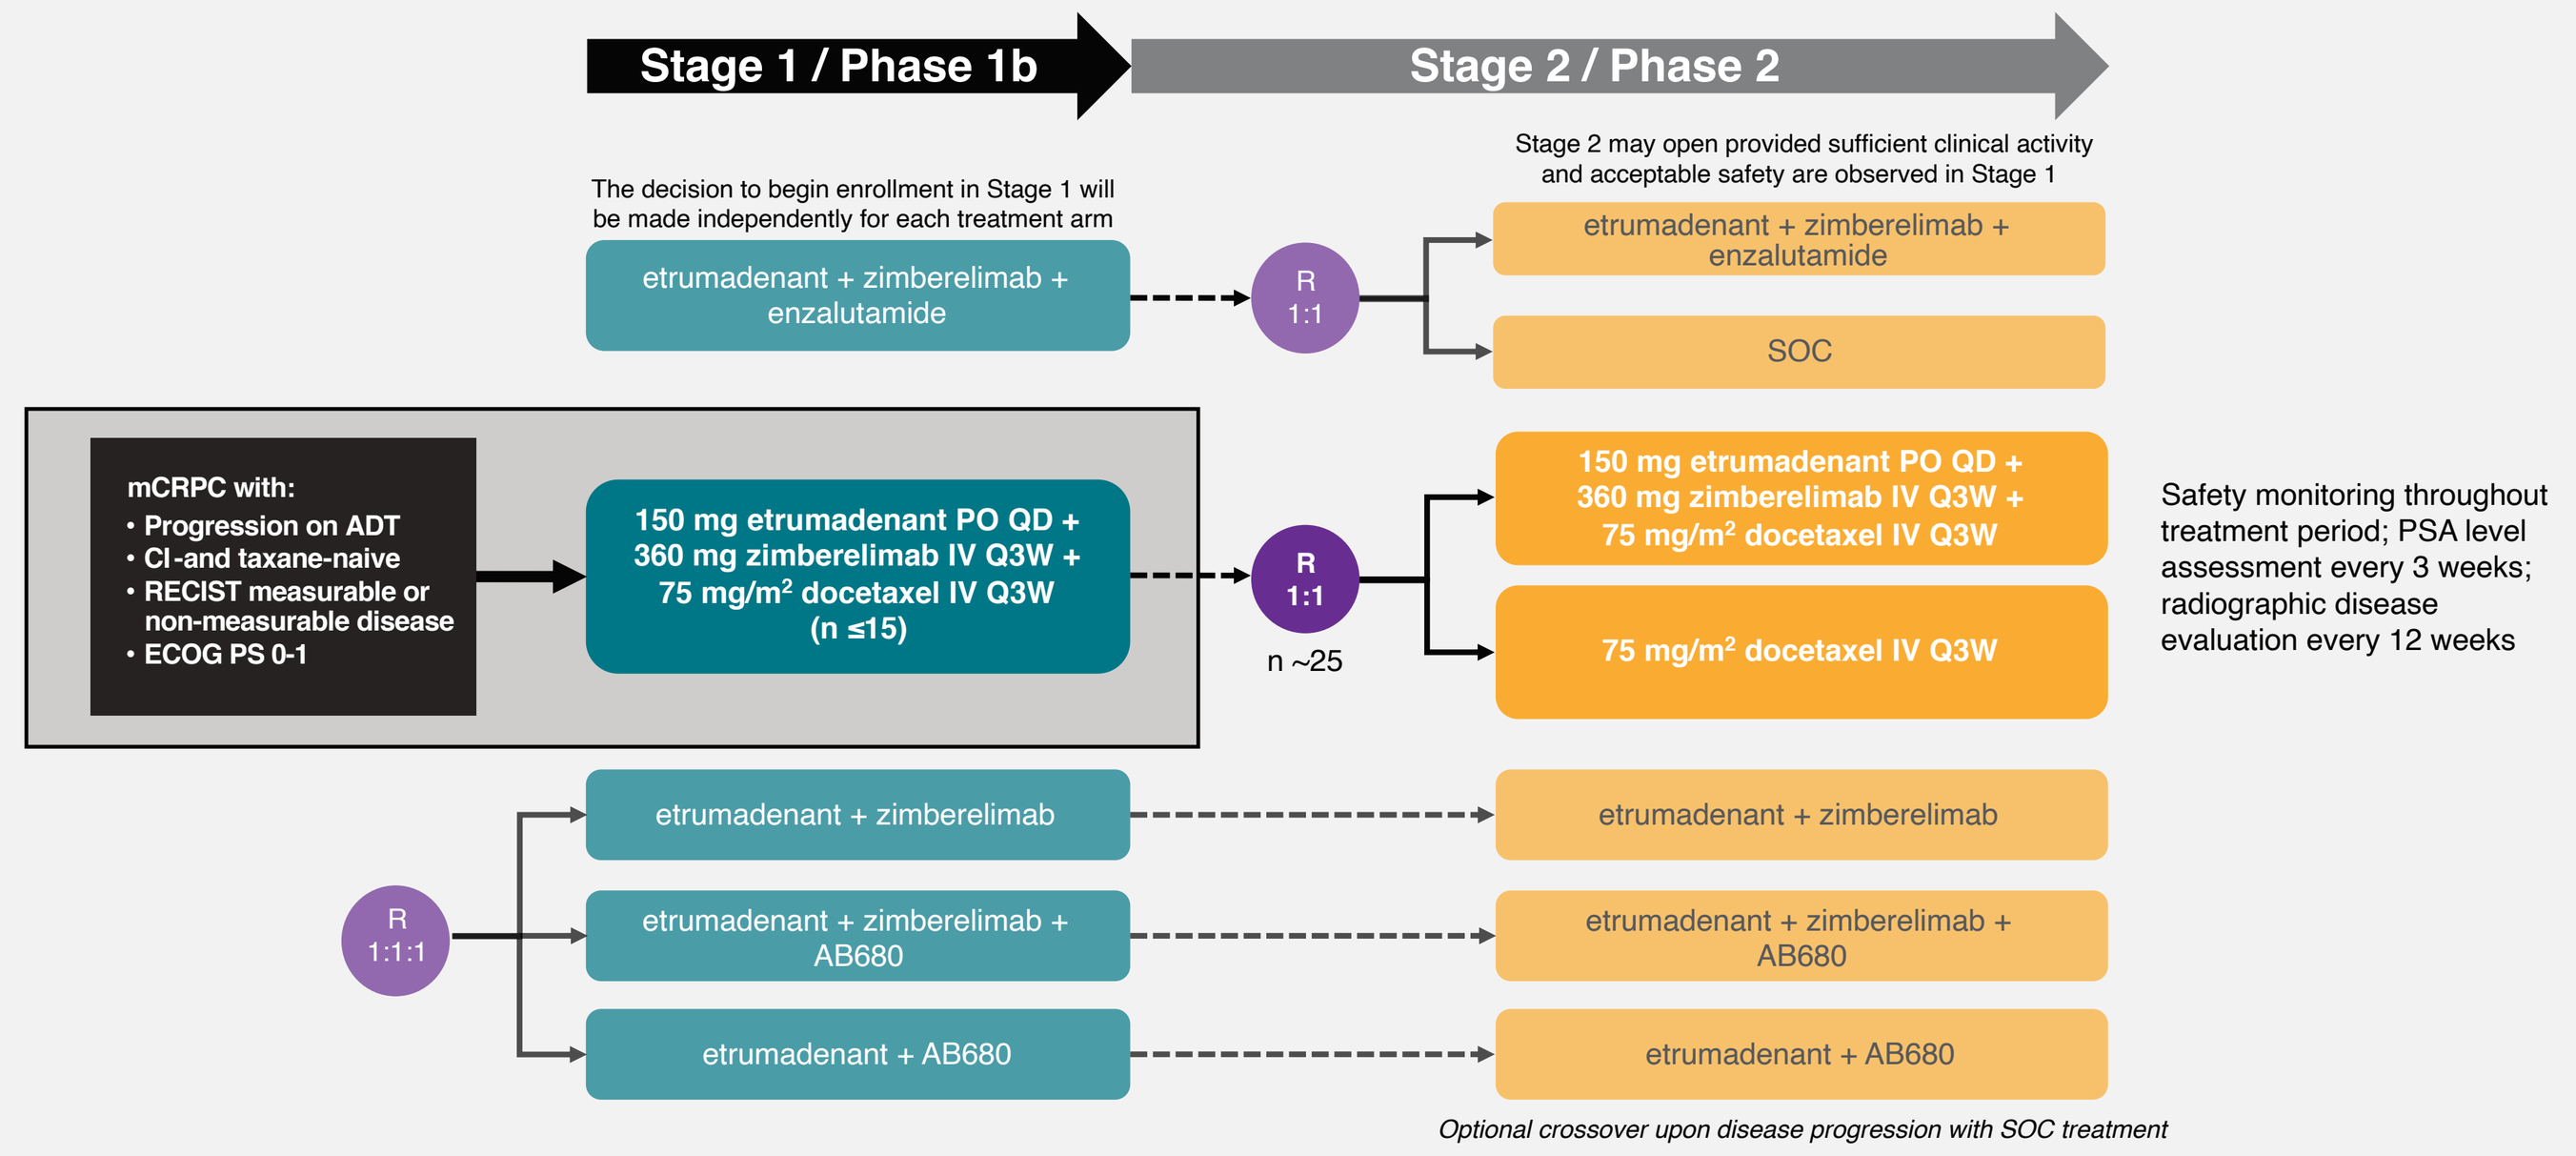
Stage 1 / Phase 1b
Stage 2 / Phase 2
The decision to begin enrollment in Stage 1 will be made independently for each treatment arm
Stage 2 may open provided sufficient clinical activity and acceptable safety are observed in Stage 1
etrumadenant + zimberelimab + enzalutamide
R 1:1
etrumadenant + zimberelimab + enzalutamide
SOC
mCRPC with:
• Progression on ADT
• CI-and taxane-naive
• RECIST measurable or non-measurable disease
• ECOG PS 0-1
150 mg etrumadenant PO QD + 360 mg zimberelimab IV Q3W + 75 mg/m² docetaxel IV Q3W (n ≤15)
R 1:1
n ~25
150 mg etrumadenant PO QD + 360 mg zimberelimab IV Q3W + 75 mg/m² docetaxel IV Q3W
75 mg/m² docetaxel IV Q3W
Safety monitoring throughout treatment period; PSA level assessment every 3 weeks; radiographic disease evaluation every 12 weeks
R 1:1:1
etrumadenant + zimberelimab
etrumadenant + zimberelimab + AB680
etrumadenant + AB680
etrumadenant + zimberelimab
etrumadenant + zimberelimab + AB680
etrumadenant + AB680
Optional crossover upon disease progression with SOC treatment

ADT, androgen deprivation therapy; CI, checkpoint inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; IV, intravenously; mCRPC, metastatic castrate-resistant prostate cancer; PO, orally; PSA, prostate-specific antigen; R, randomization; QD, once daily; Q3W, once every 3 weeks; SOC, standard of care.

ARC-6 Design Features

- The primary objectives are to evaluate safety and antitumor activity (prostate-specific antigen [PSA], radiographic, and composite objective response rates) of EZD
- For the EZD arm, eligible patients have pathologically-confirmed mCRPC that has progressed while on androgen deprivation therapy (ADT), no prior treatment with immune checkpoint inhibitors or taxane chemotherapy, measurable or non-measurable disease per RECIST v1.1, and an ECOG performance status 0–1
- Patients are allowed to receive study treatment until disease progression, unacceptable toxicity, or investigator decision
- PSA levels are assessed every 3 weeks and radiographic scans are performed every 12 weeks; responses are assessed according to PCWG3 criteria

Statistical Analysis

- Safety analyses included all enrolled patients who received any amount of study treatment
 - Summary statistics were provided for treatment-emergent adverse events (TEAEs) and serious TEAEs (TESAEs), TEAE relationship to study drugs, and TEAE severity based on NCI CTCAE v5.0
- Efficacy analyses included all enrolled patients who received ≥1 dose of each drug in the EZD combination regimen
 - PSA response-evaluable patients have baseline and ≥2 consecutive post-baseline PSA assessments
 - Radiographic response-evaluable patients have RECIST measurable or non-measurable disease per baseline imaging and ≥1 post-baseline radiographic assessment
 - Composite ORR is defined as the proportion of patients with a PSA response or radiographic complete response (CR) or partial response (PR) according to PCWG3 criteria

RESULTS

Patient Baseline Characteristics

- As of April 9, 2021, 17 patients have received EZD in the Phase 1b portion of the study
- For all patients, the mean age at baseline was 70 years and most were white (11/17; 65%); the majority of patients with available data (10/15; 67%) had a total Gleason score ≥8 at initial diagnosis **(Table 1)**

Table 1. Patient Demographics and Characteristics

Parameter	N=17[a]
Age, mean (SD), years	70 (8)
Race, n (%)	
Black	5 (29)
White	11 (65)
Not reported	1 (6)
Total Gleason score at initial diagnosis, n (%)	
6	2 (12)
7	3 (18)
8	3 (18)
9	7 (41)
Unknown	2 (12)
Type of progression at study entry[b], n (%)	
PSA elevation	17 (100)
Radiographic progression	17 (100)
ECOG performance status, n (%)	
0	10 (59)
1	7 (41)

[a] Percentages in this column may not equal 100 due to rounding; [b] As defined by PCWG3 criteria. ECOG, Eastern Cooperative Oncology Group; PSA, prostate-specific antigen; PCWG3, Prostate Cancer Working Group 3; SD, standard deviation.

- All patients had received prior systemic therapy; 16/17 (94%) had received ≥3 prior lines including ADT **(Table 2)**
- Most patients (13/17; 76%) had received ≥1 prior anti-androgen and 11/17 patients (65%) had received prior abiraterone

Table 2. Prior Therapy for Metastatic Disease

Parameter	n (%)[a]
Prior systemic anticancer therapy	17 (100)
Previous lines of treatment[b]	
1	0
2	1 (6)
3	3 (18)
>3	13 (76)
Prior radiotherapy	10 (59)
Prior anti-androgens	13 (76)
Previous lines of anti-androgens	
1	4 (24)
2	8 (47)
≥3	1 (6)
Prior abiraterone	11 (65)

[a] Percentages in this column may not equal 100 due to rounding; [b] Including prior androgen deprivation therapy.

Safety Analyses

- All patients reported ≥1 TEAE; the most common TEAEs were alopecia (53%), lymphocyte count decreased (53%), and fatigue (47%; **Table 3**)
- Grade 3 or 4 related TEAEs were reported by 6/17 (35%) patients; all of these events were related to etrumadenant and may also be attributed to zimberelimab and/or docetaxel
 - 1 patient reported Grade 4 TEAEs of lymphocyte count decreased, neutrophil count decreased, and white blood cell count decreased during the first treatment cycle; the events were deemed related to all 3 study drugs, but no changes in study treatment were made and all events resolved
- TESAEs occurred in 5/17 (29%) of patients; none were considered related to etrumadenant
 - Only 1 patient reported any study treatment-related TESAE (Grade 4 anemia); the event prompted EZD interruption, the patient received supportive care, and the event resolved
 - 1 patient died from a TESAE of cardiac arrest on day 107 of the trial; this event was not considered related to any study treatment
- Two patients (12%) discontinued 1 study drug due to AEs: 1 patient reported zimberelimab-related diarrhea and continued on ED and the other reported docetaxel-related peripheral neuropathy and continued on EZ; as of April 9, 2021, both patients are still on study

Table 3. Treatment-Emergent Adverse Events

Parameter	N=17, n (%)
Any TEAE	17 (100)
Grade ≥3 TEAEs	13 (76)
Any TESAE	5 (29)
Grade ≥3 TESAEs	5 (29)
Etruma-related TEAEs	13 (76)
Etruma-related TESAEs	0
All study treatment d/c due to TEAEs	2 (12)
Deaths due to TEAEs	1 (6)[a]
TEAEs in >30% of all patients	
Alopecia	9 (53)
Lymphocyte count decreased	9 (53)
Fatigue	8 (47)
Constipation	7 (41)
Hyponatremia	7 (41)
Neutrophil count decreased	7 (41)
Hyperglycemia	6 (35)
Hypophosphatemia	6 (35)
White blood cell count decreased	6 (35)

[a] Death was due to cardiac arrest that was considered unrelated to study drug. d/c, discontinuation; etruma, etrumadenant; TEAE, treatment-emergent adverse event; TESAE, treatment-emergent serious adverse event.

Clinical Activity

- As of April 9, 2021, the median time on treatment for all patients was 4.2 months (range: 2.1–8.3+ months; **Figure 3**)
- Eleven patients remain on study treatment as of the data cut off

Figure 3. Time on EZD and PCWG3 Response

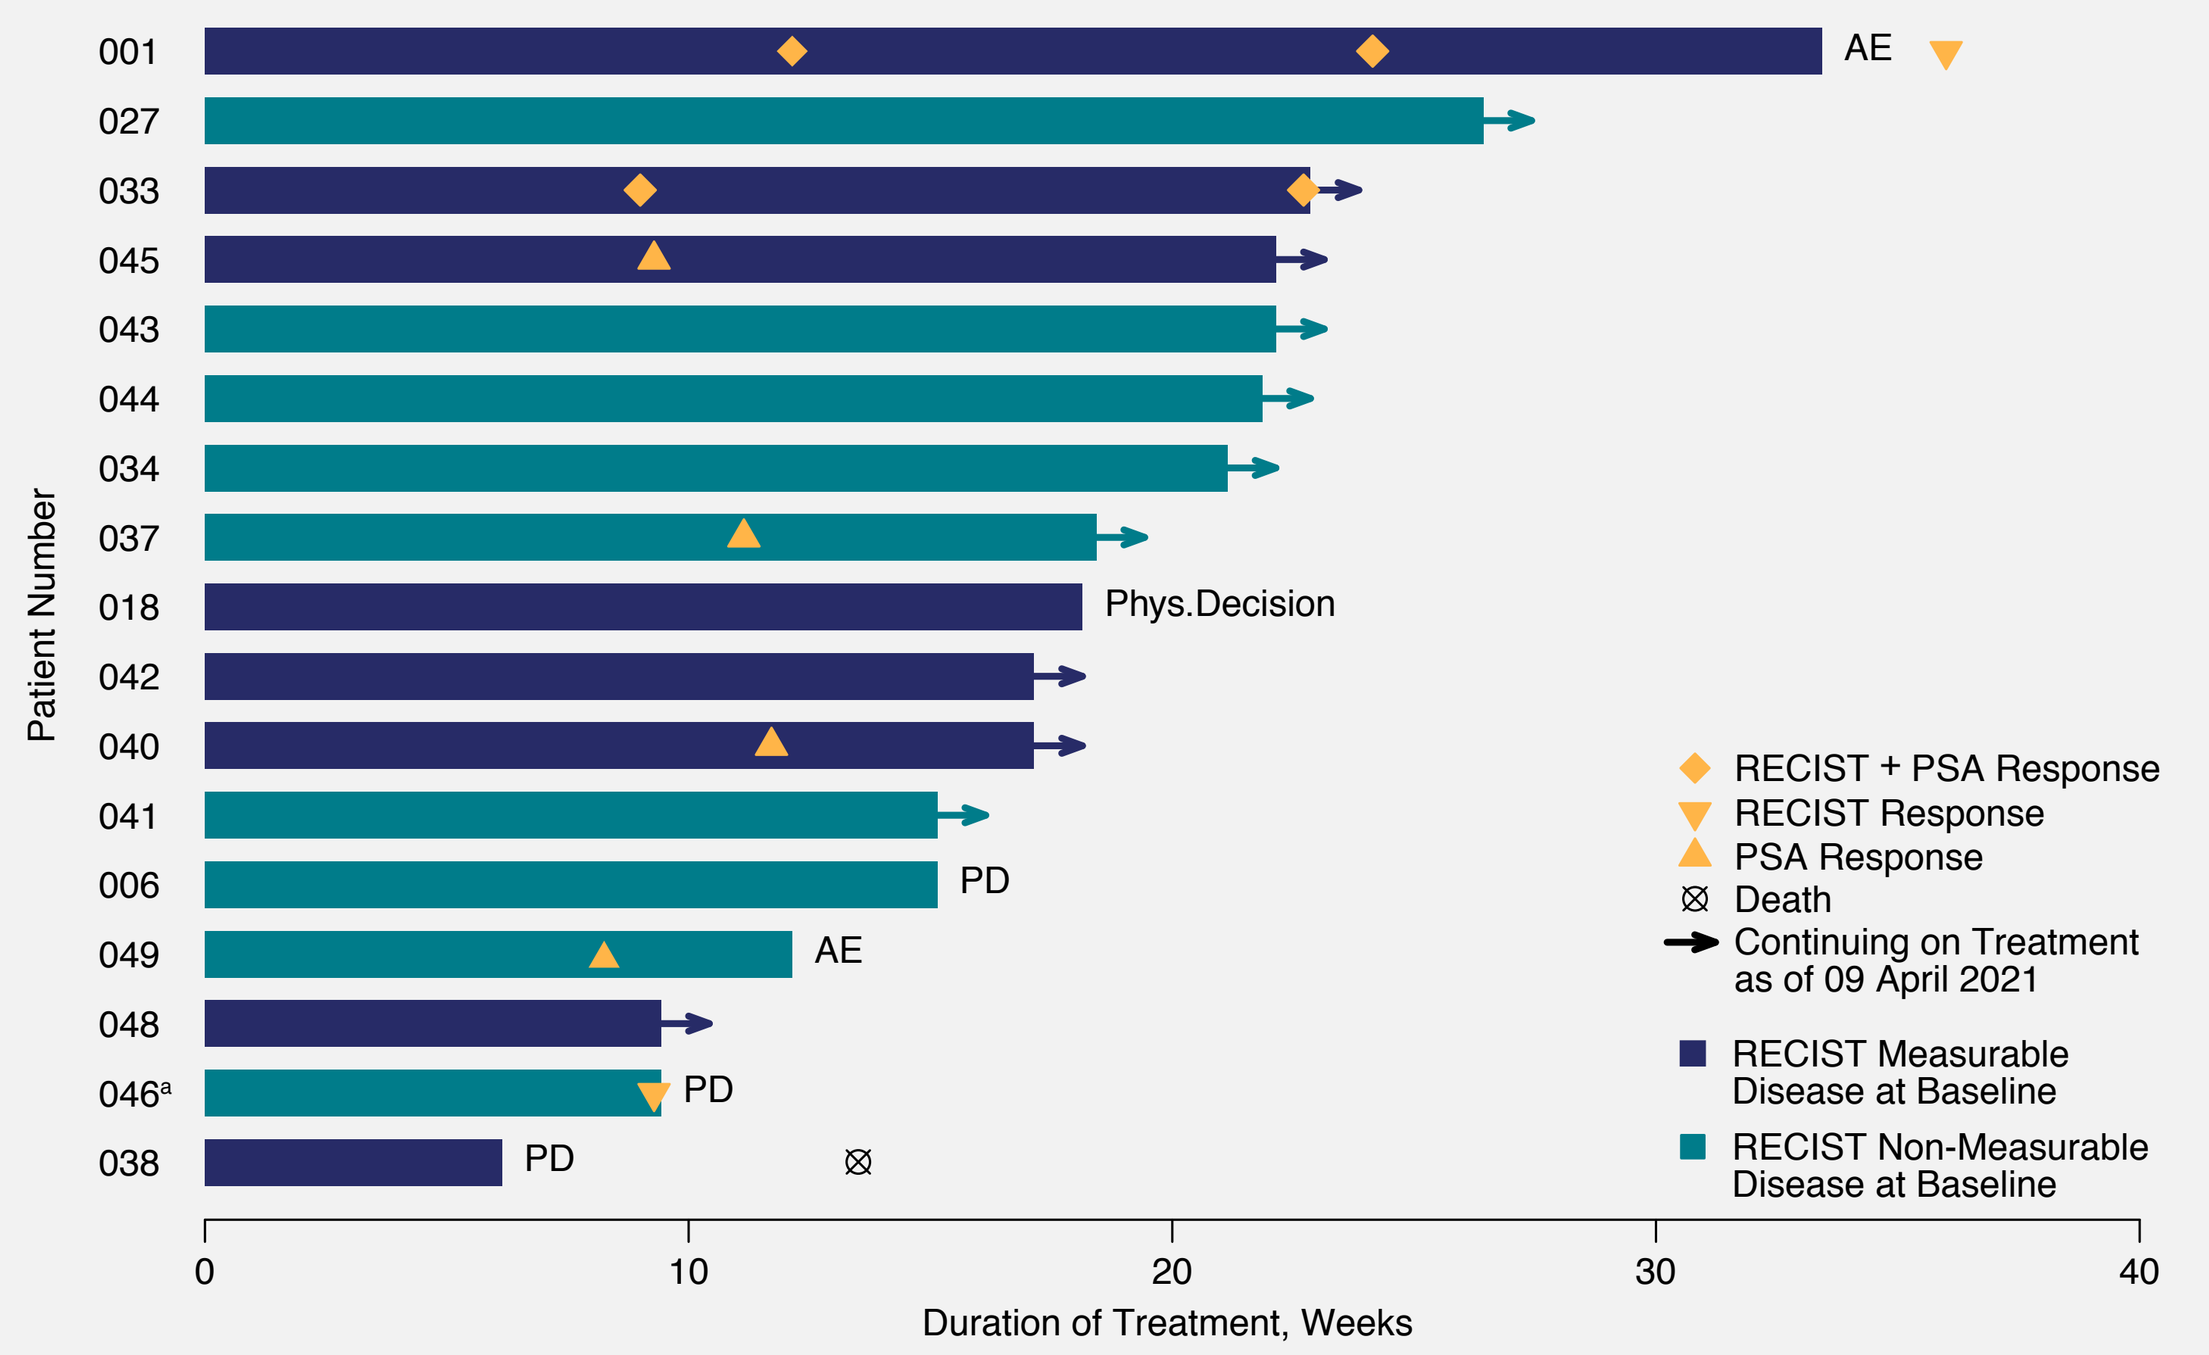
001
027
033
045
043
044
034
037
018
042
040
041
006
049
048
046[a]
038
AE
Phys.Decision
PD
AE
PD
PD
Patient Number
0
10
20
30
40
Duration of Treatment, Weeks
RECIST + PSA Response
RECIST Response
PSA Response
Death
Continuing on Treatment as of 09 April 2021
RECIST Measurable Disease at Baseline
RECIST Non-Measurable Disease at Baseline

[a] Patient 046 developed new, symptomatic bone metastases prompting an unscheduled scan subsequent to the first post-baseline radiographic disease evaluation and was taken off treatment due to PD. AE, adverse event; EZD, etrumadenant + zimberelimab + docetaxel; PCWG3, Prostate Cancer Working Group 3; PD, progressive disease; PSA, prostate-specific antigen.

Best Overall Response

- As shown in **Table 4**, 7/17 (41%) patients achieved either radiographic and/or PSA response
- All 11 patients with RECIST measurable or non-measurable disease experienced clinical benefit and achieved a best overall response (BOR) of SD or better, including 1 CR (unconfirmed) in a patient with non-target disease only (046) and 2 PRs (both confirmed; 033 and 001); 1 patient with PR also had improvement in bone disease burden (001)

Table 4. Summary of Best Overall Responses

Parameter	
BOR by RECIST v1.1[a]	**n=11**
CR, n (%)	1 (9)
PR, n (%)	2 (18)
SD, n (%)	8 (73)
PD, n (%)	0
BOR by PSA[b]	**n=17**
>50% decrease, n (%)	6 (35)
Non-response/non-progression, n (%)	7 (41)
Composite BOR [CR + PR + PSA response], n (%)	7 (41)

[a] Evaluable patients have RECIST measurable or non-measurable disease per baseline imaging and ≥1 post-baseline radiographic assessment;
[b] PSA response-evaluable patients have baseline and ≥2 consecutive post-baseline PSA assessments. BOR, best overall response; CR, complete response; PD, progressive disease; PR, partial response; PSA, prostate-specific antigen; SD, stable disease.

- Six patients had a >50% decrease in PSA level and 7 patients had non-response/non-progression (**Figure 4**)

Figure 4. PSA Percentage Change from Baseline

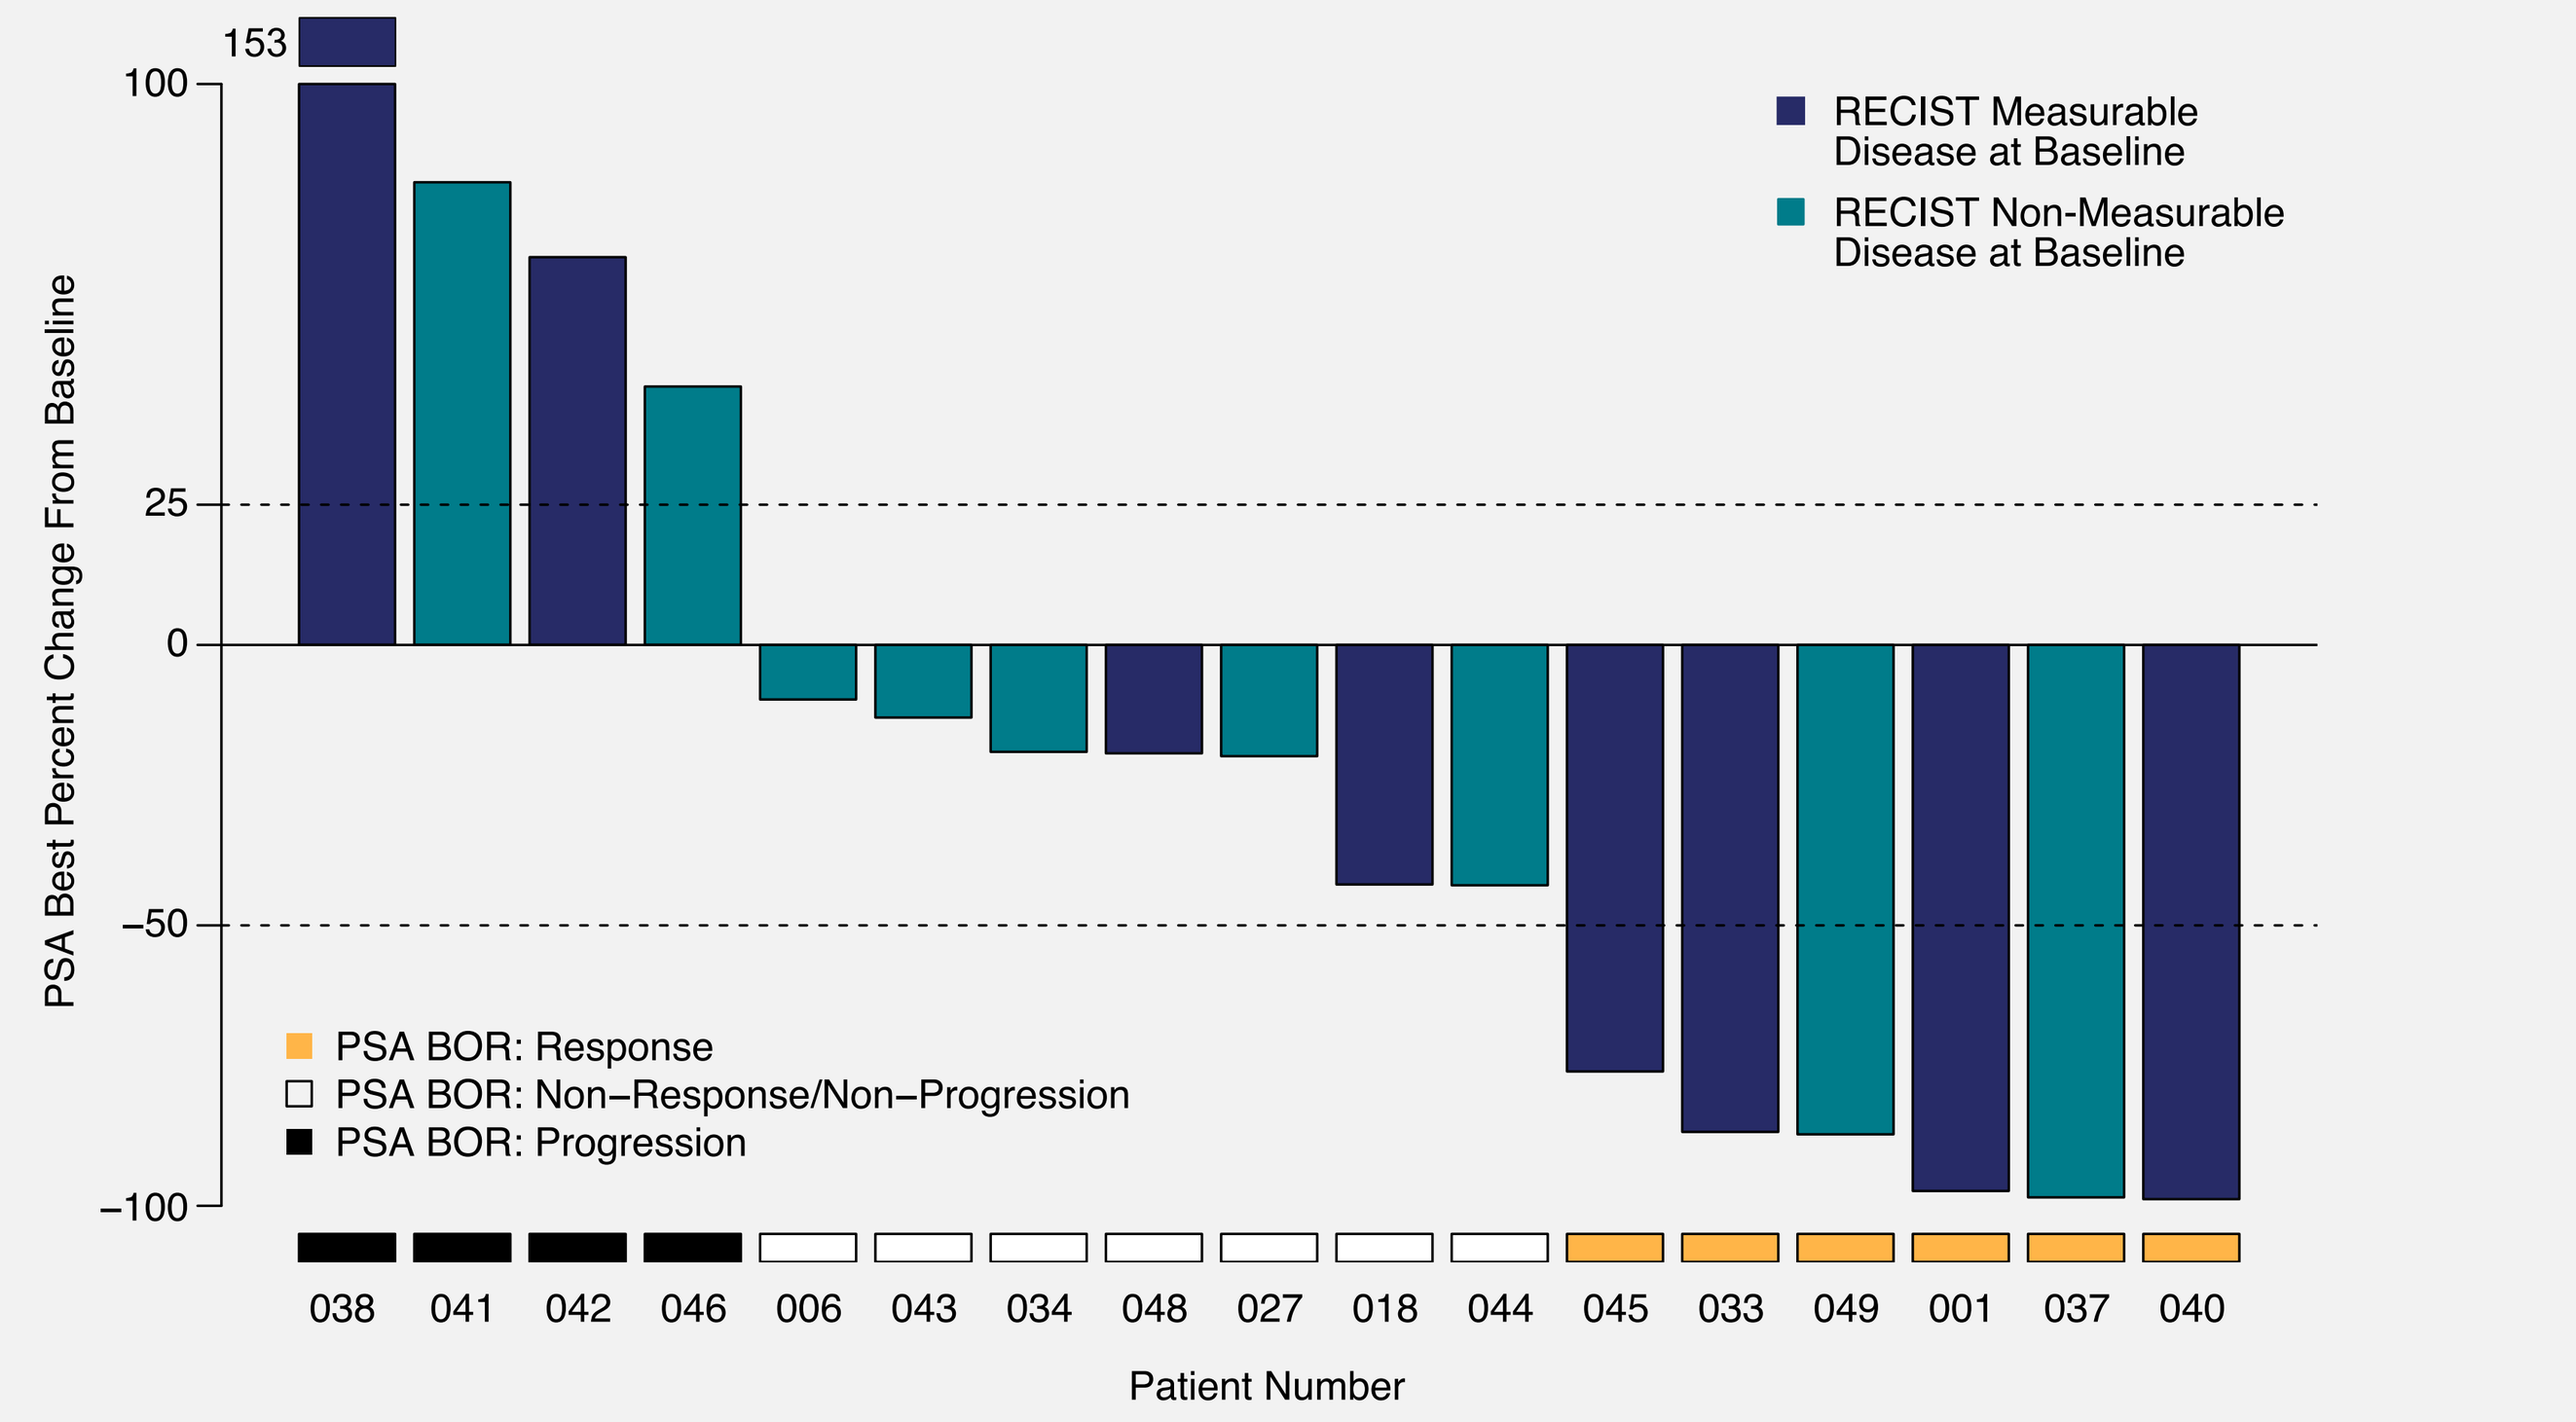
153
100
25
0
−50
−100
PSA Best Percent Change From Baseline
RECIST Measurable Disease at Baseline
RECIST Non-Measurable Disease at Baseline
PSA BOR: Response
PSA BOR: Non–Response/Non–Progression
PSA BOR: Progression
038
041
042
046
006
043
034
048
027
018
044
045
033
049
001
037
040
Patient Number

BOR, best overall response; PSA, prostate-specific antigen.

- Two patients had at least a 30% reduction in the sum of target lesions as a best percentage change from baseline (033 and 001; **Figure 5**)
- Patient 001 had disappearance of all target lesions (lymph nodes) with only non-target disease remaining
- Patient 046 with RECIST BOR of CR (**Table 4**) had non-measurable disease only at baseline and is thus not represented in this figure

Figure 5. Best Percentage Change from Baseline in Sum of Target Lesions in Patients with Measurable Disease

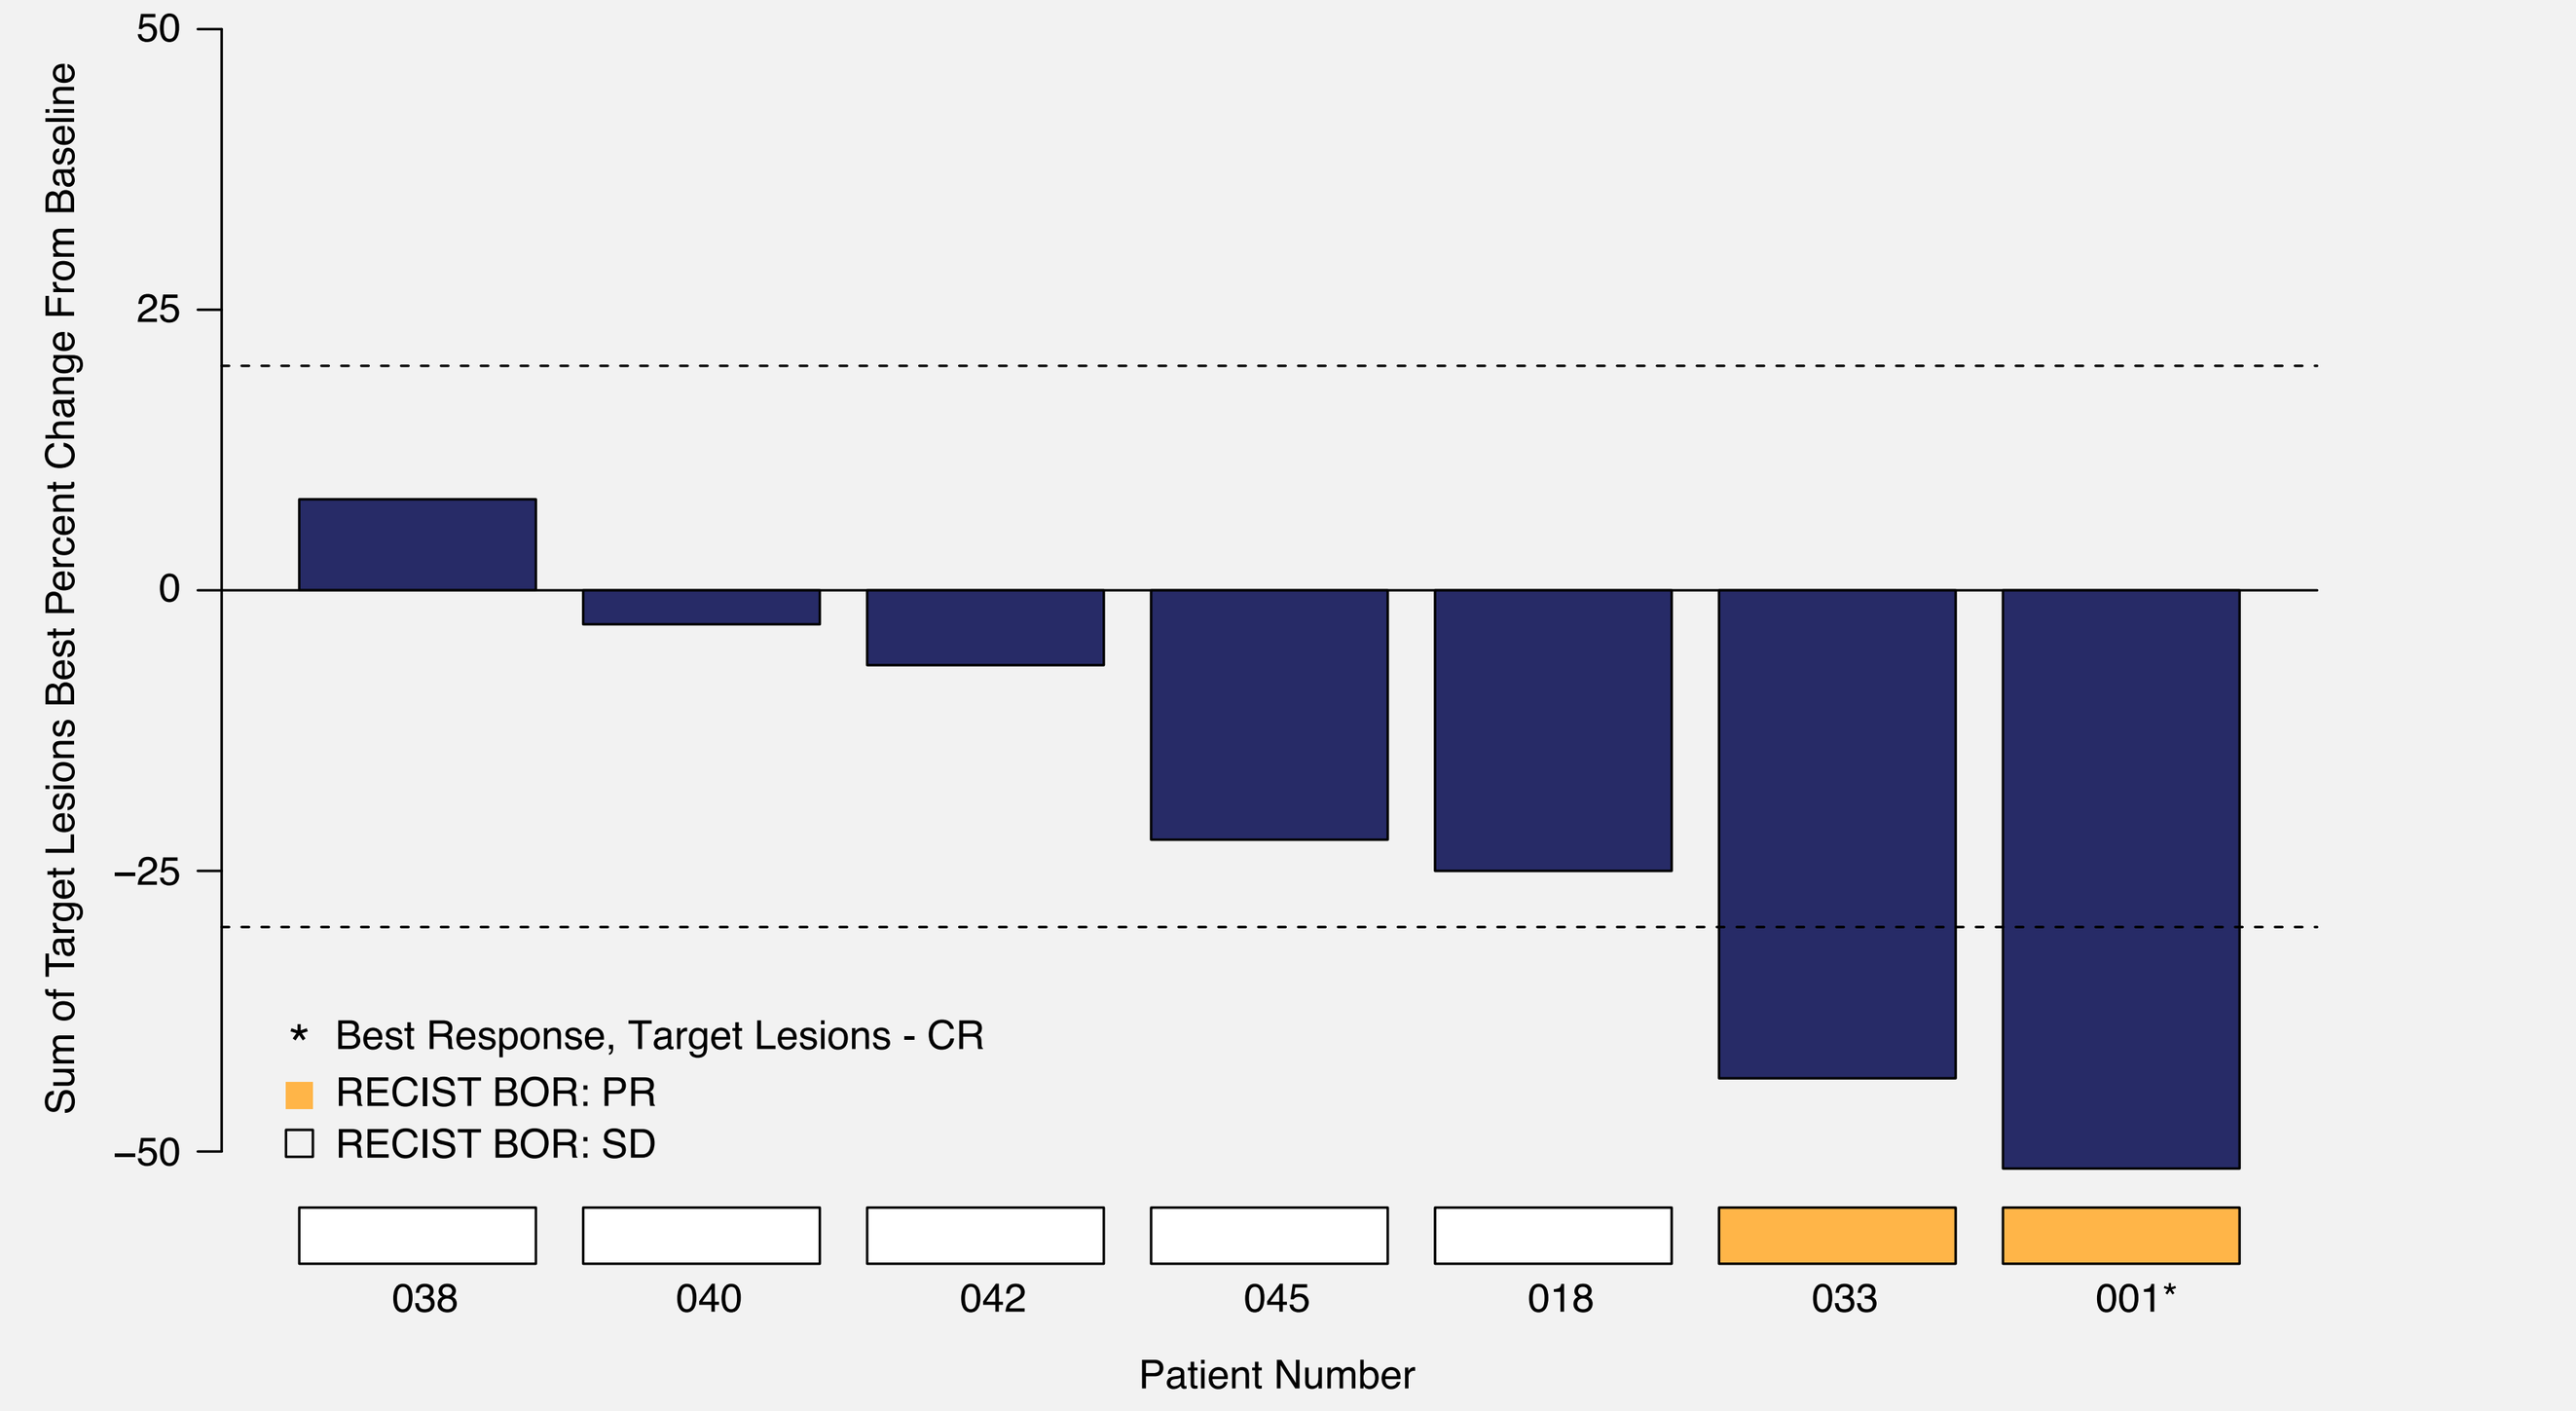
50
25
0
−25
−50
Sum of Target Lesions Best Percent Change From Baseline
* Best Response, Target Lesions - CR
RECIST BOR: PR
RECIST BOR: SD
038
040
042
045
018
033
001*
Patient Number

BOR, best overall response; CR, complete response; PR, partial response; SD, stable disease.

CONCLUSIONS

- EZD shows promising activity in patients previously treated with anti-androgens and/or abiraterone and is associated with a 41% composite ORR in chemotherapy-naive mCRPC
- The safety of the EZD combination is consistent with the known profiles of each individual agent
- These promising Phase 1b data satisfy protocol-defined advancement criteria
- As a result, randomized enrollment EZD vs docetaxel is currently ongoing in the Phase 2 part of the study

ACKNOWLEDGMENTS AND DISCLOSURES

The authors gratefully acknowledge the patients, their families, and their caregivers for their participation in this clinical trial. Additionally, they would like to thank the ARC-6 Principal Investigators and study staff for their efforts in conducting the study. LM Kopp, JR Scott, M Grady, and OS Gardner are employees of Arcus Biosciences, Inc. and may own company stock. SK Subudhi has had a consulting/advisory role for Amgen, Apricity Health, AstraZeneca, Bayer, Bristol Myers Squibb, DAVA Oncology, Janssen, Polaris, and Valeant/Dendreon; has received honoraria from Amgen, Apricity Health, AstraZeneca, Bayer, Bristol Myers Squibb, DAVA Oncology, Dendreon, Exelixis, Janssen, Polaris, the Parker Institute of Cancer Immunotherapy, and the Society for Immunotherapy of Cancer; has ownership interest in Apricity Health; has received travel reimbursements from Compugen; and has received research funding from AstraZeneca, Bristol Myers Squibb, and Janssen. JC Bendell has received travel reimbursements from ARMO BioSciences, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Daiichi Sankyo, FORMA Therapeutics, Gilead Sciences, Ipsen, Lilly, MedImmune, Merck, Novartis, OncoMed, Roche/Genentech, and Taiho Pharmaceutical. M Carducci has had a consulting/advisory role for Exelixis, Foundation Medicine, Pfizer, and Roche/Genentech; and has received research funding from Arcus Biosciences, AstraZeneca, Bristol Myers Squibb, Pfizer, and EMD Serono. DR Wise has had a consulting/advisory role for Alphasights, Foundation Medicine, GLG, Guidepoint Global, Leap Therapeutics, and Pfizer; has received honoraria from OncLive; and has received travel reimbursements from Pfizer. Medical writing assistance was provided by Amanda Martin, PhD, and Kendall Foote, PhD, of Medical Expressions (Chicago, IL) and was funded by Arcus Biosciences.

REFERENCES

1. Martins I, et al. *Cell Cycle*. 2009;8(22):3723-3728; 2. Vijayan D, et al. *Nat Rev Cancer.* 2017;17(12):709-724; 3. Cekic C and Linden J. *Nat Rev Immunol.* 2016;16(3):177-192;
4. Gao ZG and Jacobson KA. *Int J Mol Sci.* 2019;20(20):5139; 5. Powderly J, et al. *ESMO 2019*. Abstract 4854; 6. Sachdev J, et al. *SABCS 2021*. Poster PS-12-12;
7. Cecchini M, et al. *AACR 2021*. Poster CT129; 8. Available at: www.arcusbio/pipeline/chart; 9. Kong HY and Byun J. *Biomol Ther*. 2013;21(1):10-20;
10. Abida W, et al. *Proc Natl Acad Sci.* 2019;116(23):11428-11436; 11. Bendell J, et al. *AACR 2019*. Abstract CT026; 12. Hansen AR, et al. *Ann Oncol.* 2018;29(8):1807-1813.

Corresponding Author: Olivia S. Gardner, ogardner@arcusbio.com

ASCO Virtual Annual Meeting 2021