AACR
Annual Meeting, April 2022
Abstract 1367

HPK1 Inhibition Enhances T Cell Activation and Relieves the Immunosuppressive Phenotype of Inhibitory Signals Found in the Tumor Microenvironment

Rajesh K. Singh, Rameshwari Rayaji, Bindi Patel, Kristen Zhang, Sachie Marubayashi, Sean Cho, Stefan Garrido-Shaqfeh, Joseph Kulusich, Cesar Meleza, Nidhi Tibrewal, Joice Thomas, Pradeep Nareddy, Ehesan Sharif, Sharon Zhao, David Green, Manmohan R Leleti, Jay P Powers, Matthew J Walters, Daniel DiRenzo
Arcus Biosciences, Inc.; 3928 Point Eden Way, Hayward, CA 94545 (USA)

Introduction

- Hematopoietic Progenitor Kinase 1 (HPK1/MAP4K1) is a member of the MAP4K family whose activity has been demonstrated to restrain T cell activation through phosphorylation of SLP-76 at Serine 376 leading to TCR disassembly.
- Mediators generated in the tumor microenvironment (TME) such as adenosine, PGE_2 and TGF-β dampen T cell activity and present a significant barrier to cancer immunotherapy. HPK1 inhibition may allow for enhanced T cell activation under such suppressive conditions.
- Herein, we describe studies to assess the effects of HPK1 inhibition on T cell activation and in alleviating the effects of suppressive signals. We also test the ability of HPK1 inhibition in combination with adenosine receptor antagonism to further amplify T cell activation.

Methods

- **T cell experiments:** $CD8^+$ T cells were isolated from healthy human blood and activated using anti-CD3/CD28 stimulation. Supernatants were assayed for IL-2 and IFN-γ using cytokine bead array. Activation markers and SLP-76 phosphorylation (S376) were assayed by flow cytometry
- **CRISPR knockout:** $MAP4K1^{KO}$, $MAP4K3^{KO}$ and $MAP4K4^{KO}$ $CD8^+$ T cells and $MAP4K1^{KO}$ Jurkat cells were generated using Amaxa nucleofection of cells with Cas9 and gene-specific sgRNA sequences. Cells were recovered for at least 7 days prior to use.
- **Dendritic cell experiments:** Dendritic cells (DC) were isolated from healthy human blood by negative selection and matured with IFN-γ or IFN-γ + LPS for 40 hours. Cells were then harvested for flow cytometry and supernatants assessed for cytokine production.
- **Gene expression analysis:** Gene expression (log2CPM) data was processed from gene count data from the Genotype-Tissue Expression (GTEx, v7) project and FASTQ files from sorted immune populations of healthy human donors (Monaco et al. *Cell Rep*. 2019, PRJNA418779).
- **Compounds:** HPK1 inhibitor tool compound from You et al. *J Immunother Cancer*. 2021. Adenosine receptor antagonist etrumadenant (etruma) was used to block the effects of the synthetic adenosine receptor agonist NECA.

Results

HPK1 is Broadly Expressed in Immune Cells and Correlates with Immune Infiltration in Normal Human Tissue

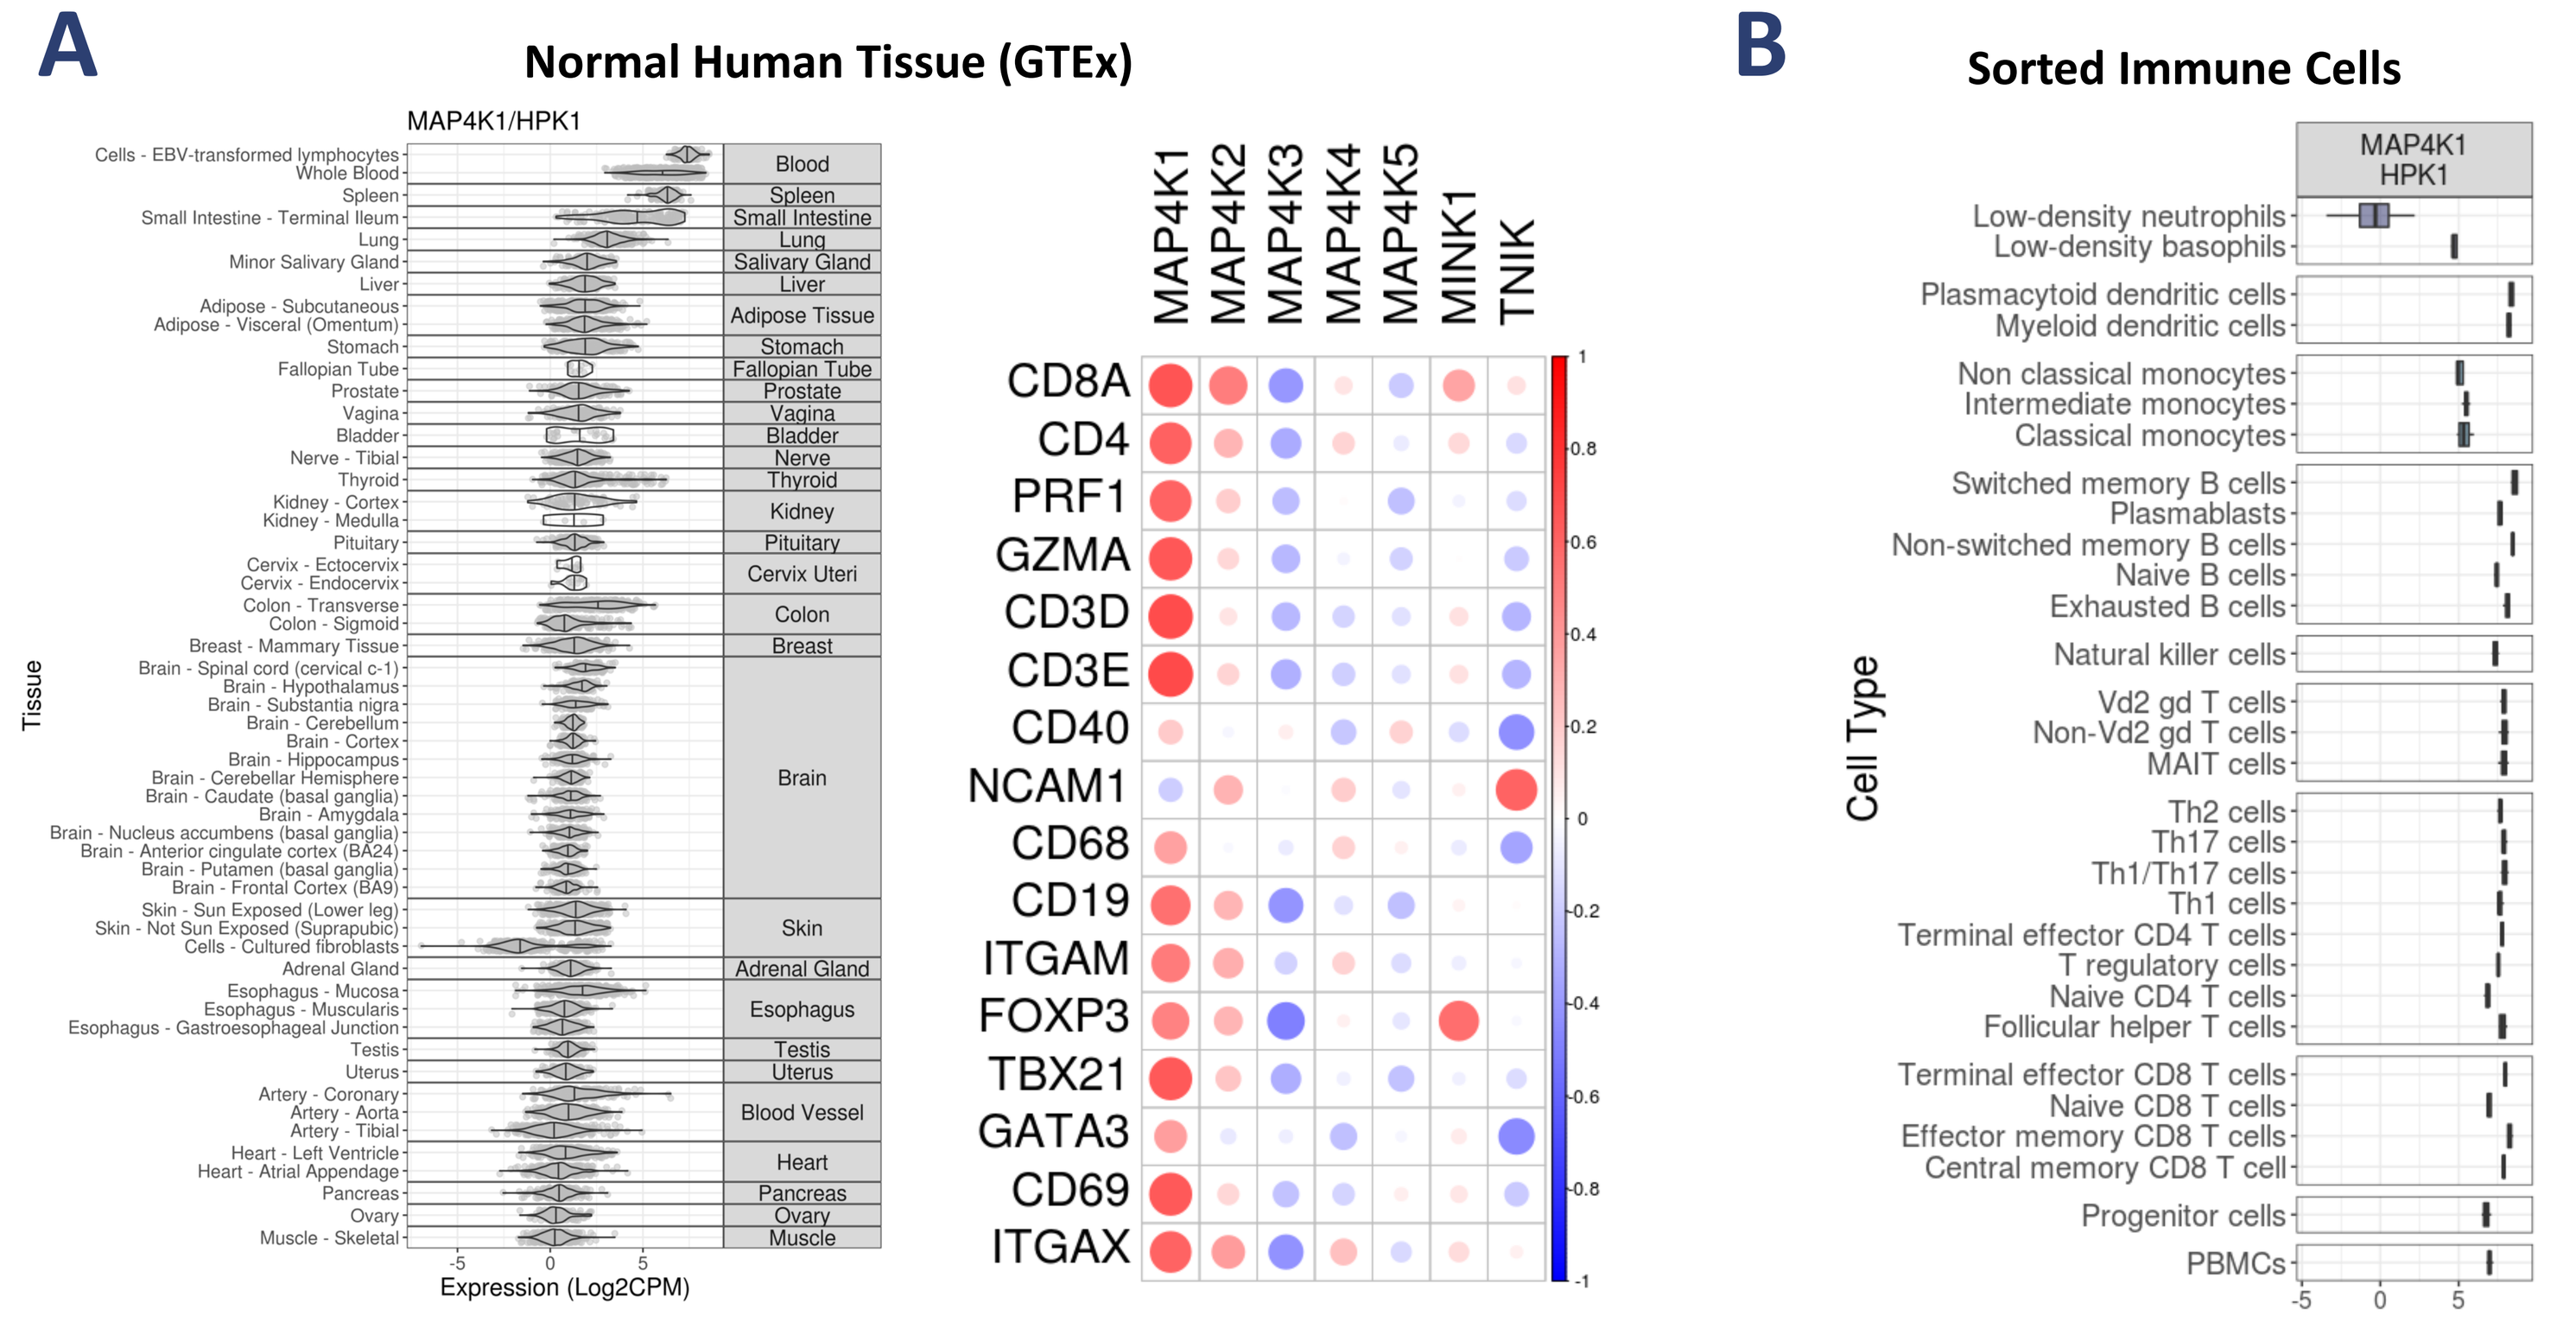
A
Normal Human Tissue (GTEx)
MAP4K1/HPK1
Tissue
Expression (Log2CPM)
MAP4K1 MAP4K2 MAP4K3 MAP4K4 MAP4K5 MINK1 TNIK
CD8A CD4 PRF1 GZMA CD3D CD3E CD40 NCAM1 CD68 CD19 ITGAM FOXP3 TBX21 GATA3 CD69 ITGAX
B
Sorted Immune Cells
MAP4K1 HPK1
Cell Type

Figure 1. **(A)** HPK1 (MAP4K1) gene expression is found at high levels in tissues with high immune cell content (left panel) and strongly correlates with immune related genes in normal tissues compared to other MAP4K family members (right panel). **(B)** Gene expression data from sorted immune cell populations shows that HPK1 expression is found at high levels in nearly all immune cells with moderate expression observed in circulating monocytes.

Genetic Deletion of HPK1 but not MAP4K3/4 Enhances Human T Cell Activity

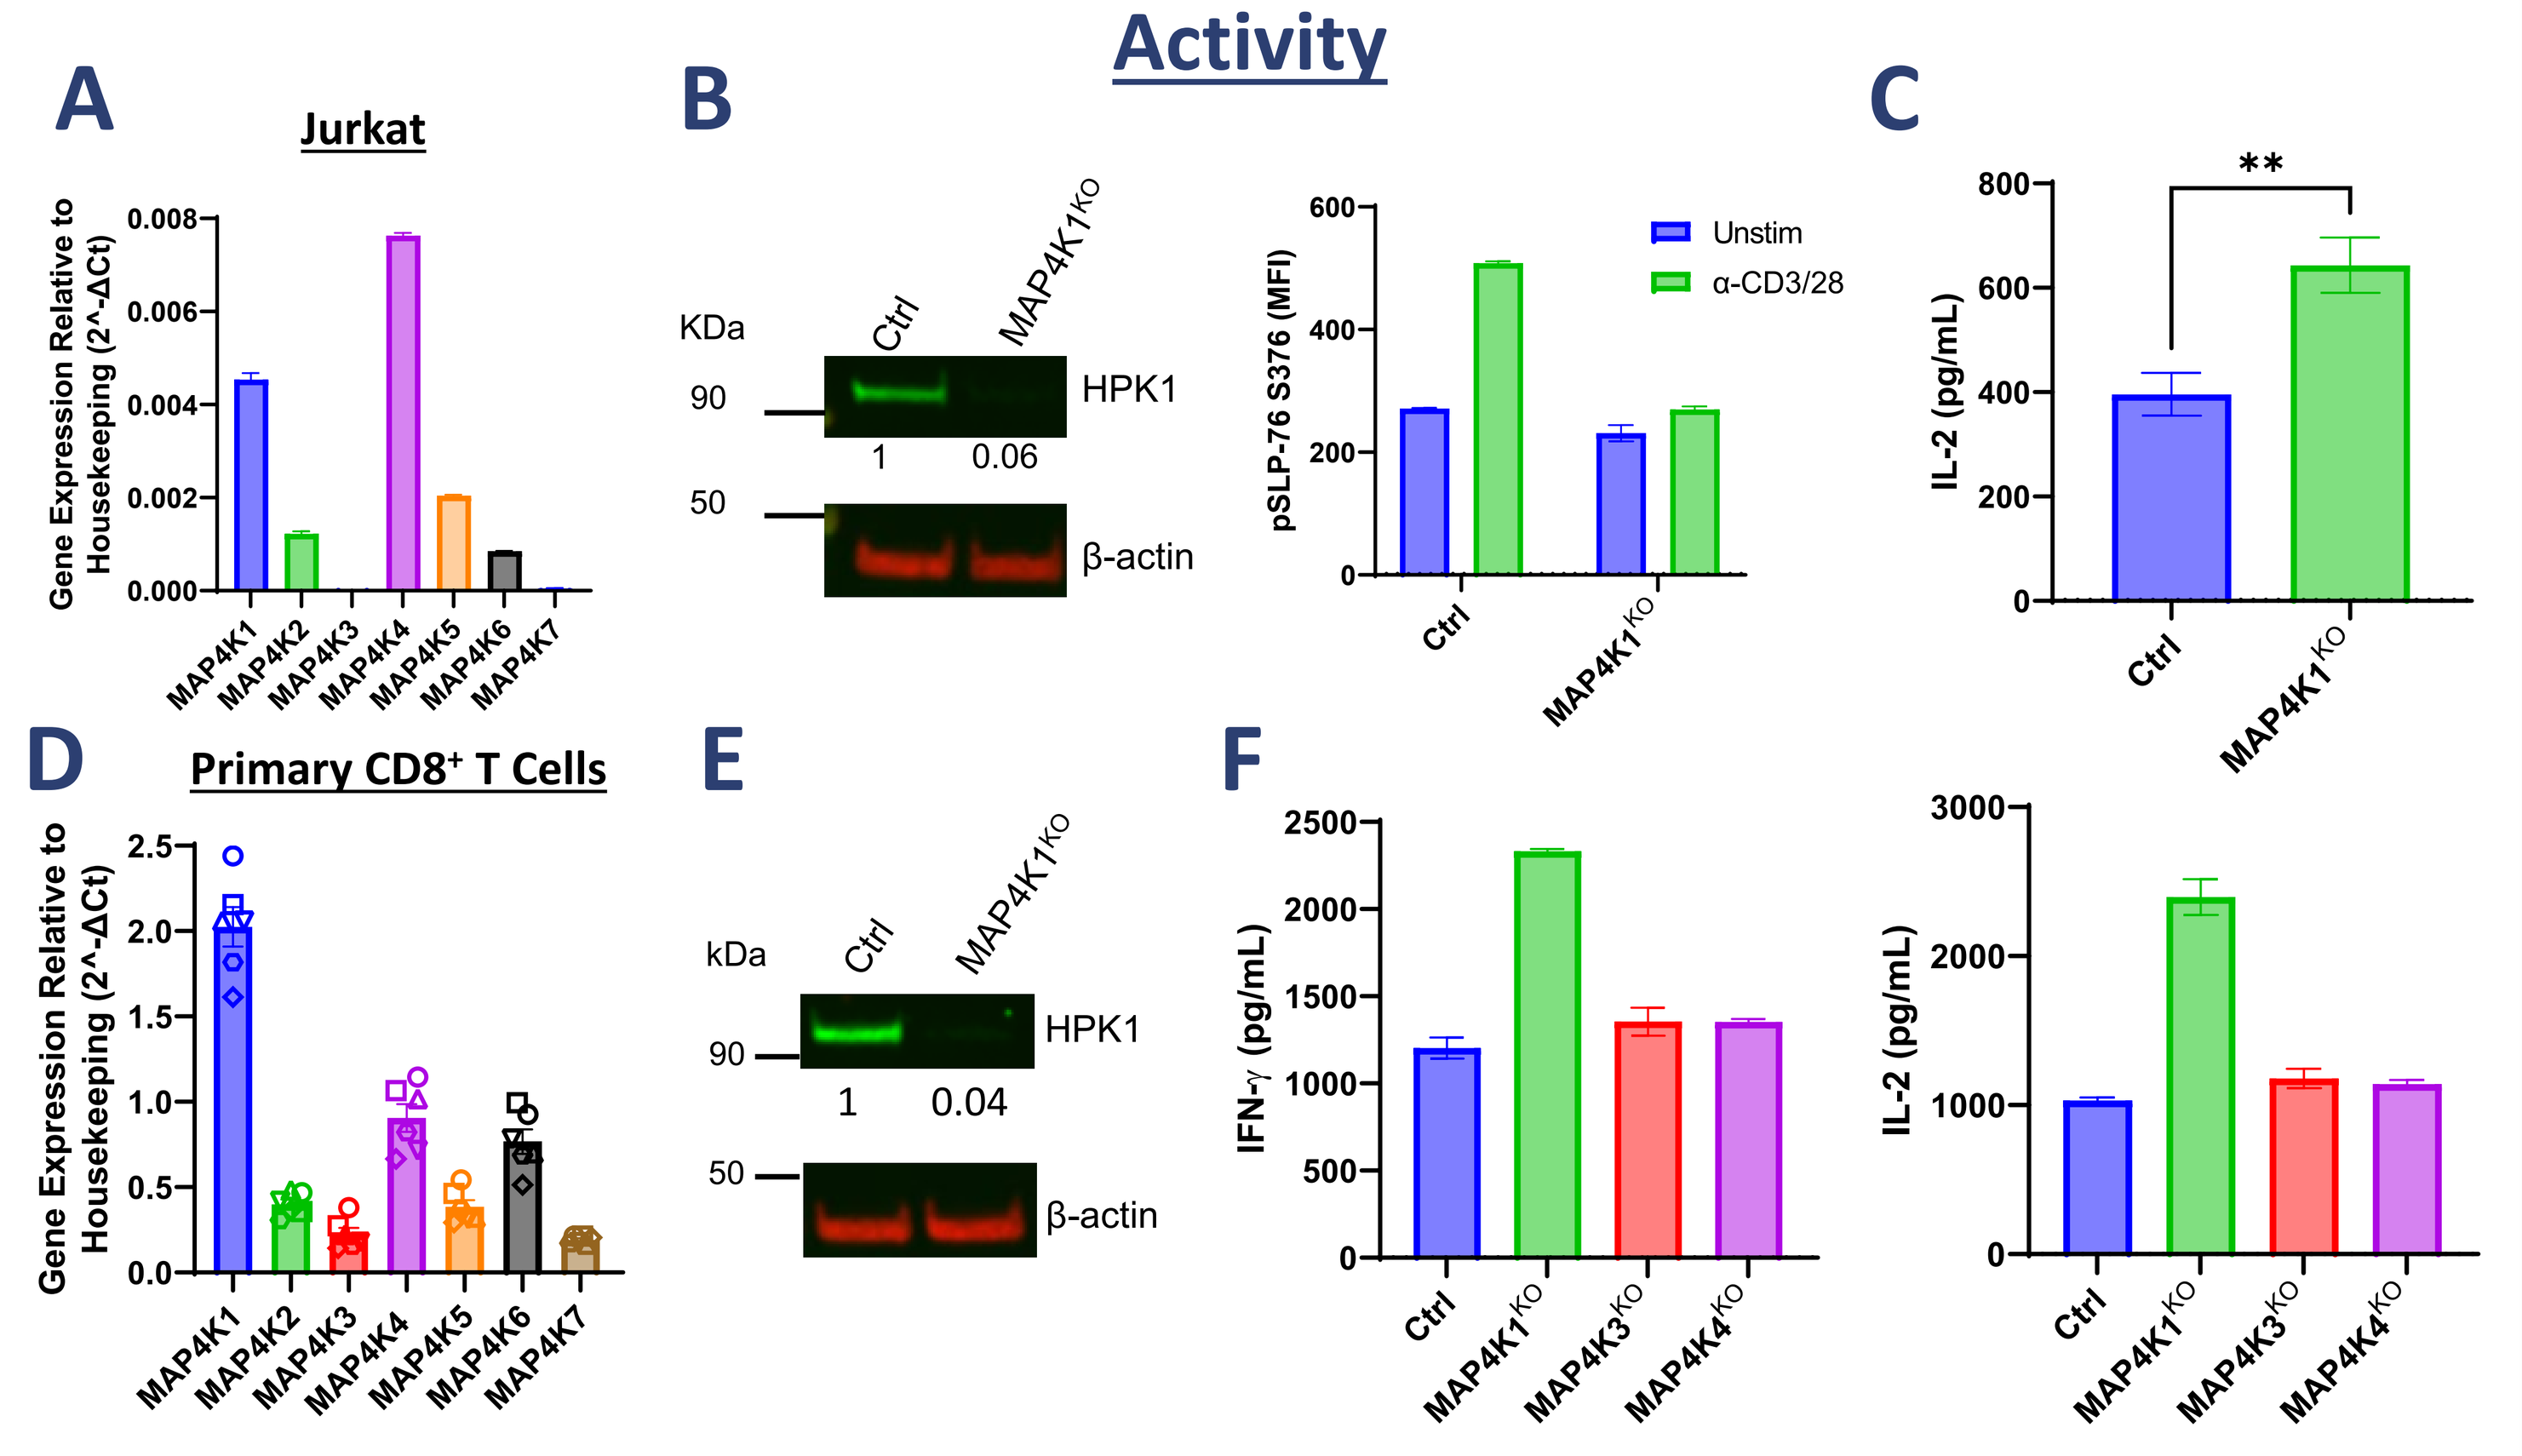
A
Jurkat
Gene Expression Relative to Housekeeping (2^-ΔCt)
MAP4K1 MAP4K2 MAP4K3 MAP4K4 MAP4K5 MAP4K6 MAP4K7
B
KDa
Ctrl
MAP4K1KO
HPK1
β-actin
1 0.06
pSLP-76 S376 (MFI)
Unstim
α-CD3/28
C
IL-2 (pg/mL)
**
D
Primary CD8+ T Cells
E
1 0.04
F
IFN-γ (pg/mL)
IL-2 (pg/mL)
Ctrl MAP4K1KO MAP4K3KO MAP4K4KO

Figure 2. **(A)** Gene expression of MAP4K1-7 in Jurkat cells. **(B)** CRISPR knockout of MAP4K1 was confirmed by Western blot of HPK1 protein in Jurkat cells (left panel) and HPK1 stimulates CD3/28 activation-induced phosphorylation of SLP-76 (S376) quantified by flow cytometry (right panel). **(C)** $MAP4K1^{KO}$ Jurkat cells display increased IL-2 secretion measured using cytokine bead array. **(D)** Gene expression of MAP4K1-7 in primary human $CD8^+$ T cells from 6 donors. **(E)** CRISPR knockout of MAP4K1 was confirmed by Western blot of HPK1 protein in human $CD8^+$ T cells. **(F)** $MAP4K1^{KO}$ but not $MAP4K3^{KO}$ or $MAP4K4^{KO}$ $CD8^+$ T cells display increased IFN-γ (left panel) and IL-2 (right panel) secretion upon anti-CD3/CD28 stimulation measured using cytokine bead array. **=p<0.01.

Inhibition of HPK1 Kinase Activity Enhances T Cell Signaling and Cytokine Production Irrespective of Stimulation Strength

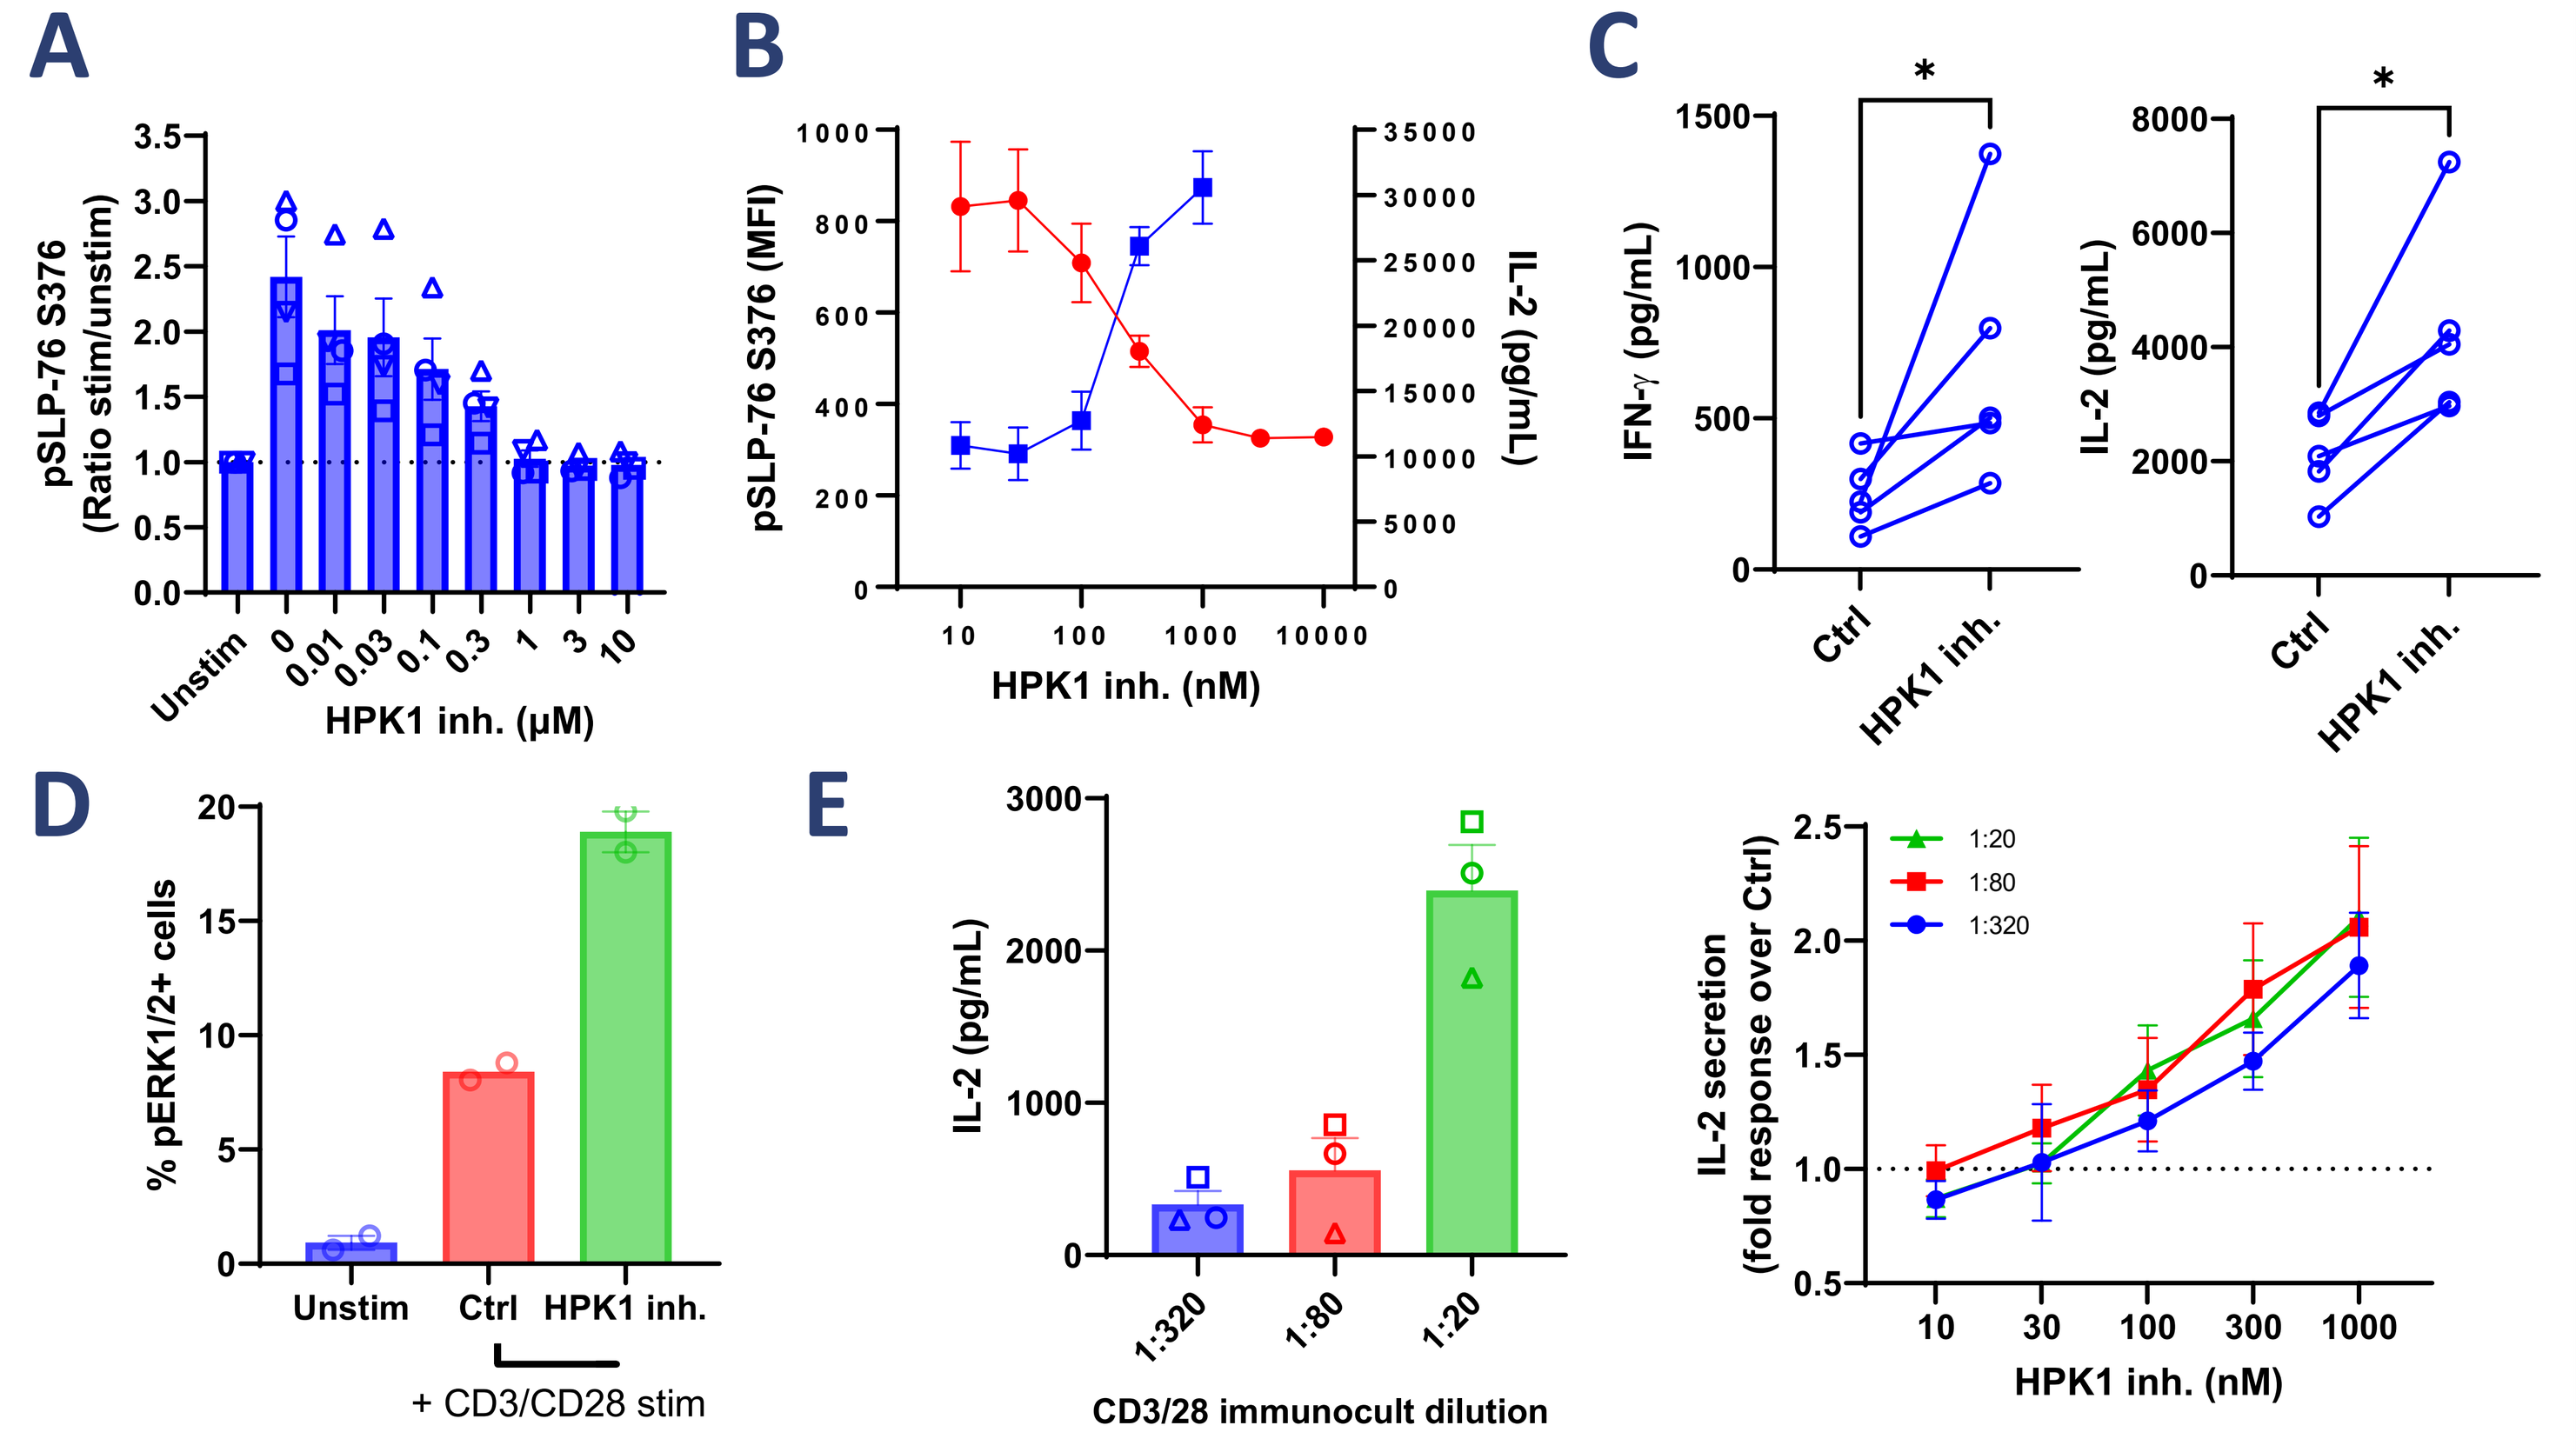
A
pSLP-76 S376 (Ratio stim/unstim)
HPK1 inh. (µM)
B
pSLP-76 S376 (MFI)
IL-2 (pg/mL)
HPK1 inh. (nM)
C
IFN-γ (pg/mL)
IL-2 (pg/mL)
Ctrl HPK1 inh.
D
% pERK1/2+ cells
Unstim Ctrl HPK1 inh.
+ CD3/CD28 stim
E
IL-2 (pg/mL)
1:320 1:80 1:20
CD3/28 immunocult dilution
IL-2 secretion (fold response over Ctrl)
HPK1 inh. (nM)

Figure 3. **(A)** HPK1 inhibitor decreases CD3/28 activation-induced phosphorylation of SLP-76 (S376) in primary human $CD8^+$ T cells from 4 donors in a concentration-dependent manner . **(B)** Inhibition of pSLP-76 S376 (red line) correlates with an increase in IL-2 secretion (blue line) in a matched human $CD8^+$ T cell donor. **(C)** HPK1 inhibition enhances IFN-γ (left panel) and IL-2 (right panel) secretion from stimulated human $CD8^+$ T cells from 5 donors treated with 1 µM HPK1 inh. **(D)** HPK1 inhibition using 1 µM HPK1 inh. increases CD3/28 activation-induced T cell downstream signaling, as measured by pERK1/2+ cells by flow cytometry. **(E)** Human $CD8^+$ T cells from 3 donors were stimulated with different dilutions of anti-CD3/CD28 stimulant to produce low (1:320), medium (1:80), and high (1:20) strength activation prior to measurement of IL-2 secretion by cytokine bead array (left panel). HPK1 inhibition in the same three donors shows similar enhancement of T cell activity across the different activation strengths (right panel). . *=p<0.05, **=p<0.01.

HPK1 Inhibition Restores Activity to T Cells Suppressed by Individual Inhibitory Signals

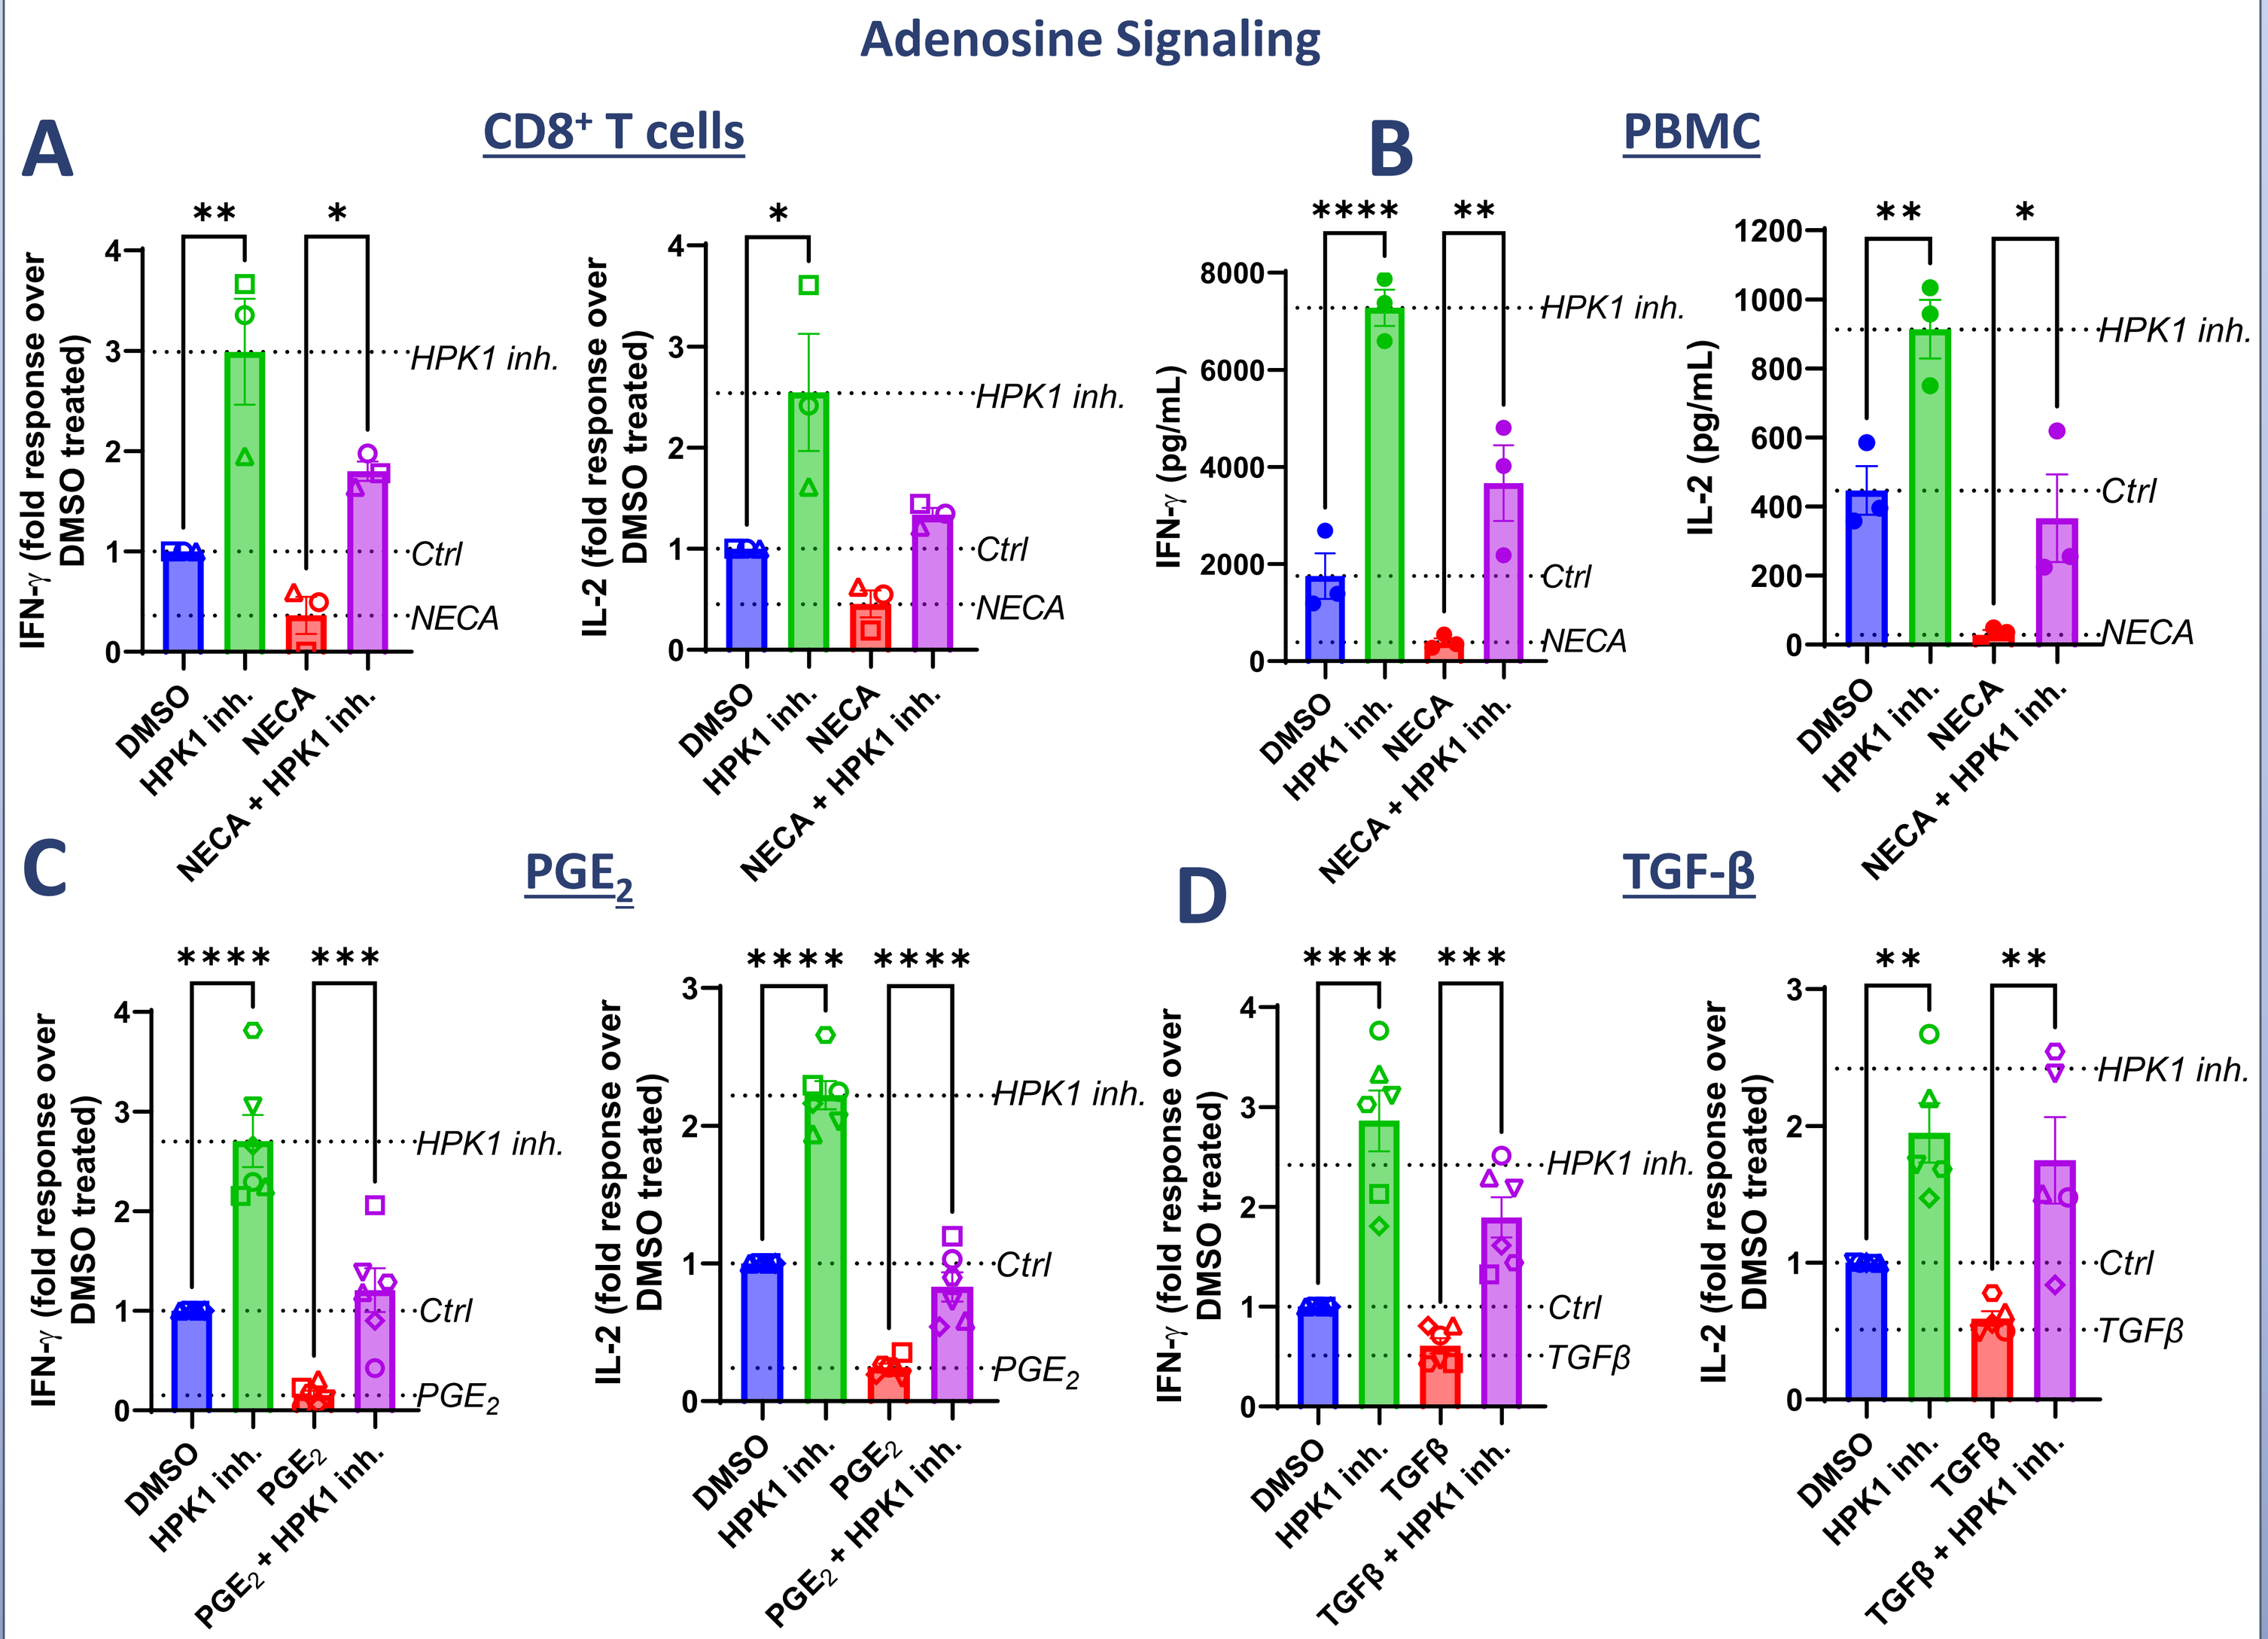
Adenosine Signaling
A
CD8+ T cells
B
PBMC
C
PGE2
D
TGF-β
IFN-γ (fold response over DMSO treated)
IL-2 (fold response over DMSO treated)
HPK1 inh.
Ctrl
NECA
TGFβ
DMSO HPK1 inh. NECA NECA + HPK1 inh.

Figure 4. **(A)** HPK1 inhibition increases IFNγ (left panel) and IL-2 (right panel) secretion alone or in the presence of suppressive adenosine signaling (NECA, 5 µM) in human $CD8^+$ T cells from 3 donors. **(B)** HPK1 inhibition increases IFNγ (left panel) and IL-2 (right panel) secretion alone or in the presence of NECA in human PBMC. **(C)** HPK1 inhibition increases IFNγ (left panel) and IL-2 (right panel) secretion alone or in the presence of suppressive prostaglandin signaling (PGE_2, 10 nM) in human $CD8^+$ T cells from 6 donors. **(D)** HPK1 inhibition increases IFNγ (left panel) and IL-2 (right panel) secretion alone or in the presence of suppressive TGFβ signaling (TGFβ , 3 ng/mL) in human $CD8^+$ T cells from 6 donors *=p<0.05, **=p<0.01, ***=p<0.001, ****=p<0.0001.

HPK1 Inhibition Restores T cell Activity in the Presence of Multiple Inhibitory Factors Found in the TME

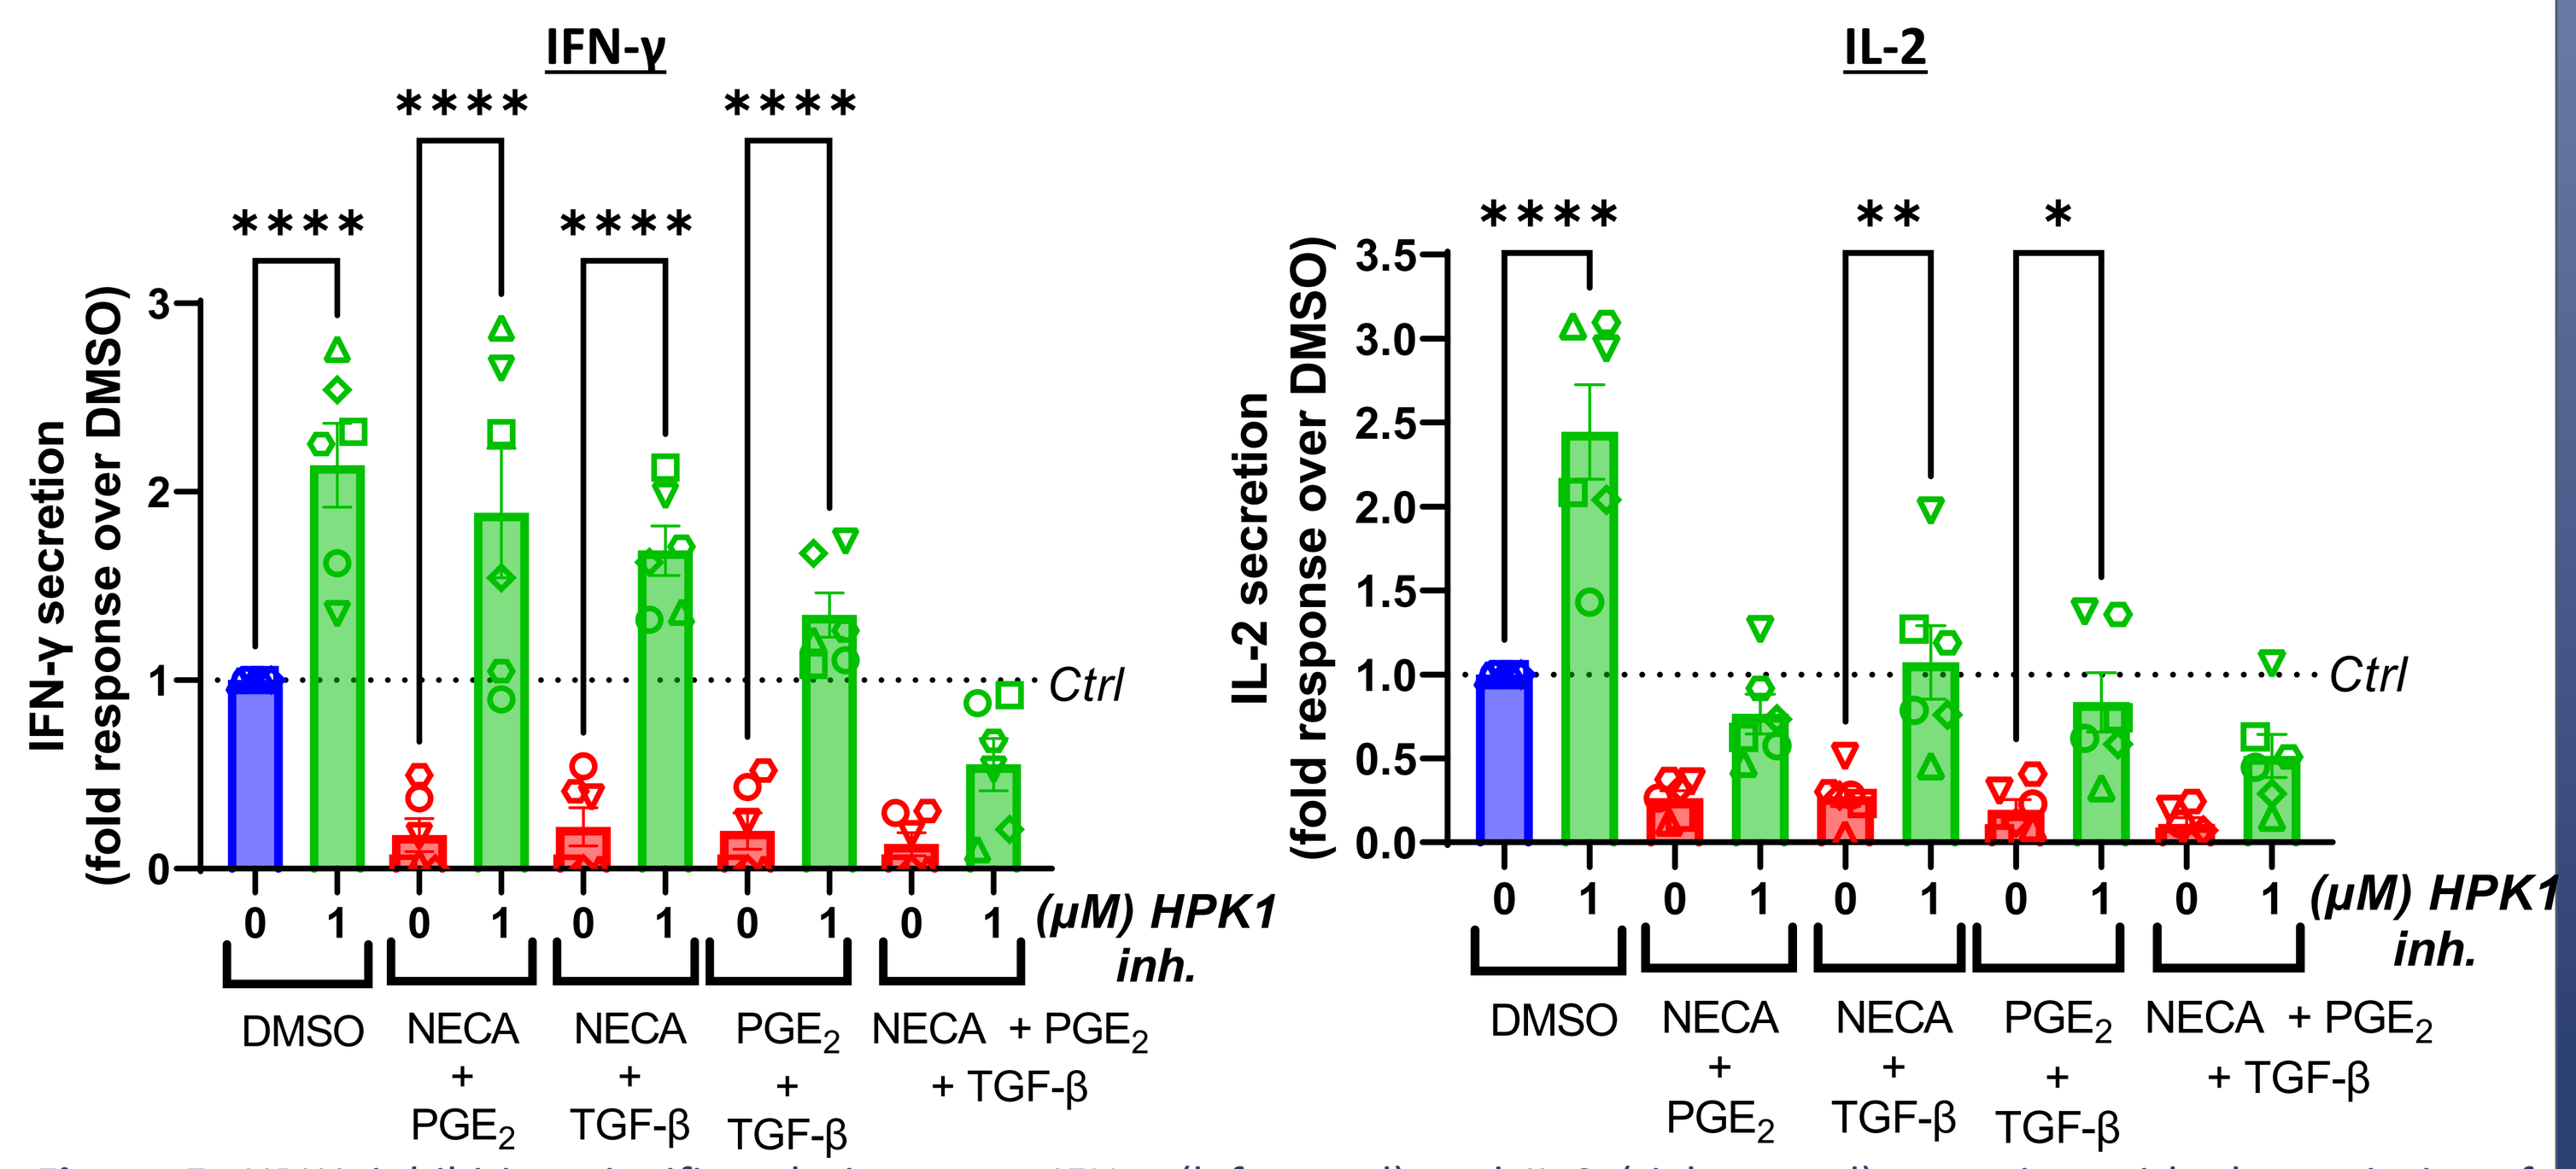
IFN-γ
IL-2
IFN-γ secretion (fold response over DMSO)
IL-2 secretion (fold response over DMSO)
Ctrl
(µM) HPK1 inh.
DMSO
NECA + PGE2
NECA + TGF-β
PGE2 + TGF-β
NECA + PGE2 + TGF-β

Figure 5. HPK1 inhibition significantly increases IFN-γ (left panel) and IL-2 (right panel) secretion with the majority of combined suppressive signals whereas HPK1 inhibition with all three suppressive signals shows a qualitative but not statistically significant increase in cytokine production. **These results suggest that while HPK1 inhibition alone can restore T cell activity in the presence of two suppressive signals, combining HPK1 inhibition with blockade of a suppressive signal, such as adenosine signaling, should enable HPK1 inhibition to significantly enhance T cell activity under highly immunosuppressive conditions**. *=p<0.05, **=p<0.01, ****=p<0.0001.

HPK1 Inhibition Combines with Etrumadenant to Fully Restore T Cell Activity and Prevent Adenosine Receptor-Mediated Suppression

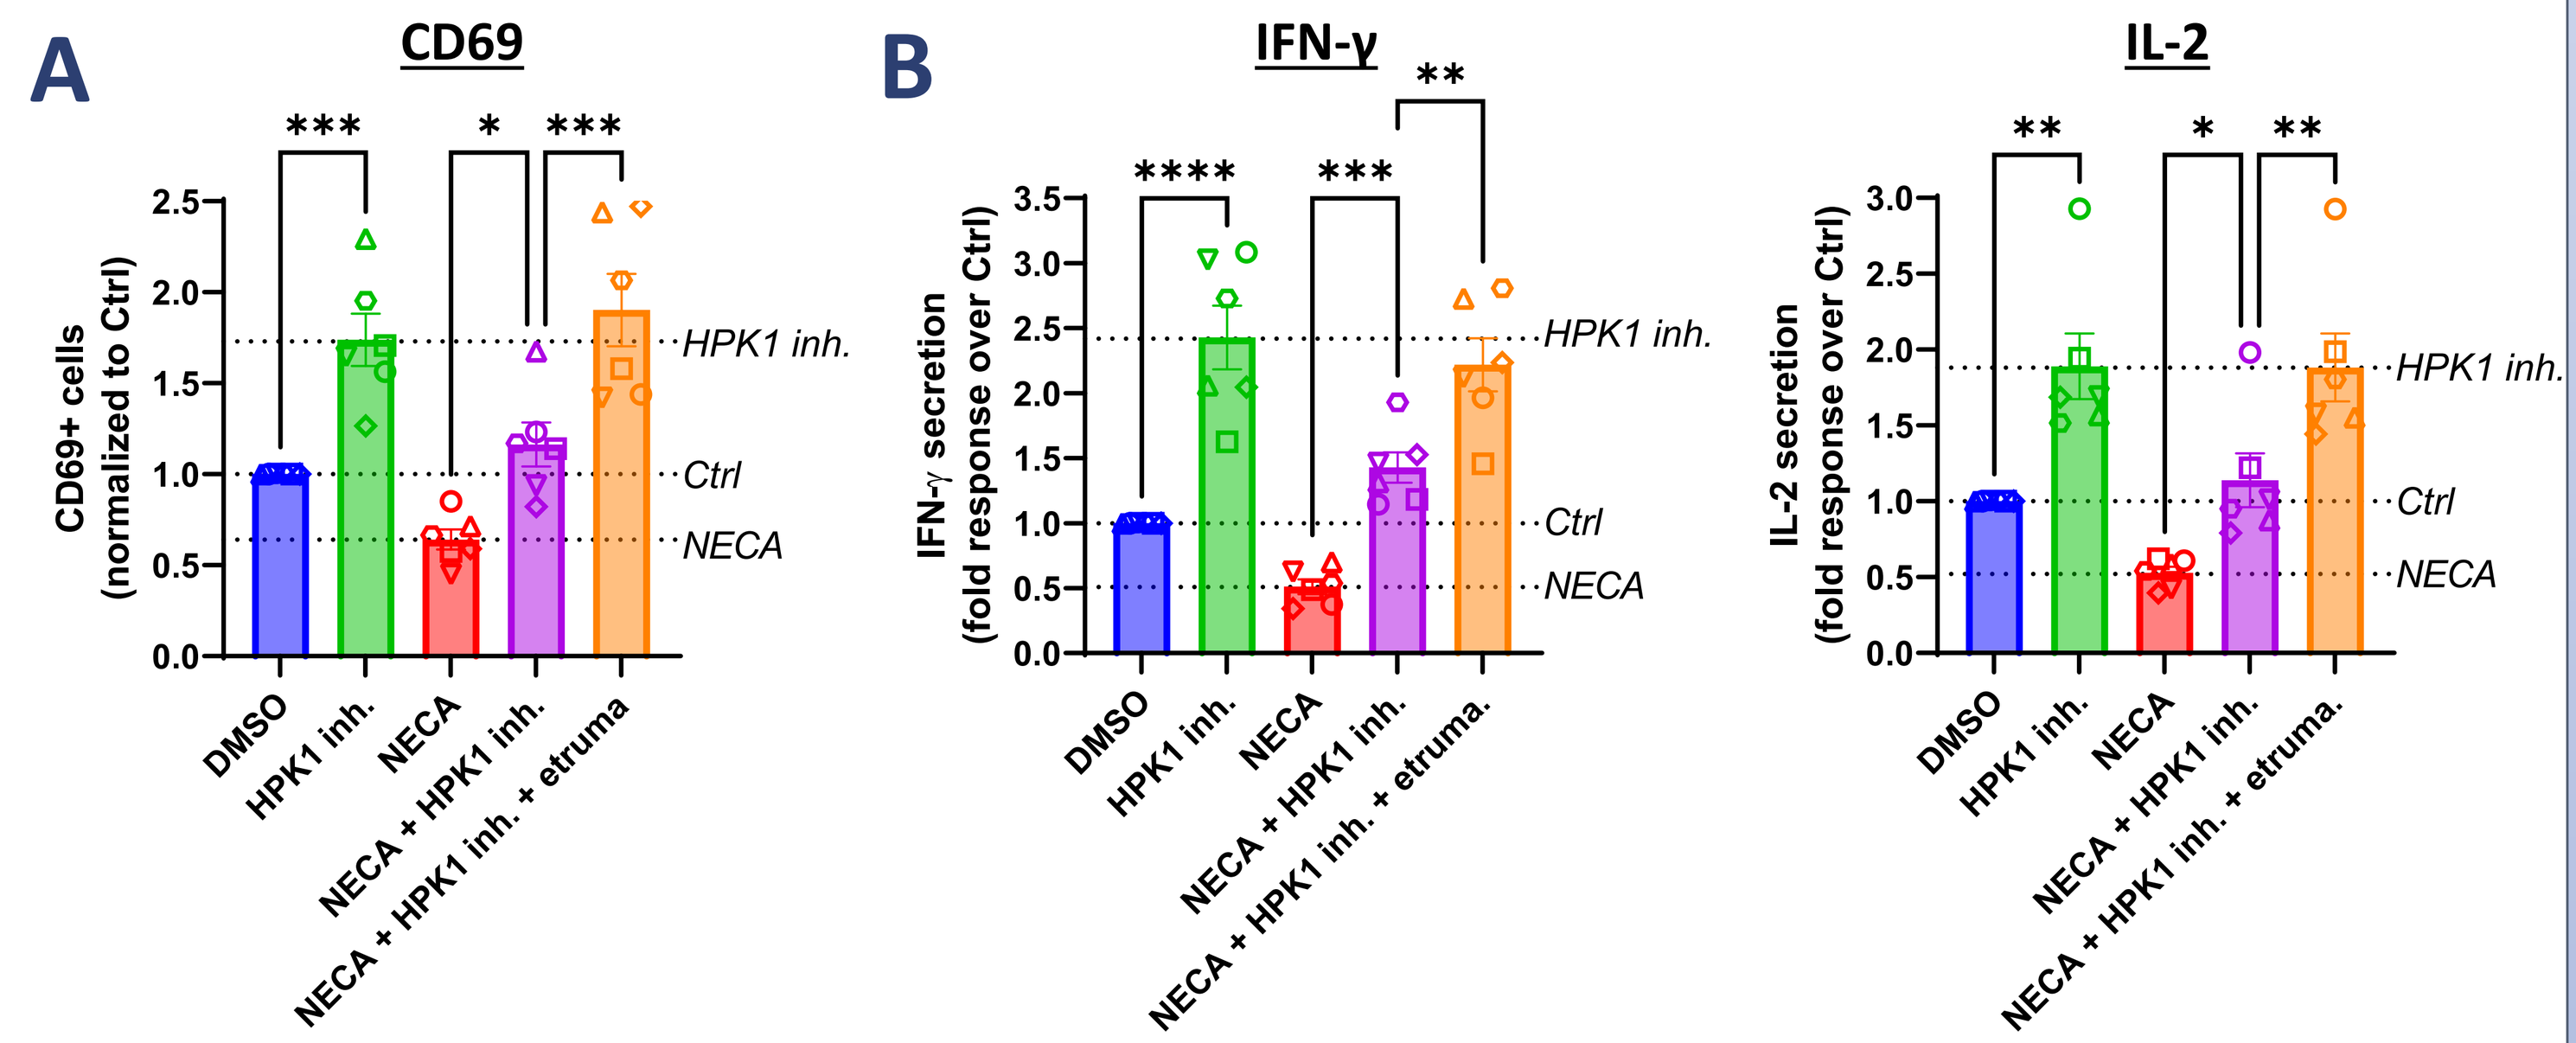
A
CD69
CD69+ cells (normalized to Ctrl)
B
IFN-γ
IFN-γ secretion (fold response over Ctrl)
IL-2
IL-2 secretion (fold response over Ctrl)
HPK1 inh.
Ctrl
NECA
DMSO HPK1 inh. NECA NECA + HPK1 inh. NECA + HPK1 inh. + etruma

Figure 6. **(A)** HPK1 inhibition increases $CD69^+$ cells alone or in the presence of suppressive adenosine signaling (NECA, 5 µM) but full restoration of T cell activity is observed with combined HPK1 inhibition and adenosine receptor antagonism (etrumadenant, 1 µM). **(B)** A similar phenotype was observed with production of IFN-γ and IL-2 showing combined HPK1 inhibition and adenosine receptor antagonism is required for full T cell activity. *=p<0.05, **=p<0.01, ***=p<0.001, ****=p<0.0001.

HPK1 Inhibition Enhances Costimulatory Molecule Expression and Cytokine Production in Freshly Isolated Human Dendritic Cells

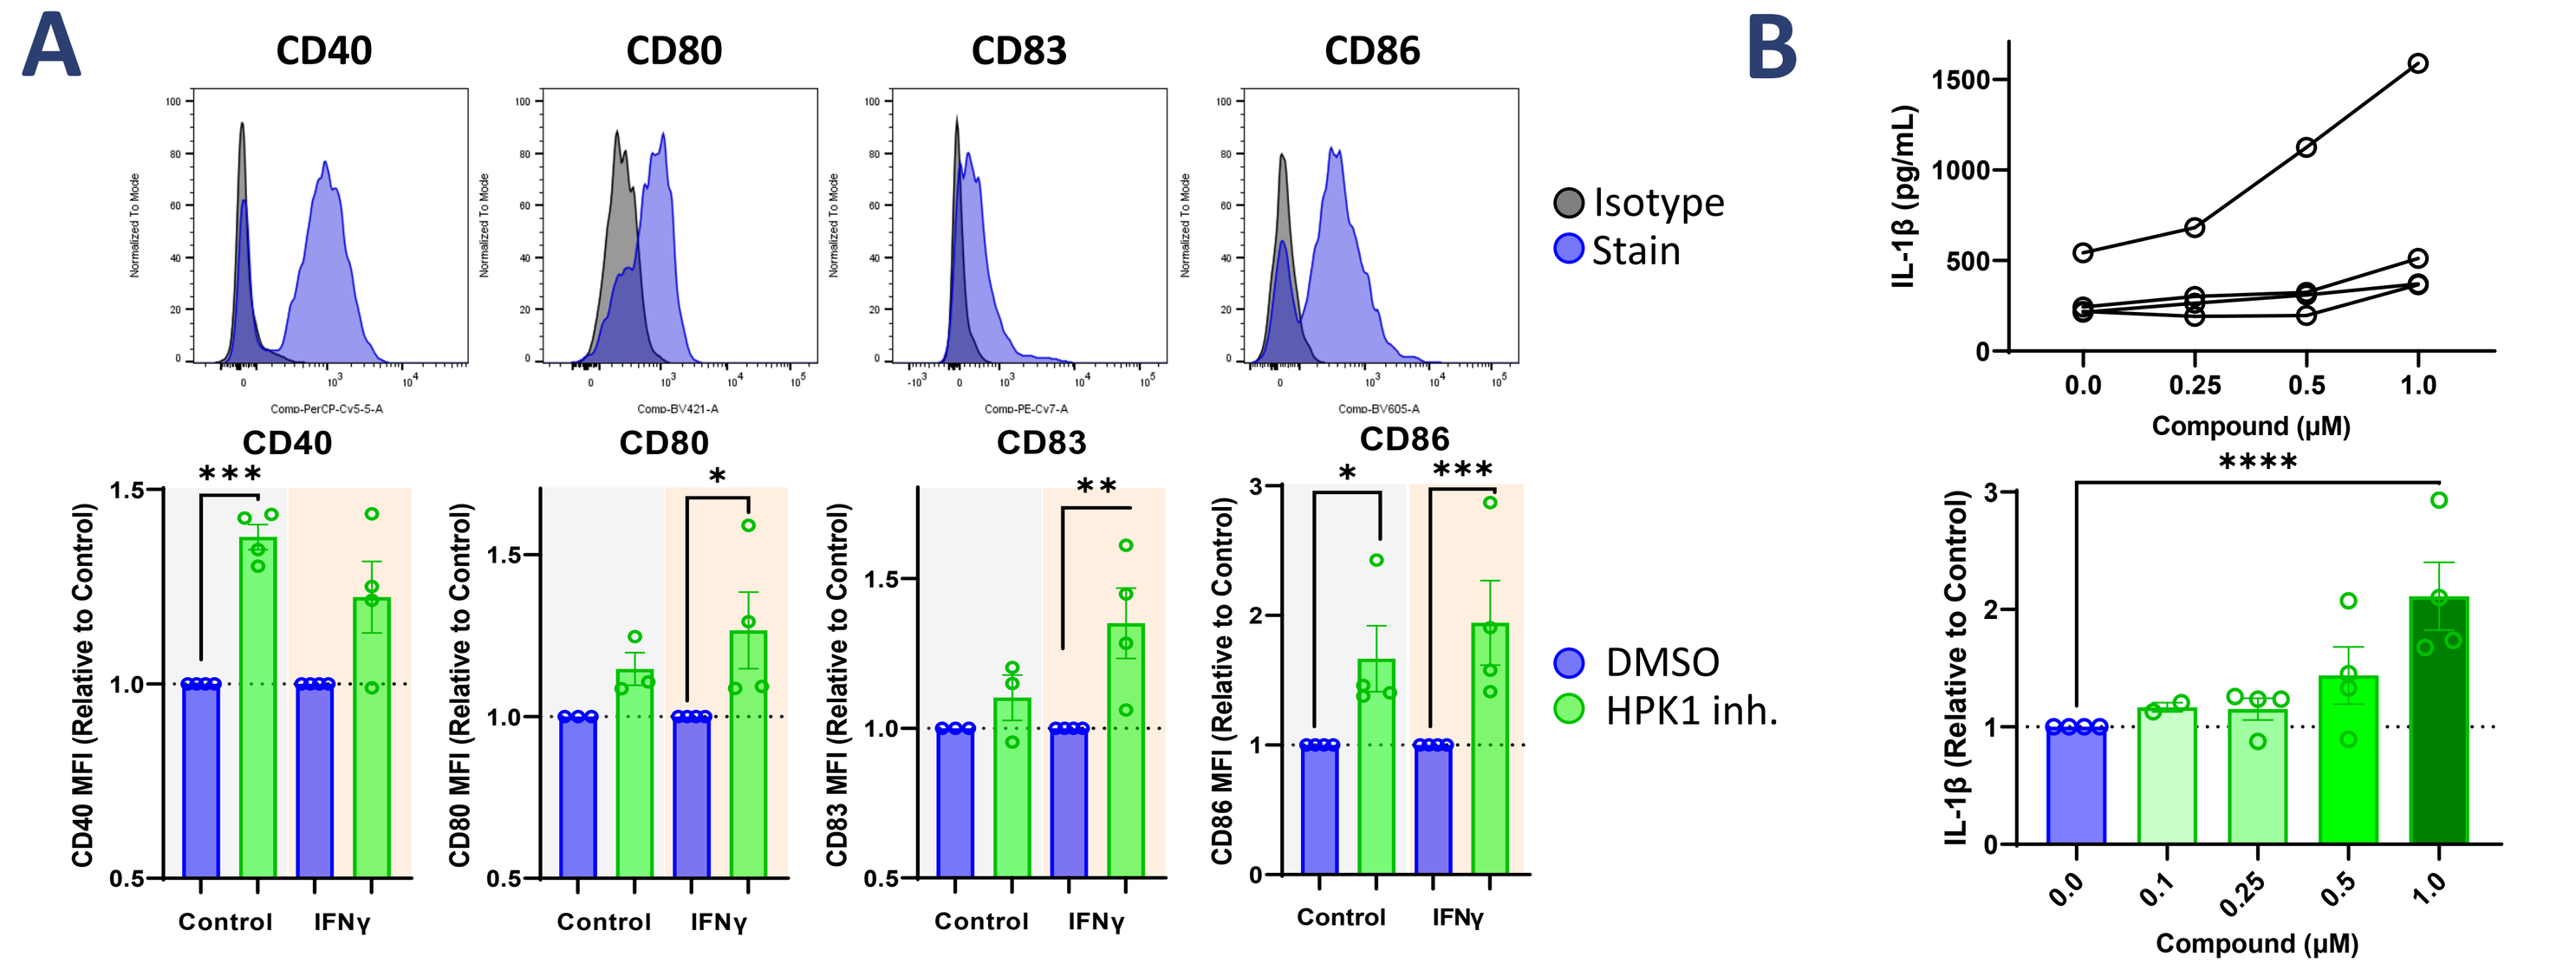
A
CD40 CD80 CD83 CD86
Isotype
Stain
DMSO
HPK1 inh.
Control IFNγ
B
IL-1β (pg/mL)
Compound (µM)
IL-1β (Relative to Control)

Figure 7. **(A)** Histograms showing the expression of co-stimulatory molecules on freshly isolated dendritic cells ($CD11c^+$ $HLA\text{-}DR^+$) (top panels). HPK1 inhibition (1 µM) enhances costimulatory molecule expression +/- maturation with IFN-γ (bottom panels). **(B)** Dendritic cells matured with LPS and IFN-γ show increased IL-1β secretion upon HPK1 inhibition (top graph) and a statistically significant increase when adjusted for donor variability (bottom graph). Dendritic cells were isolated from 4 healthy human donors. *=p<0.05, **=p<0.01, ***=p<0.001, ****=p<0.0001.

Conclusions

- HPK1 (MAP4K1) is primarily expressed in immune cells and strongly correlates with the immune composition of normal human tissue.
- HPK1 is the highest expressed MAP4K family member in $CD8^+$ T cells and genetic deletion of HPK1 but not MAP4K3 or MAP4K4 enhances T cells activity upon stimulation.
- As expected, inhibiting HPK1 kinase activity reproduces the phenotypes of genetic deletion in T cells and enhances cytokine production and costimulatory molecule expression in freshly isolated dendritic cells.
- HPK1 inhibition enhances T cell activation in the presence of suppressive signals alone or in combination. However, overall cellular activity is still suppressed compared to HPK1 inhibition without suppression.
- HPK1 inhibition combines with etrumadenant (dual $A_{2a}R/A_{2b}R$ antagonist) to further enhance T cell activity in the presence of suppressive adenosine signaling.