Limited Drug Interaction Supported by a Clinical Drug Interaction Study and Physiologically Based Pharmacokinetic Modeling of Etrumadenant (AB928), a Novel Dual Adenosine Receptor Antagonist, Under Coadministration with a Strong CYP3A4 and P-glycoprotein Inhibitor (Itraconazole) in Healthy Participants

Lian Zhou,[1] Kara Schmelzer,[2] Nikunjkumar Patel,[2] Lixia Jin,[1] Balaji Agoram[1]

[1]Arcus Biosciences, Hayward, CA, USA; [2]Certara, Princeton, NJ, USA

Poster Number: PII-057

BACKGROUND

- Etrumadenant (AB928) is an orally bioavailable selective dual antagonist of the adenosine 2a receptor ($A_{2a}R$) and the adenosine 2b receptor ($A_{2b}R$) being developed for cancer therapy (**Figure 1**)
 - Most tumors contain high extracellular levels of adenosine, leading to activation of $A_{2a}R$ on T lymphocytes and $A_{2b}R$ myeloid cells and impaired T-cell activation and proliferation
 - Inhibition of ligand-dependent signaling by etrumadenant reverses these immunosuppressive effects without causing any immune activation effects
- In vitro data suggest etrumadenant metabolism is mediated by CYP3A4 (to the *N*-dealkylated metabolite), and by CYP2C8 and UDP-glucuronosyltransferase ([UGT] to the glucuronide metabolite)
- Etrumadenant is also an in vitro substrate of P-glycoprotein (P-gp) and breast cancer resistance protein (BCRP)

Figure 1. Critical Role of the Adenosine Pathway in the Immunosuppressive Tumor Microenvironment

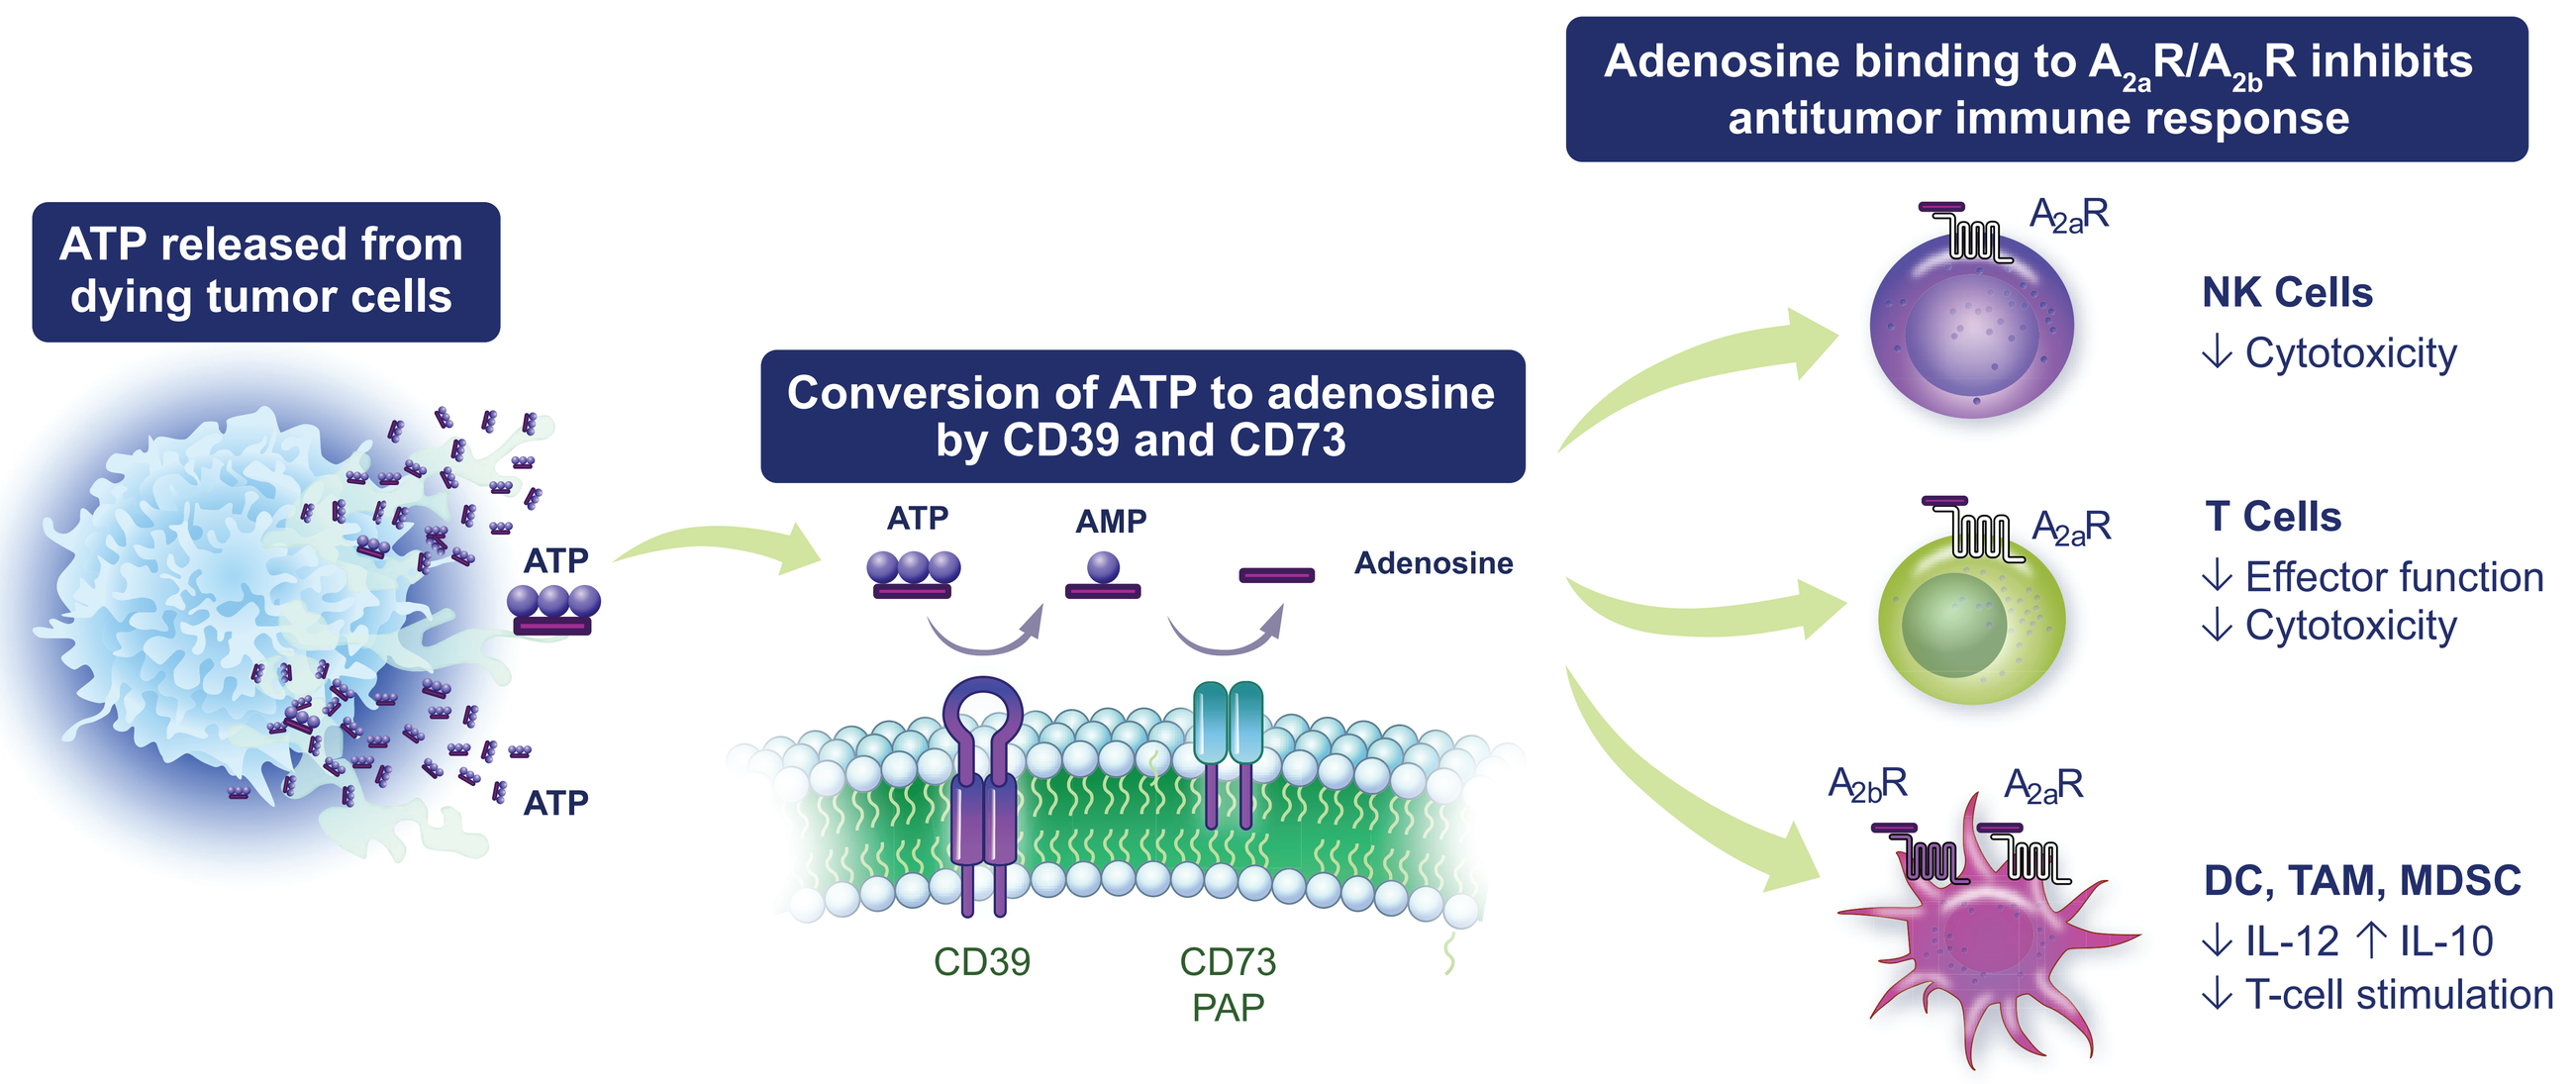
ATP released from dying tumor cells
ATP
Conversion of ATP to adenosine by CD39 and CD73
ATP
AMP
Adenosine
CD39
CD73
PAP
Adenosine binding to A2aR/A2bR inhibits antitumor immune response
A2aR
NK Cells
↓ Cytotoxicity
T Cells
↓ Effector function
↓ Cytotoxicity
A2bR
DC, TAM, MDSC
↓ IL-12 ↑ IL-10
↓ T-cell stimulation

AMP, adenosine monophosphate; ATP, adenosine triphosphate; $A_{2a}R/A_{2b}R$, adenosine receptors 2a/2b; DC, dendritic cell; IL, interleukin; MDSC, myeloid-derived suppressor cells; NK, natural killer; PAP, prostatic acid phosphatase; TAM, tumor-associated macrophage.

OBJECTIVES

- To evaluate the effect of itraconazole (a strong CYP3A4/P-gp inhibitor) on the pharmacokinetics (PK) of etrumadenant by conducting a direct drug-drug interaction (DDI) study in healthy participants
- To assess the CYP3A4 contribution to overall metabolism and to predict DDI using a physiologically based pharmacokinetic (PBPK) model

METHODS

Clinical Study Design

- An open-label, fixed-sequence, 2-period DDI study to evaluate the effect of multiple doses of itraconazole on the single-dose PK of etrumadenant (n = 20) (ClinicalTrials.gov Identifier: NCT05154136) (**Figure 2**)

PBPK Modelling

- The PBPK base model for etrumadenant was developed from physicochemical, in vitro experimental, and clinical data sets (**Figure 3**)
- Predictive performance of the base model was verified by comparing model PK predictions with the observed clinical PK of etrumadenant

Figure 2. ARC-18 Study Design

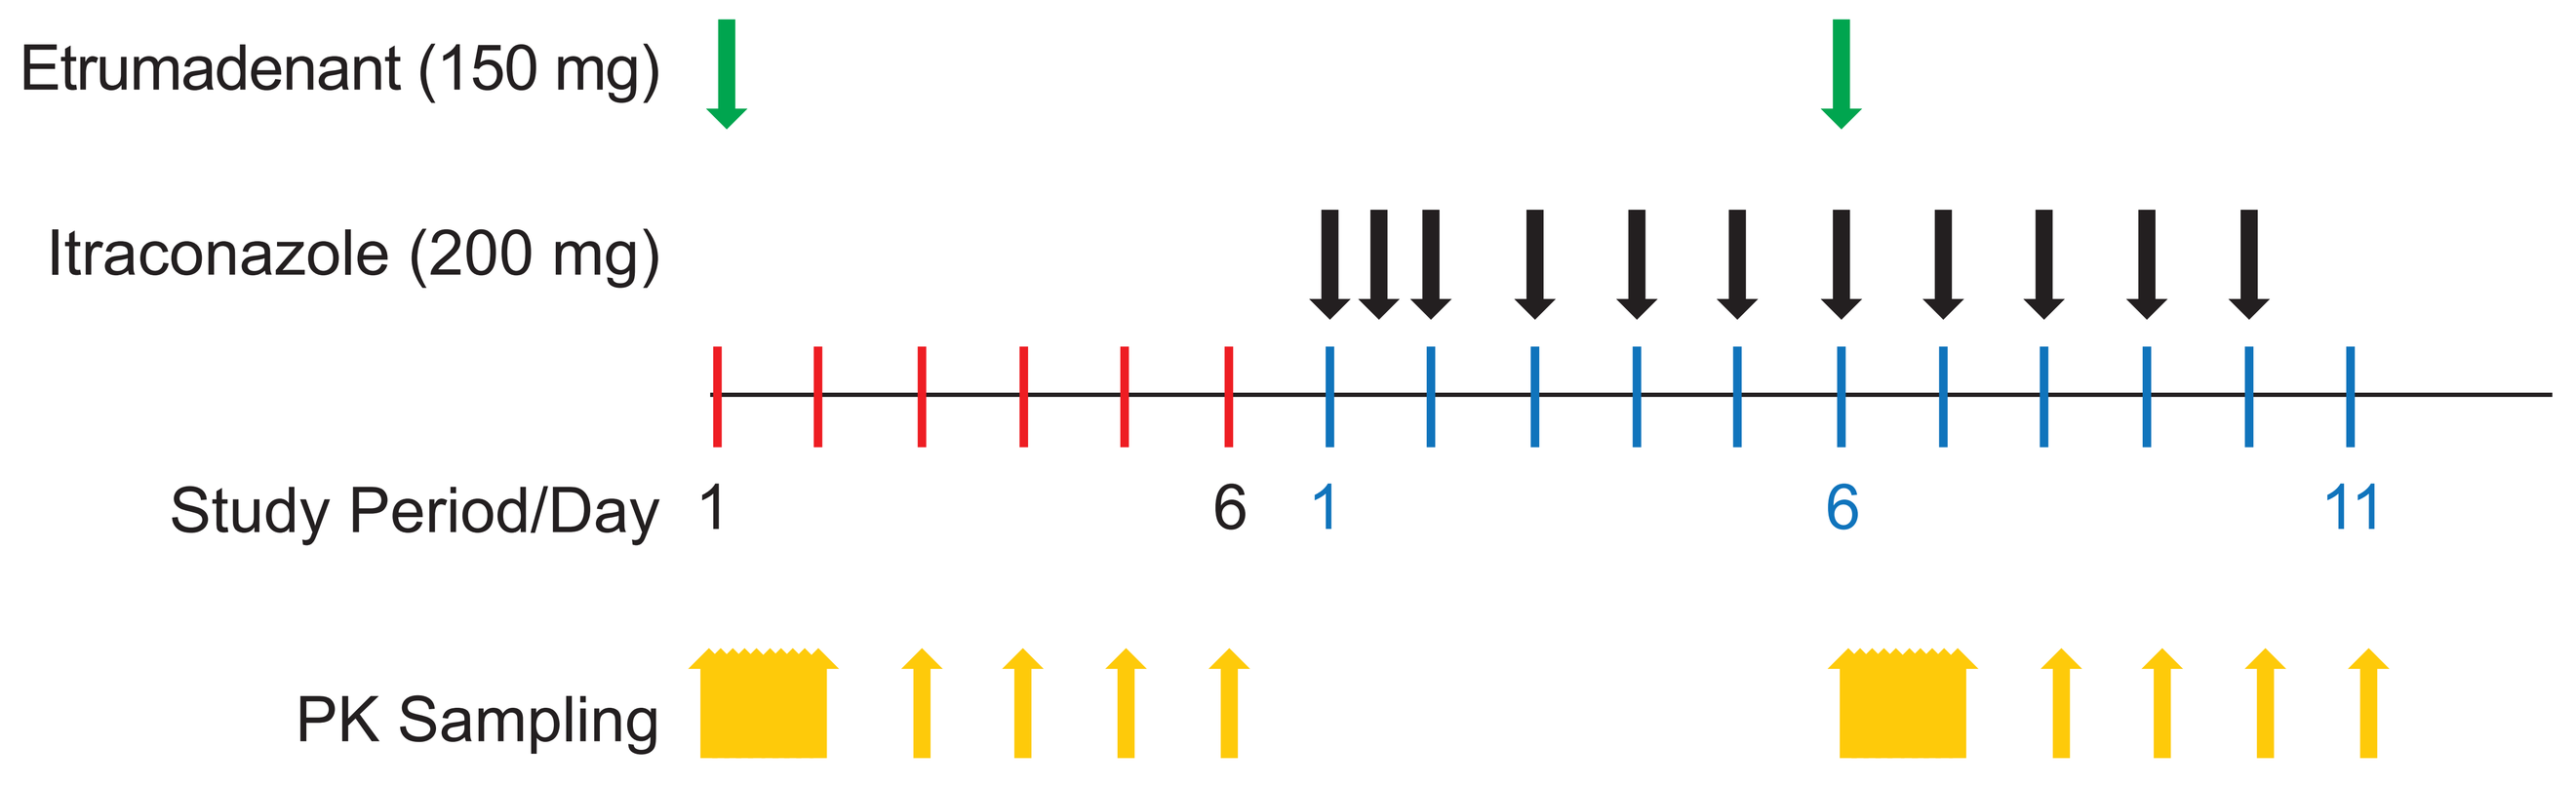
Etrumadenant (150 mg)
Itraconazole (200 mg)
Study Period/Day
1
6
1
6
11
PK Sampling

PK, pharmacokinetics. Green arrow, etrumadenant dosing; black arrow, itraconazole dosing; red, days in period 1; blue, days in period 2; gold arrow, PK sampling times.

Figure 3. (A) Advanced Dissolution, Absorption, and Metabolism Model and (B) Full PBPK Distribution Model

A

B

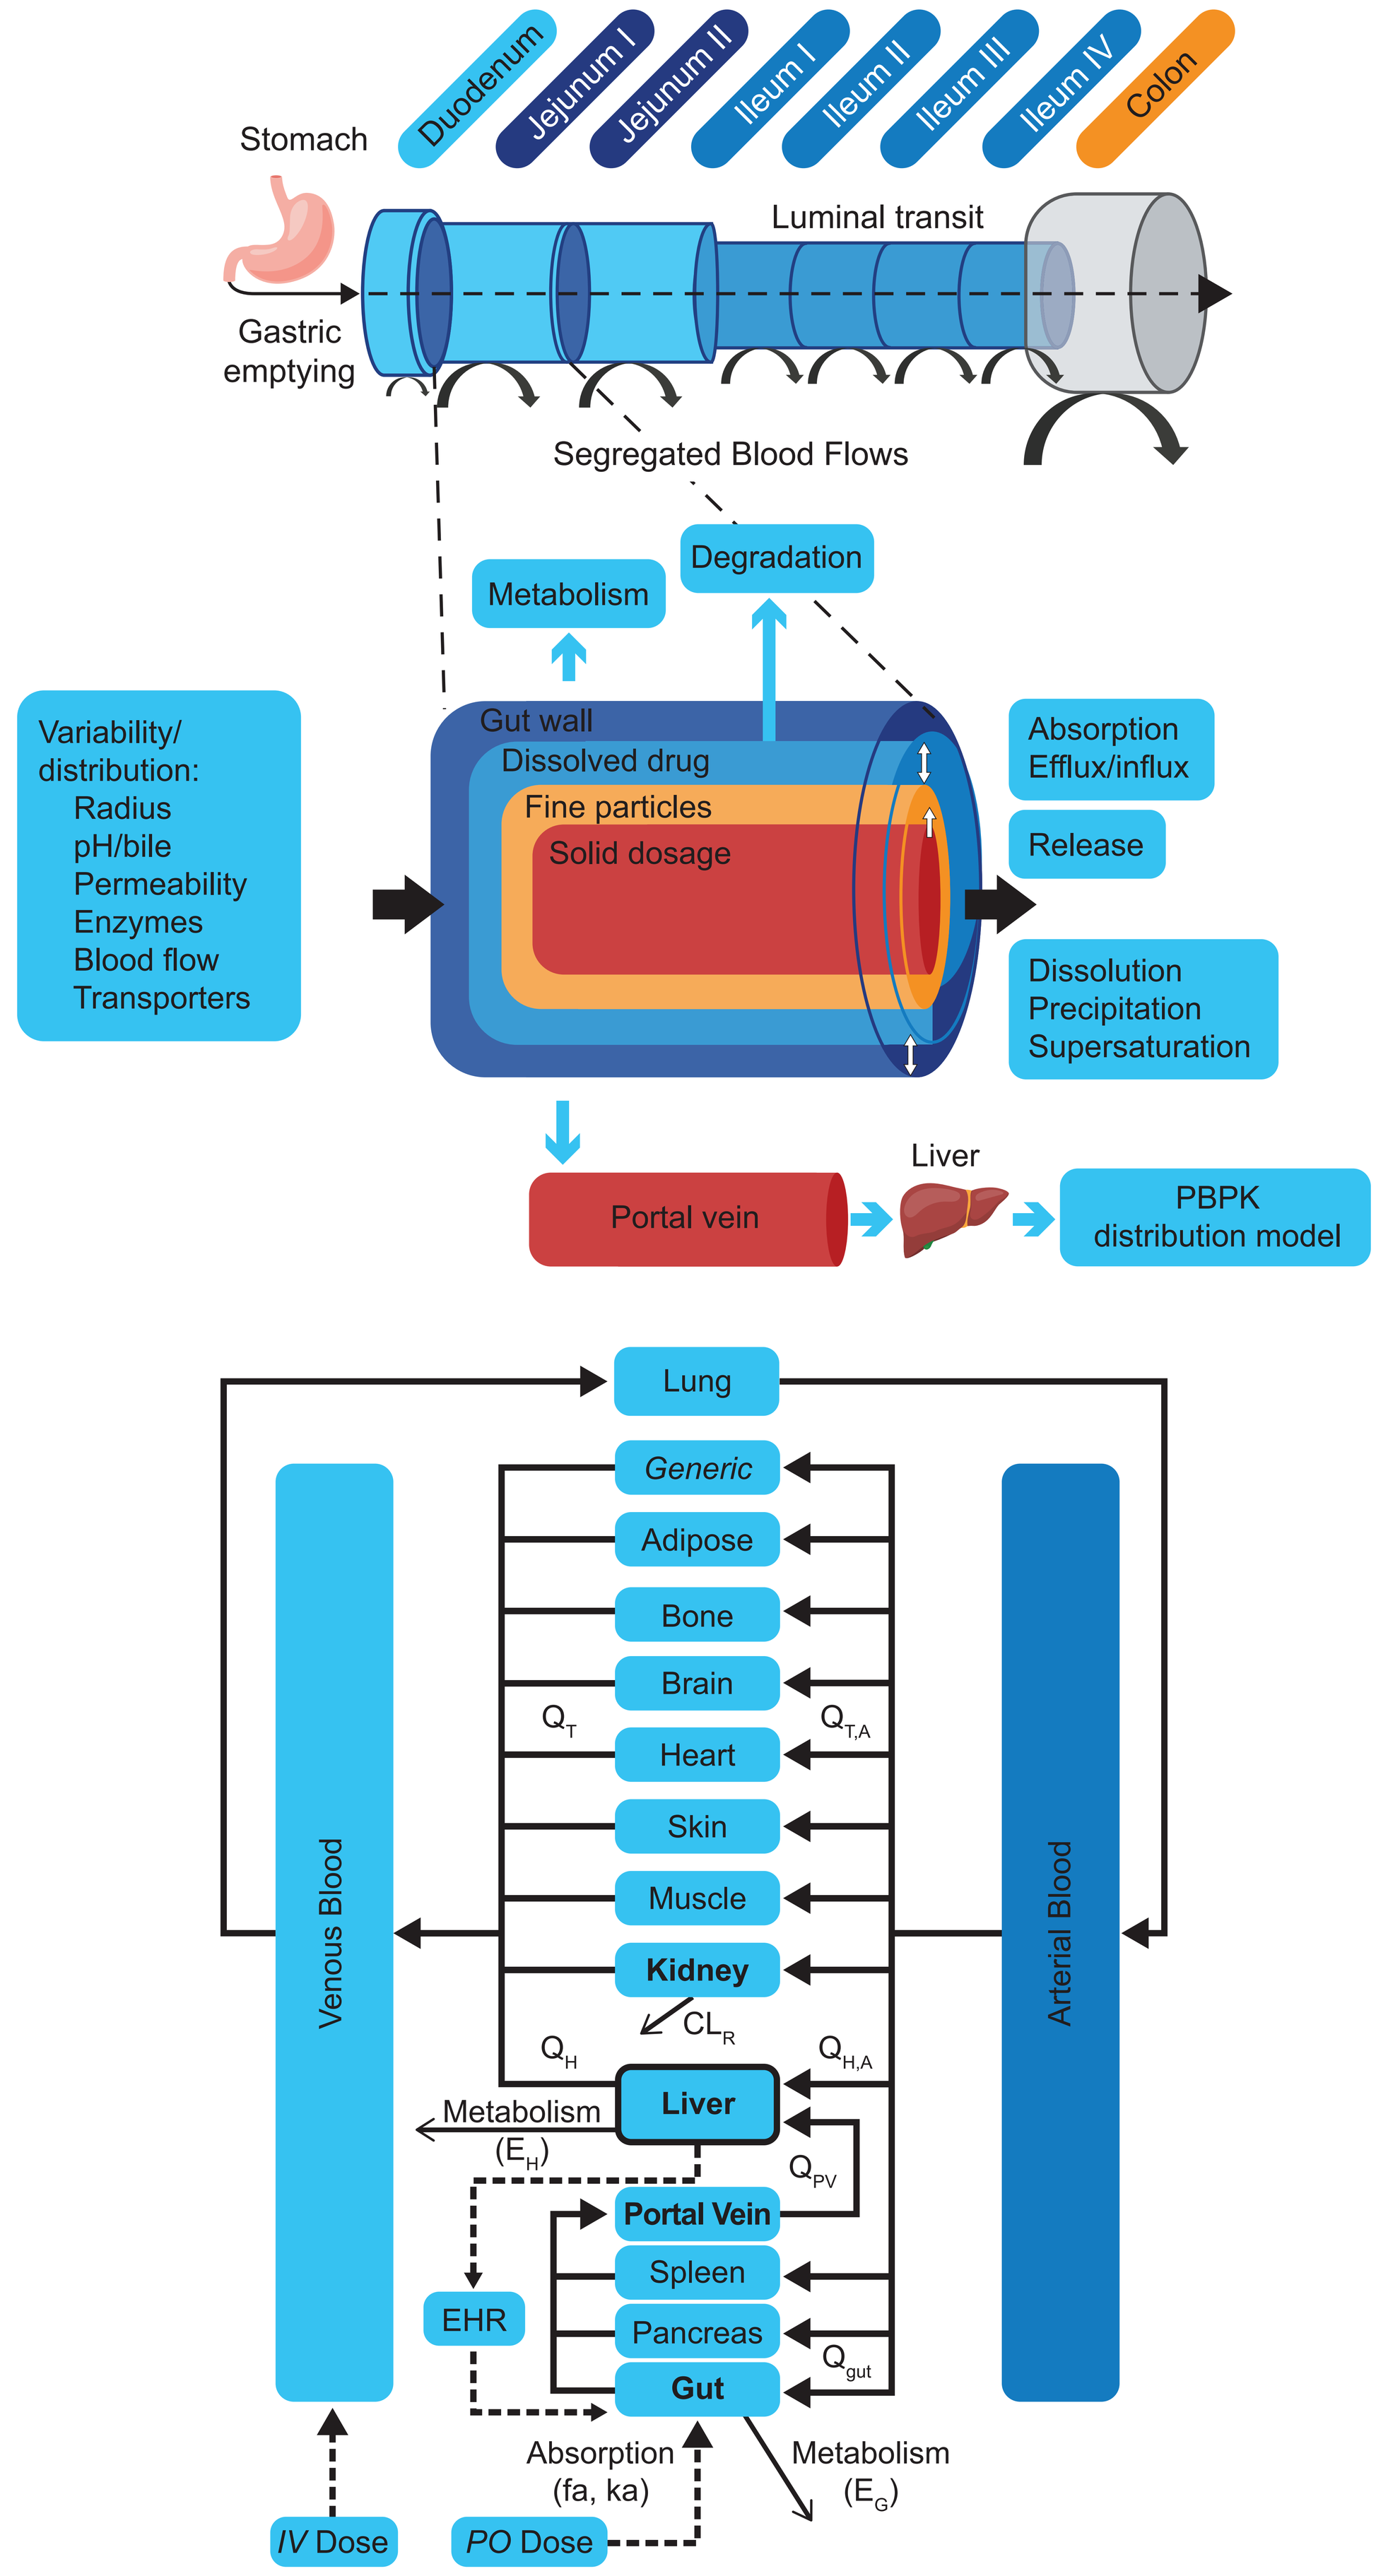
Stomach
Duodenum
Jejunum I
Jejunum II
Ileum I
Ileum II
Ileum III
Ileum IV
Colon
Luminal transit
Gastric emptying
Segregated Blood Flows
Metabolism
Degradation
Variability/ distribution: Radius pH/bile Permeability Enzymes Blood flow Transporters
Gut wall
Dissolved drug
Fine particles
Solid dosage
Absorption Efflux/influx
Release
Dissolution Precipitation Supersaturation
Portal vein
Liver
PBPK distribution model
Lung
Generic
Adipose
Bone
Brain
Heart
Skin
Muscle
Kidney
CLR
Liver
Portal Vein
Spleen
Pancreas
Gut
Venous Blood
Arterial Blood
QT
QT,A
QH
QH,A
QPV
Qgut
Metabolism (EH)
EHR
Absorption (fa, ka)
Metabolism (EG)
IV Dose
PO Dose

CL_R, renal clearance; E_G and E_H, fractions undergoing first-pass metabolism in the gut and liver, respectively; EHR, enterohepatic recirculation; fa, fractions absorbed; ka, first-order absorption rate constant; IV, intravenous administration; PO, oral administration; Q_H, Q_{HA}, Q_{PV}, Q_{gut}, Q_{TA} and Q_T, blood flows in the hepatic vein, hepatic artery, hepatic portal vein, gut and blood flows into and out of other tissue (T) compartments, respectively.

The effect of a strong CYP3A4 inhibitor on the PK of etrumadenant is limited, and P-gp may not influence etrumadenant oral absorption

RESULTS

- Following multiple doses of itraconazole:
 - Etrumadenant area under the curve to infinity (AUC_{inf}) and maximum concentration (C_{max}) increased approximately 62% and 18%, respectively, compared to etrumadenant given alone (**Table 1; Figure 4A**)
 - AUCs and C_{max} of the *N*-dealkylated metabolite (metabolism mediated by CYP3A4) decreased by 3.4- to 9.0-fold respectively, compared to etrumadenant given alone (**Figure 4B**)
 - AUCs of the glucuronide metabolite (metabolism mediated by UDP-glucuronosyltransferase) increased 1.6- to 1.9-fold whereas the C_{max} increased 1.3-fold, compared to etrumadenant given alone (**Figure 4C**)

Table 1. Summary of Statistical Comparisons of Plasma Etrumadenant PK Parameters With and Without Itraconazole (observed clinical results)

	Etrumadenant + Itraconazole		Etrumadenant Alone				
Parameter	Geometric LSMs	n	Geometric LSMs	n	GMR (%)	90% Confidence Interval	Intrasubject CV%
AUC_{last} (ng*hr/mL)	59,300	19	36,500	20	162.58	149.83-176.40	14.64
AUC_{inf} (ng*hr/mL)	62,400	19	38,600	20	161.61	148.30-176.11	15.43
C_{max} (ng/mL)	3540	19	3000	20	117.87	107.04-129.79	17.31

AUC, area under the curve; C_{max}, maximum concentration; CV, coefficient of variation; GMR, geometric mean ratio; LSM, least-squares mean; PK, pharmacokinetics. One patient (etrumadenant + itraconazole) was not administered itraconazole on days 8-10 and was excluded from PK analysis.

- The PBPK model predicted a similar level of interaction as the observed values. The model was then used to simulate changes in exposure with other CYP3A4 inhibitors (**Table 2**)

Table 2. Summary of PBPK Modeling Comparisons of Plasma Etrumadenant Pharmacokinetic Parameters With and Without Various CYP3A4 Inhibitors

	GMR (%) Observed/Predicted		
CYP3A4 Inhibitor (Category)	C_{max}	AUC_{0-24h}	AUC_{last}
Itraconazole (Strong)	118/112	143/140	163/172
Fluconazole (Moderate)	NA/108	NA/125	NA/136
Cimetidine (Weak)	NA/104	NA/106	NA/106

AUC, area under the curve; C_{max}, maximum concentration; GMR, geometric mean ratio; NA, not applicable. The observed data is from the clinical trial, predicted values are derived from PBPK model.

Scan this QR Code to download the full paper

FPO

Figure 4. Mean (SD) Plasma Concentration of (A) Etrumadenant 150 mg, (B) *N*-Dealkylated Etrumadenant, and (C) Glucuronide Etrumadenant Over Time With and Without Itraconazole

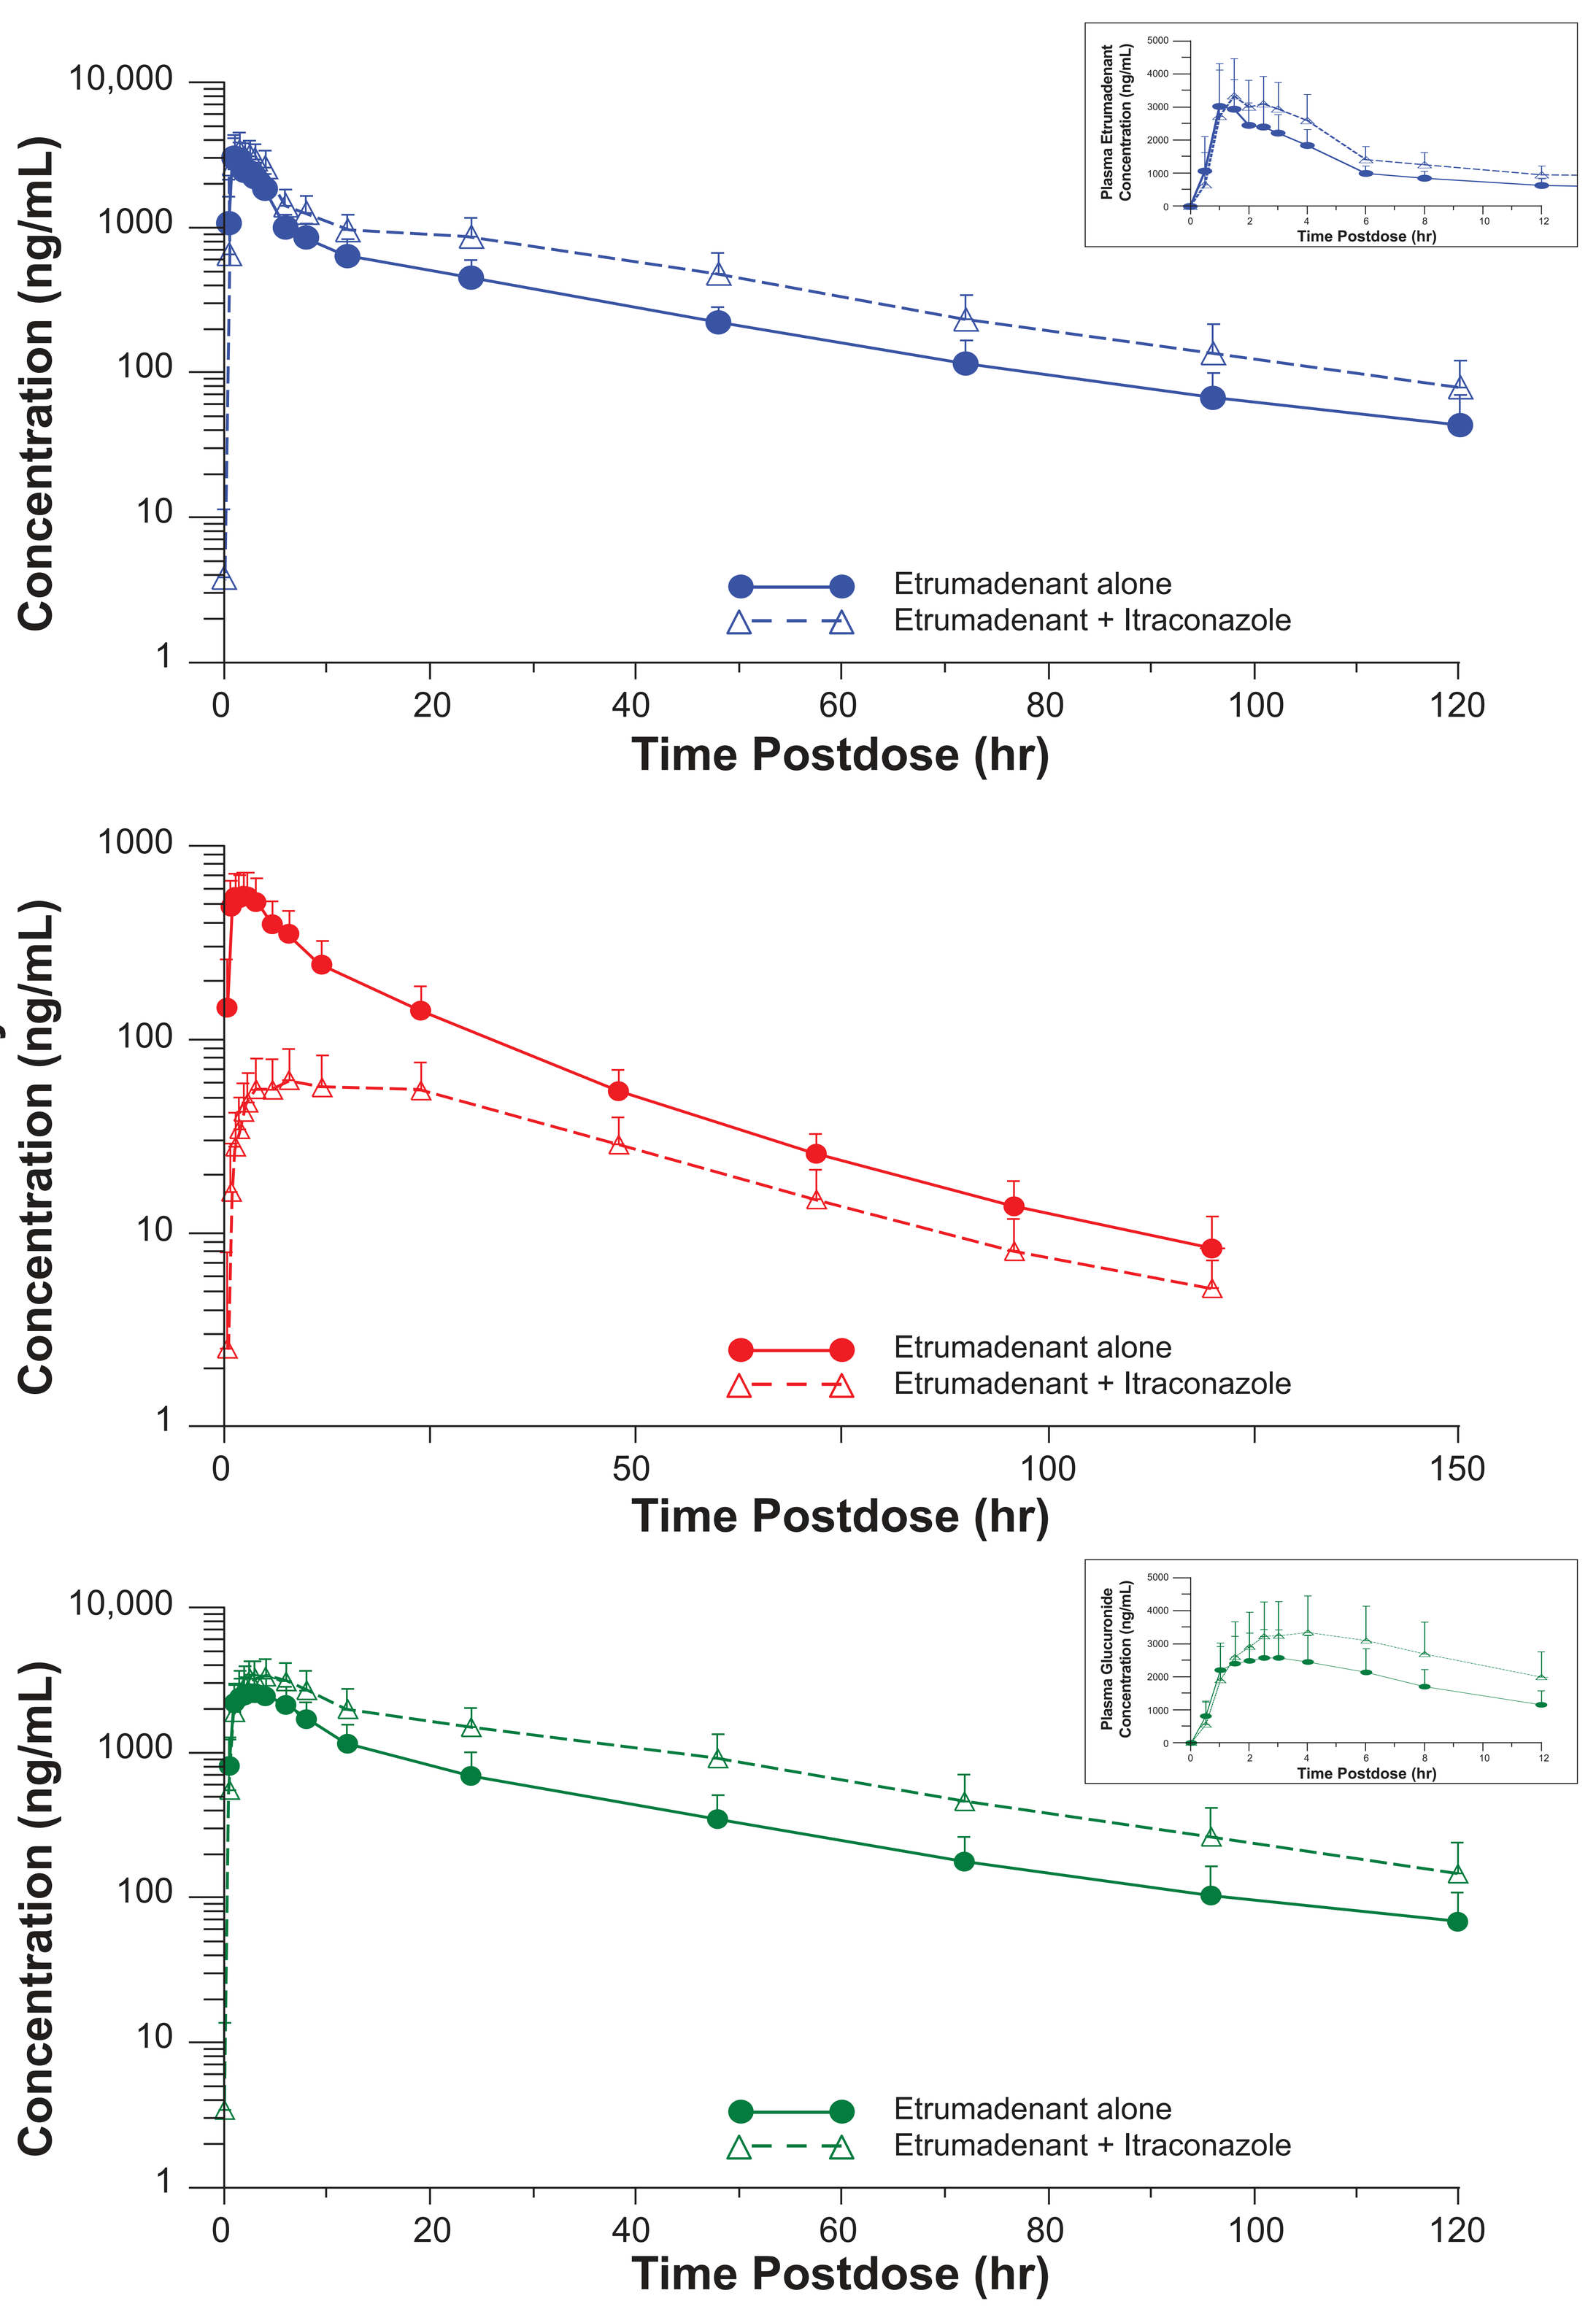
A
Plasma Etrumadenant Concentration (ng/mL)
10,000
1000
100
10
1
0
20
40
60
80
100
120
Time Postdose (hr)
Etrumadenant alone
Etrumadenant + Itraconazole
B
Plasma N-Dealkylated Concentration (ng/mL)
1000
100
10
1
0
50
100
150
Time Postdose (hr)
Etrumadenant alone
Etrumadenant + Itraconazole
C
Plasma Glucuronide Concentration (ng/mL)
10,000
1000
100
10
1
0
20
40
60
80
100
120
Time Postdose (hr)
Etrumadenant alone
Etrumadenant + Itraconazole

CONCLUSIONS

- The limited increase (62% increase in AUC_{inf}) when co-administered with itraconazole indicates that CYP3A4 is partially responsible for the elimination of etrumadenant
- The minimal increase (18%) in etrumadenant C_{max} suggests coadministration of a P-gp inhibitor (itraconazole) does not cause meaningful differences in etrumadenant PK
- PBPK modelling suggests that the fraction of etrumadenant metabolized through CYP3A4 is approximately 0.4. No clinically significant differences in etrumadenant PK were predicted when coadministered with CYP moderate and weak inhibitors

ACKNOWLEDGMENTS

This study was sponsored by Arcus Biosciences (Hayward, CA, USA). Medical writing assistance was provided by ApotheCom (San Diego, CA, USA) and funded by Arcus Biosciences.

Presented at the Annual Meeting of the American Society for Clinical Pharmacology and Therapeutics (ASCPT); March 22-24, 2023; Atlanta, GA, USA