SITC Annual Meeting
Nov. 8-12, 2022
Abstract 801

The Anti-Tumor Efficacy of Immunogenic Chemotherapy is Enhanced by the Dual $A_{2a}R/A_{2b}R$ Antagonist Etrumadenant, Relieving the Necessity for an Extended Chemotherapy Regimen

Daniel DiRenzo, Dana Piovesan, Sachie Marubayashi, Bindi Patel, Ferdie Soriano, Lisa Seitz, Livia Yamashiro, Janine Kline, Dillon Miles, Ehesan Sharif, Manmohan R Leleti, Matthew J Walters

Arcus Biosciences, Inc.; 3928 Point Eden Way, Hayward, CA 94545 (USA)

INTRODUCTION

- High levels of extracellular adenosine generated in the tumor microenvironment (TME) engage A_{2a} and A_{2b} adenosine receptors on immune cells, resulting in immunosuppression (Figure 1).
- Chemotherapy releases adenosine triphosphate (ATP) into the tumor microenvironment, where it is rapidly converted into adenosine, primarily by the ectoenzymes CD39 and CD73.
- Etrumadenant (etruma) is a selective, small-molecule, dual $A_{2a}R/A_{2b}R$ antagonist. It was specifically designed to potently block adenosine-induced immunosuppression in the TME. We have previously shown that etrumadenant blocks the immunosuppressive effects of adenosine in immune cells and enhances anti-tumor immune responses in mouse syngeneic tumors.
- Herein, we describe the capacity for etruma to drive enhanced tumor control and immune activity in mouse tumor models using immunogenic chemotherapeutic agents with different chemotherapy doses or with reduced dosing regimens.
- Etruma is currently being studied in several Ph2 studies in combination with chemotherapy or other immune-enhancing regimens: NCT04381832, NCT04660812, NCT04262856.

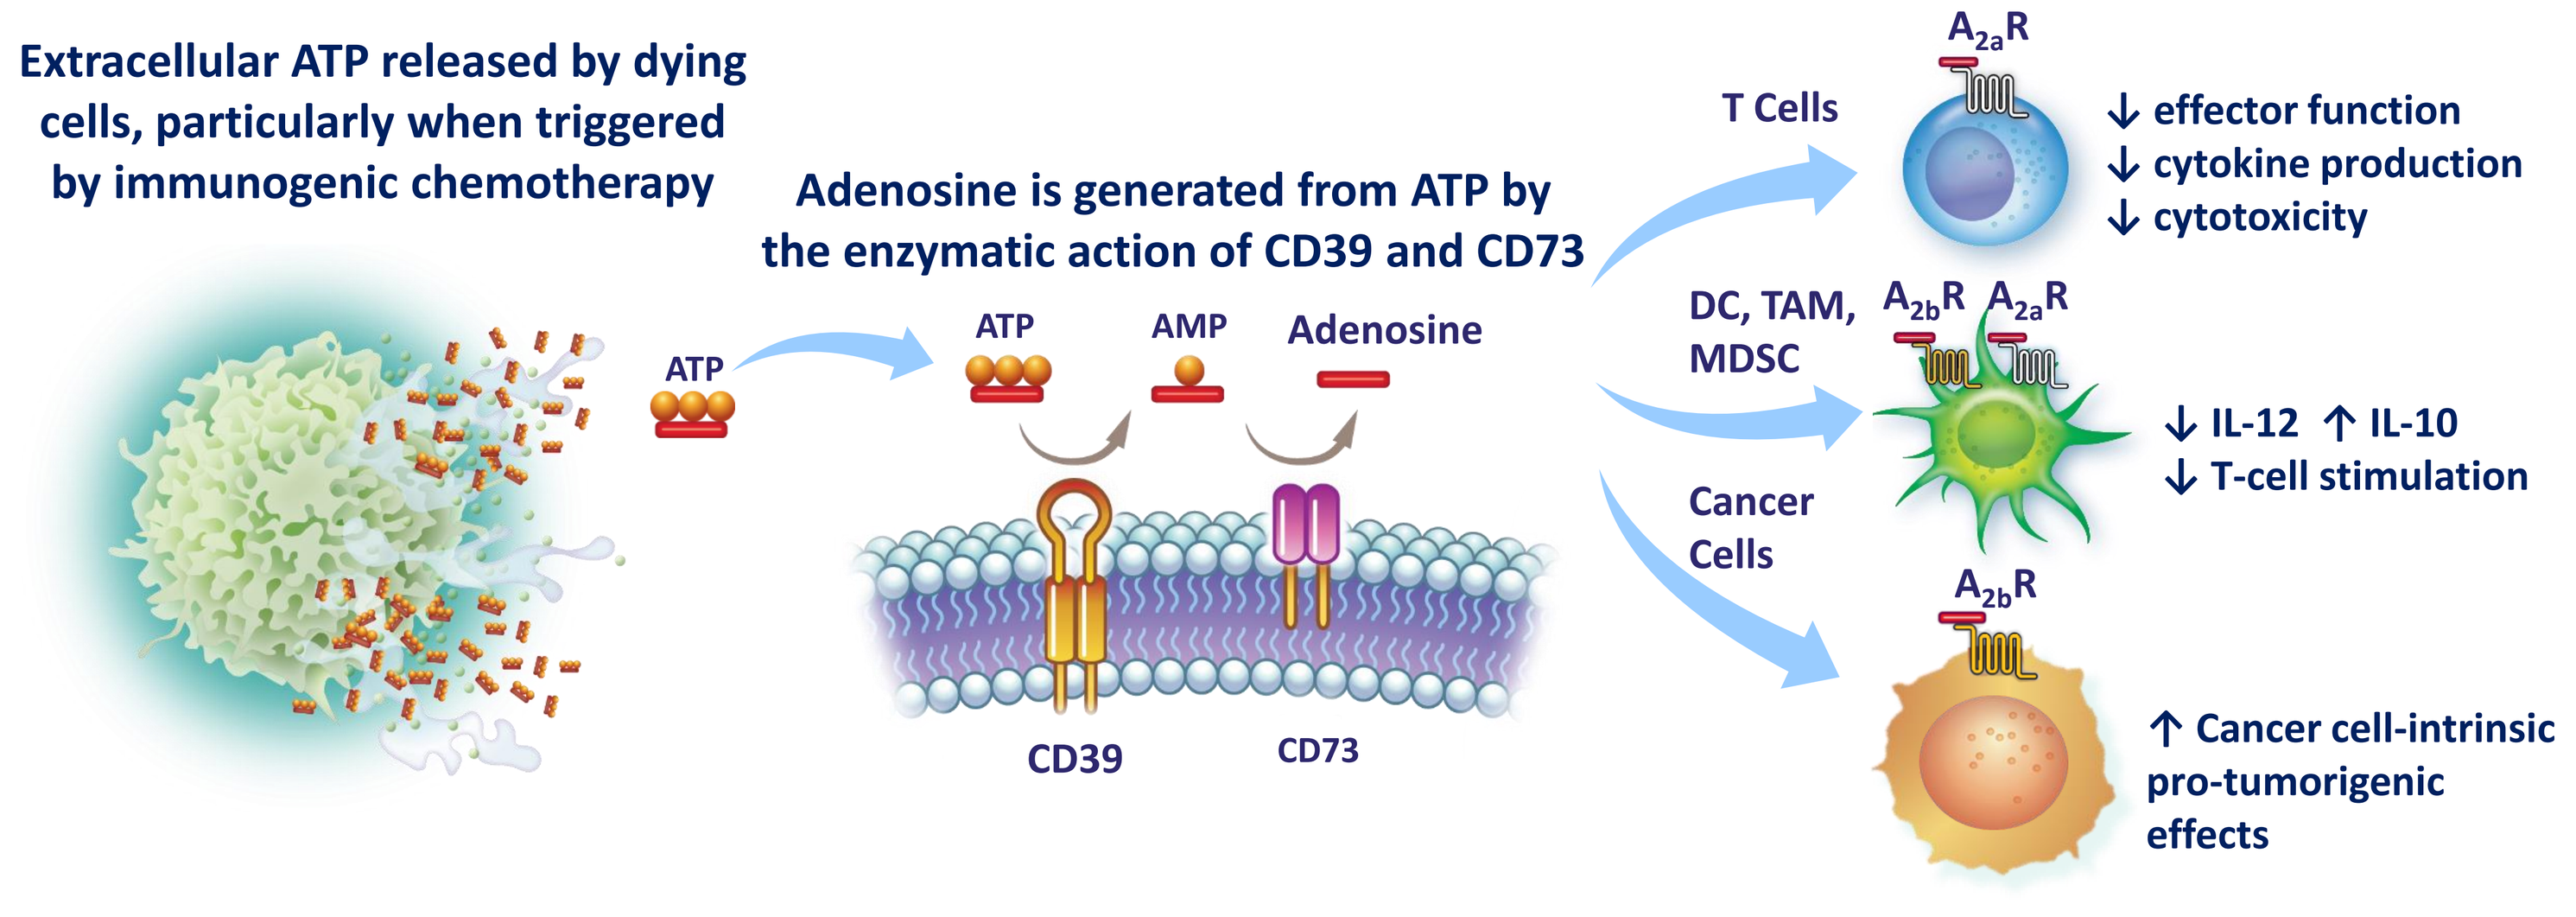
Extracellular ATP released by dying cells, particularly when triggered by immunogenic chemotherapy
Adenosine is generated from ATP by the enzymatic action of CD39 and CD73
ATP
ATP
AMP
Adenosine
CD39
CD73
T Cells
A2aR
↓ effector function
↓ cytokine production
↓ cytotoxicity
DC, TAM, MDSC
A2bR A2aR
↓ IL-12 ↑ IL-10
↓ T-cell stimulation
Cancer Cells
A2bR
↑ Cancer cell-intrinsic pro-tumorigenic effects

Figure 1. Diagram of adenosine production from ATP released into the TME by immunogenic chemotherapy. Hydrolysis of ATP by the ectoenzymes CD39 and CD73 produces adenosine, which exerts immunosuppressive effects by binding to adenosine receptors expressed on immune cells and may promote cell growth via-A_{2b}R-mediated signaling on cancer cells.

METHODS

- **Isolated immune cell experiments:** CD8⁺ T cells and dendritic cells (DC) were isolated from healthy human blood by negative selection. DC were matured with LPS/IFN-γ for 24 hours in the presence of NECA (synthetic adenosine receptor agonist) +/- antagonists.
- **CREB phosphorylation assay:** CREB phosphorylation (pCREB) in CD8⁺ T cells was determined by phospho-flow cytometry using mouse whole blood stimulated with NECA (5 µM) for 20 min at 37°C *ex* vivo. The dose of etruma used in anti-tumor efficacy studies was selected based upon full inhibition of adenosine receptor-mediated pCREB between dose intervals (100 mg/kg, BID)
- **Mouse *in vivo* studies:** C57BL/6 female mice (6-8 weeks old) were injected subcutaneously on the right flank with B16F10 or AT3-OVA cells. 4T1 cells were implanted in the mammary fat pad of 6-8 week old female Balb/c mice. Etruma was administered at 100 mg/kg BID or 200 mg/kg QD at the initiation of the chemotherapy regimen.

Table 1: Potency and Selectivity of Adenosine Receptor Antagonists *

Potency (nM)	Etrumadenant (etruma)	A_{2a}R antagonist[a]	A_{2a}R antagonist[b]
A_{2a}R	1.4	1.5	0.2
A_{2b}R	2.0	123	141

[a] Proprietary A_{2a}R-selective adenosine receptor antagonist
[b] Compound 35 from patent application WO2018178338 (iTeos Therapeutics)
* Potency data generated by measuring cAMP levels in CHO cells stably expressing A_{2a}R or A_{2b}R following stimulation with NECA

RESULTS

Adenosine Signaling Drives Suppression Through A_{2a}R and A_{2b}R in Immune Cells Which is Reversed by Etrumadenant

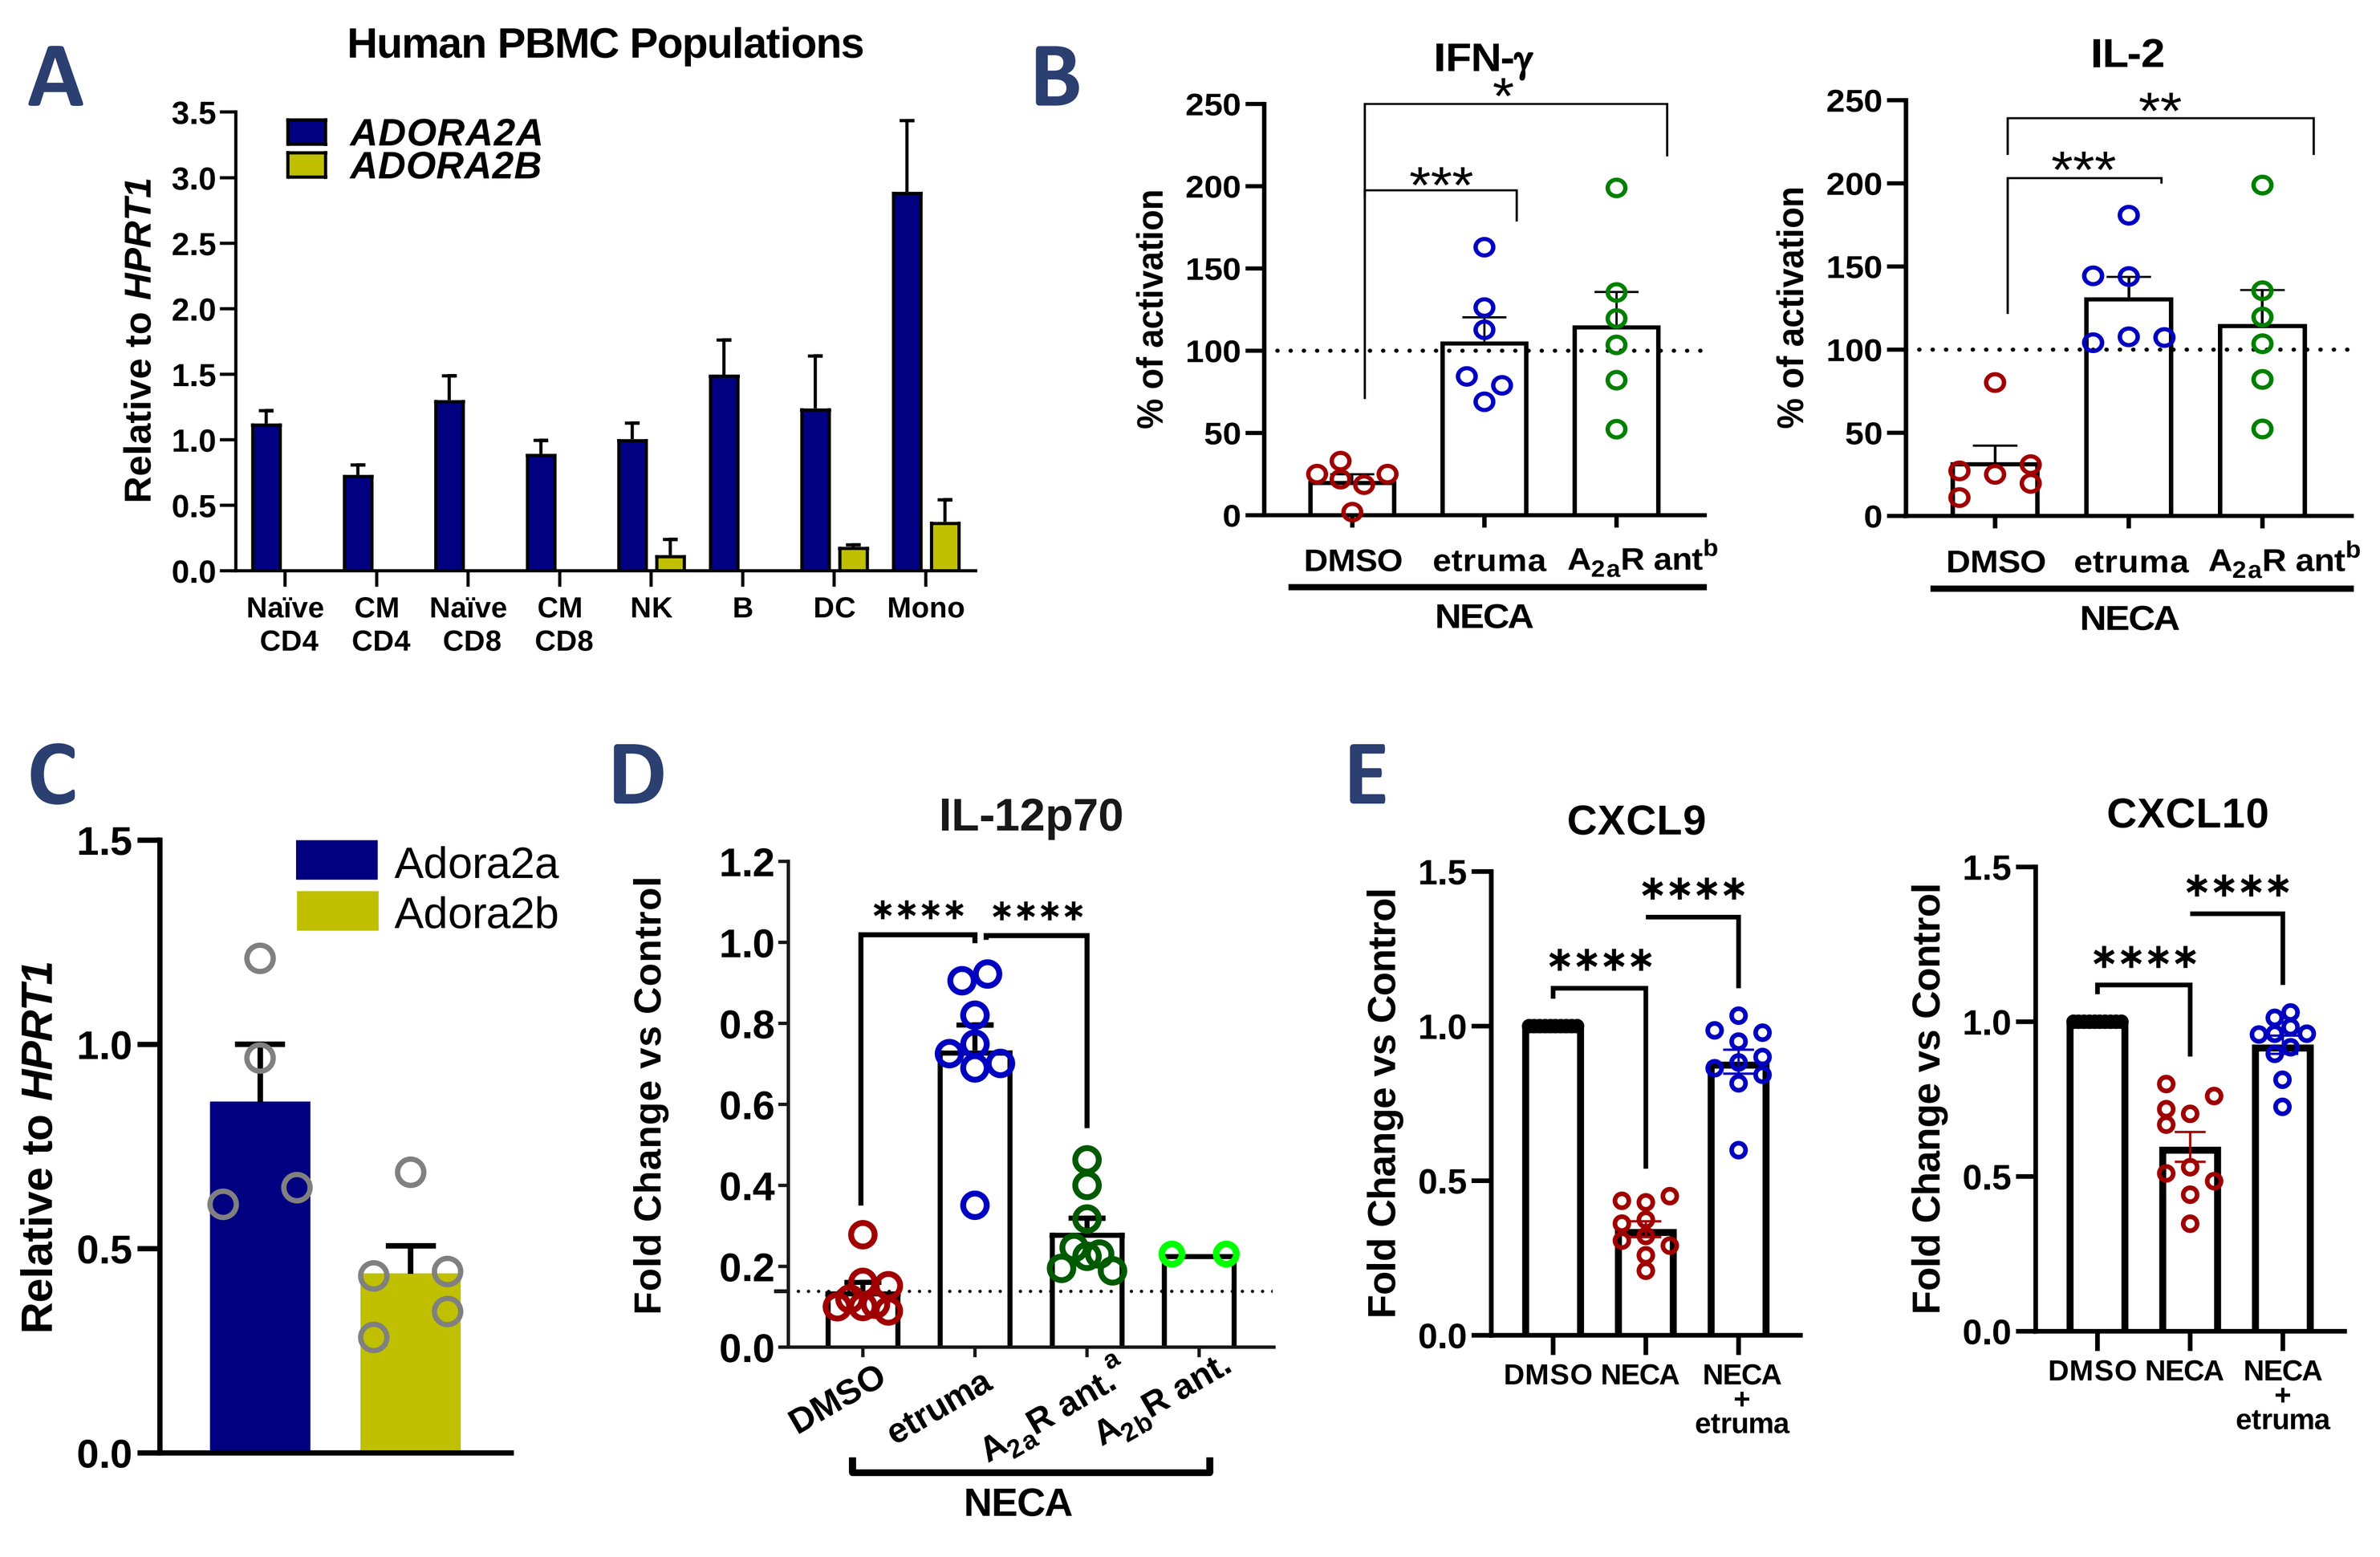
A
Human PBMC Populations
ADORA2A
ADORA2B
Relative to HPRT1
Naïve CD4
CM CD4
Naïve CD8
CM CD8
NK
B
DC
Mono
B
IFN-γ
IL-2
% of activation
DMSO
etruma
A2aR ant[b]
NECA
C
Adora2a
Adora2b
Relative to HPRT1
D
IL-12p70
Fold Change vs Control
DMSO
etruma
A2aR ant.[a]
A2bR ant.
NECA
E
CXCL9
CXCL10
Fold Change vs Control
DMSO
NECA
NECA + etruma

Figure 2. (A) Adenosine receptor gene expression of sorted human immune cells from healthy donors. (B) IFN-γ and IL-2 production from primary human CD8⁺ T cells activated for 72 h in the presence of NECA (5 µM) and adenosine receptor antagonists (1 µM). **These results demonstrate that 1 µM of the adenosine receptor antagonists used in this experiment are capable of fully suppressing adenosine-mediated suppression driven by 5 µM NECA.** (C) Real-time PCR for adenosine receptors ADORA2A and ADORA2B expression in DCs enriched from healthy human blood shows near equal expression of each receptor. (D) IL-12p70 and (E) CXCL9 and CXCL10 suppression by NECA (5 µM) in DCs was rescued by etrumadenant (1 µM) after 24 hours in culture whereas blockade of A_{2a}R or A_{2b}R alone was less effective. *p<0.05, **p<0.01, ***p<0.001, ****p<0.0001. A_{2b}R antagonist: GS-6201 purchased from TOCRIS. See Table 1 for details on A_{2a}R ant a and b.

A_{2b}R is Highly Expressed on Cancer Cell Lines and Drives Gene Expression Changes Which are Blocked by Etrumadenant

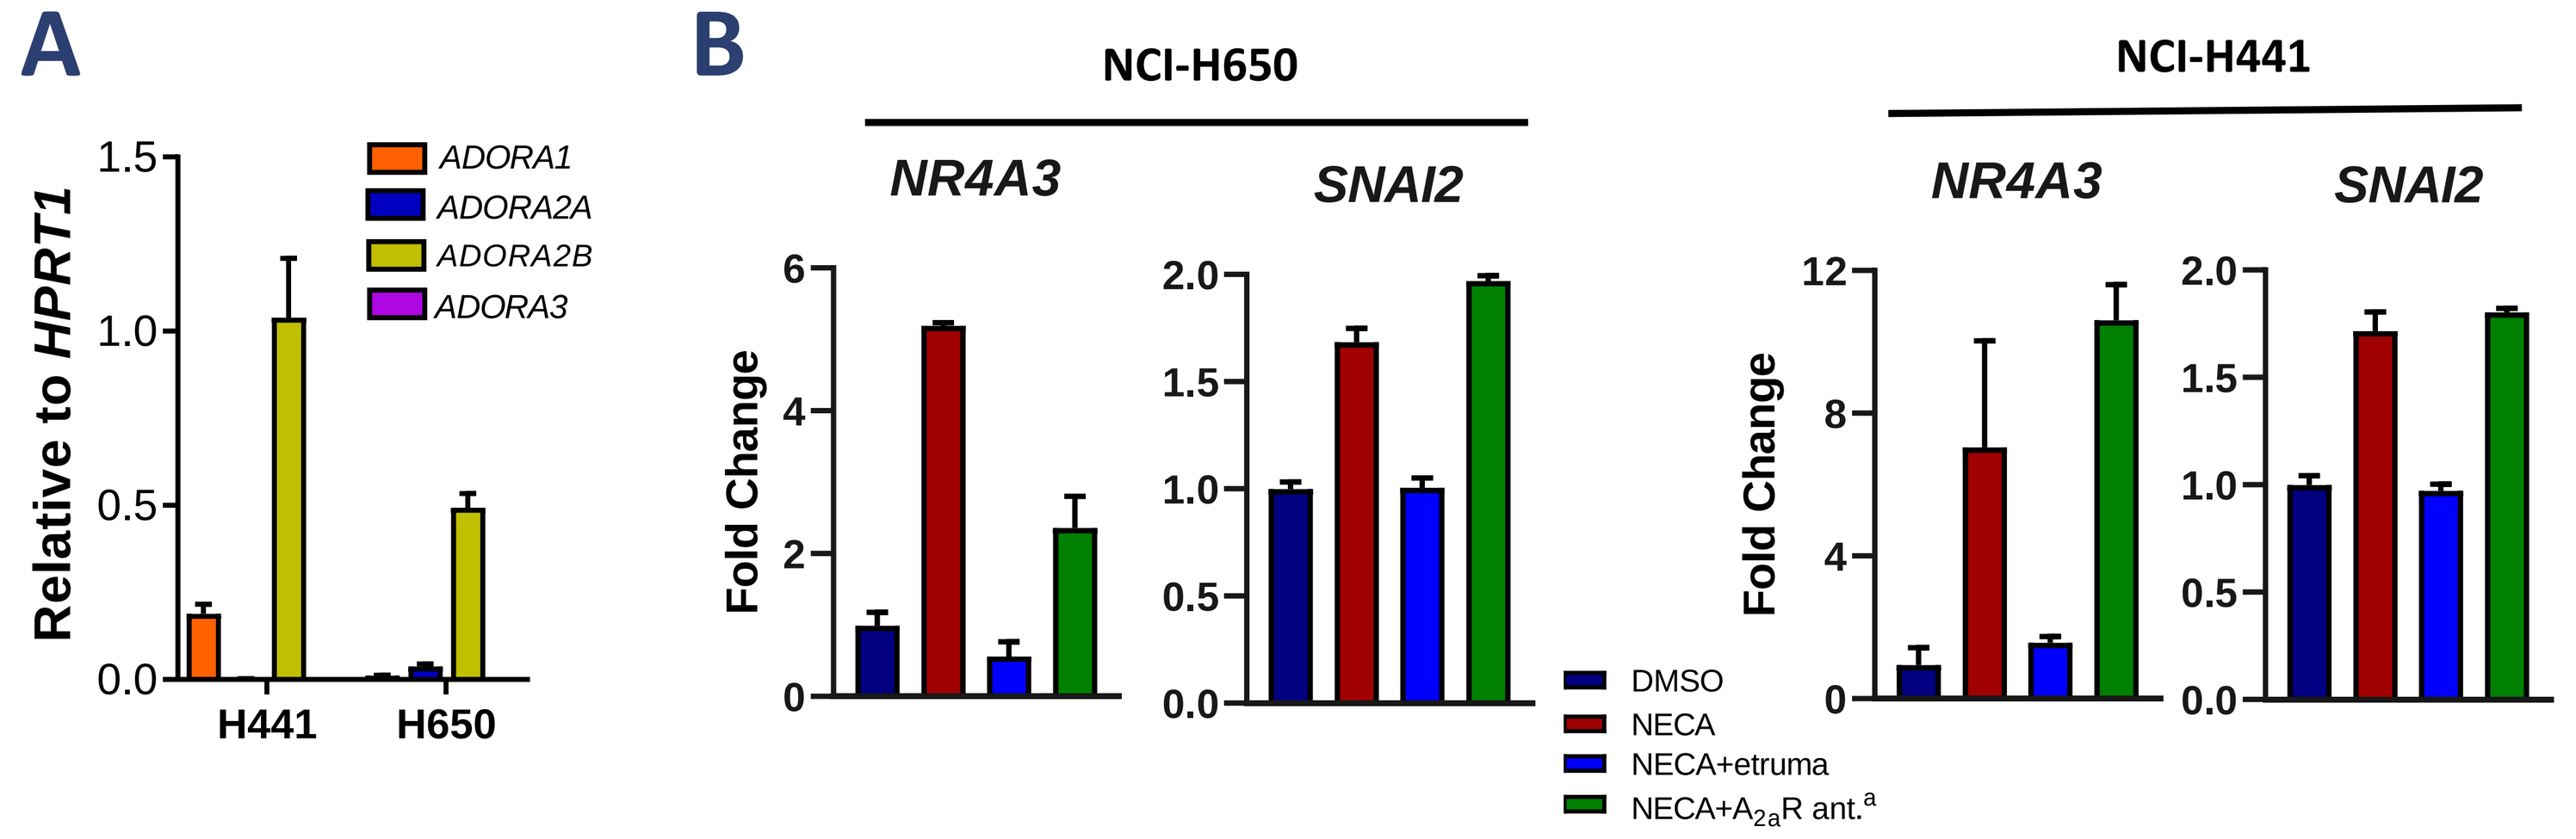
A
ADORA1
ADORA2A
ADORA2B
ADORA3
Relative to HPRT1
H441
H650
B
NCI-H650
NCI-H441
NR4A3
SNAI2
Fold Change
DMSO
NECA
NECA+etruma
NECA+A2aR ant.[a]

Figure 3. (A) Real-time PCR for adenosine receptor expression from NSCLC cell lines showing high expression of A_{2b}R and substantially lower levels of expression for all other adenosine receptors. (B) Real-time PCR from NSCLC cell lines that were stimulated with NECA (5 µM) in the presence of dual vs A_{2a}R-selective antagonists (1 µM, see table 1 for details) showing that etruma is capable of blocking NECA-stimulated gene expression changes driven by A_{2b}R.

Etrumadenant Inhibits Peripheral CREB Phosphorylation and Suppresses B16F10 Tumor Growth

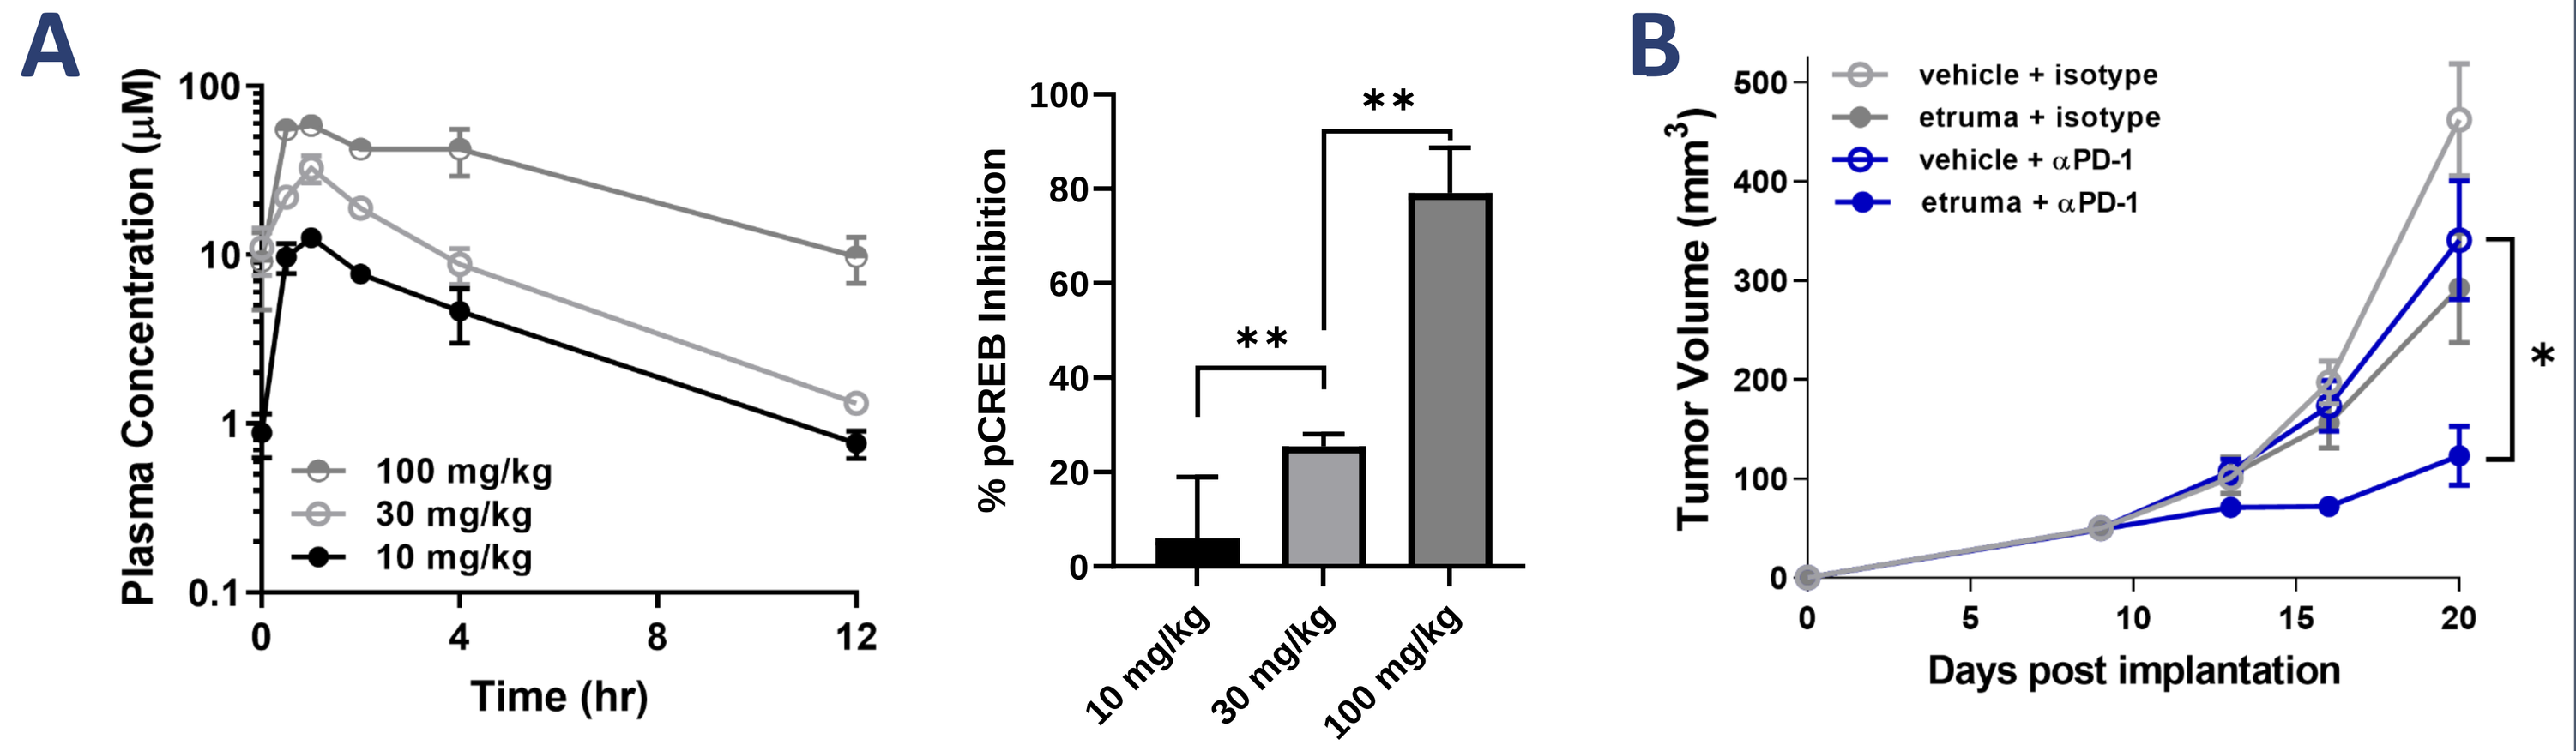
A
Plasma Concentration (µM)
100 mg/kg
30 mg/kg
10 mg/kg
Time (hr)
% pCREB Inhibition
10 mg/kg
30 mg/kg
100 mg/kg
B
Tumor Volume (mm³)
vehicle + isotype
etruma + isotype
vehicle + αPD-1
etruma + αPD-1
Days post implantation

Figure 4. (A) Pharmacokinetic profile of etrumadenant administered orally to C57Bl/6 mice (left panel). NECA was utilized to stimulate pCREB on T cells in whole blood from mice 12 hours after etrumadenant dosing (right panel). (B) Mice bearing B16F10 tumors were treated with α-PD-1 antibodies (Q3Dx4) beginning at a tumor volume of 50 mm³ concurrently with etrumadenant (100 mg/kg PO, BID) or vehicle control. *p<0.05, **p<0.01.

Etrumadenant Combines with Platinum-Based Chemotherapy to Enhance AT3-OVA Tumor Control and Immune Infiltration

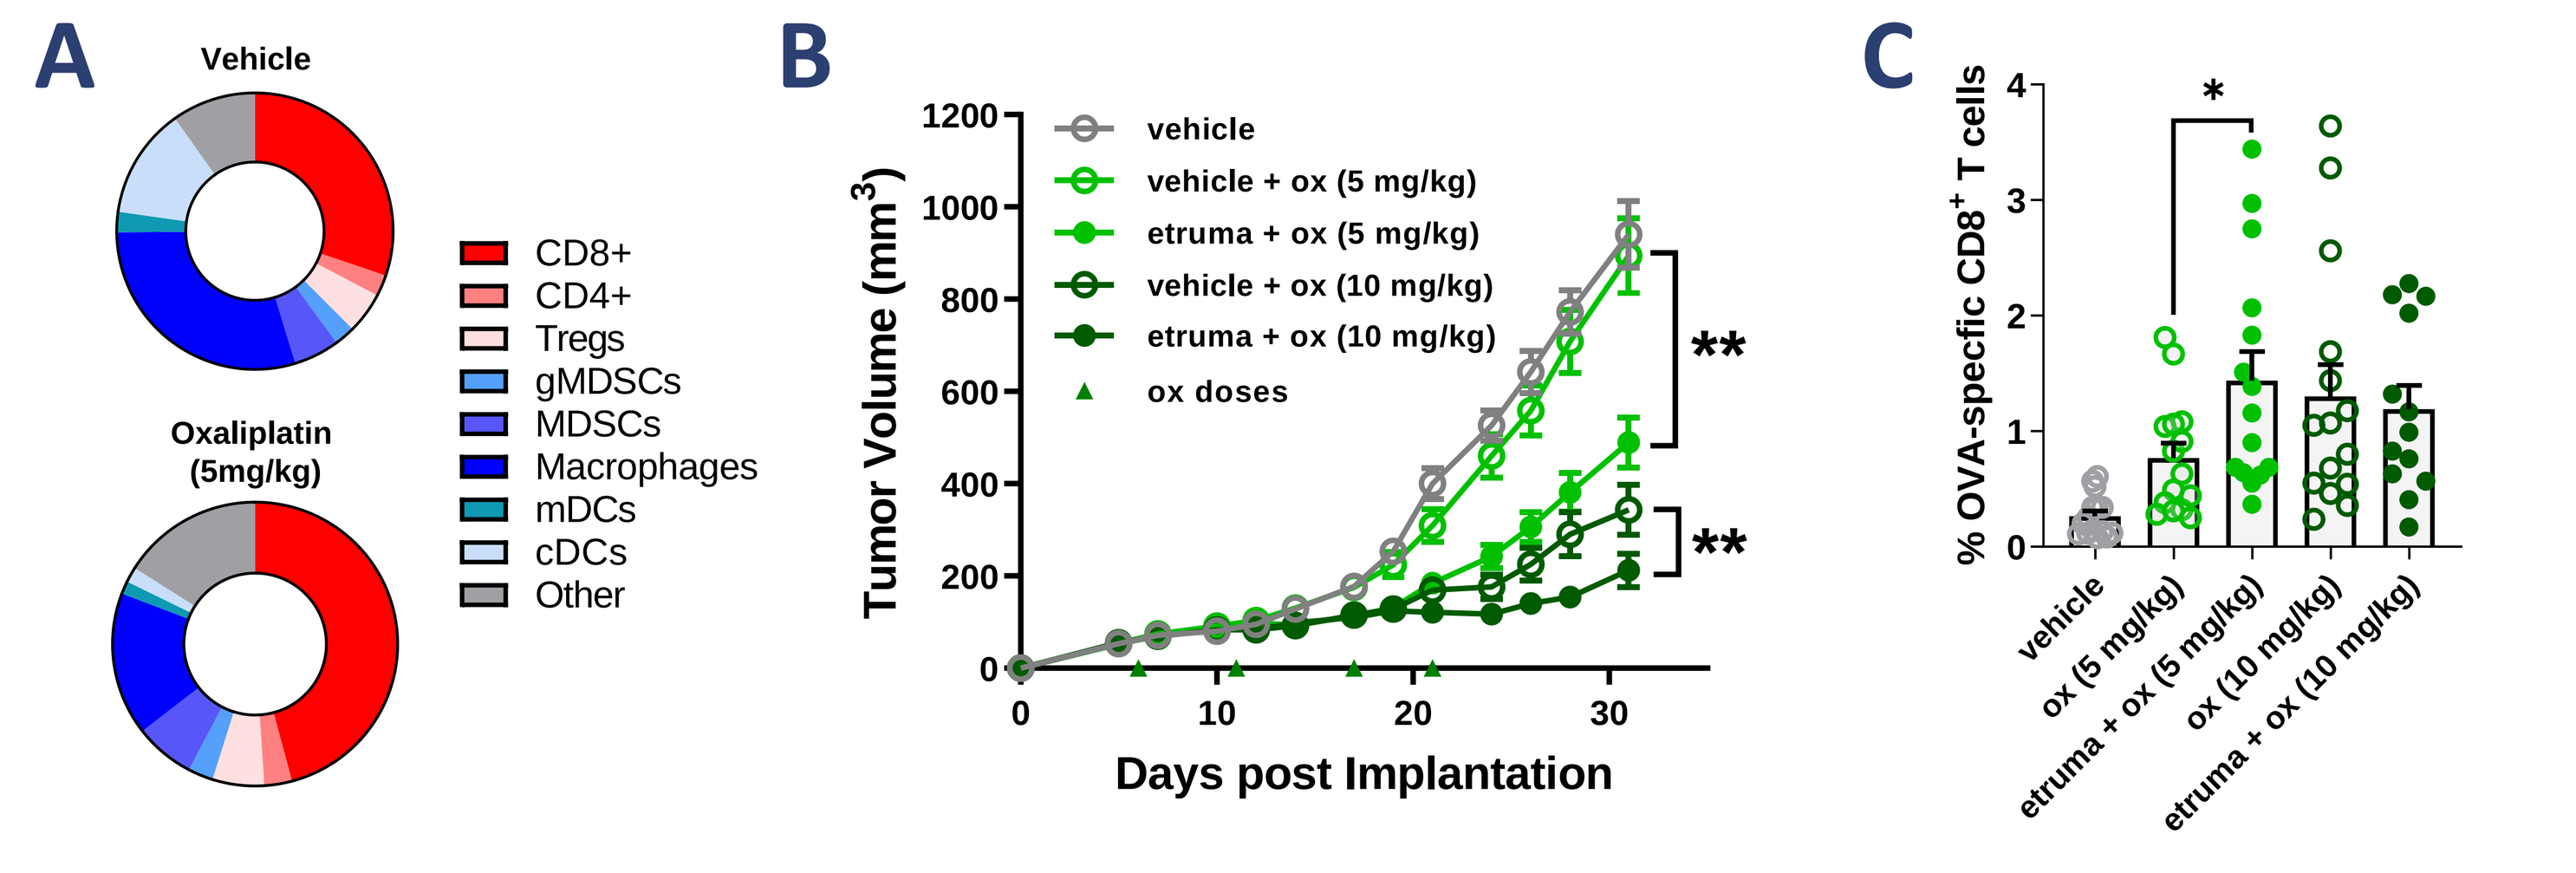
A
Vehicle
Oxaliplatin (5mg/kg)
CD8+
CD4+
Tregs
gMDSCs
MDSCs
Macrophages
mDCs
cDCs
Other
B
Tumor Volume (mm³)
vehicle
vehicle + ox (5 mg/kg)
etruma + ox (5 mg/kg)
vehicle + ox (10 mg/kg)
etruma + ox (10 mg/kg)
ox doses
Days post Implantation
C
% OVA-specfic CD8⁺ T cells
vehicle
ox (5 mg/kg)
etruma + ox (5 mg/kg)
ox (10 mg/kg)
etruma + ox (10 mg/kg)

Figure 5. (A) Profiling of tumor-infiltrating immune cells in the AT3-OVA tumor in vehicle vs oxaliplatin-treated tumors. (B) Tumor volume from AT3-OVA model. Dosing started at 50 mm³, etrumadenant (100 mg/kg, PO, BID) and oxaliplatin dosing Q4Dx4, as indicated. (C) Flow cytometric analysis of OVA-specific tumor infiltrating CD8⁺ T cells displayed as % of total CD45+ cells. *p<0.05, **p<0.01.

Etrumadenant in Combination with Doxorubicin Reduces 4T1 Syngeneic Tumor Growth and Lung Metastases

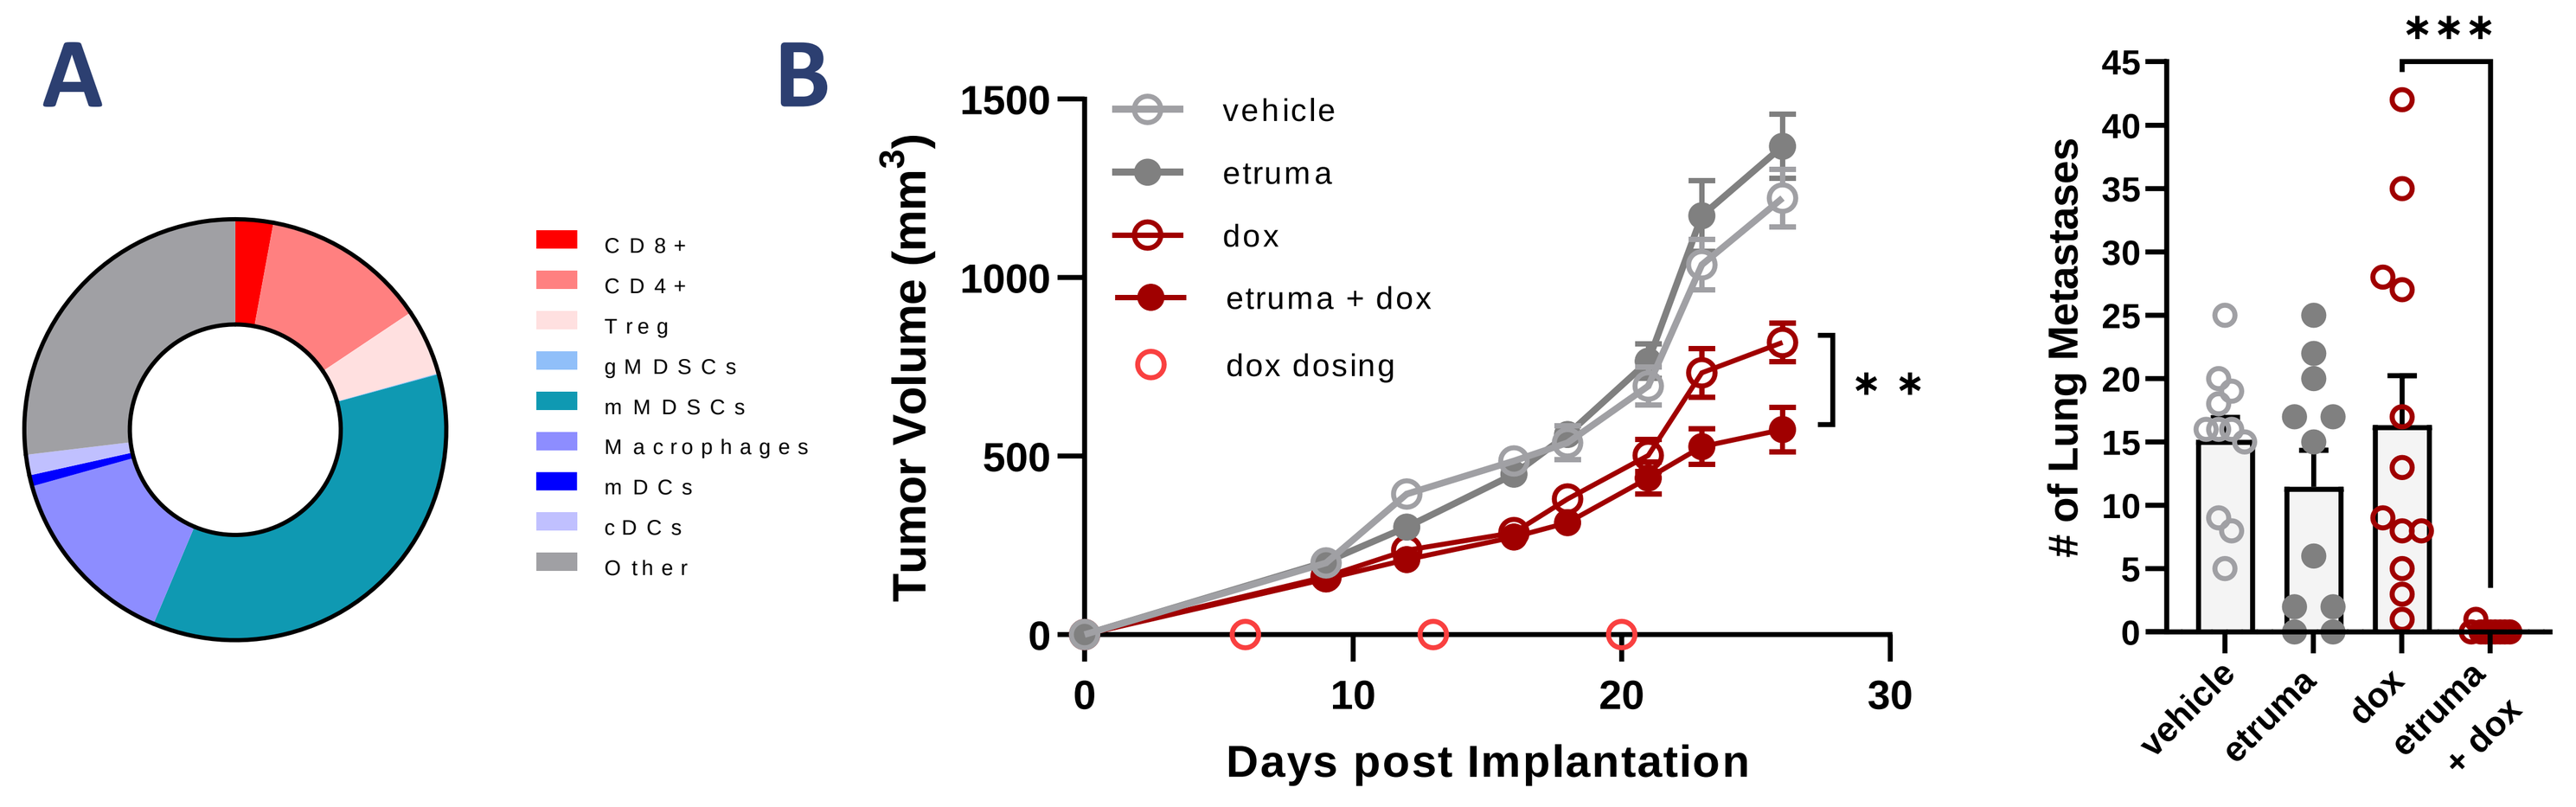
A
CD8+
CD4+
Treg
gMDSCs
mMDSCs
Macrophages
mDCs
cDCs
Other
B
Tumor Volume (mm³)
vehicle
etruma
dox
etruma + dox
dox dosing
Days post Implantation
of Lung Metastases
vehicle
etruma
dox
etruma + dox

Figure 6. (A) Profiling of tumor-infiltrating immune cell shows that myeloid cells comprise a large portion of the immune cell infiltrate in 4T1 tumors. (B) Tumor volume from 4T1 model. Dosing started at 50 mm³, etrumadenant (100 mg/kg, PO, BID) and doxorubicin (6 mg/kg) dosing as indicated. Number of lung metastases were quantified at the end of the study (right panel). **p<0.01, ***p<0.001.

Etrumadenant Combines with Doxorubicin Chemotherapy to Enhance AT3-OVA Tumor Control and Immune Infiltration

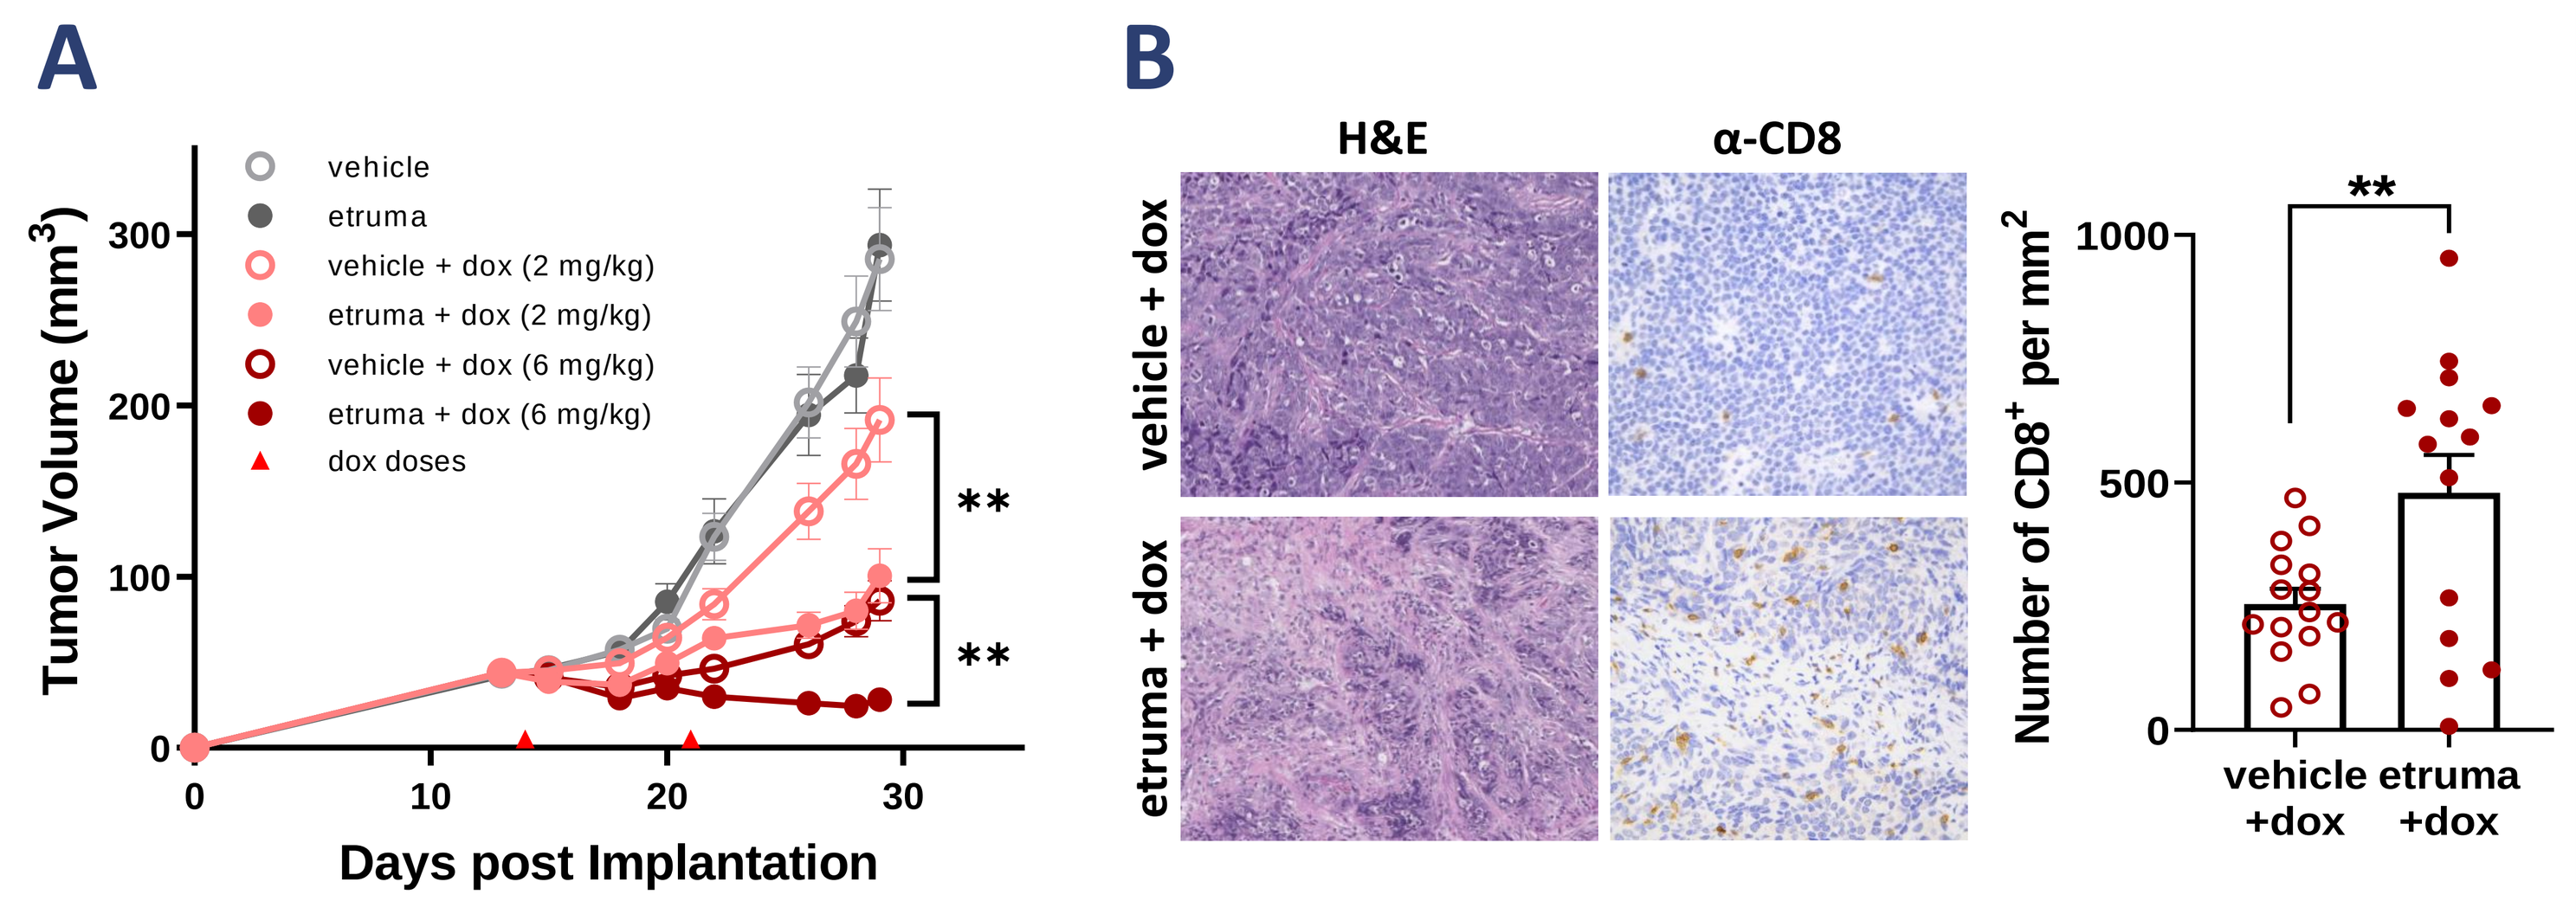
A
Tumor Volume (mm³)
vehicle
etruma
vehicle + dox (2 mg/kg)
etruma + dox (2 mg/kg)
vehicle + dox (6 mg/kg)
etruma + dox (6 mg/kg)
dox doses
Days post Implantation
B
H&E
α-CD8
vehicle + dox
etruma + dox
Number of CD8⁺ per mm²
vehicle +dox
etruma +dox

Figure 7. (A) Tumor volume from AT3-OVA model. Dosing started at 50 mm³ with etrumadenant (100 mg/kg, PO, BID) and doxorubicin (administered Q4Dx2) initiated on the same day. Tumor growth was significantly reduced in the etruma + dox group at both doses compared to vehicle + dox. (B) Tumors from the 6 mg/kg doxorubicin treated group were taken for histologic analysis. H&E staining (images on the left) showed an enhanced stromal compartment in the etruma + dox group versus dox alone, suggesting an enhanced immune infiltrate. CD8⁺ cells were identified by chromogenic immunohistochemistry (images on the right) and quantified (graph) demonstrating that the combination of etruma and dox led to a significant enhancement of infiltrating CD8⁺ cells. **p<0.01.

Etrumadenant in Combination with Doxorubicin Provides Similar Tumor Control as an Extended Course of Chemotherapy

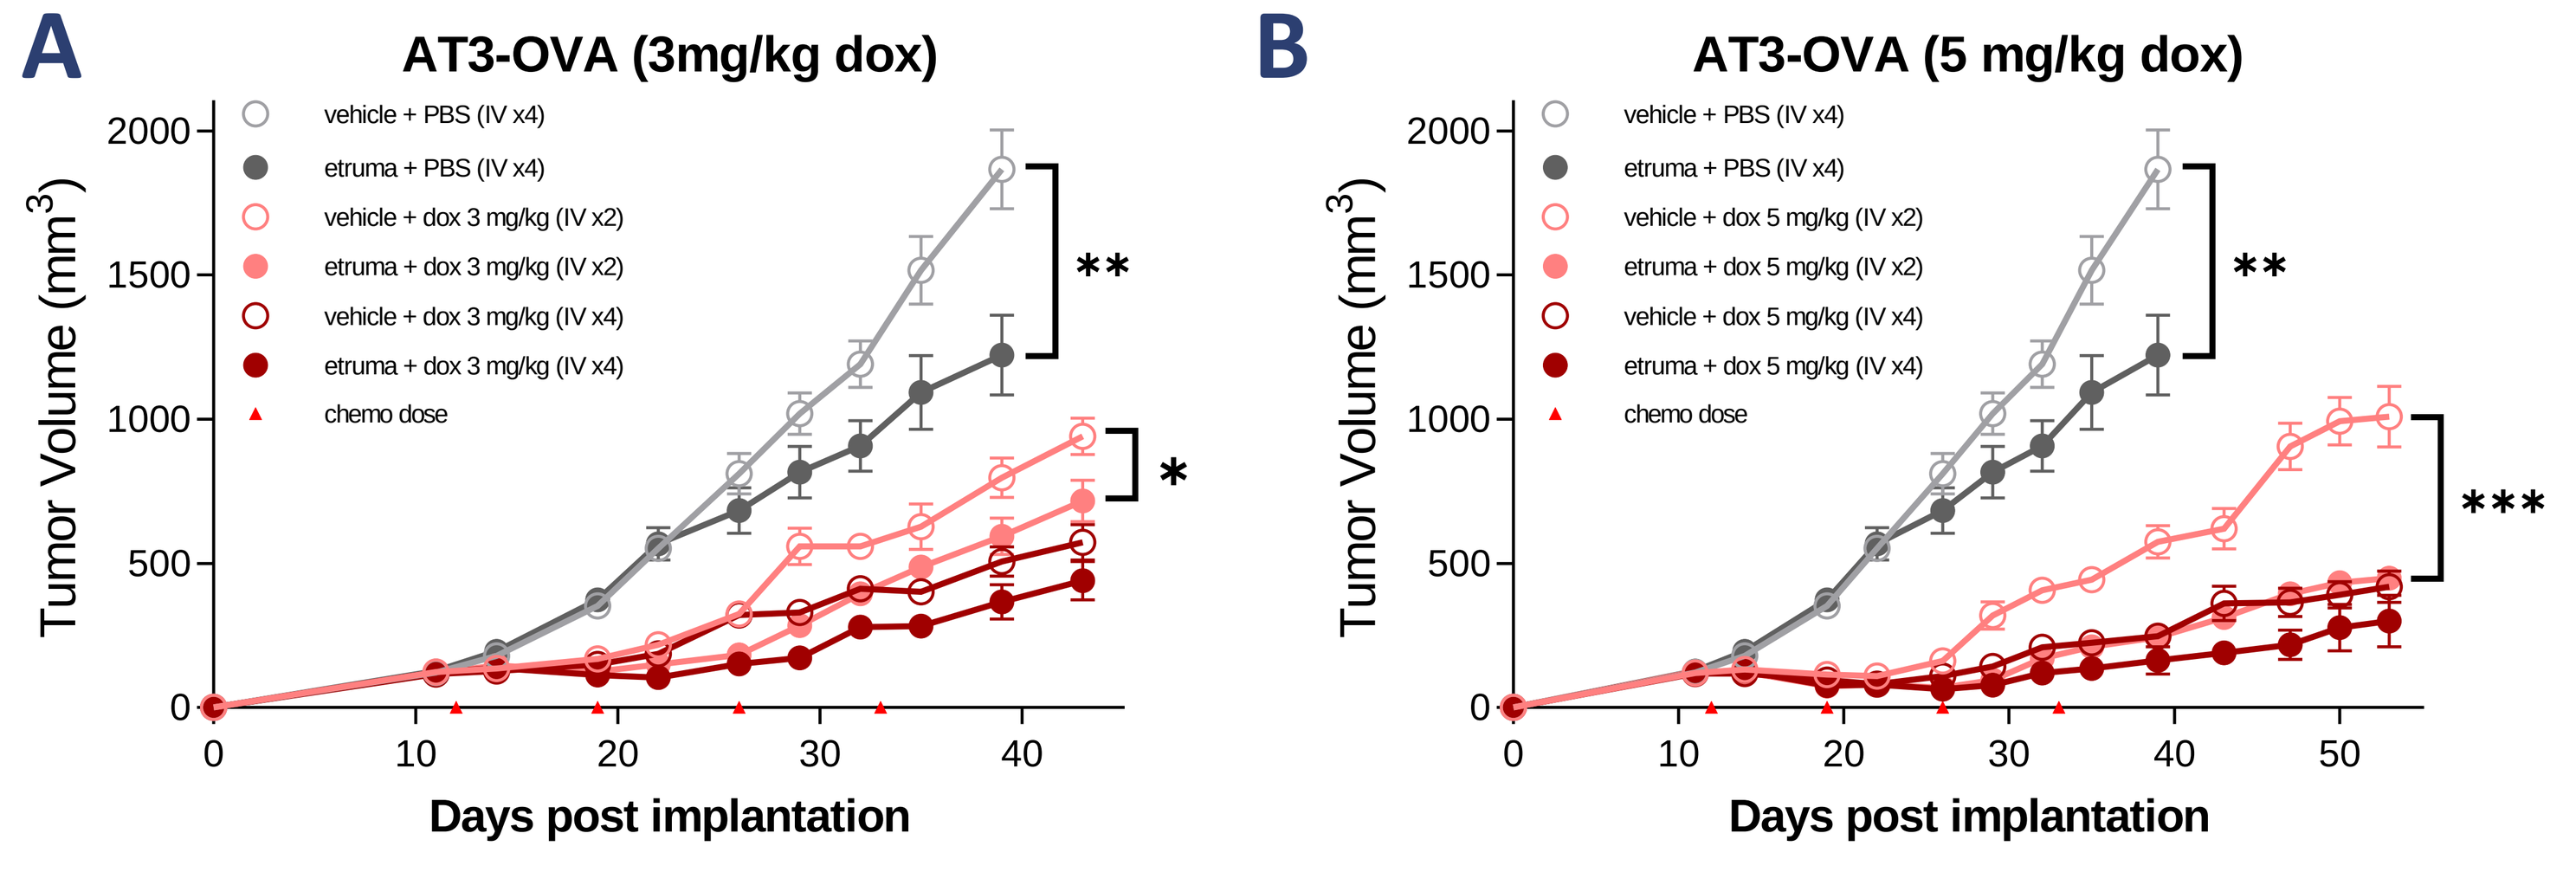
A
AT3-OVA (3mg/kg dox)
vehicle + PBS (IV x4)
etruma + PBS (IV x4)
vehicle + dox 3 mg/kg (IV x2)
etruma + dox 3 mg/kg (IV x2)
vehicle + dox 3 mg/kg (IV x4)
etruma + dox 3 mg/kg (IV x4)
chemo dose
Tumor Volume (mm³)
Days post implantation
B
AT3-OVA (5 mg/kg dox)
vehicle + PBS (IV x4)
etruma + PBS (IV x4)
vehicle + dox 5 mg/kg (IV x2)
etruma + dox 5 mg/kg (IV x2)
vehicle + dox 5 mg/kg (IV x4)
etruma + dox 5 mg/kg (IV x4)
chemo dose
Tumor Volume (mm³)
Days post implantation

Figure 8. (A) AT3-OVA tumors were administered doxorubicin (3 mg/kg) as a 2 or 4 dose regimen in combination with etrumadenant (200 mg/kg, PO, QD) starting at 100 mm³, as indicated. A significant reduction in tumor volume was observed with etrumadenant as a monotherapy and a small but significant reduction was observed in the etruma + dox group vs vehicle + dox in the abbreviated chemotherapy group. (B) A substantially larger reduction in tumor growth was observed in the etruma + dox group vs vehicle + dox in the abbreviated chemotherapy group at the higher (5 mg/kg) dose of doxorubicin. These data suggest that the greatest therapeutic benefit of doxorubicin, when combined with etruma, may result from the initial, priming, doses of chemotherapy. . *p<0.05, **p<0.01, ***p<0.001.

CONCLUSIONS

- These studies demonstrate the ability of etrumadenant to block adenosine-driven immunosuppression and to combine with chemotherapeutic agents in preclinical models.
- Furthermore, our studies suggest that combination of etrumadenant with a truncated course of immunogenic chemotherapy (either fewer cycles or a reduced dose level) may be sufficient to enhance immune activation and yield similar anti-tumor effects as a full course of chemotherapy.