Anti-TIGIT Antibodies Promote Immune Activation Relevant to Targeting Stem-like and Tumor-specific T Cells in Combination With Anti-PD-1

IMMUNOLOGY2022
#2719

Kelsey E Sivick Gauthier[1], Amita Patnaik[2], Amy E Anderson[1], Amber E de Groot[1], Dana Piovesan[1], Ferdie Soriano[1], Ada Chen[1], Xiaoning Zhao[1], Lisa Seitz[1], Anita Reddy[1], Stephen W Young[1], Nigel Walker[1], Paul Foster[1], Matthew J Walters[1]

[1]Arcus Biosciences, 3928 Point Eden Way, Hayward, CA 94545 (USA)
[2]START San Antonio, San Antonio, TX 78229 (USA)

Abstract

Background. TIGIT is an inhibitory receptor expressed primarily on NK and T cell subsets, and binding to its cognate receptor ligand, CD155, results in multiple mechanisms of immunosuppression. Blocking the TIGIT-CD155 interaction in the context of cancer promotes anti-tumor immunity.

Methods. We characterized cellular subsets that TIGIT blockade may impact and the pharmacology of two anti-TIGIT antibodies - representing functional (AB308) and non-functional (AB154, domvanalimab, dom) Fc domain classes - undergoing clinical evaluation. Surrogate antibodies were leveraged to interrogate TIGIT biology in mouse syngeneic tumor models. Human tumor-infiltrating lymphocytes from a variety of cancer types expressed appreciable levels of TIGIT on relevant immune populations, including T_{regs} and $CD8^+$ T cells with tumor reactive or pre-dysfunctional stem-like phenotypes.

Results. Both antibodies potently bound TIGIT and blocked the TIGIT-CD155 interaction as well as displayed the predicted phenotypes in terms of Fcγ receptor (FcγR) engagement. In line with FcγRIII binding, AB308 demonstrated a capacity to induce ADCC against TIGIT-expressing target cells. Combination of anti-TIGIT antibodies with other therapeutic approaches that promote T cell activation resulted in enhanced immune responses. In mice, while combining Fc-silent or Fc-enabled anti-mouse TIGIT antibody with anti-PD-1 resulted in greater tumor growth inhibition than with anti-PD-1 alone, the activity of Fc-enabled anti-TIGIT was associated with intratumoral T_{reg} depletion.

Conclusion. These data provide a rationale for combination with immune-activating agents and support ongoing clinical evaluation of AB154 and AB308 with biomarker strategies focused on understanding the role of Fc functionality.

TIGIT Blockade Promotes T and NK Cell Activation

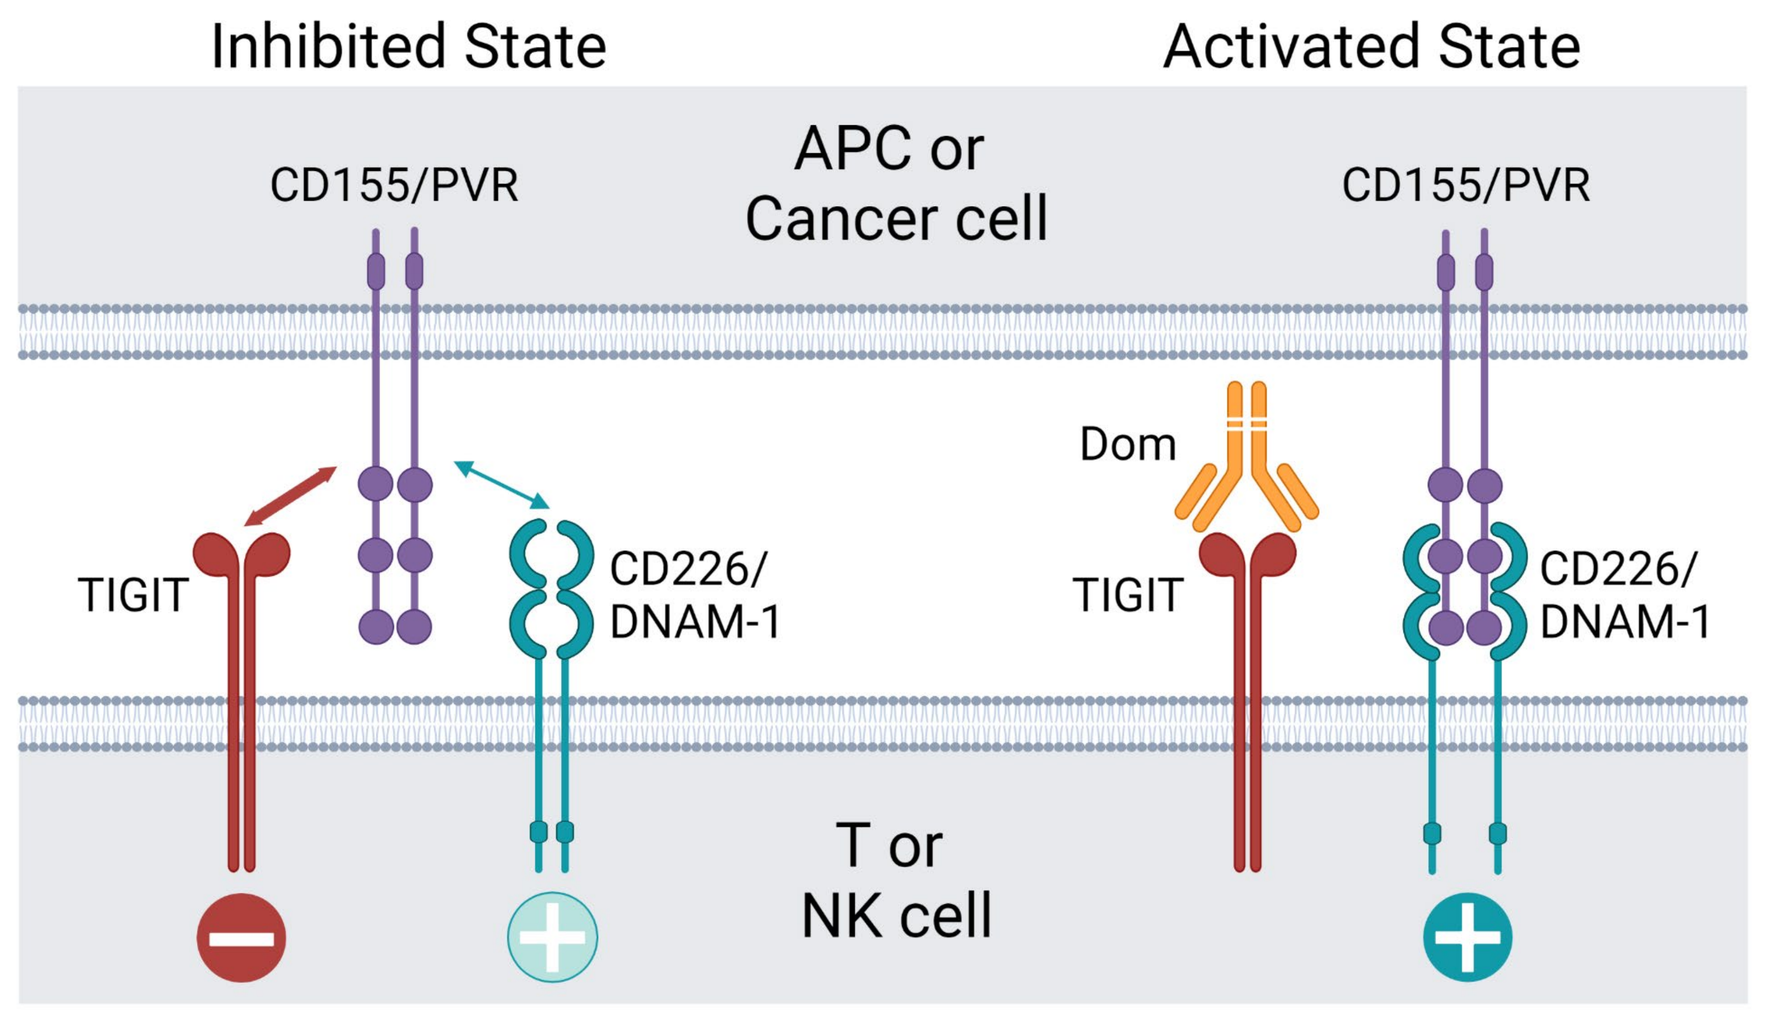
Inhibited State
Activated State
APC or
Cancer cell
CD155/PVR
CD155/PVR
TIGIT
CD226/
DNAM-1
Dom
TIGIT
CD226/
DNAM-1
T or
NK cell

Figure 1. Schematic depicting primary mechanism of action of anti-TIGIT antibodies. On the surface of NK and T cell subsets, TIGIT out-competes CD226/DNAM-1 for binding to CD155/PVR, delivering inhibitory signals and blocking activation. When TIGIT is blocked, CD226 promotes activation and effector function. Thickness of arrows indicates binding affinity; white lines in dom signify mutated Fc domain to silence antibody effector function. Created with Biorender.

PD-1, TIGIT, and CD226 are Expressed on Human Tumor Infiltrating T cells Representing Various Phenotypic Profiles and Differentiation States

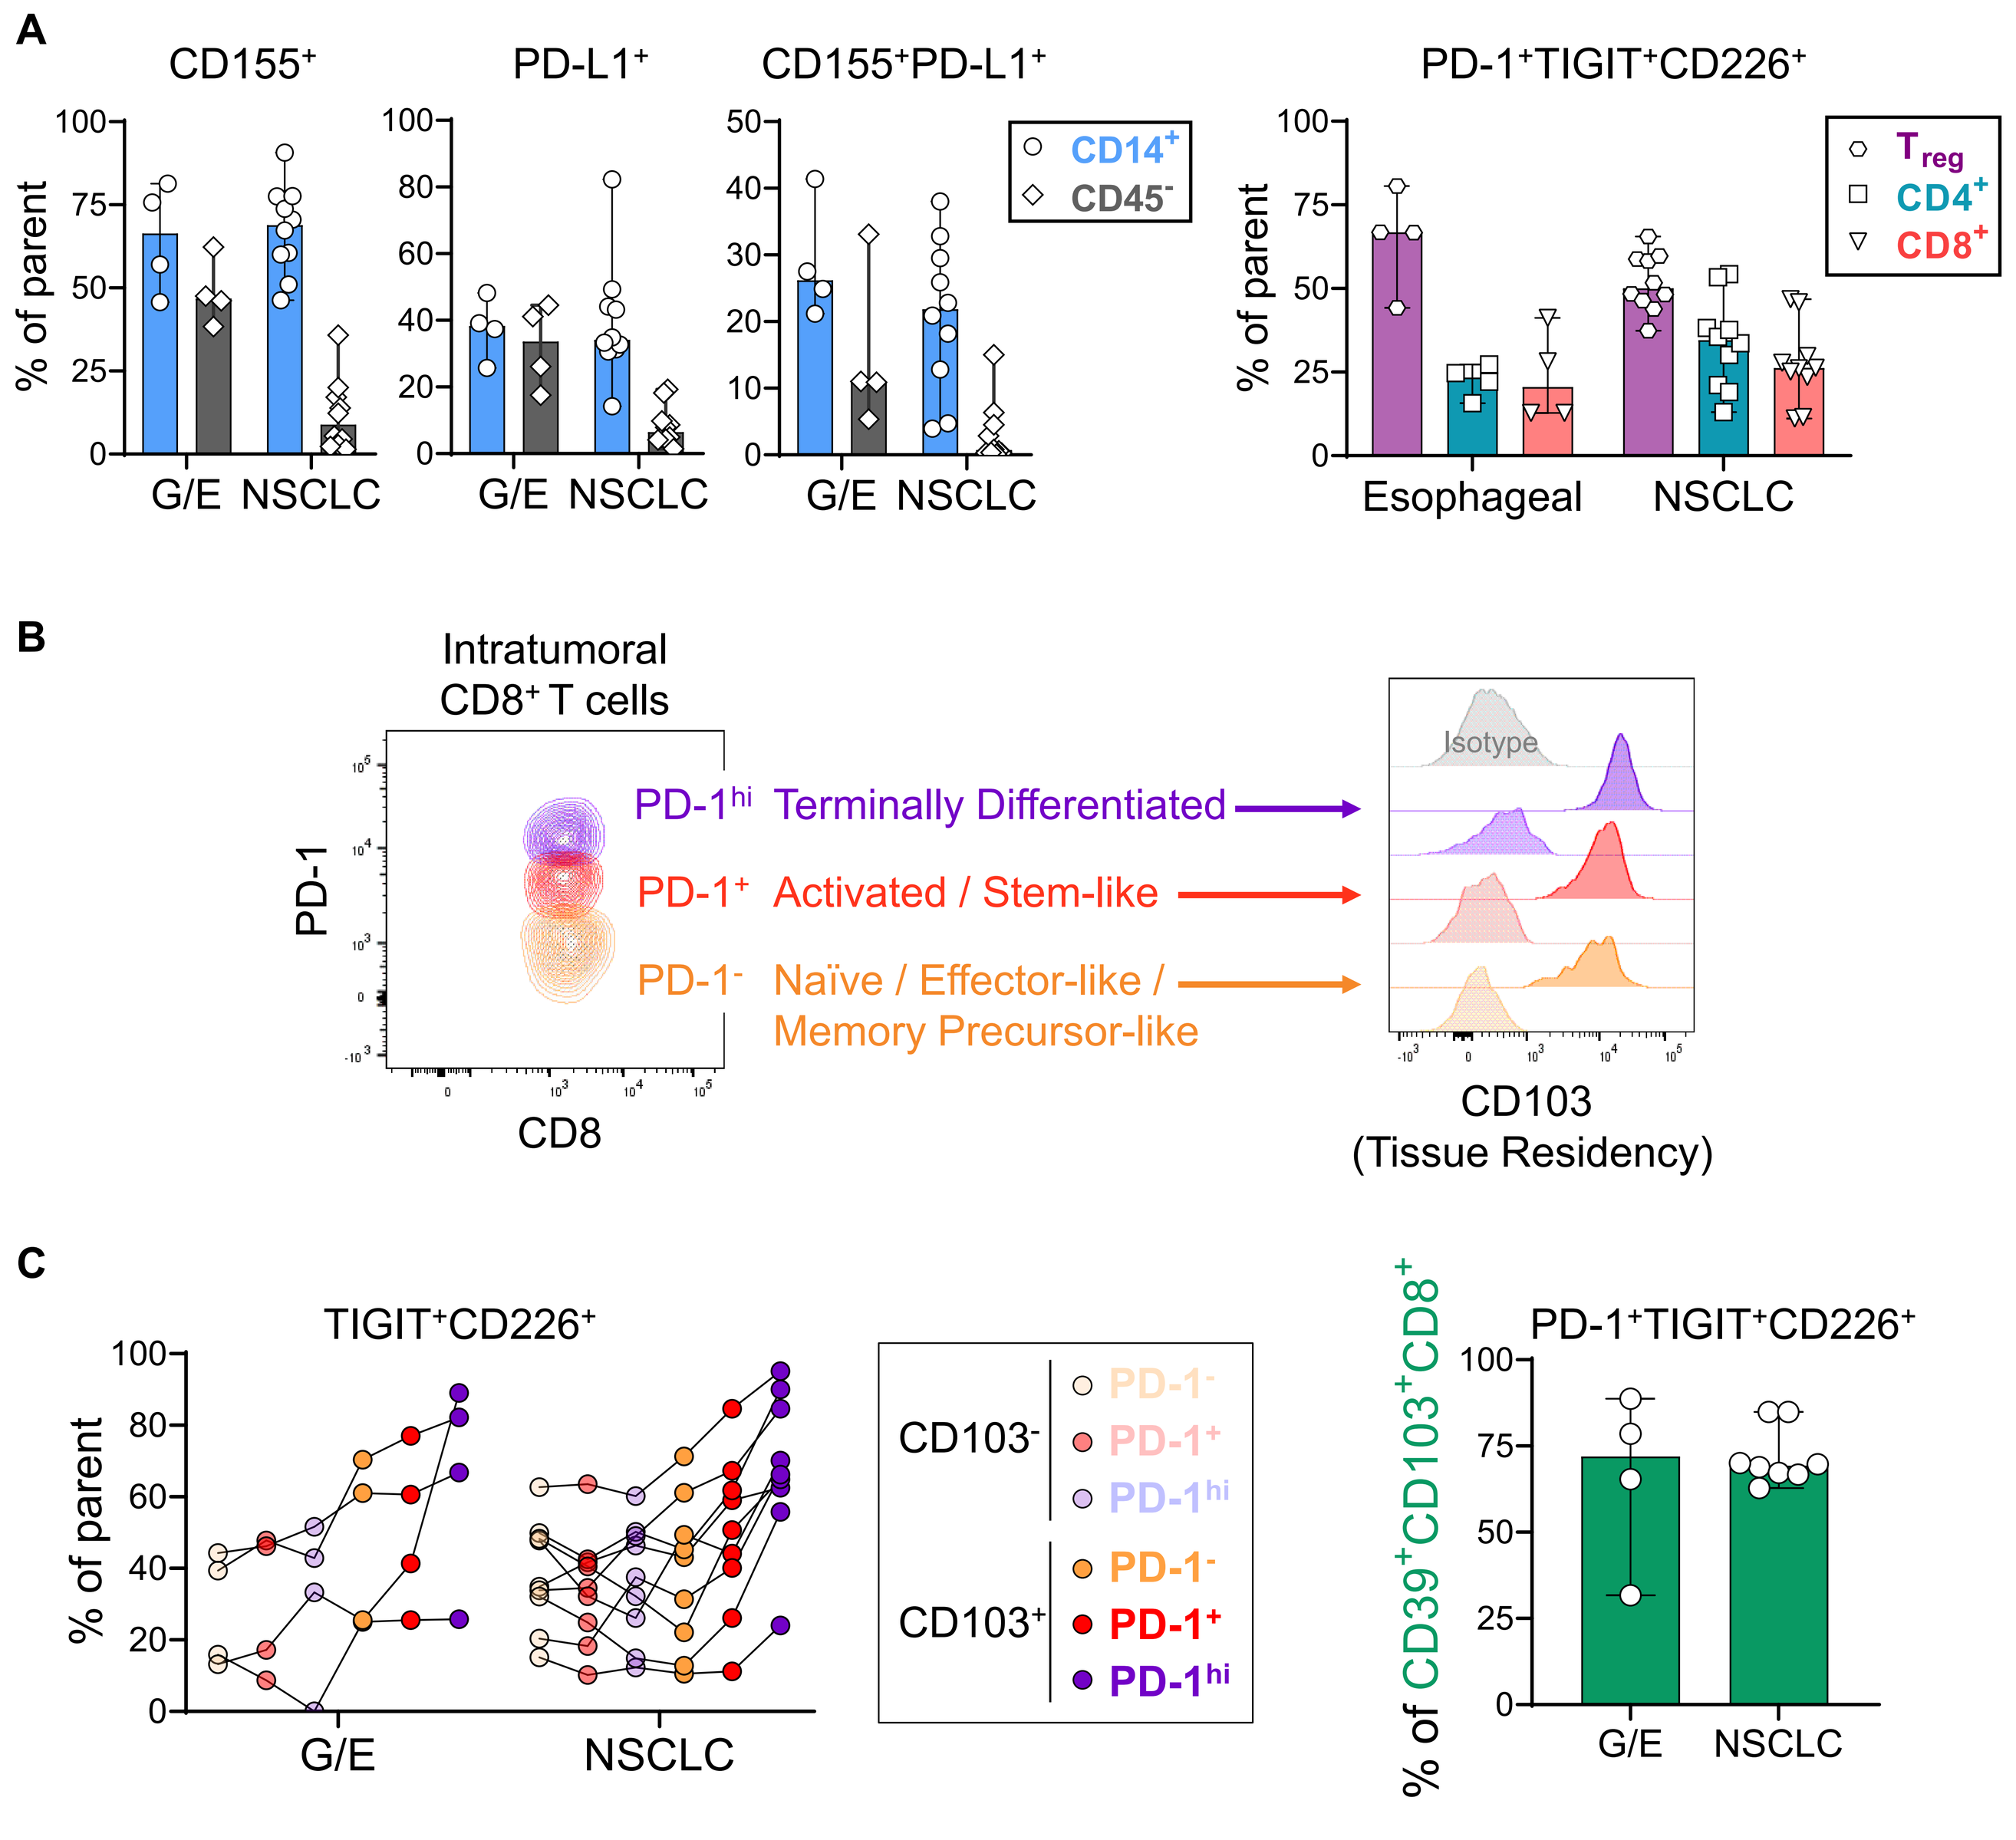
A
CD155+
PD-L1+
CD155+PD-L1+
PD-1+TIGIT+CD226+
% of parent
G/E NSCLC
CD14+
CD45-
Treg
CD4+
CD8+
Esophageal
NSCLC
B
Intratumoral
CD8+ T cells
PD-1
CD8
PD-1hi Terminally Differentiated
PD-1+ Activated / Stem-like
PD-1- Naïve / Effector-like / Memory Precursor-like
Isotype
CD103
(Tissue Residency)
C
TIGIT+CD226+
% of parent
G/E
NSCLC
CD103-
CD103+
PD-1-
PD-1+
PD-1hi
% of CD39+CD103+CD8+
PD-1+TIGIT+CD226+

Figure 2. Evaluation of TIGIT, CD226, and CD155 expression in the tumor microenvironment. Cell subsets from frozen dissociated gastric/esophageal (G/E) and lung (NSCLC) cancer patient tumors (sourced from Discovery Life Sciences) were phenotyped using flow cytometry. **(A)** CD155 and PD-L1 ligand expression on $CD14^+$ monocytes ($CD45^+CD3^-CD16^-CD56^-CD14^+$) and cancer/stromal cells ($CD45^-$) and co-expression of PD-1, TIGIT, and CD226 on broad T cell populations. **(B)** Contour plot (NSCLC subject) of PD-1 expression on $CD8^+$ T cells encompassing various states of activation and differentiation. Each population was further characterized into tissue resident ($CD103^+$) and circulating ($CD103^-$) $CD8^+$ populations, resulting in six subsets. **(C)** Co-expression of TIGIT and CD226 on the six $CD8^+$ T cell populations from (B) and co-expression of PD-1, TIGIT, and CD226 on probable tumor-specific $CD39^+CD103^+CD8^+$ T cells. Lines connect populations from the same subject. Each symbol represents a unique subject while bars and error denote median ± range. PD-$1^{-/+/hi}$ population descriptions based on published literature[1,2].

Fc-silent Anti-TIGIT Enhances Tumor Control by Anti-PD-1 Without Intratumoral Treg Depletion in Mice

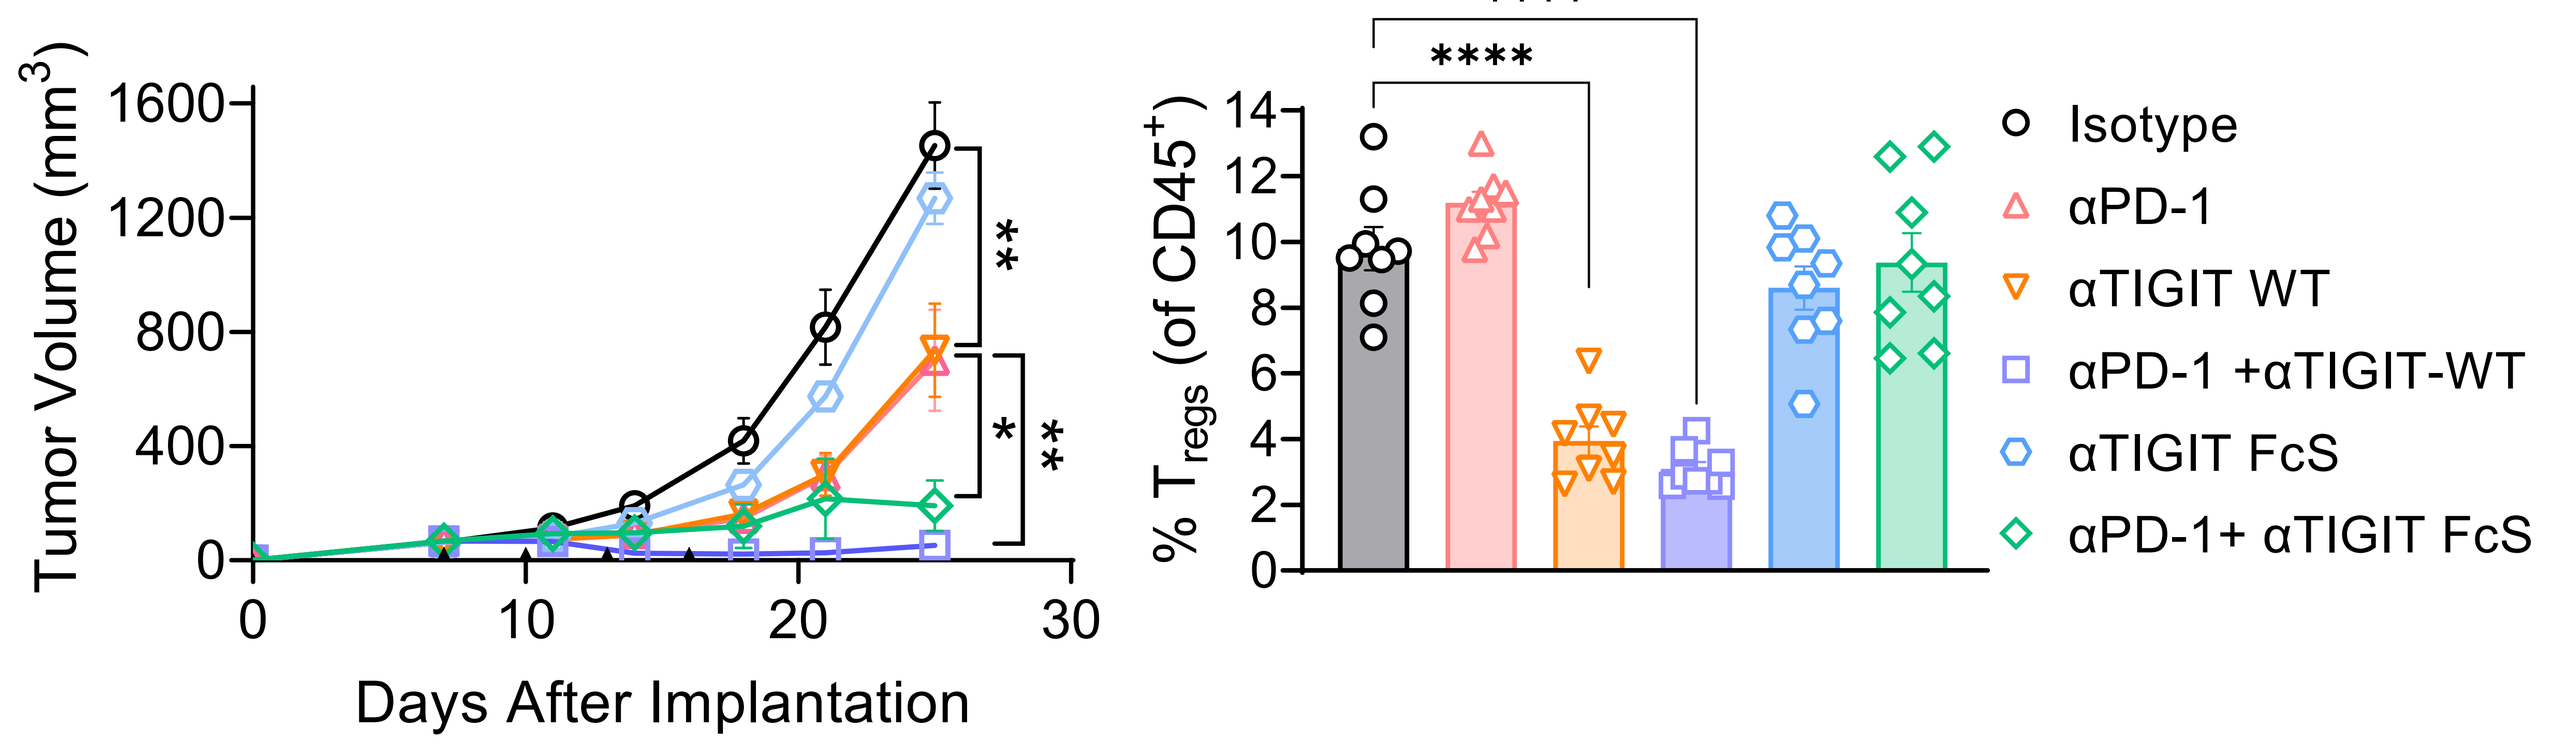
Tumor Volume (mm3)
Days After Implantation
% Tregs (of CD45+)
Isotype
αPD-1
αTIGIT WT
αPD-1 +αTIGIT-WT
αTIGIT FcS
αPD-1+ αTIGIT FcS

Figure 3. Differential effects of surrogate mouse Fc-enabled (WT) and Fc-silent (FcS) anti-TIGIT antibodies in the MC38 syngeneic tumor model. Mice with established MC38 tumors were dosed with 5 mg/kg anti-PD-1 once and 10 mg/kg anti-TIGIT four times as indicated by solid triangles (left). The percentage of intratumoral T_{regs} ($CD4^+Foxp3^+CD25^+$) was quantified by flow cytometry 3 days post-dose (right). Lines or bars and error denote the mean ± standard error of the mean.

Domvanalimab Does Not Induce Fcγ Receptor-mediated Signaling or Promote NK-Mediated ADCC *in vitro*

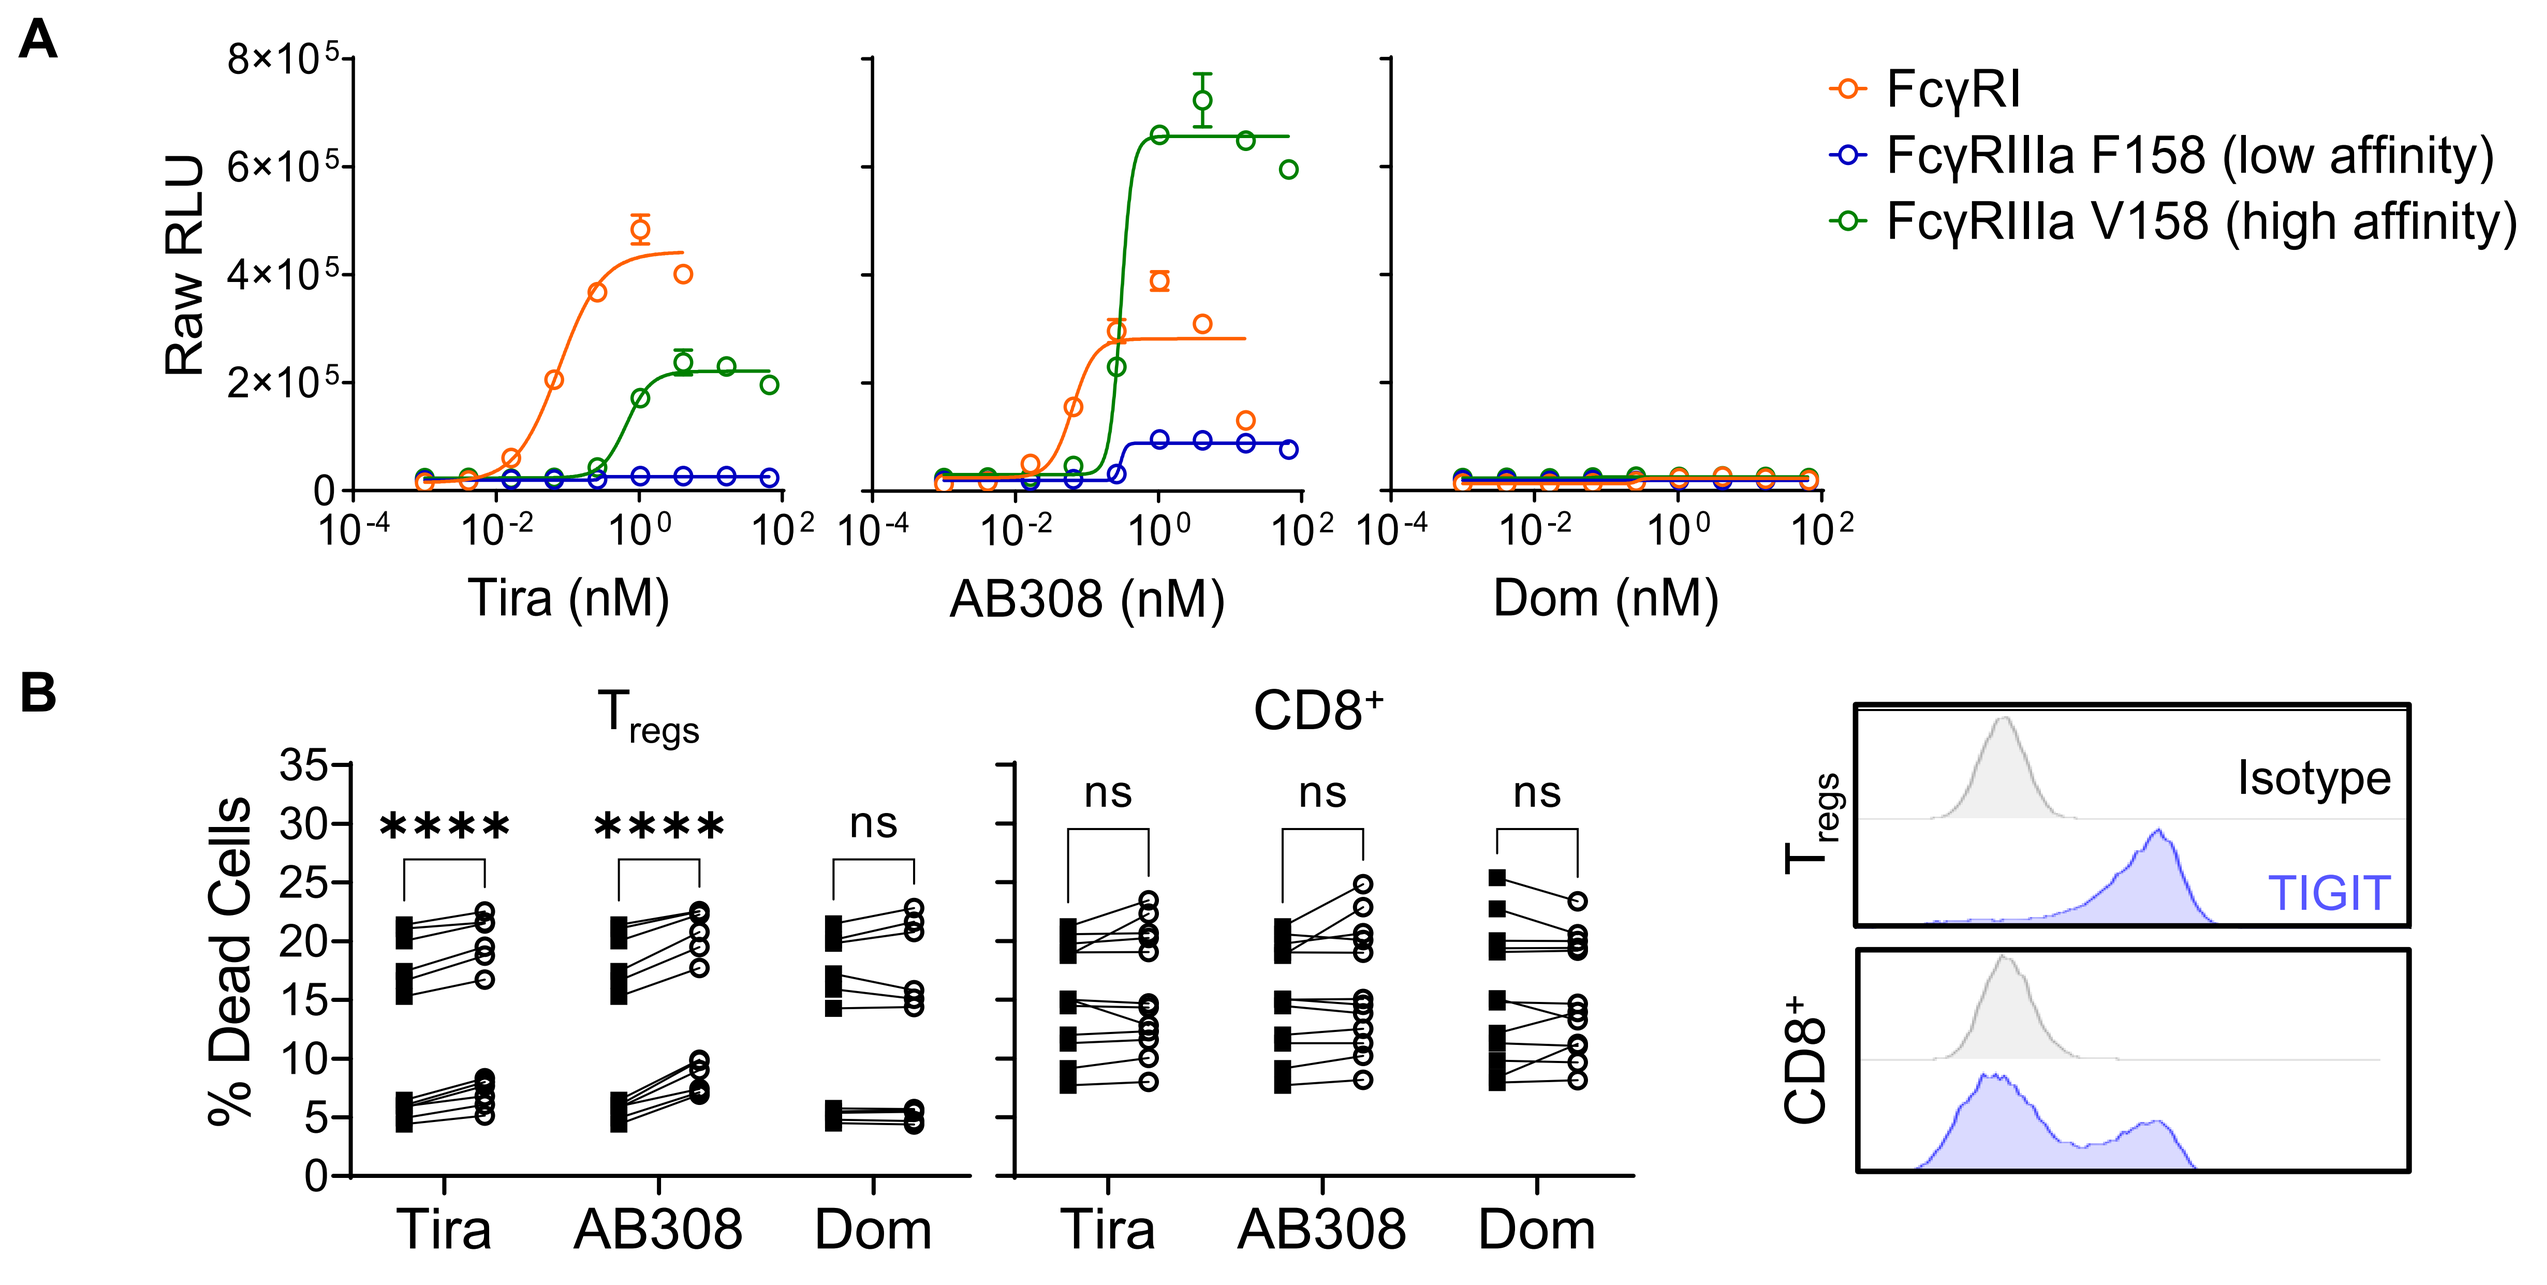
A
Raw RLU
Tira (nM)
AB308 (nM)
Dom (nM)
FcγRI
FcγRIIIa F158 (low affinity)
FcγRIIIa V158 (high affinity)
B
Tregs
CD8+
% Dead Cells
Tira
AB308
Dom
Isotype
TIGIT

Figure 4. Characterization of anti-TIGIT antibody Fc functionality *in vitro*. (A) Antibody-dependent cell-mediated cytotoxicity (ADCC) Luciferase Reporter Assay Kit (Promega). Lines and error denote the mean ± standard deviation (SD). **(B)** NK-mediated ADCC against T_{reg} or $CD8^+$ target cells isolated from peripheral blood mononuclear cell suspensions with representative histograms showing TIGIT expression. Paired symbols indicate treatment with human IgG isotype (■) or anti-TIGIT antibodies (○) tiragolumab (tira), AB308, or domvanalimab (dom) for each NK-T cell donor pair. Tira was produced by Arcus using sequences disclosed in the WHO Drug Information proposed INN publication[3].

Domvanalimab Does Not Deplete Peripheral Immune Cells in Patients

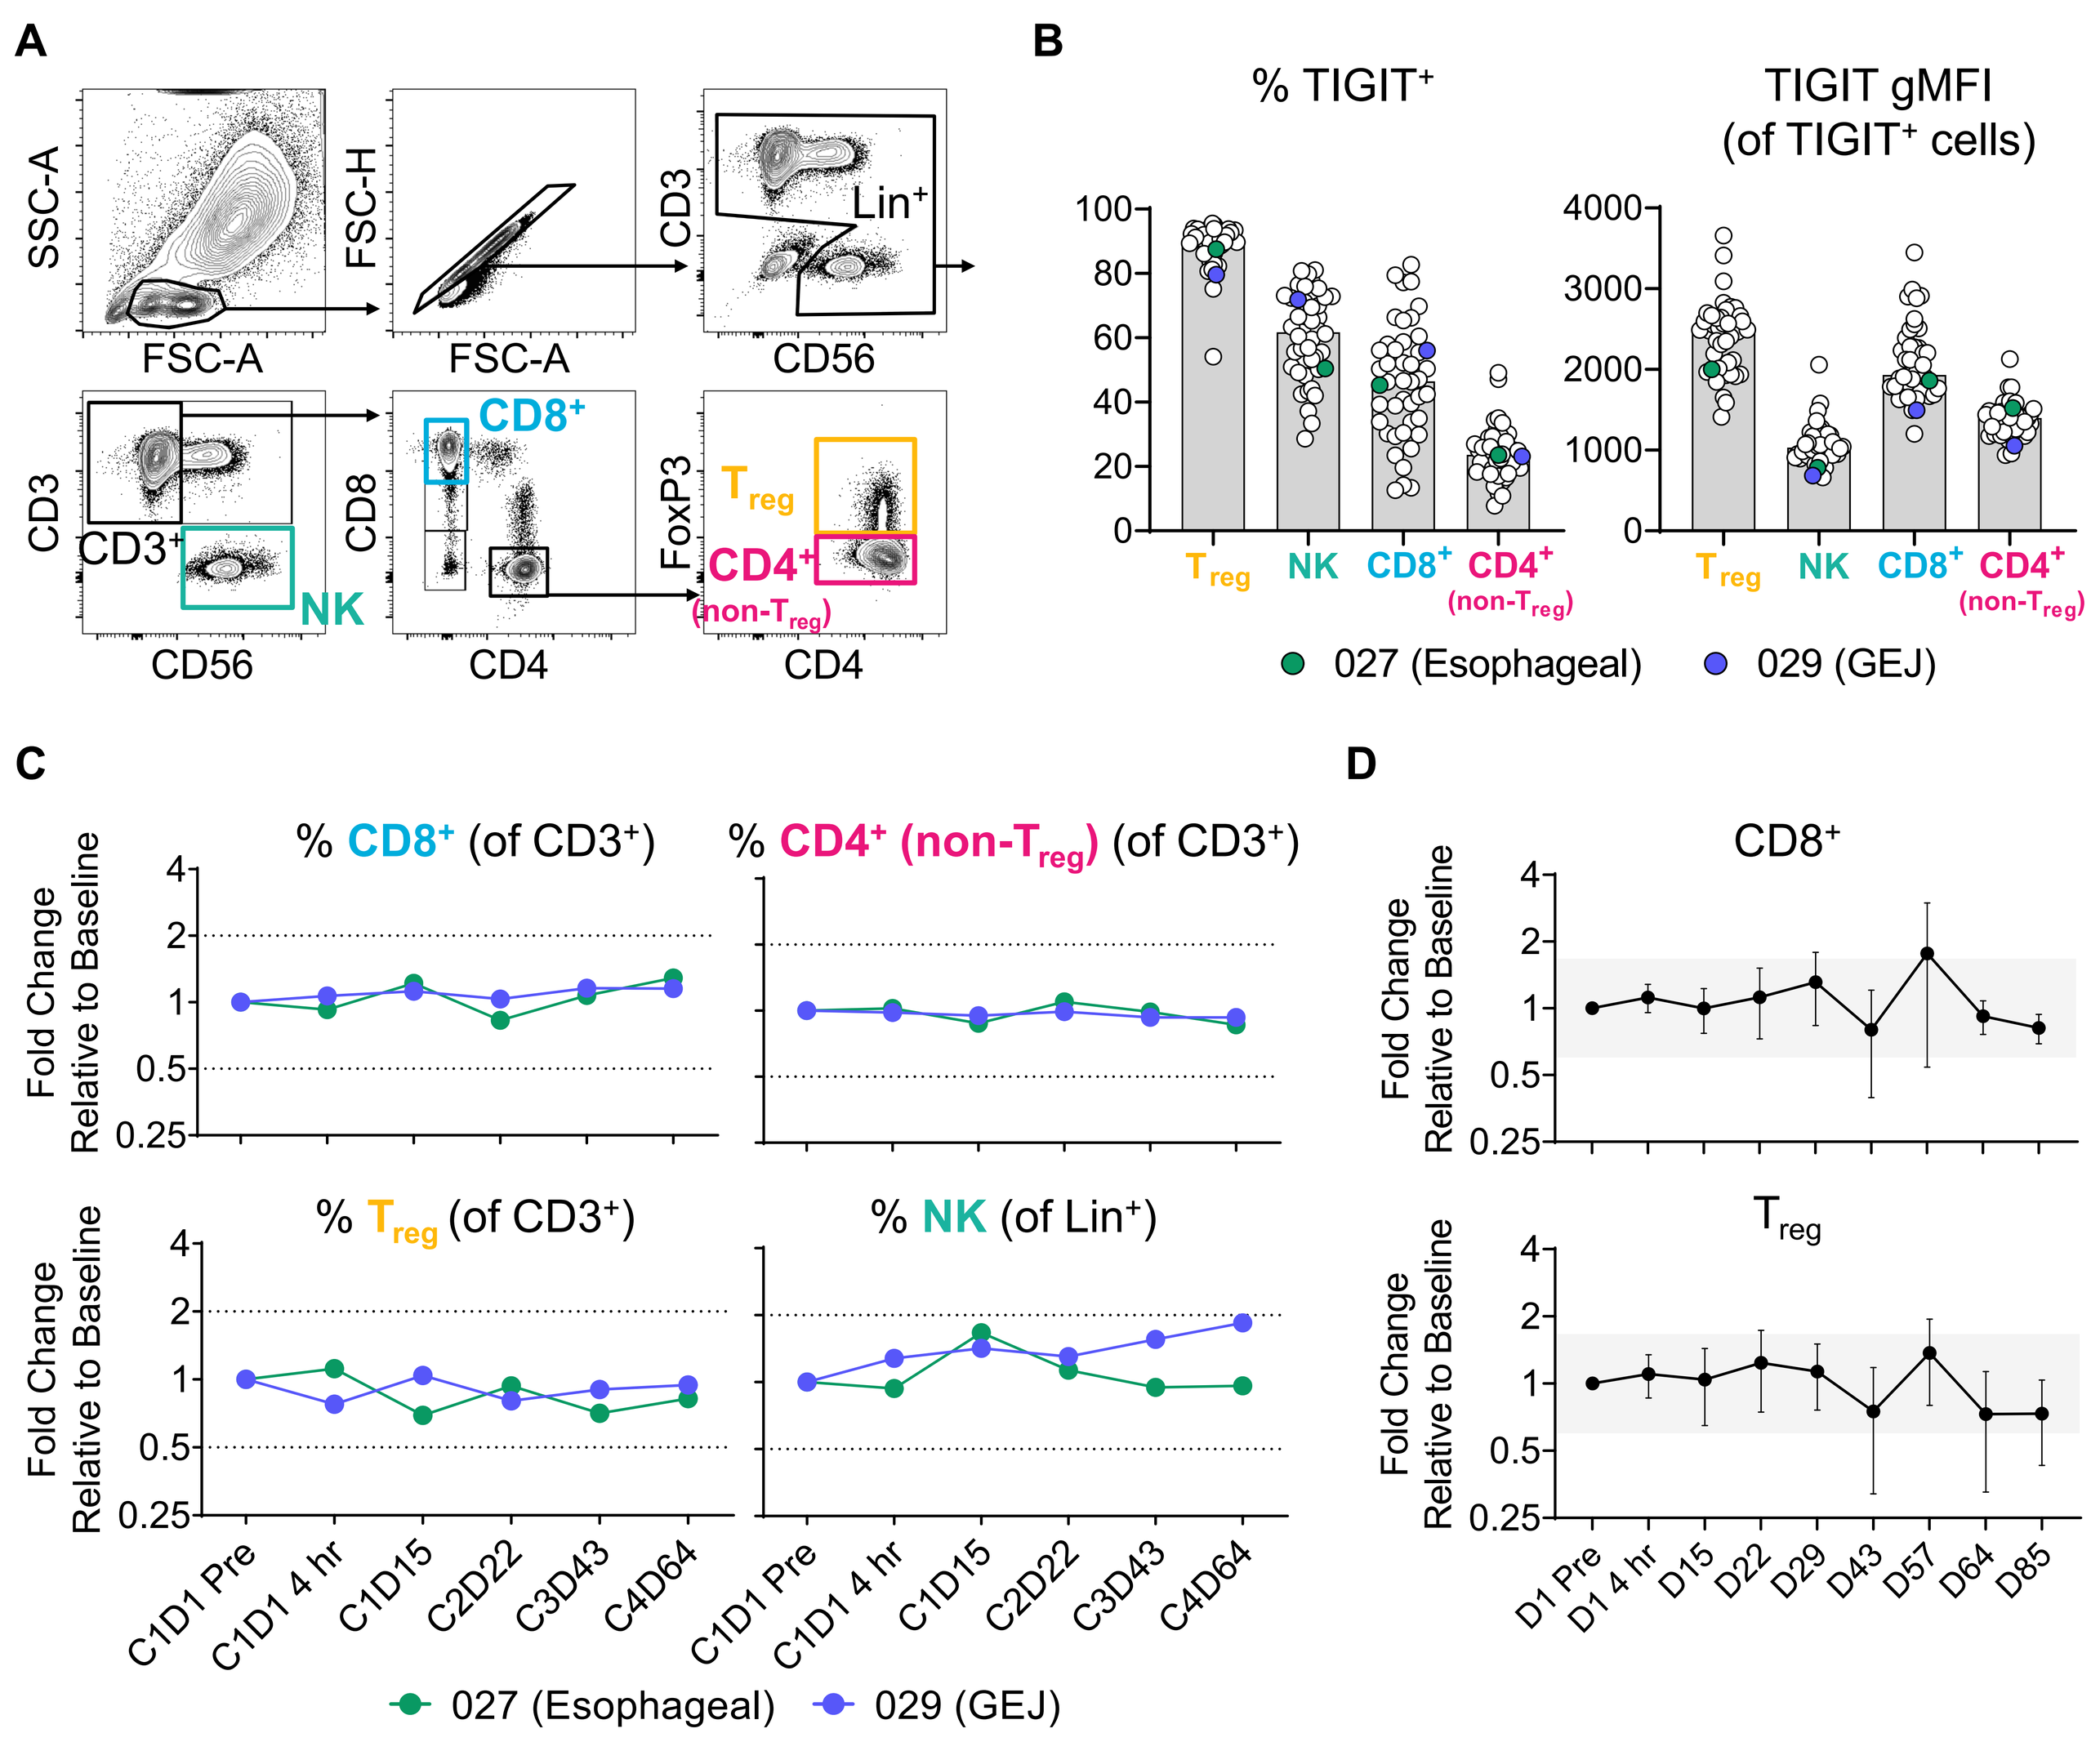
A
SSC-A
FSC-A
FSC-H
CD3
CD56
Lin+
CD3+
NK
CD8+
CD4
FoxP3
Treg
CD4+ (non-Treg)
B
% TIGIT+
TIGIT gMFI
(of TIGIT+ cells)
027 (Esophageal)
029 (GEJ)
C
% CD8+ (of CD3+)
% CD4+ (non-Treg) (of CD3+)
% Treg (of CD3+)
% NK (of Lin+)
Fold Change Relative to Baseline
C1D1 Pre
C1D1 4 hr
C1D15
C2D22
C3D43
C4D64
D
CD8+
Treg
D1 Pre
D1 4 hr
D15
D22
D29
D43
D57
D64
D85

Figure 5. Peripheral lymphocyte frequencies and absolute cell counts were assessed in whole blood (WB) samples from Phase 1 patients (NCT03628677). (A) Representative gating strategy. Lin, lineage. **(B)** Population and per-cell TIGIT expression levels at baseline. Bars denote median and each symbol an individual subject. gMFI, geometric mean fluorescence intensity. **(C)** Fold change in peripheral $CD8^+$, T_{reg}, $CD4^+$ (non-T_{reg}), and NK cell frequencies for patient 027 (esophageal cancer) and 029 (gastroesophageal junction cancer, GEJ) treated with 10 mg/kg domvanalimab (dom) Q3W + 360 mg zimberelimab (zim) Q3W. Dotted lines indicate two-fold change, and the x-axis indicates cycle ("C") and study day ("D") for each time point. **(D)** Phase 1 subjects were treated with 15 mg/kg dom Q3W and 960 mg zim Q6W (n=2), 1500 mg dom Q4W and 480 mg zim Q4W (n=5), or 1200 mg dom Q3W and 720 mg zim Q6W (n=4). Peripheral $CD8^+$ ($CD3^+$) and T_{reg} ($CD4^+CD25^+CD127^{lo}$) absolute counts (cells per µL WB) were assessed using TruCount Absolute-Count Tubes (BD) and are displayed as fold change. Grey shading indicates the healthy donor range (n=6). Lines and error denote mean ± SD. Longitudinal changes were not statistically significant by log transformed parametric repeated measures analysis.

Immune-related Adverse Events (irAEs) for the Combination of Domvanalimab + Zimberelimab

Phase 1 (n = 56)			
	dom + zim n (%)		dom + zim n (%)
Hypothyroidism	5 (8.9%)	Pneumonitis	2 (3.6%)
Pruritus	**4 (7.1%)**	Arthritis	2 (3.6%)
Rash	**4 (7.1%)**	Hyperthyroidism	2 (3.6%)
Maculopapular rash	**3 (5.4%)**	Psoriasis	1 (1.8%)
Infusion-related reaction	**3 (5.4%)**	Swelling face	1 (1.8%)
Immune-mediated hepatitis	2 (3.6%)		

Table 1. irAEs in the ongoing Phase 1 trial of domvanalimab (NCT03628677) as of 01Apr2022. Dom doses equivalent to 700-1400 mg on a Q2 to Q4 week schedule in combination with zim. All events are Grade 1 or 2. Regimens containing Fc-enabled anti-TIGIT monoclonal antibodies, including those reported to deplete peripheral T_{regs}[4,5,6], have reported incidences in the following ranges: pruritus (~20-38%), rash (~21-40%), maculopapular rash (~0-9%), infusion-related reaction (~10-31%)[4,5,7,8].

Case Studies from Phase 1 Patients Exhibiting Partial Responses in the Absence of Peripheral T_{reg} Depletion

A Case Study #1: Phase 1 Subject 027 (68 yo male)

- Stage IV esophageal adenocarcinoma; PD-L1 (CPS) ~2%
- Prior treatment: (1) FOLFOX; (2) Carboplatin/Paclitaxel; (3) Pembrolizumab
- Study regimen: 10 mg/kg domvanalimab Q3W + 360 mg zimberelimab Q3W

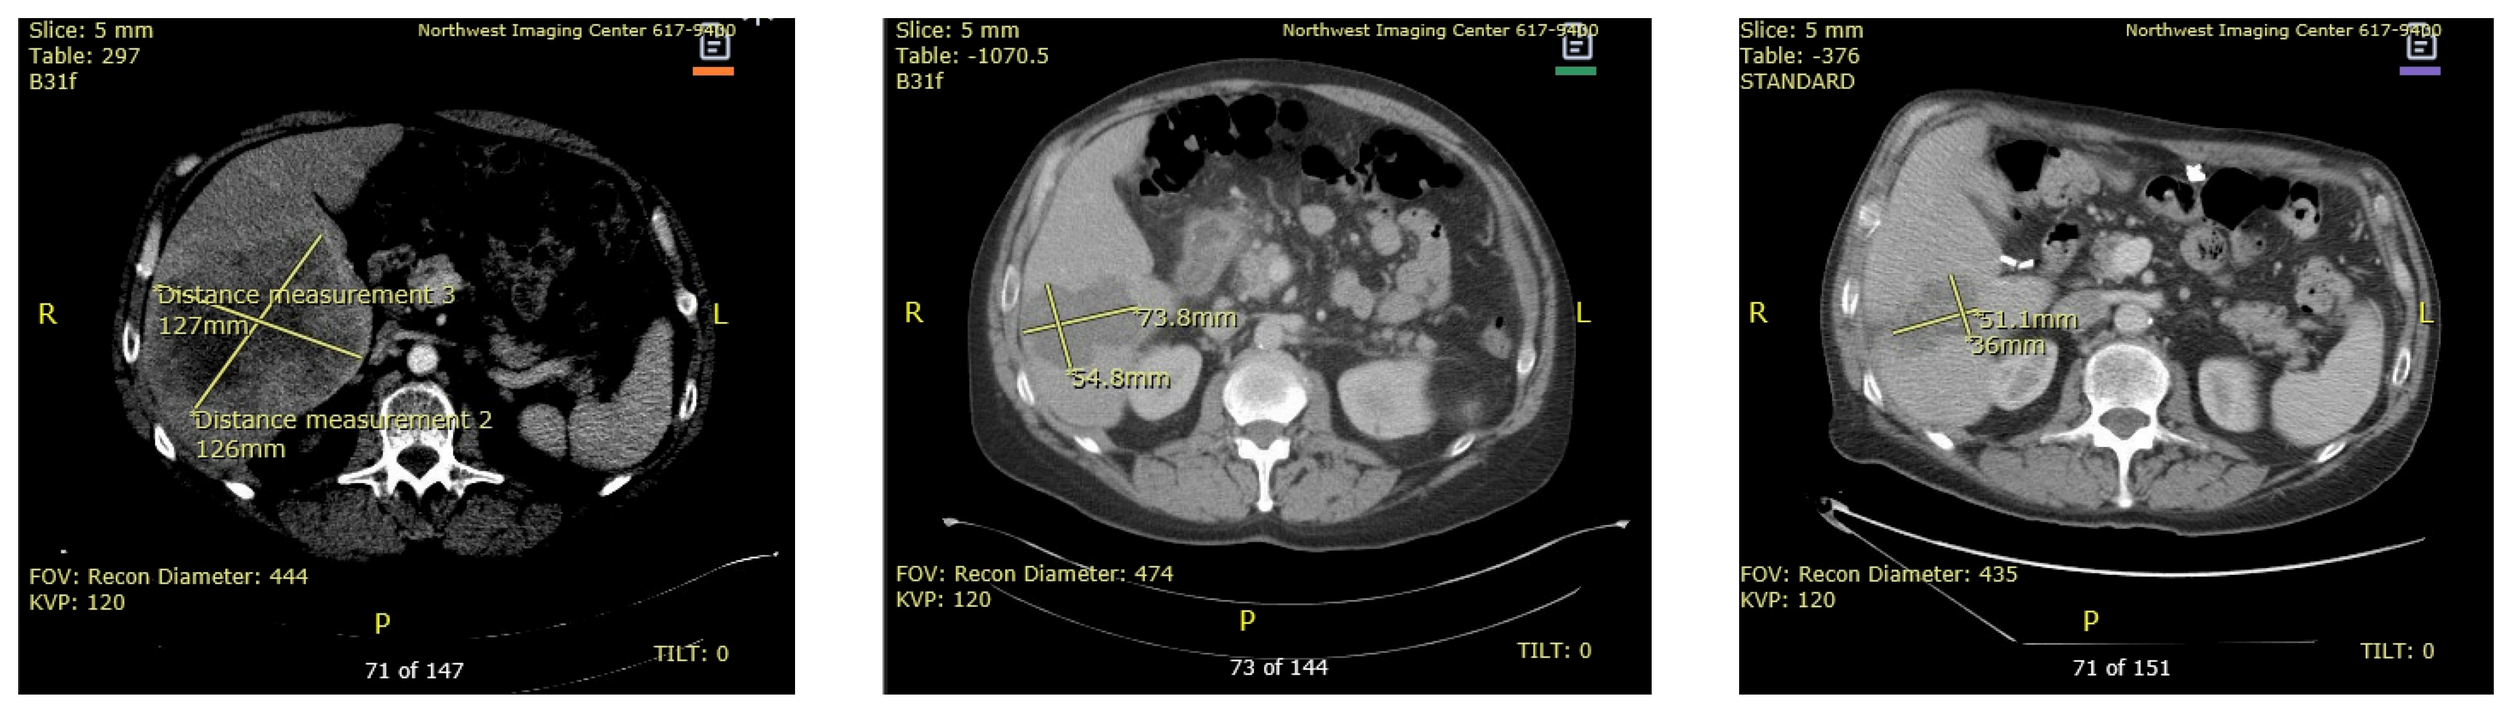

Baseline scan
Target lesion #1: **127 mm long axis**

Post-Cycle 6 scan
Target lesion #1: **76 mm long axis**

Post-Cycle 30 scan
Target lesion #1: **62 mm long axis**

B Case Study #2: Phase 1 Subject 029 (69 yo male)

- Stage IVb gastroesophageal; PD-L1 status unknown
- Prior treatment: FOLFOX
- Study regimen: 10 mg/kg domvanalimab Q3W + 360 mg zimberelimab Q3W

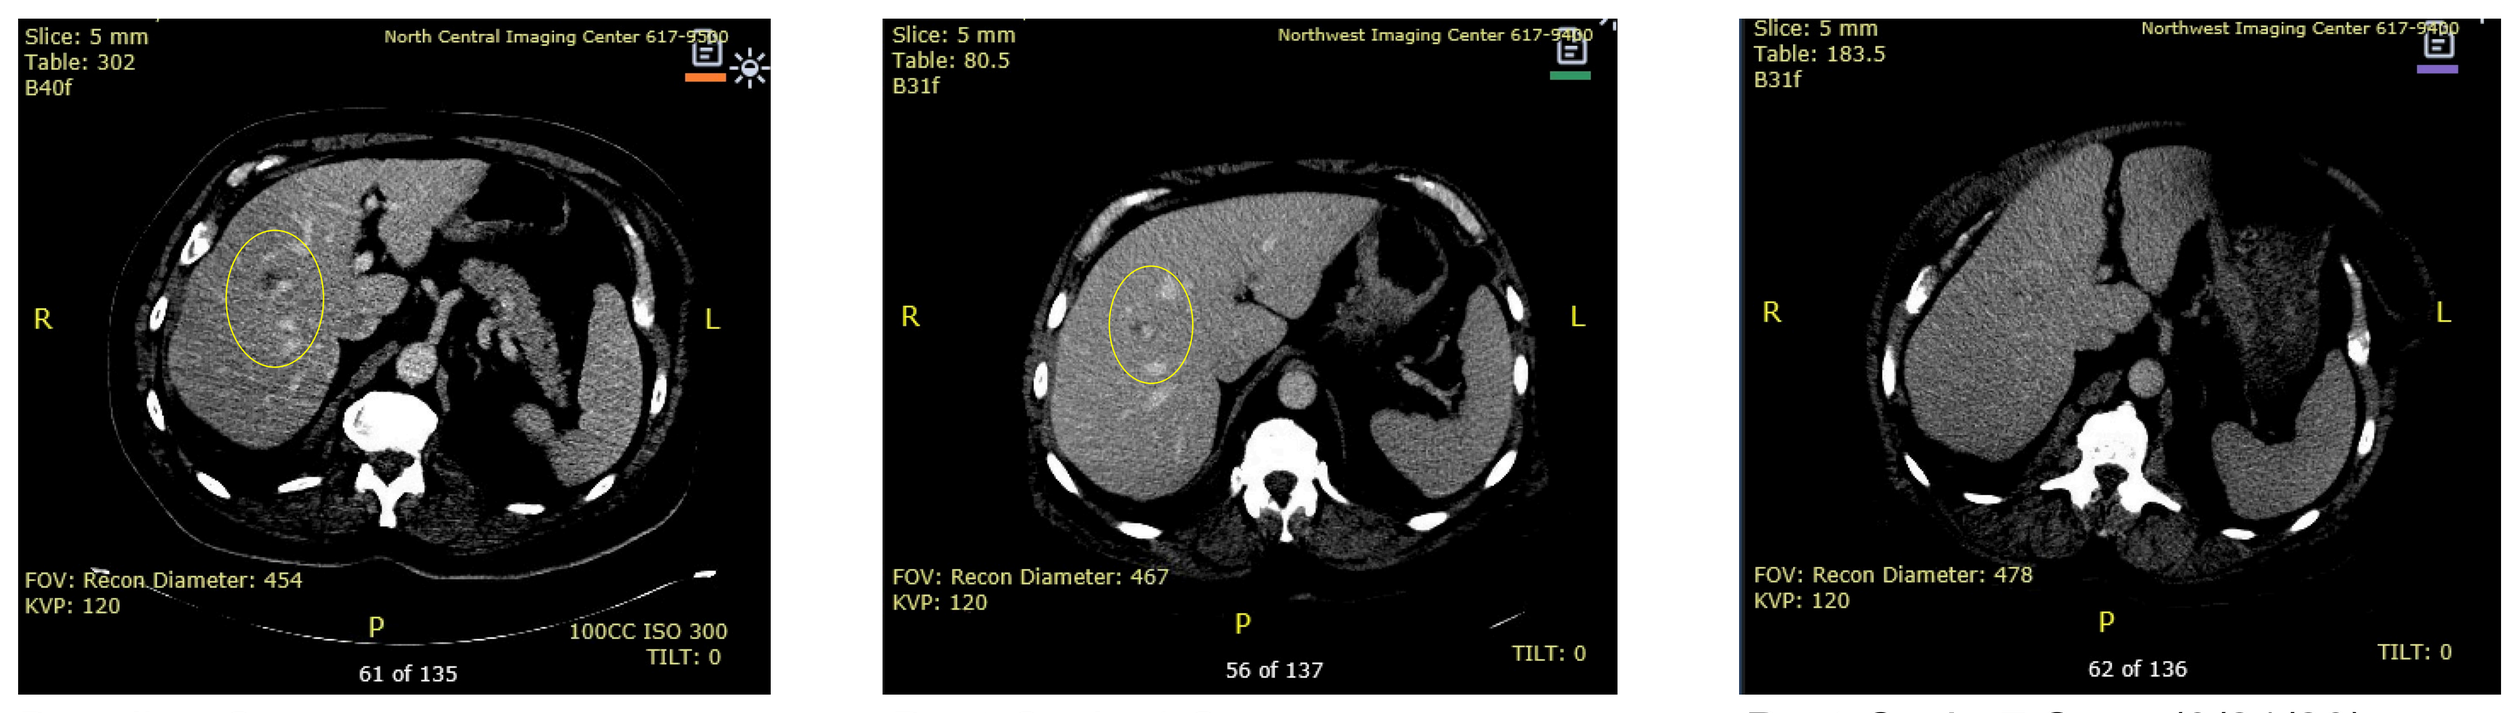

Baseline Scan
Target lesion #2: **14 mm long axis**

Post-Cycle 4 Scan
Target lesion #2: **8 mm long axis**

Post-Cycle 7 Scan (9/21/20)
Target lesion #2: **0 mm long axis**

C

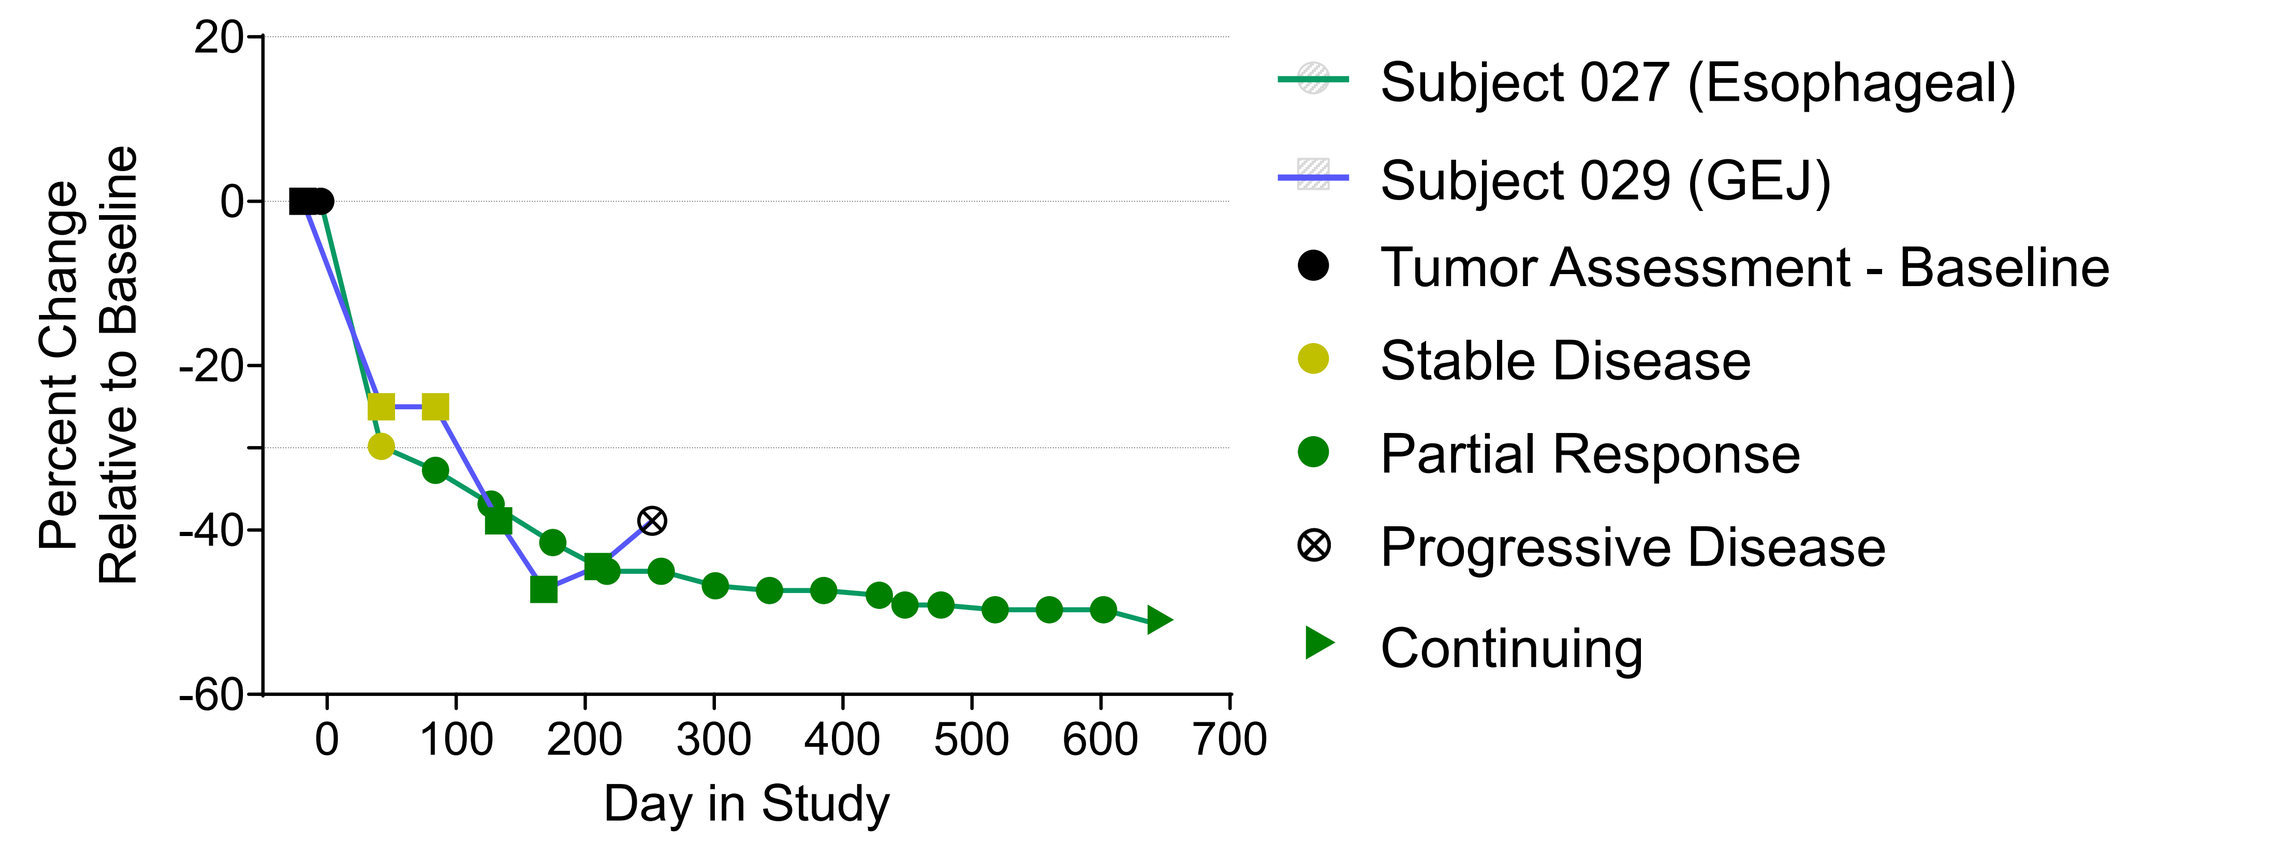
Percent Change Relative to Baseline
Day in Study
Subject 027 (Esophageal)
Subject 029 (GEJ)
Tumor Assessment - Baseline
Stable Disease
Partial Response
Progressive Disease
Continuing

Figure 6. Clinical evaluation of domvanalimab + zimberelimab in AB154CSP0001 (NCT03628677). The primary objective of the study was to evaluate safety and tolerability of dom + zim in a heterogenous patient population representing multiple advanced malignancies. **(A and B)** Scans demonstrating a decrease in target lesion size in two partial responders in AB154CSP0001. Scans show one of two target lesions with each patient having lesions not documented in the scans shown here. **(C)** Percent change from baseline for measurable target lesions per RECIST 1.1 over time. +20% = progressive disease; -30% = partial response. Of those patients for whom peripheral blood results are available, an additional melanoma patient with prior progression on ipilimumab + nivolumab achieved a clinical response without a decrease in peripheral T_{regs}.

Summary and References

- PD-1, TIGIT and CD226 are co-expressed on tumor-resident and circulating $CD8^+$ T cells that represent stem-like populations (akin to reported cellular targets of anti-PDx)[2] and terminally differentiated dysfunctional populations[2] in gastric/esophageal and lung tumor samples (**Figure 2**).
- In mice, Fc-silent anti-TIGIT antibody enhances tumor control without intratumoral T_{reg} depletion (**Figure 3**) in combination with anti-PD-1.
- Domvanalimab binds human TIGIT[9], blocks the TIGIT-CD155 interaction[9], and does not exhibit Fc-dependent cytotoxicity of $TIGIT^+$ cells *in vitro* (**Figure 4**) or *in vivo* (**Figure 5**).
- Competitor molecules with Fc domain functionality have reported depletion of peripheral lymphocyte populations[4,5,6]. In contrast, domvanalimab does not deplete peripheral lymphocyte populations, including regulatory and $CD8^+$ T cells (**Figure 5**).
- The incidence of pruritis, rash, maculopapular rash and infusion-related reactions for domvanalimab in an ongoing Phase 1 trial in combination with anti-PD-1 (zimberelimab) appear to be lower than Fc-enabled anti-TIGIT monoclonal antibodies (**Table 1**).
- Phase 1 patients unlikely to respond to single-agent anti-PD-1 have had deep partial responses to dom + zim (**Figure 6A**) without evidence of peripheral lymphocyte depletion (**Figure 5**) supporting continued clinical development of the dom + zim combination.

[1]Kurtulus *et al.* 2019 Immunity DOI: 10.1016/j.immuni.2018.11.014; [2]Dolina *et al.* 2021 Front Immunol DOI:10.3389/fimmu.2021.715234; [3]List 117; Vol 31 No. 2, 2017; [4]Mettu *et al.* 2022 Clin Cancer Res DOI:10.1158/1078-0432.CCR-21-2780 (etigilimab); [5]Van den Mooter *et al.* 2021 Cancer Res DOI:10.1158/1538-7445.AM2021-CT118 (EOS-448). [6]Cuende *et al.* 2022 AACR #LB189 (EOS-448); [7]Niu *et al.* 2022 Ann Oncol DOI:10.1016/j.annonc.2021.11.002 (vibostolimab); [8]Cho *et al.* 2021 Ann Oncol DOI:10.1016/j.annonc.2021.10.217 (tiragolumab); [9]https://arcusbio.com/wp-content/uploads/2020/09/2019.03.25-AACR-2019-AB154-Poster-AA-FINAL.pdf