A Mechanistic Pharmacokinetic-Pharmacodynamic (PK-PD) Model of Quemliclustat (AB680), a Small-Molecule Inhibitor of CD73, in Healthy Volunteers and Patients with Gastrointestinal Malignancies

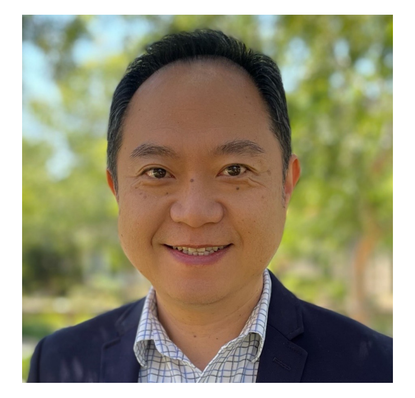

PRESENTER:

Kai Hsin (Ken) Liao

INTRODUCTION

- The tumor microenvironment contains high levels of immunosuppressive adenosine, which binds to and activates the A2a and A2b receptors on immune cells, leading to an ineffective anti-tumor response.
- CD73 enzyme and tissue non-specific alkaline phosphatase (TNAP) are primarily responsible for the conversion of extracellular adenosine mono-phosphate (AMP) to adenosine.
- Quemliclustat is a potent, selective, small-molecule inhibitor of soluble and membrane-bound CD73 developed with the aim of eliminating adenosine-mediated immunosuppression in the tumor microenvironment.

OBJECTIVES

- Develop a population PK-PD model capturing the relationship between dose, quemliclustat PK profiles, soluble CD73 (sCD73) measurements, and CD73 enzyme activity
- Use model simulations to guide dose selection for future clinical trials

METHODS: Clinical Study Design

Study	AB680CSP0001		ARC-8 (dose escalation)
Study design	Single- and multiple-ascending dose, placebo controlled		Open labeled, dose escalation
Population	Healthy volunteers		Patients with gastrointestinal (GI) malignancies
Number of subjects	64		22
Quemliclustat dosing regimen	Single dose, IV infusion	Weekly, IV infusion	Every 2 week (Q2W), IV infusion
Quemliclustat dose (mg)	0.1, 0.6, 2, 4, 8, 16, 25	8	25, 50, 75, 100, 125
Number of subjects per cohort	6 + 2 (quemliclustat + placebo)		3 to 6
Data* analyzed	Quemliclustat PK, soluble CD73 (sCD73) concentrations		Quemliclustat PK, sCD73 concentrations, plasma CD73 enzyme activities

**Preliminary data for quemliclustat PK from ARC-8, sCD73 concentrations, and CD73 enzyme activities as of 06-Aug-2021*

METHODS: Population PK-PD Model Structure

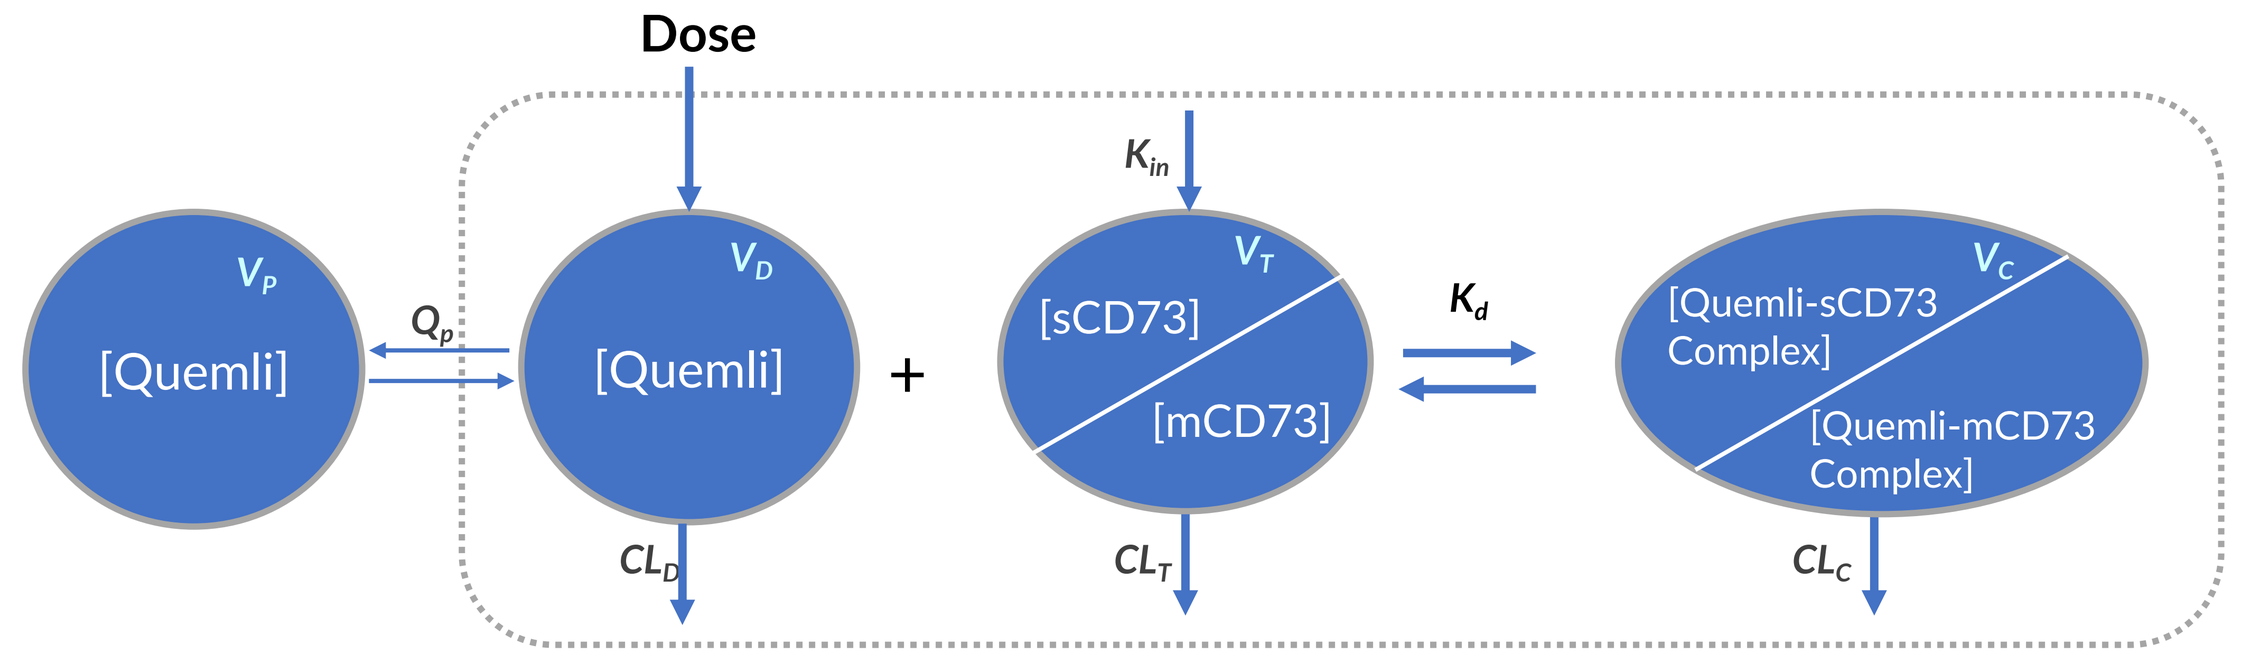
Dose
V_P
[Quemli]
Q_p
V_D
[Quemli]
CL_D
+
K_{in}
V_T
[sCD73]
[mCD73]
CL_T
K_d
V_C
[Quemli-sCD73 Complex]
[Quemli-mCD73 Complex]
CL_C

Abbreviations: Quemli: Quemliclustat; sCD73: soluble CD73; mCD73: membrane-bound CD73; CL_D, CL_T, CL_C: Clearance of unbound Drug (Quemli), Target (CD73), and drug-target Complex, respectively; V_D, V_T, V_C: Distribution volume of unbound Drug (Quemli), Target (CD73), and drug-target Complex, respectively; Q_p: Distribution clearance of unbound drug; V_p: Peripheral volume for unbound drug; K_d: Dissociation constant; k_{in}: Production rate for target

RESULTS

- A preliminary population PK-PD model, consistent with target-mediated disposition of quemliclustat, adequately described the quemliclustat PK and sCD73 profiles over the dose range of 0.1 to 125 mg.
- The model established a power relationship between predicted free CD73 concentrations and functional measurements of residual CD73 enzyme activity.
- Simulations indicated that at steady-state, the recommended dose for expansion of 100 mg Q2W would yield quemliclustat concentrations above the level required to achieve ≥90% inhibition of CD73 enzyme activity in majority of patients with GI malignancies.

A mechanism-based model was developed to adequately describe quemliclustat PK, sCD73, and its inhibition of CD73 enzyme activity. The model provided rationale for selecting a dose of 100 mg Q2W for future clinical trials for quemliclustat.

Model adequately described the quemliclustat PK and sCD73 profiles over the dose range of 0.1 to 125 mg

Visual Predictive Check: Quemliclustat PK

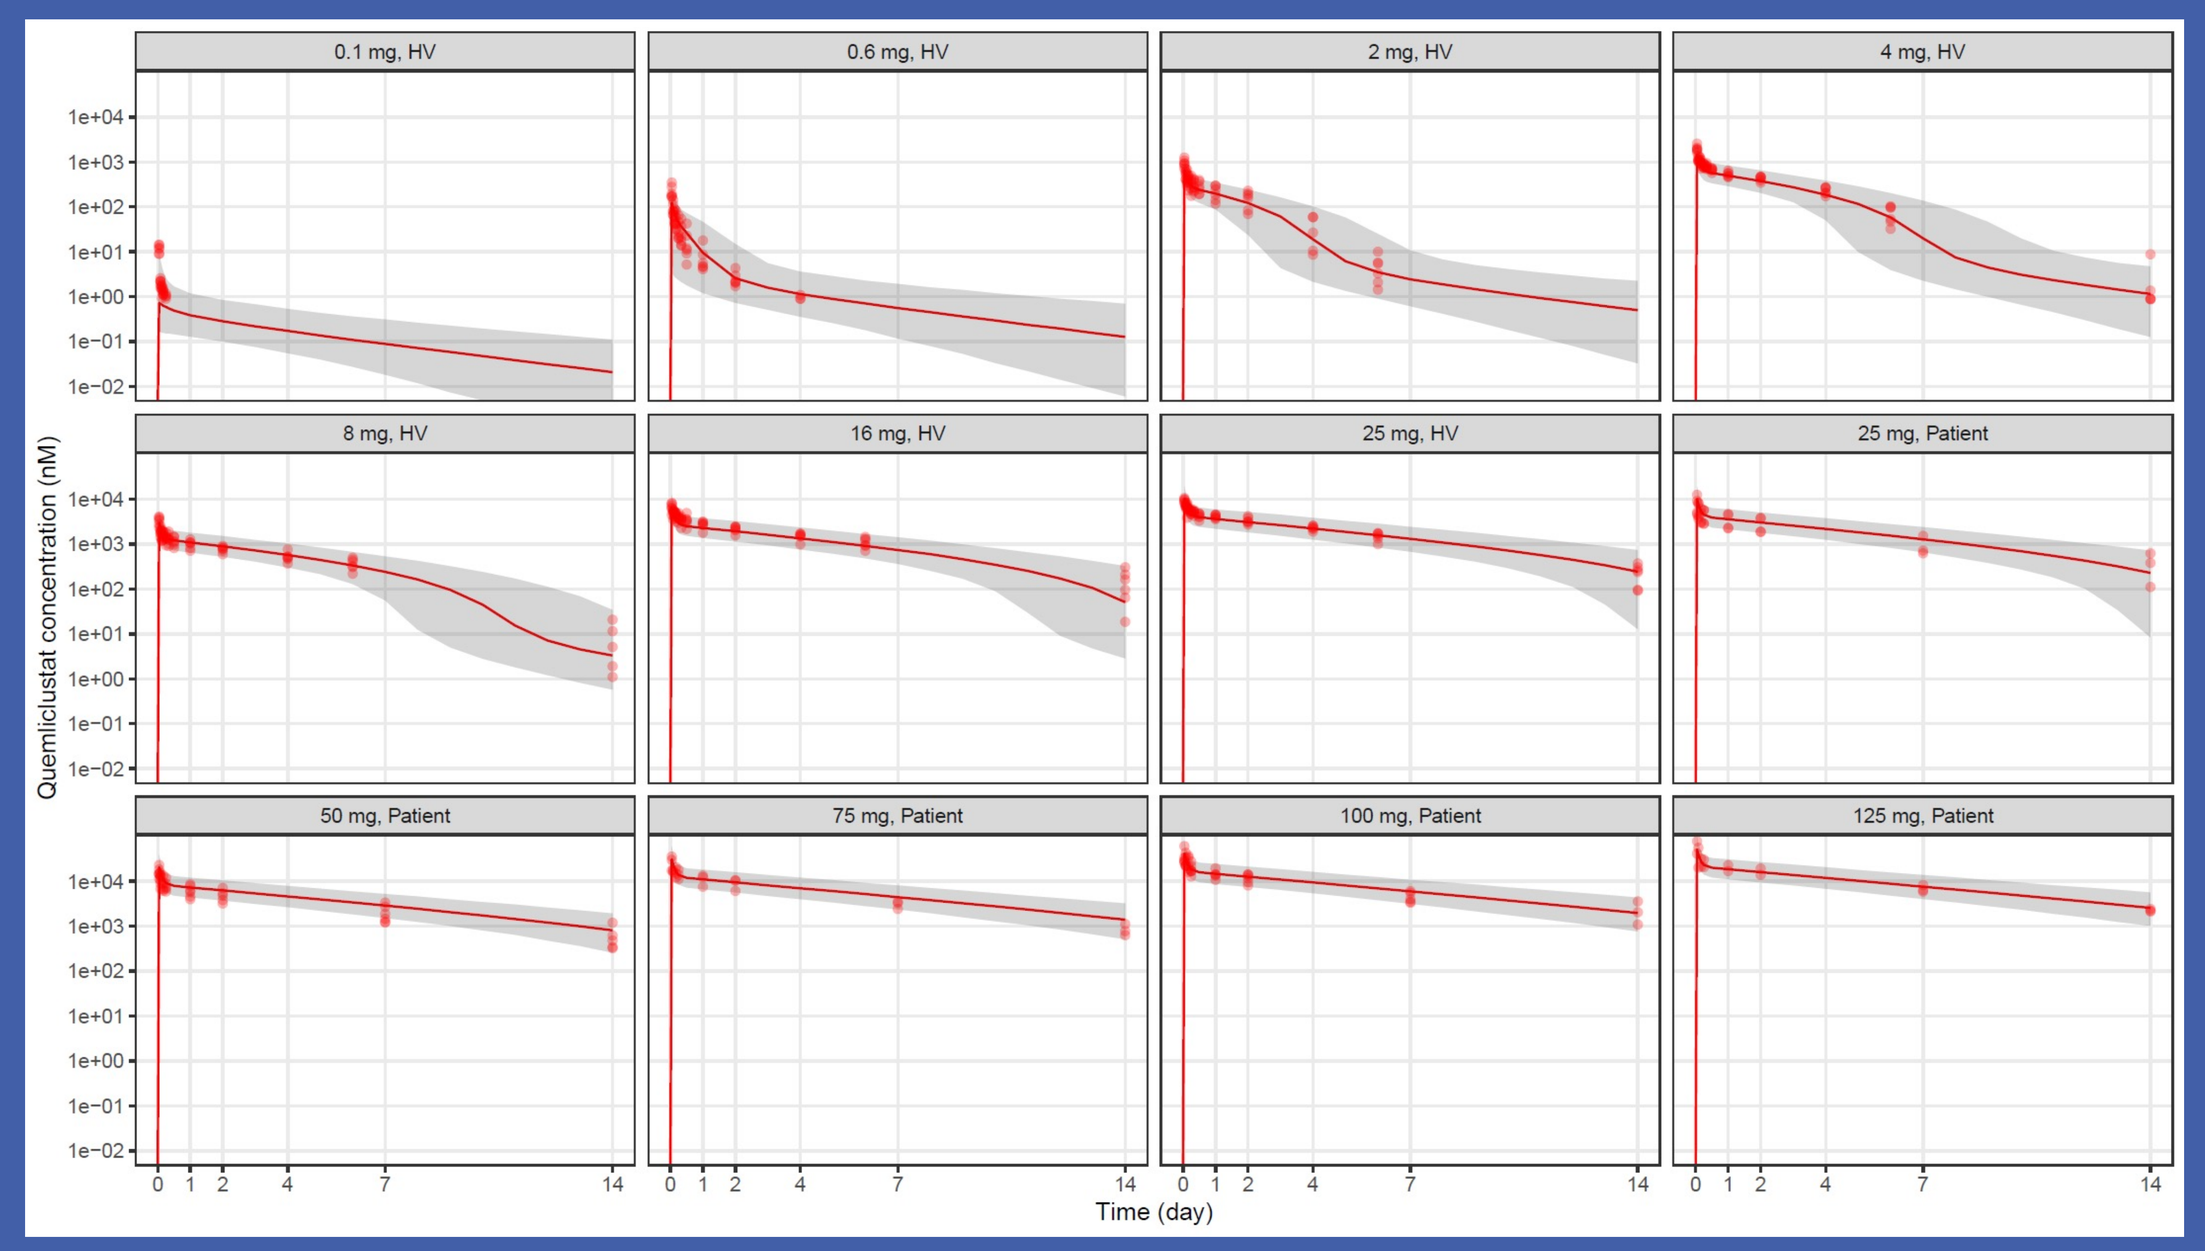
0.1 mg, HV
0.6 mg, HV
2 mg, HV
4 mg, HV
8 mg, HV
16 mg, HV
25 mg, HV
25 mg, Patient
50 mg, Patient
75 mg, Patient
100 mg, Patient
125 mg, Patient
Quemliclustat concentration (nM)
Time (day)

<u>Symbols</u>: Observed concentrations
<u>Lines</u>: Median of simulated concentrations
<u>Shaded areas</u>: 5th and 95th percentiles of simulated concentrations

Visual Predictive Check: Soluble CD73

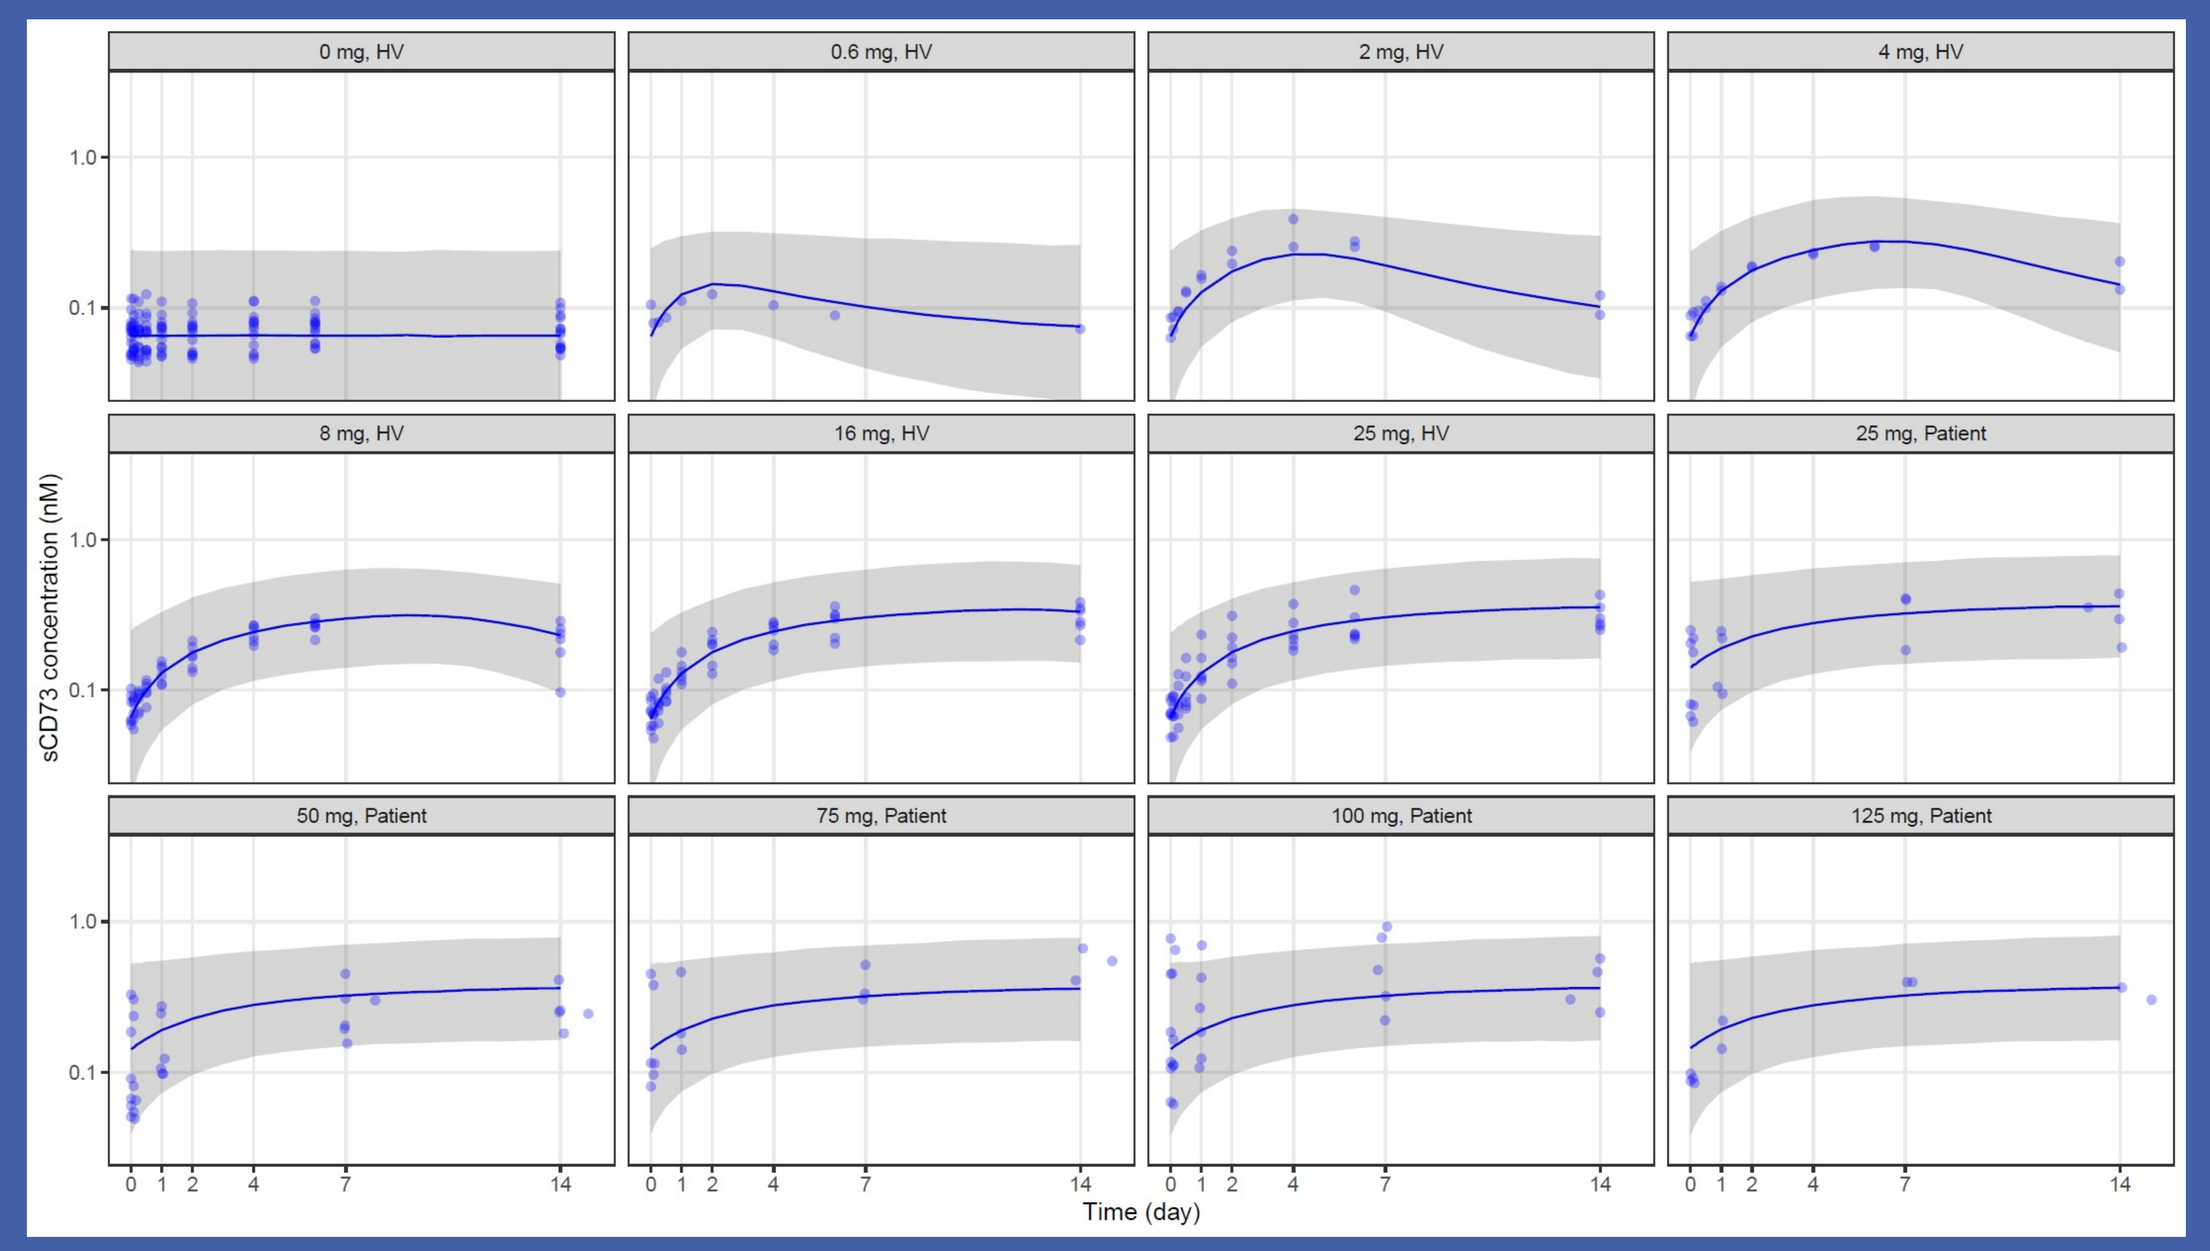
0 mg, HV
0.6 mg, HV
2 mg, HV
4 mg, HV
8 mg, HV
16 mg, HV
25 mg, HV
25 mg, Patient
50 mg, Patient
75 mg, Patient
100 mg, Patient
125 mg, Patient
sCD73 concentration (nM)
Time (day)

Dose selection rationale based on population PK-PD model

- The model predicted quemliclustat at doses ≥100 mg would yield exposure above the IC_{90} of plasma CD73 activity throughout the dosing interval in majority of patients
- The relationship between the inhibition of plasma CD73 activity and antitumor effects is being investigated in ongoing clinical trials

Observed quemliclustat concentration vs plasma CD73 activity measurement

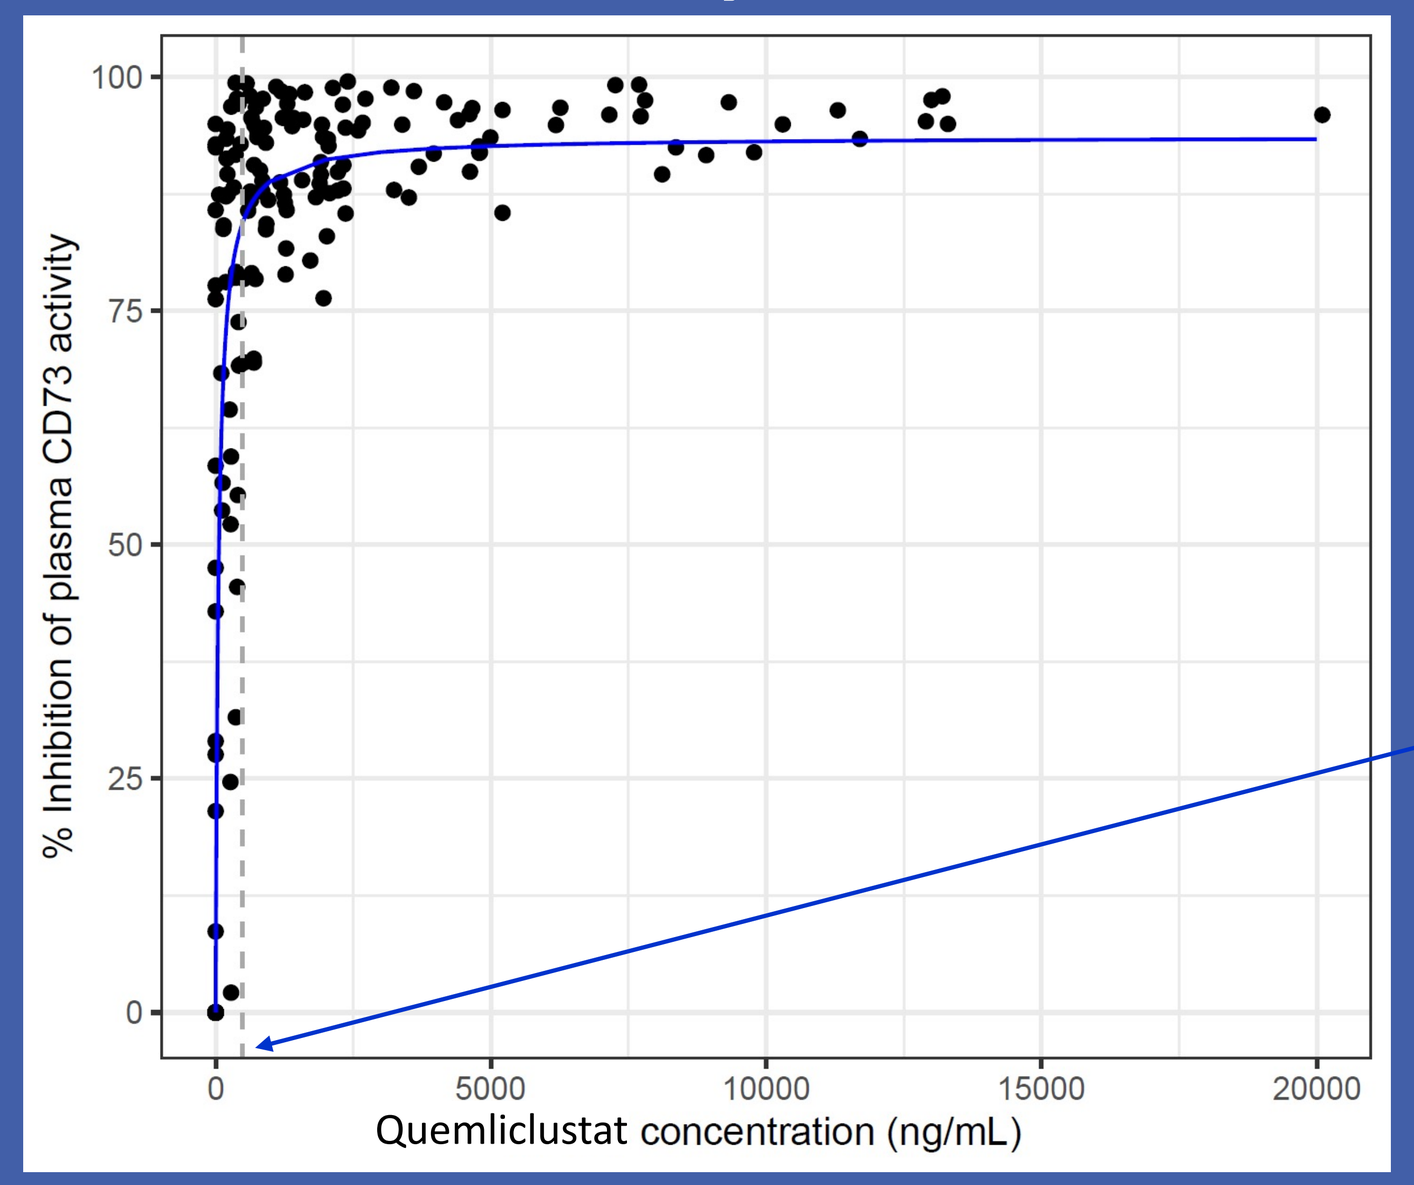
% Inhibition of plasma CD73 activity
Quemliclustat concentration (ng/mL)

IC_{90} (480 ng/mL) for plasma CD73 activity

Predicted quemliclustat PK profiles at steady state after Q2W dosing at 25, 100, and 125 mg

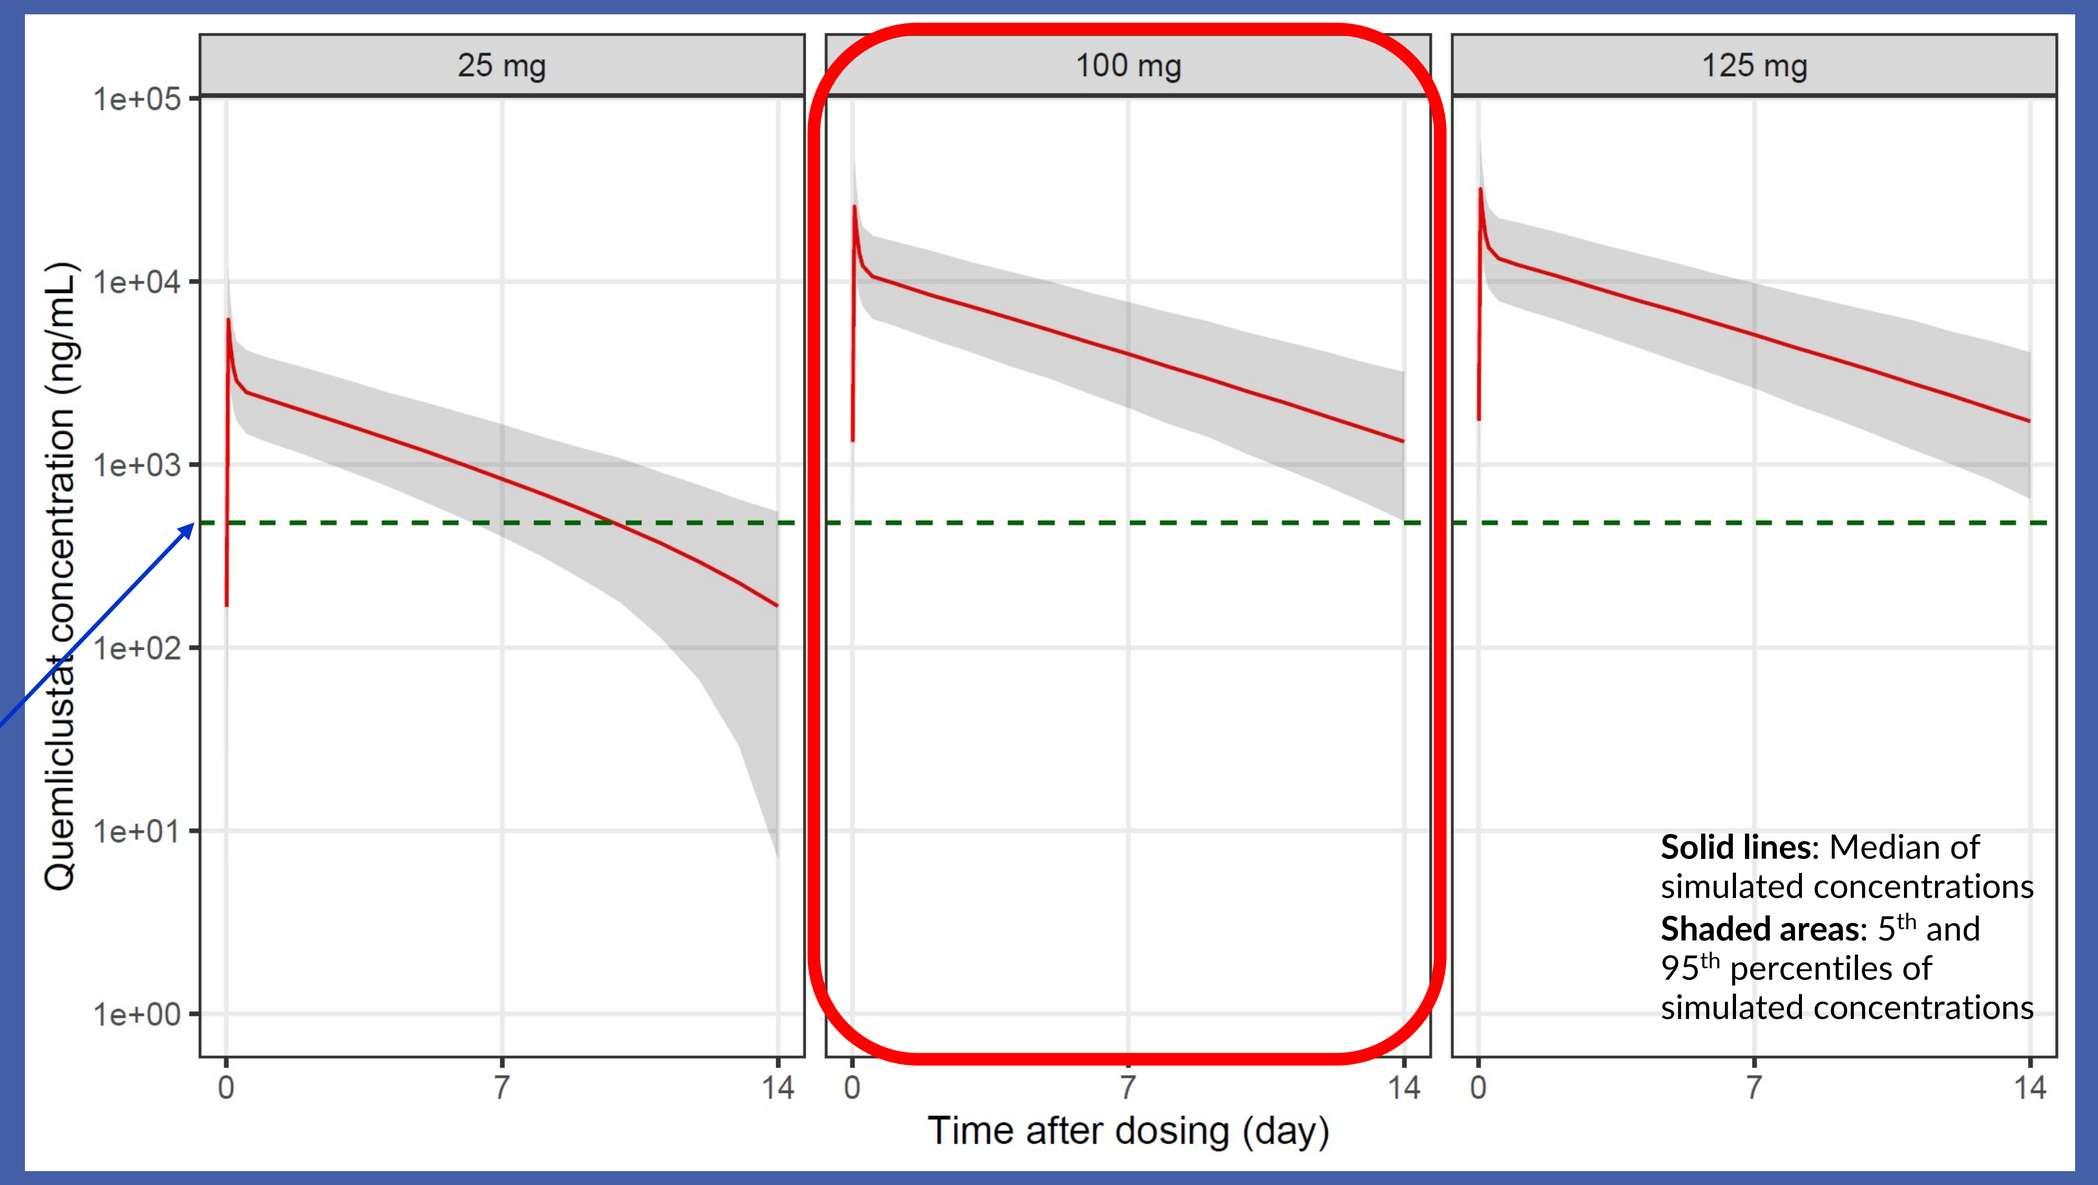
25 mg
100 mg
125 mg
Quemliclustat concentration (ng/mL)
Time after dosing (day)
Solid lines: Median of simulated concentrations
Shaded areas: 5th and 95th percentiles of simulated concentrations

Correlation between model-predicted target saturation and observed plasma CD73 activity

- The PK-PD model leverages the dose-dependent PK pattern to capture the saturation of CD73 targets throughout of body, with verification against sCD73 measurements in blood
- A power relationship was established between the predicted degree of target saturation in the body vs observed CD73 activity in plasma samples

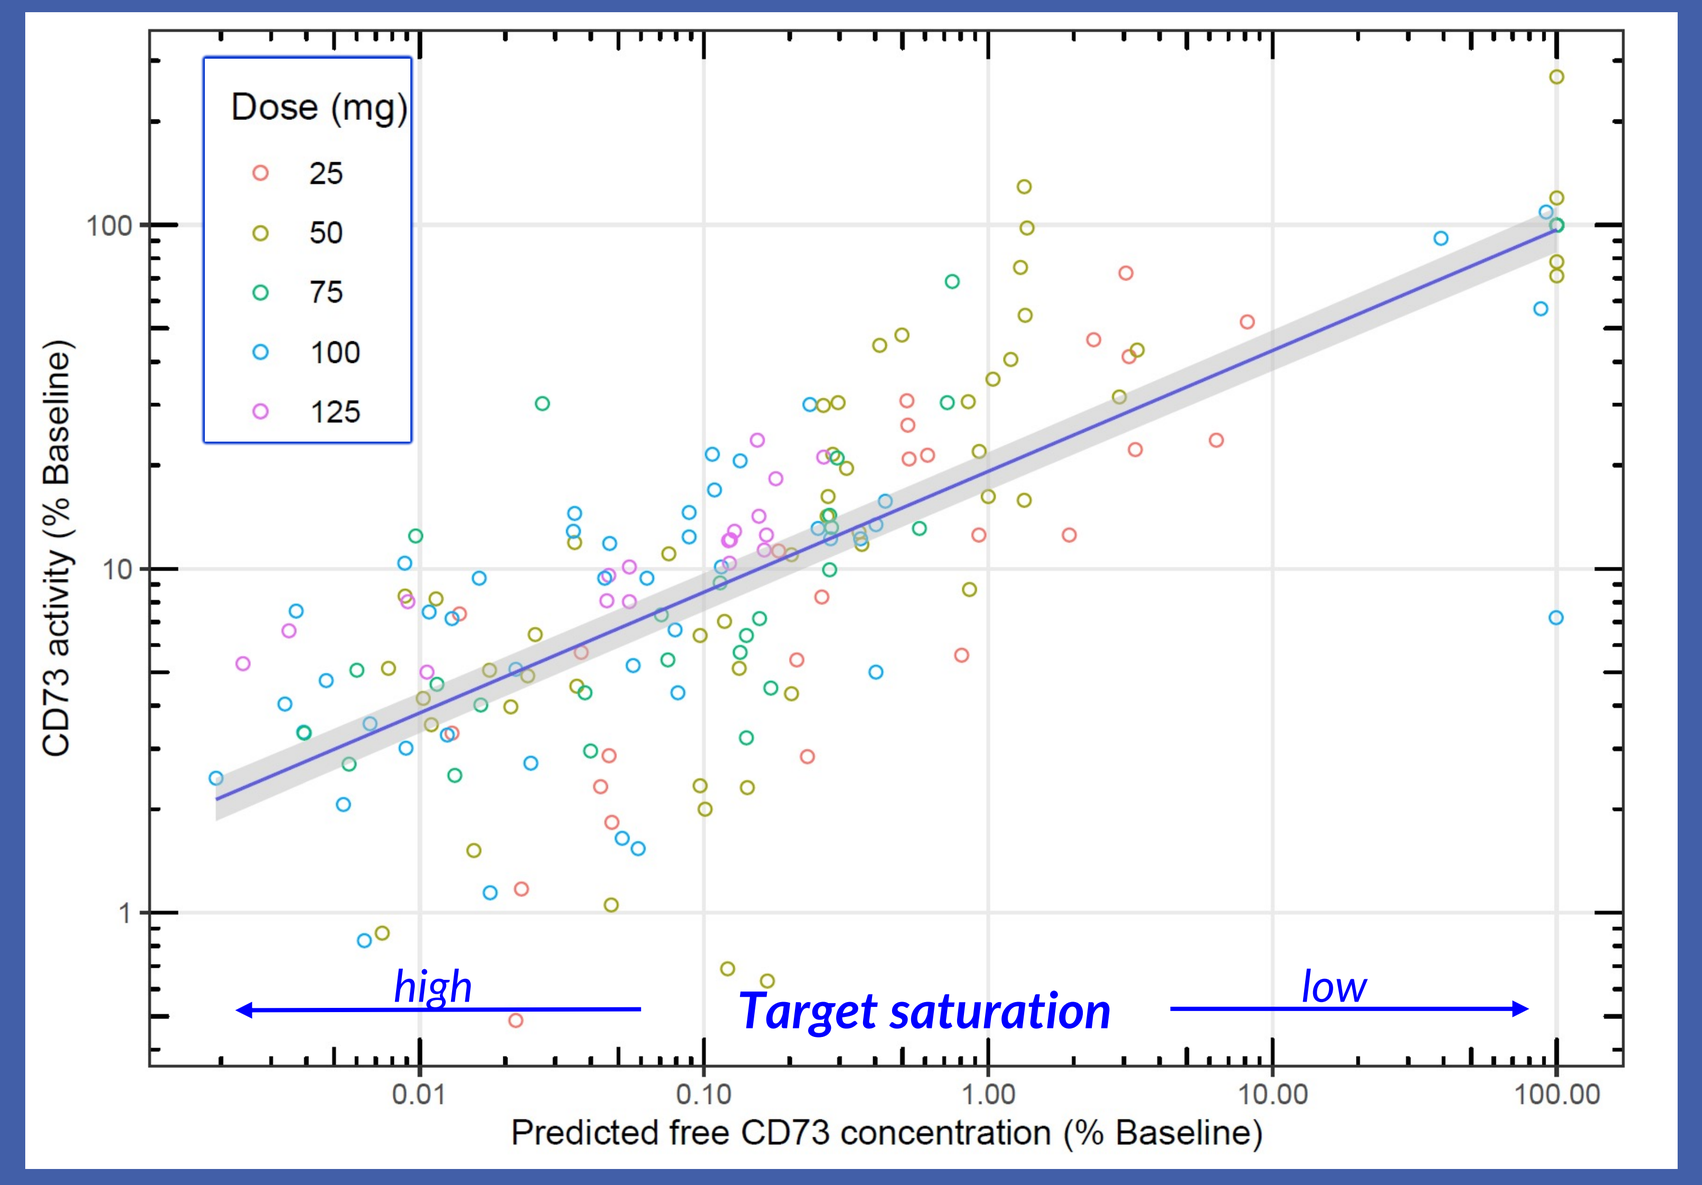
Dose (mg)
25
50
75
100
125
CD73 activity (% Baseline)
high
Target saturation
low
Predicted free CD73 concentration (% Baseline)

Take a picture to **download** the **full paper**

Parameter Estimates

Parameter	Unit	Estimate (RSE)	IIV (RSE)
CL_D (clearance of unbound drug)	L/h	0.058 (6.3%)	24% (14%)
V_D (distribution volume of unbound drug)	L	3.6 (6.7%)	33% (23%)
Correlation between $\eta(CL_D)$ & $\eta(V_D)$	--	0.74 (18%)	--
Q_p (distribution clearance of unbound drug)	L/h	1.1 (10%)	--
V_p (peripheral volume for unbound drug)	L	5.9 (4.2%)	--
CL_C (clearance of drug-target complex)	L/h	0.032 (8.4%)	36% (17%)
V_C and V_T (distribution volume for complex & target)	L	Same as V_D	Same as V_D
CL_T/V_T (ratio of clearance to volume for target)	1/h	0.053 (16%)	63% (13%)
Ratio: Patient to HV	--	0.46 (21%)	--
Correlation between $\eta(CL_T/V_T)$ & $\eta(CL_C)$	--	0.58 (21%)	--
K_d (dissociation constant)	nM	1.0 (FIX)[a]	--
Fraction of soluble target as total target	%	0.057% (5.9%)	--
Production rate for target	nmol/h	22 (8.2%)	33% (14%)
Residual variability for drug concentration	--	28% (0.9%)	--
Residual variability for soluble target concentration	--	11% (3.1%)	--

IIV: Inter-individual variability; RSE: Relative standard error
[a]Various K_d values in half-log increment were explored and the final value was selected based on objective function values

Goodness-of-Fit Plots for Quemliclustat PK

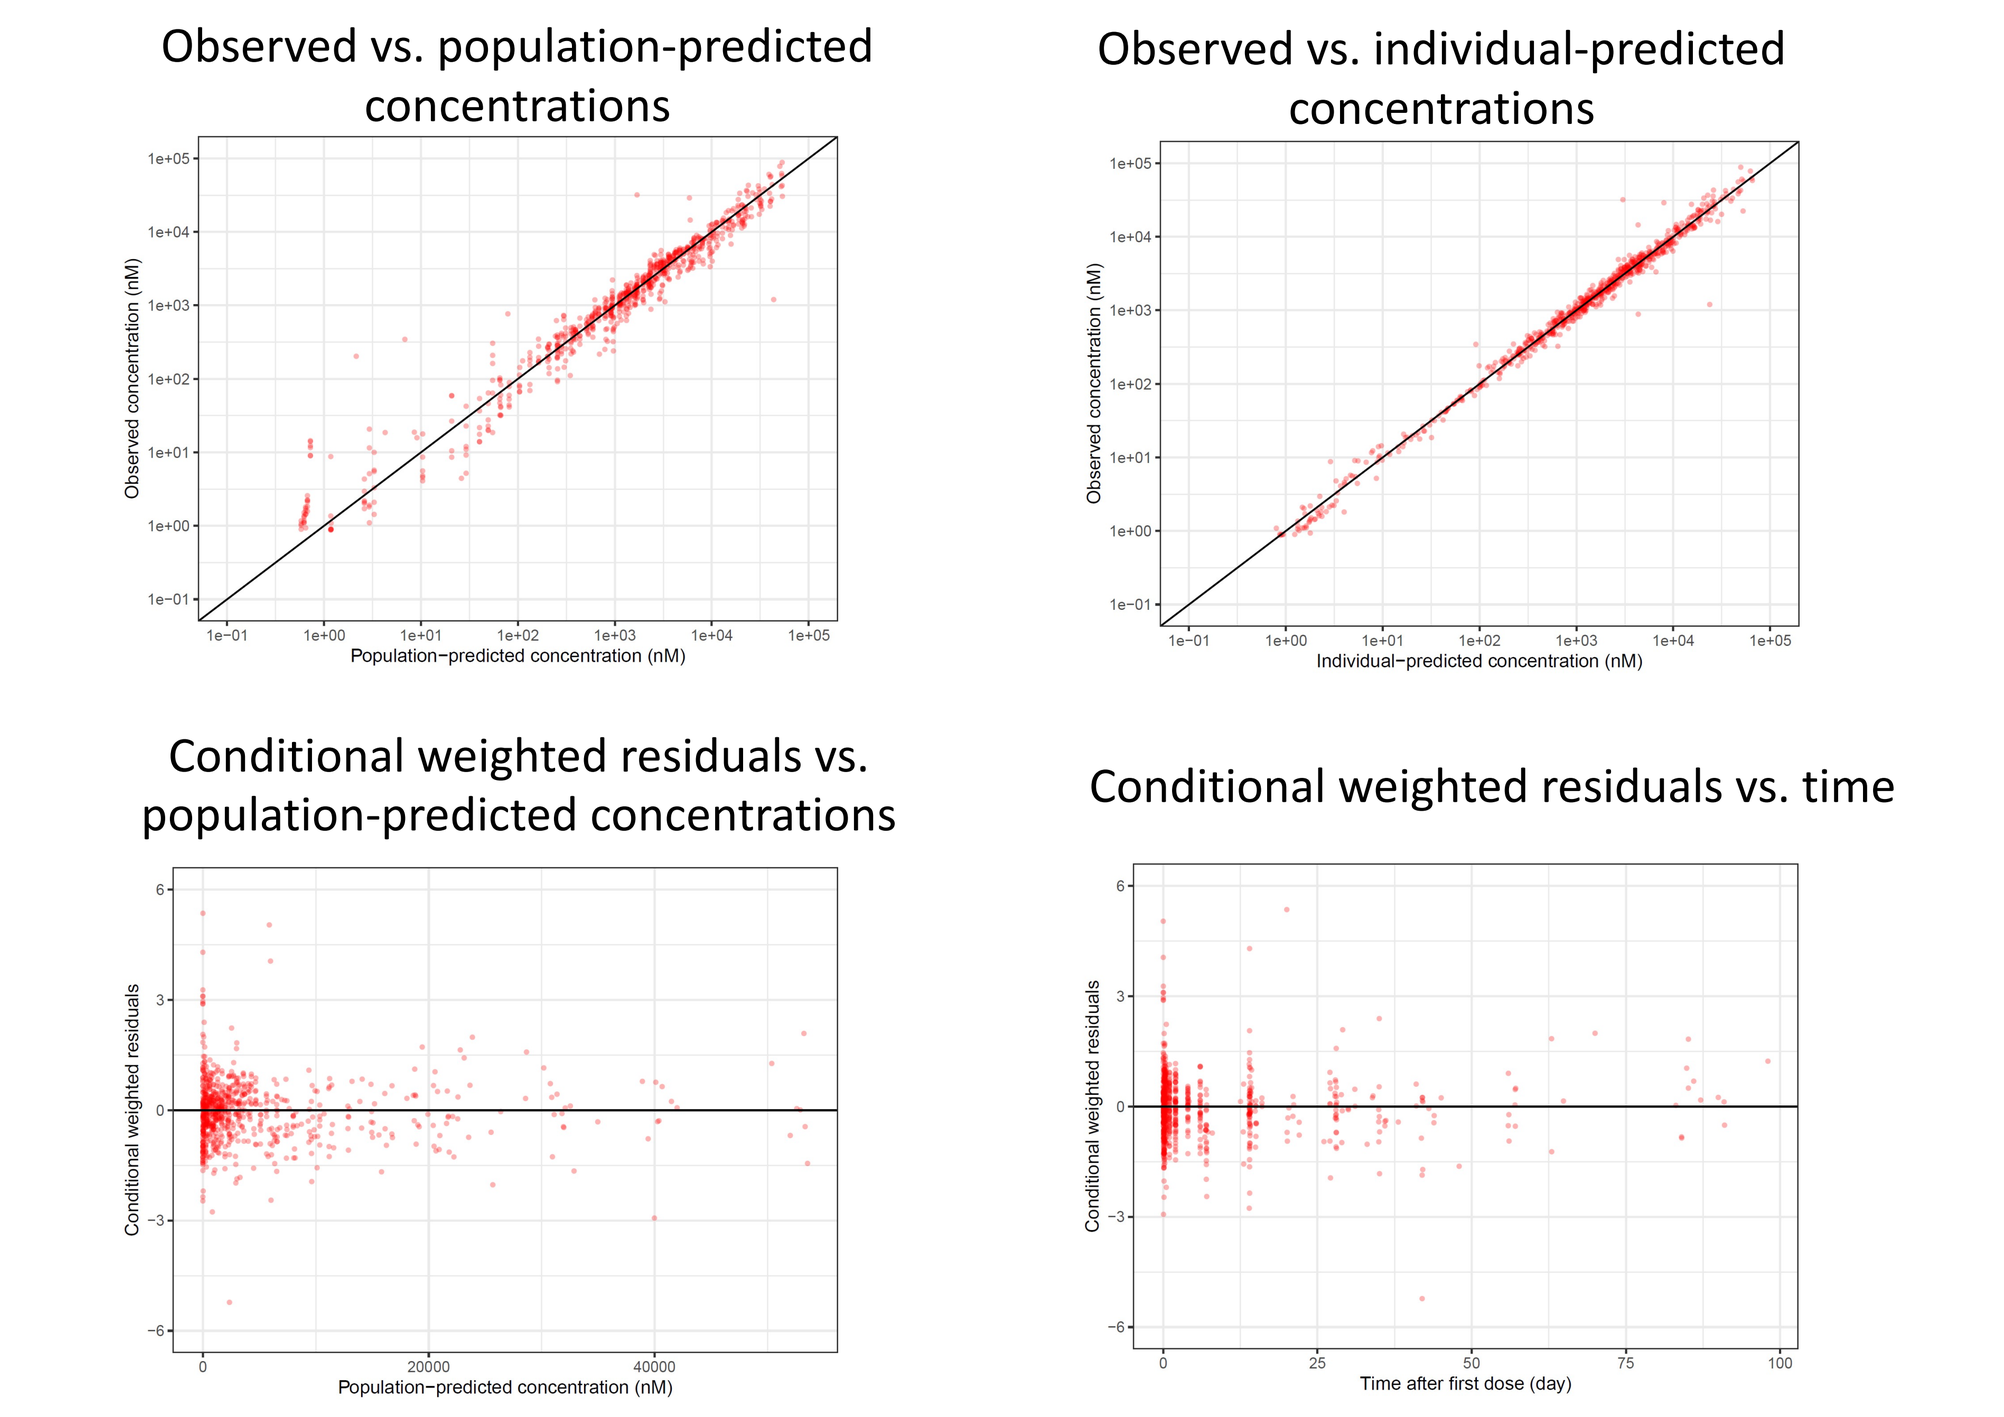
Observed vs. population-predicted concentrations
Observed vs. individual-predicted concentrations
Conditional weighted residuals vs. population-predicted concentrations
Conditional weighted residuals vs. time

Goodness-of-Fit Plots for sCD73 concentrations

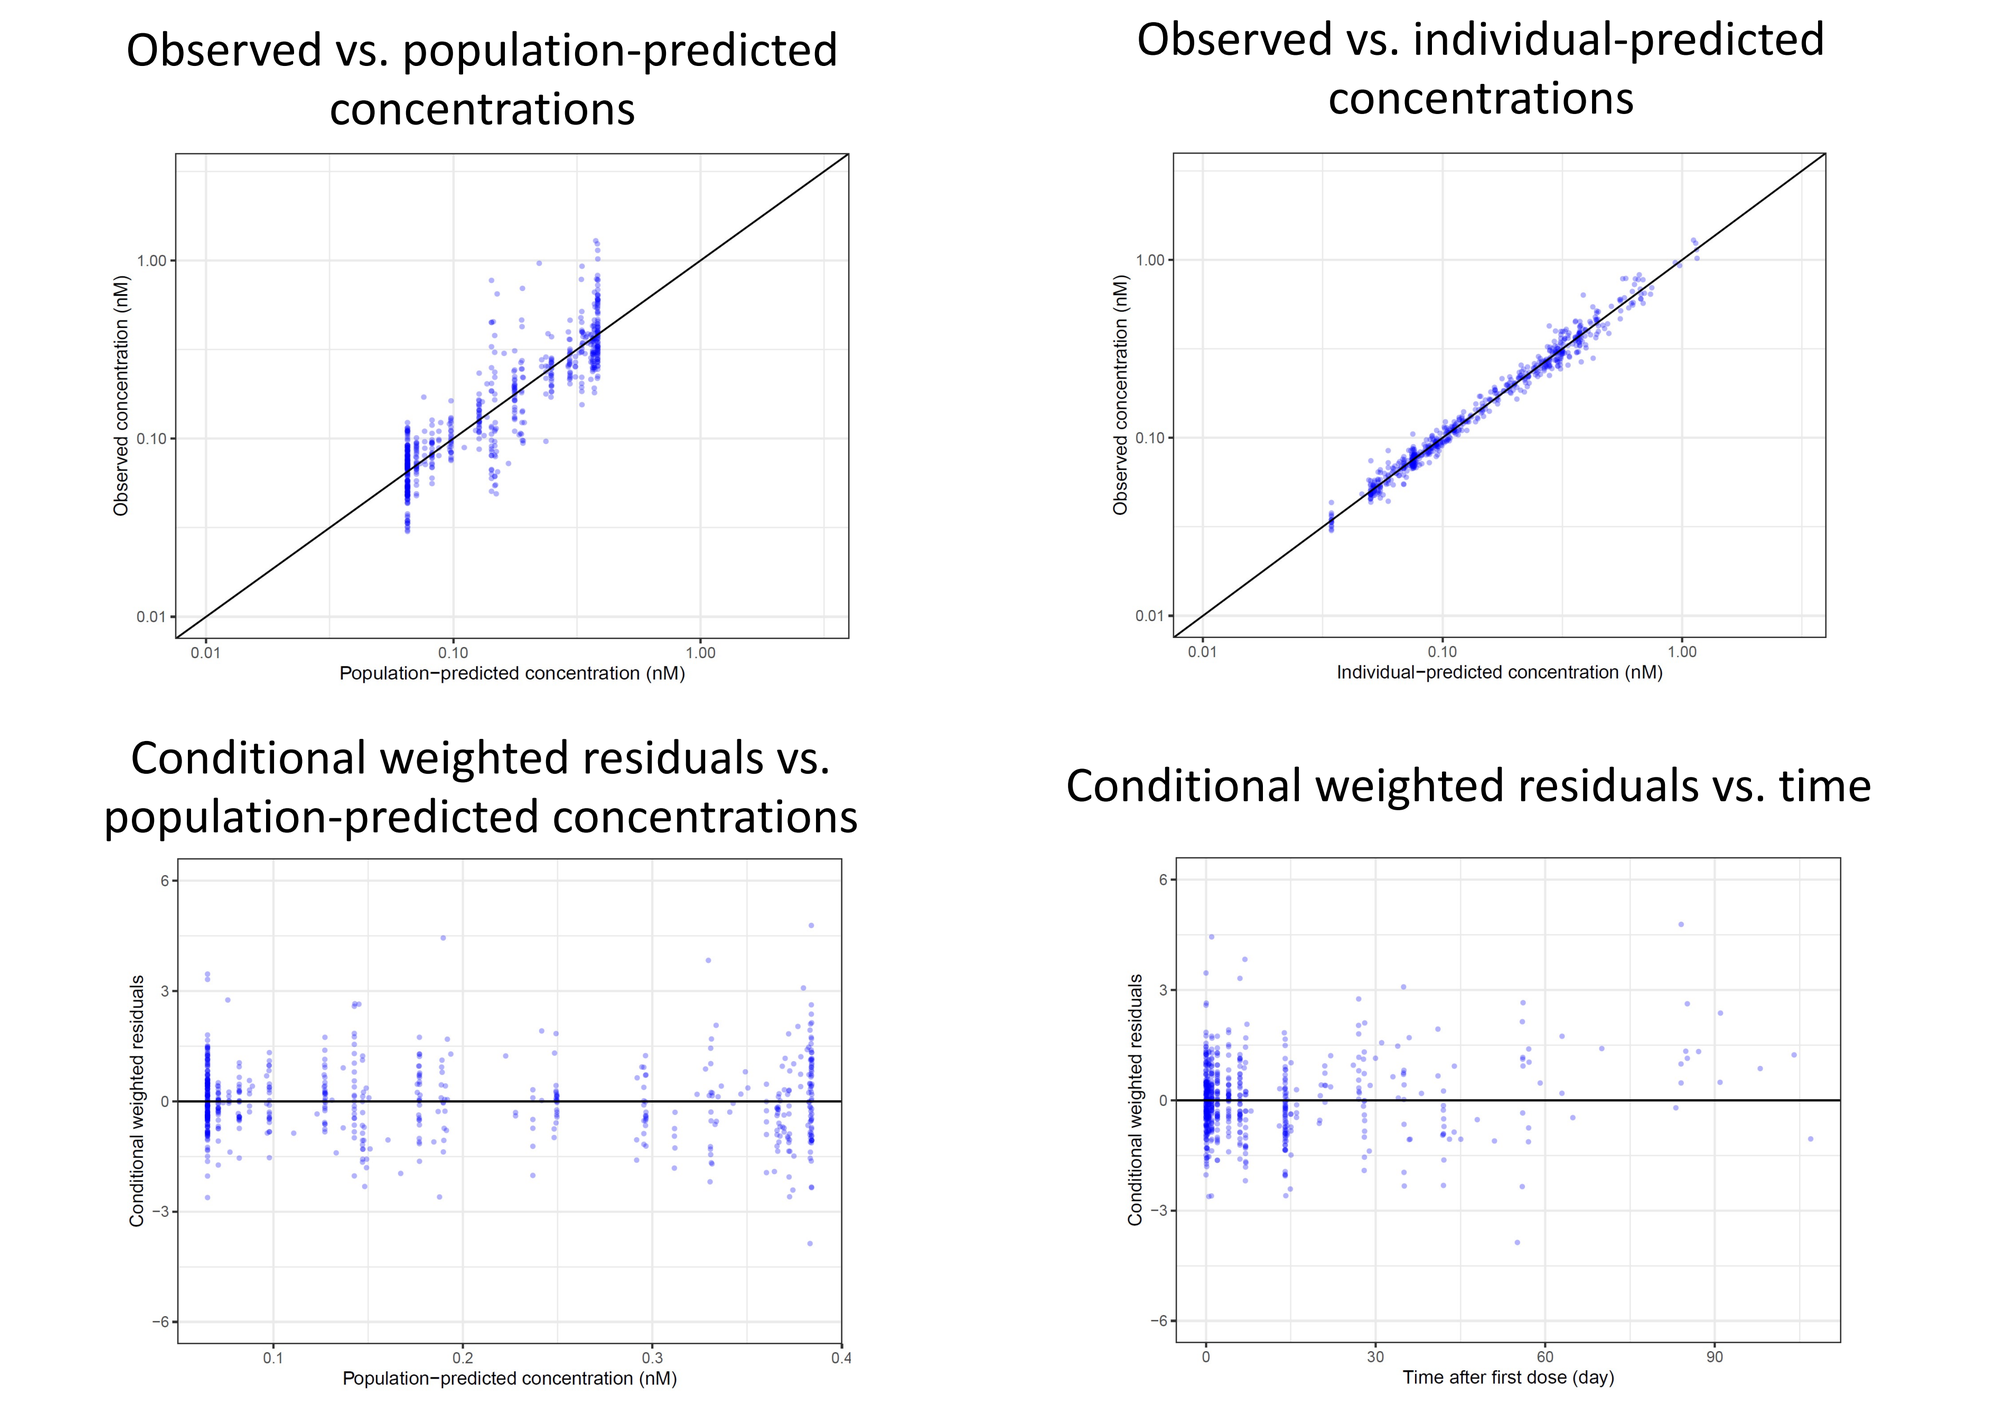
Observed vs. population-predicted concentrations
Observed vs. individual-predicted concentrations
Conditional weighted residuals vs. population-predicted concentrations
Conditional weighted residuals vs. time

METHODS: Software

- The population PK-PD analysis was conducted using nonlinear mixed-effects modeling with the NONMEM software, version 7.5
- Graphical and all other statistical analyses, including evaluation of NONMEM outputs, were performed using R version 3.6.2 for Windows

Kai H. Liao, Linh Nguyen, Lisa Seitz, Lixia Jin, Amy E. Anderson, Elaine Ginn, Salema Jafri, Balaji Agoram

ARCUS BIOSCIENCES